Deglycosylated CD8-specific immunoPET probe prevents antibody-mediated depletion of CD8+ immune cells

Jordan M. White[1,2], Outi Keinanen[2], Brendon E. Cook[3], Brian Zeglis[3,4], Heather M. Gibson[2], Nerissa T. Viola[2]

[1]Cancer Biology Program, Wayne State University School of Medicine, Detroit, MI 48201; [2] Department of Oncology, Karmanos Cancer Institute, Detroit, MI 48201; [3]Department of Chemistry, Hunter College of the City University of New York, New York, NY 10021; [4]Department of Radiology, Memorial Sloan Kettering Cancer Center, New York, NY, 10065

Introduction

A drawback to the development of imaging probes targeting immune cell surface molecules is the biantennary glycans present on the heavy chain of IgG antibodies (mAbs). These glycans are required for FcγR recognition and have the potential to deplete the effector cells facilitating tumor clearance. Eliminating heavy chain glycans by antibody deglycosylation can resolve immune cell depletion. To examine the effects of antibody deglycosylation and tracer development, we compared the PK, specificity and depletion properties of a known murine anti-CD8 T cell depleting mAb (α-CD8-wt, clone 2.43) against a deglycosylated construct (α-CD8-degly).

PNGaseF α-CD8 Deglycosylation

The mAbs were deglycosylated with PNGaseF for 24 h. α-CD8-PNGaseF was purified by incubation with magnetic chitin beads at 4 ℃ for 10 min, followed by magnetic rack separation. Analysis by denaturing SDS-PAGE showed no glycosylated antibody remained (lane 2) as compared with lane 3, and an absence of detectable PNGaseF (lanes 4 and 5).

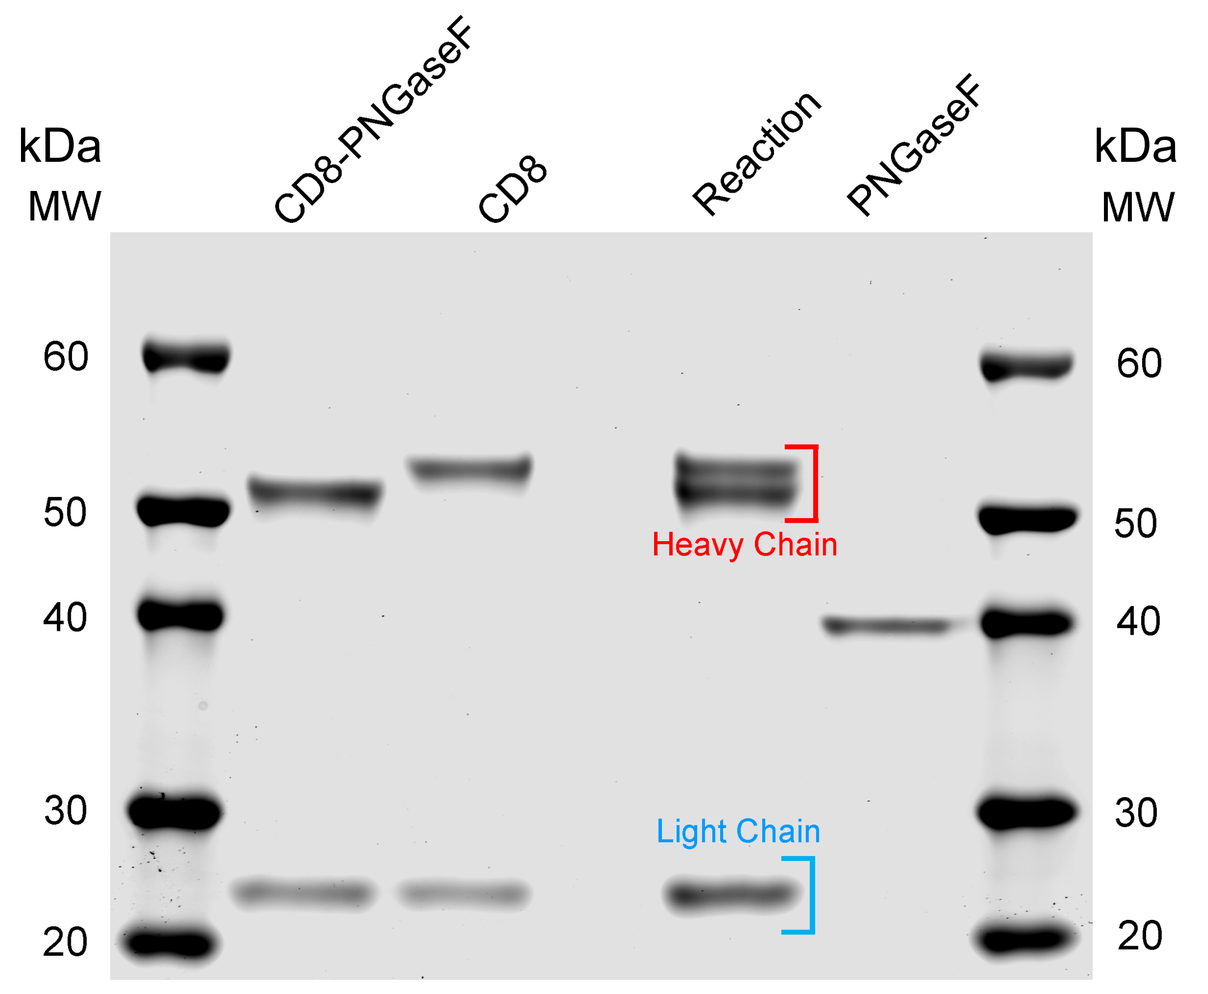
kDa MW
CD8-PNGaseF
CD8
Reaction
PNGaseF
60
50
40
30
20
Heavy Chain
Light Chain

α-CD8-degly does not deplete CD8+ T cells

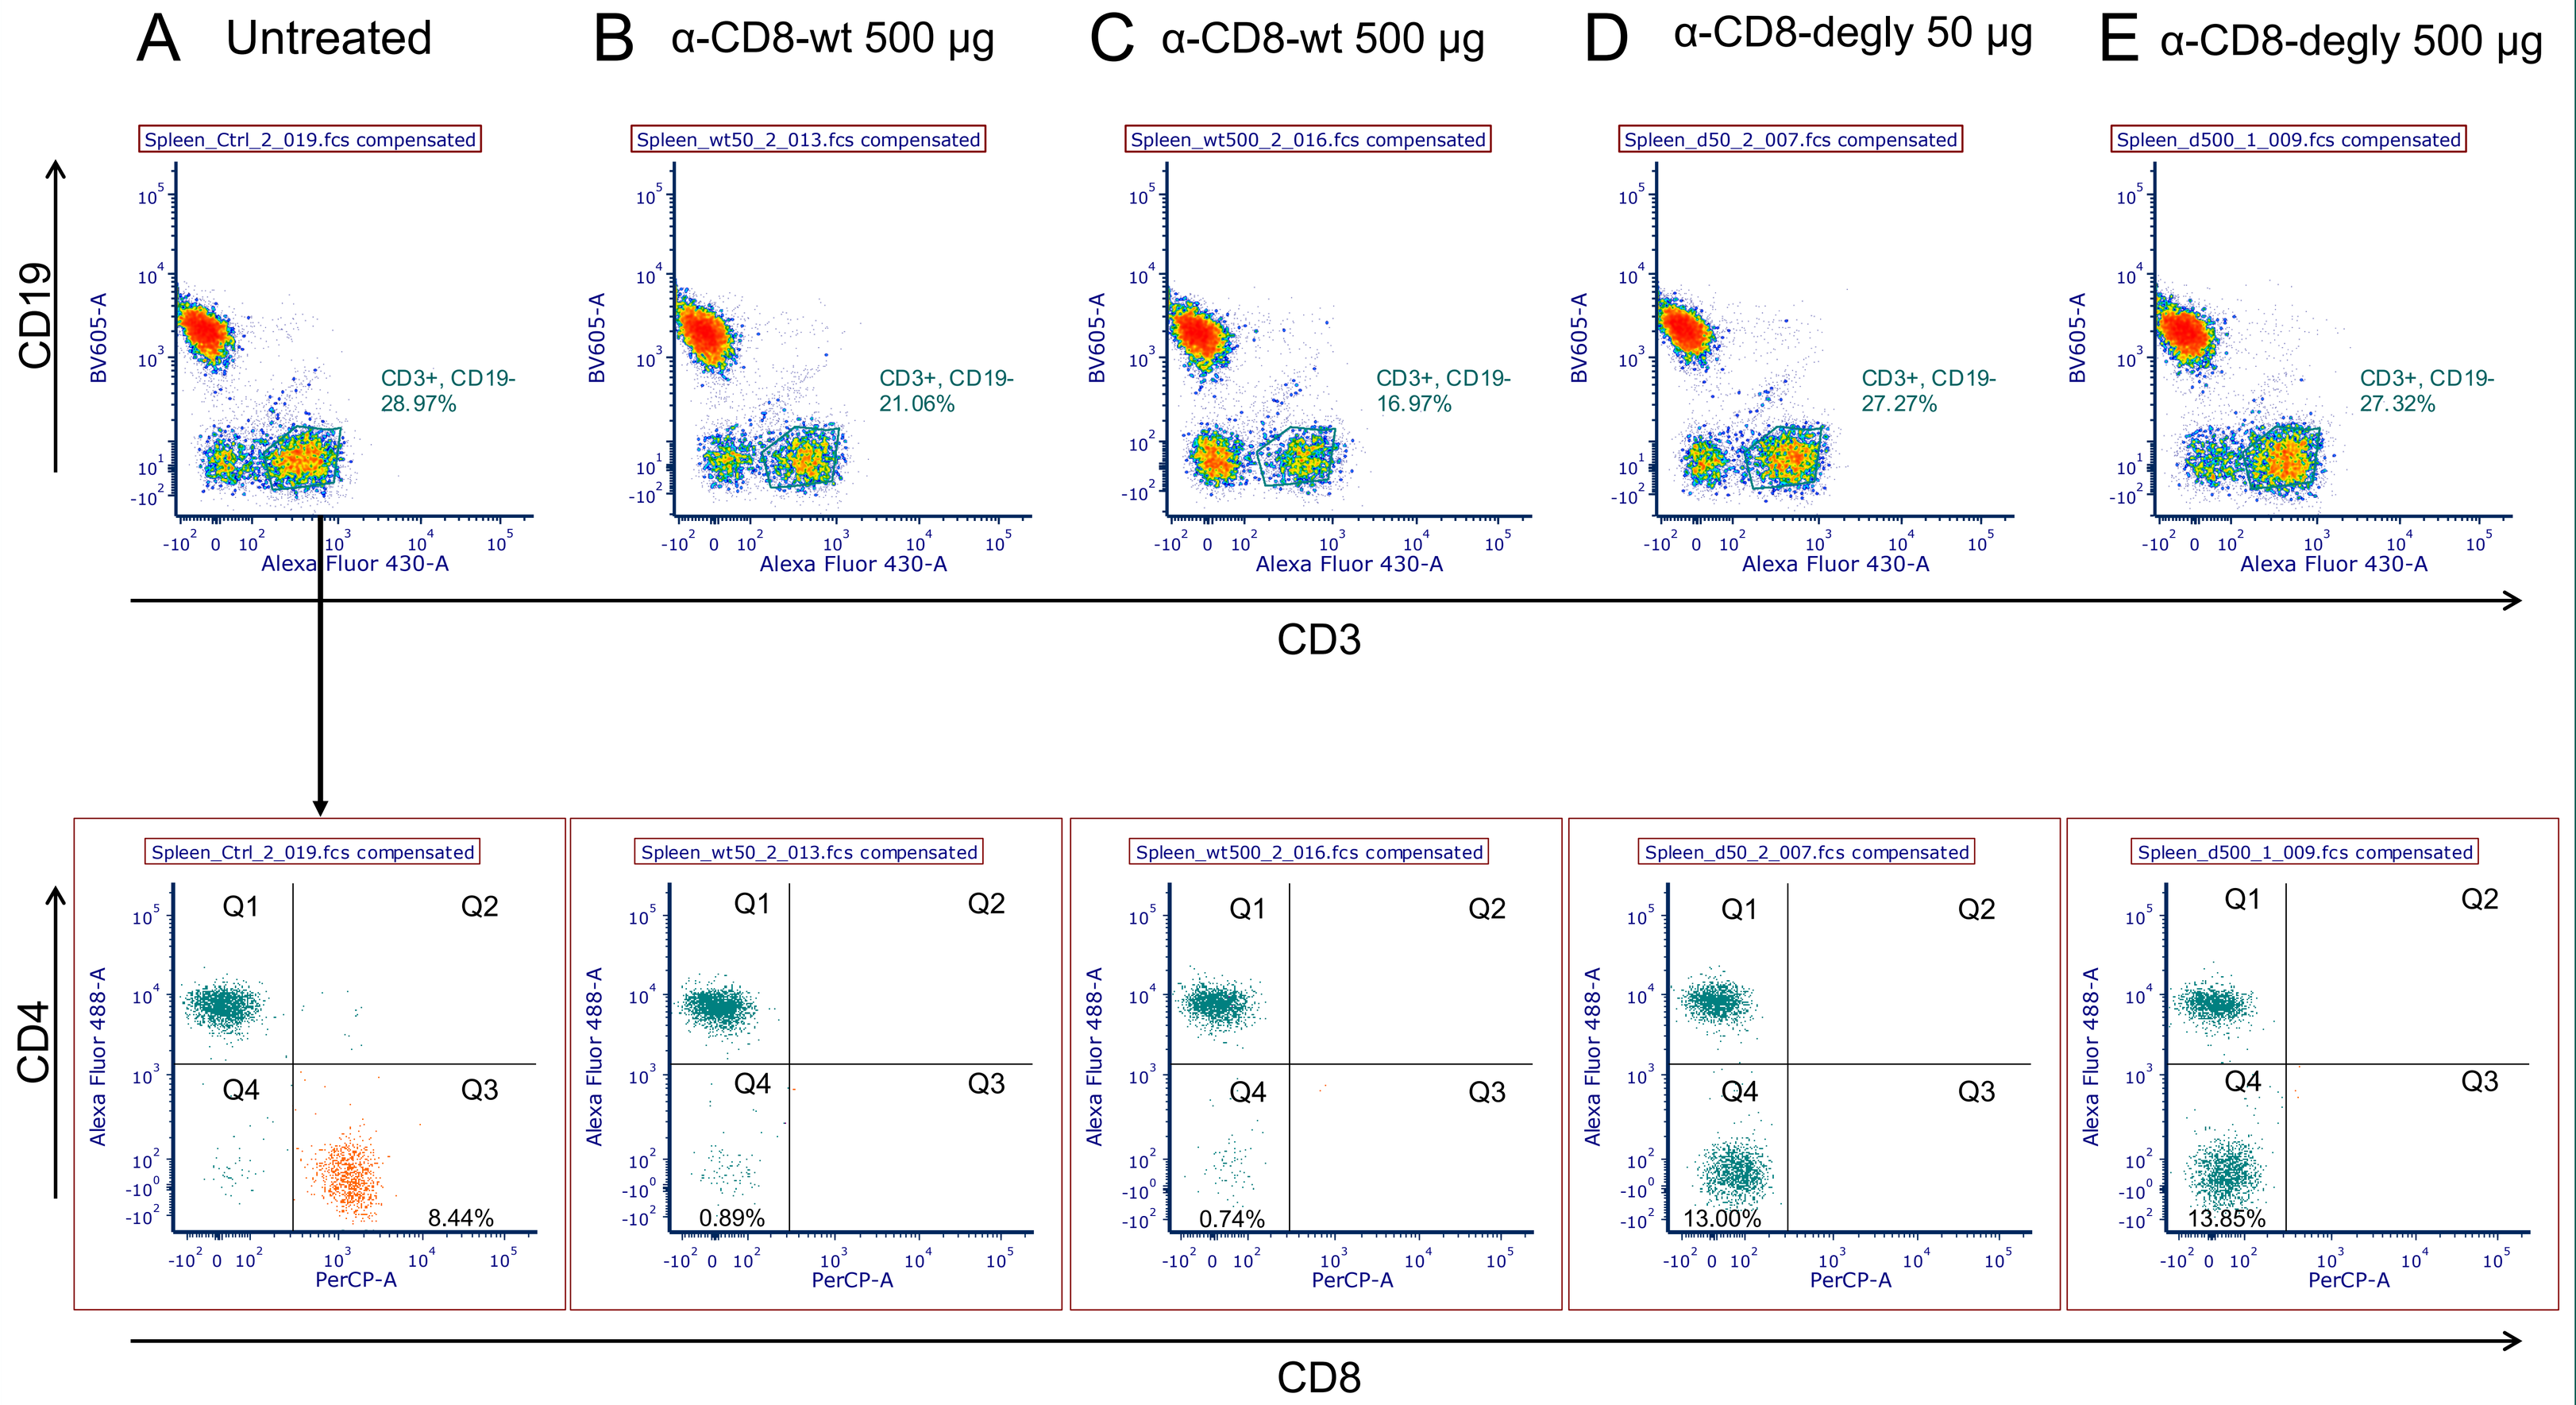
A Untreated
B α-CD8-wt 500 µg
C α-CD8-wt 500 µg
D α-CD8-degly 50 µg
E α-CD8-degly 500 µg
CD19
CD3
CD4
CD8
Q1
Q2
Q3
Q4
8.44%
0.89%
0.74%
13.00%
13.85%

F

Treatment	% CD8+ Splenocytes (Mean ± SD)
Control Mice	7.41 ± 1.46 %
α-CD8-WT 50 µg	0.91 ± 0.12 %
α-CD8-WT 500 µg	0.81 ± 0.14 %
α-CD8-degly 50 µg	15.60 ± 2.25 %
α-CD8-degly 500 µg	13.09 ± 1.49 %

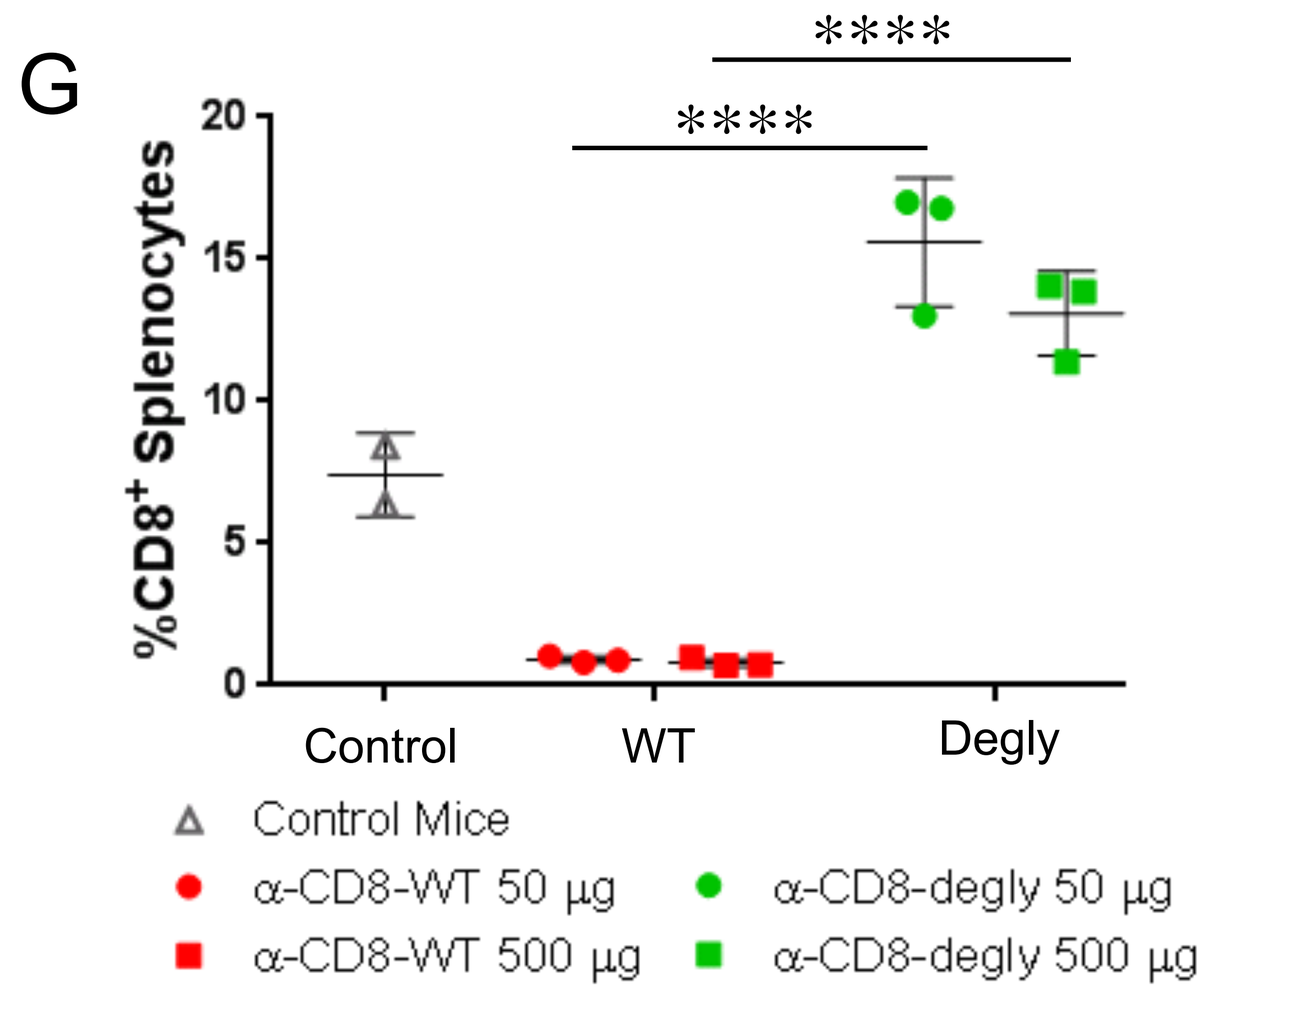
G
%CD8+ Splenocytes

Control
WT
Degly
Control Mice
α-CD8-WT 50 µg
α-CD8-WT 500 µg
α-CD8-degly 50 µg
α-CD8-degly 500 µg

Flow cytometry analysis of spleens of (A) untreated mice, mice treated with (B) α-CD8-wt 50 µg or (C) 500 µg and mice treated with (D) α-CD8-degly 50 µg or (E) 500 µg. In the untreated mice, a CD8+ T cell population was observed in Q3. The CD8+ T cell population was not present in the α-CD8-wt mice as exhibited by the decrease in the cell population in Q4. However, the α-CD8-degly exhibited a CD8+ T cell population present in Q4 due to the interference of the antibody with the flow mAb. This exhibits a change in the ability of the mAb to deplete its target population dependent on the glycosylation status.

^{89}Zr-α-CD8 development

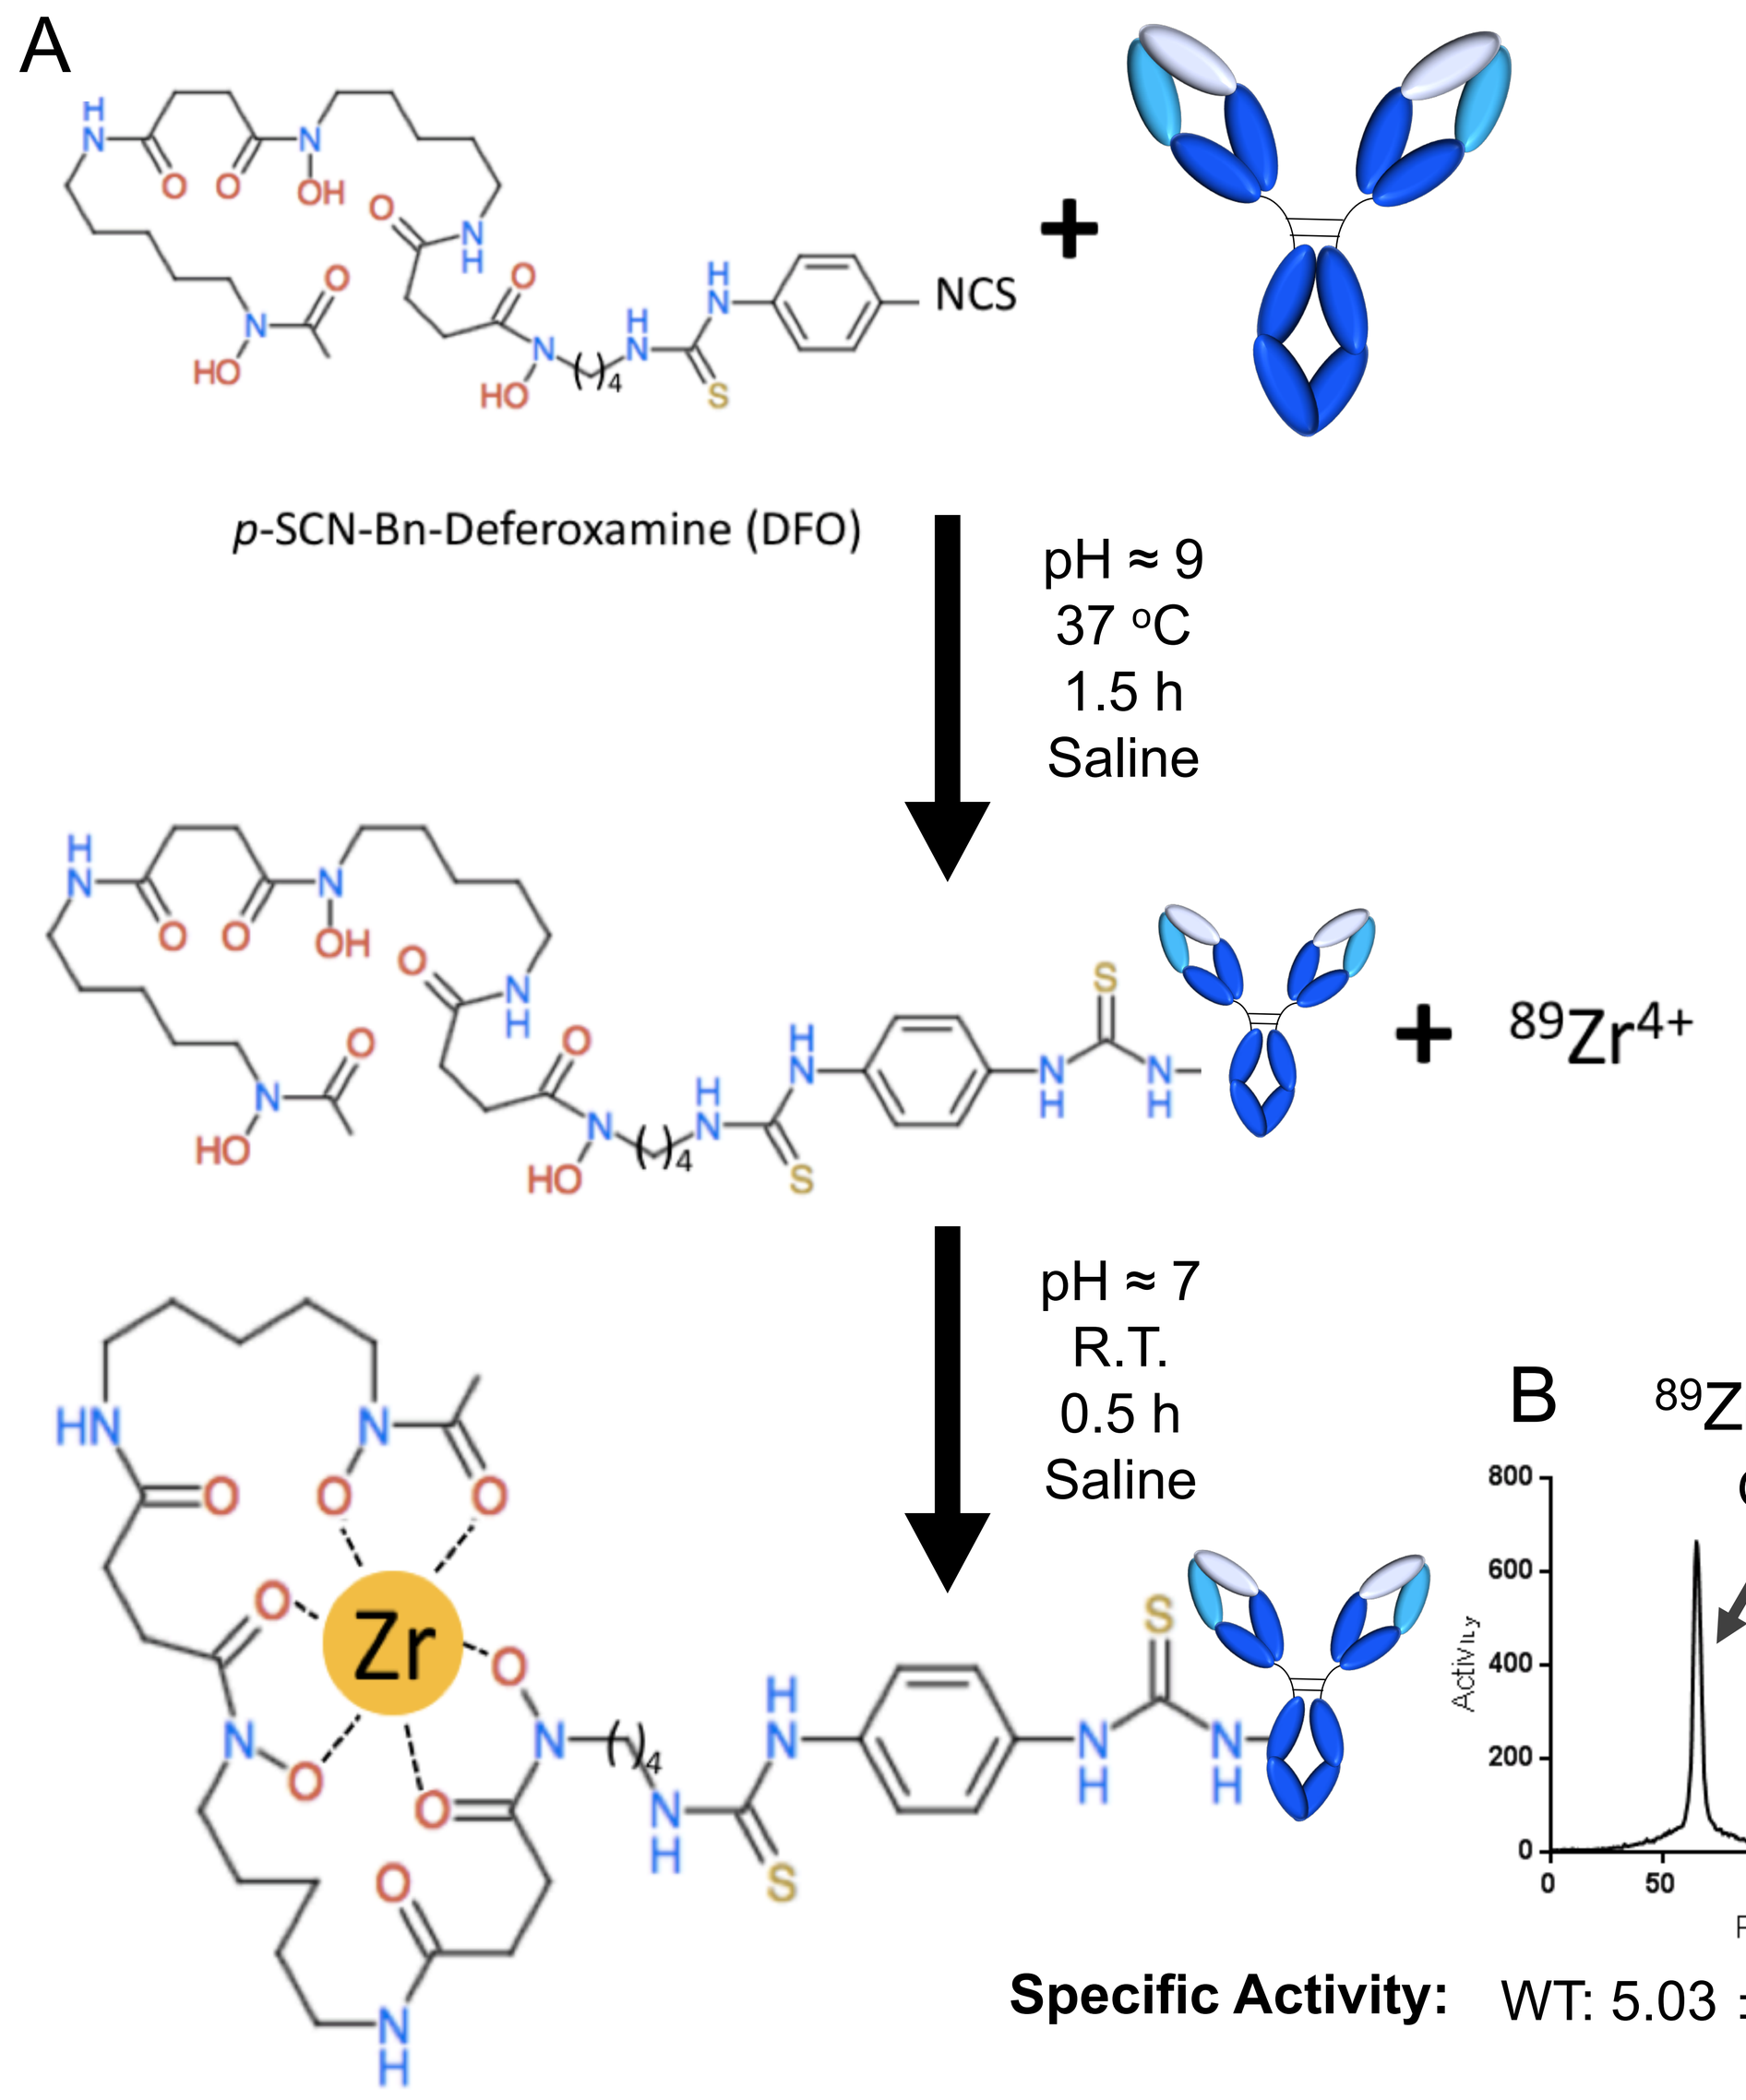
A
NCS
p-SCN-Bn-Deferoxamine (DFO)
pH ≈ 9
37 °C
1.5 h
Saline
89Zr4+
pH ≈ 7
R.T.
0.5 h
Saline
Zr

All antibodies were developed following the same method. (A) Both α-CD8-wt and α-CD8-degly were conjugated with a 1:10 mole ratio of mAb:DFO. The mAb-DFO conjugates were labeled with $^{89}Zr^{4+}$ at a neutral pH. ^{89}Zr-α-CD8 labeling was then assessed by instant thin layer chromatography (iTLC). Both ^{89}Zr-α-CD8-wt (B) and ^{89}Zr-α-CD8-degly (C) exhibited sufficient labeling at the origin and with no free ^{89}Zr peak at the solvent front.

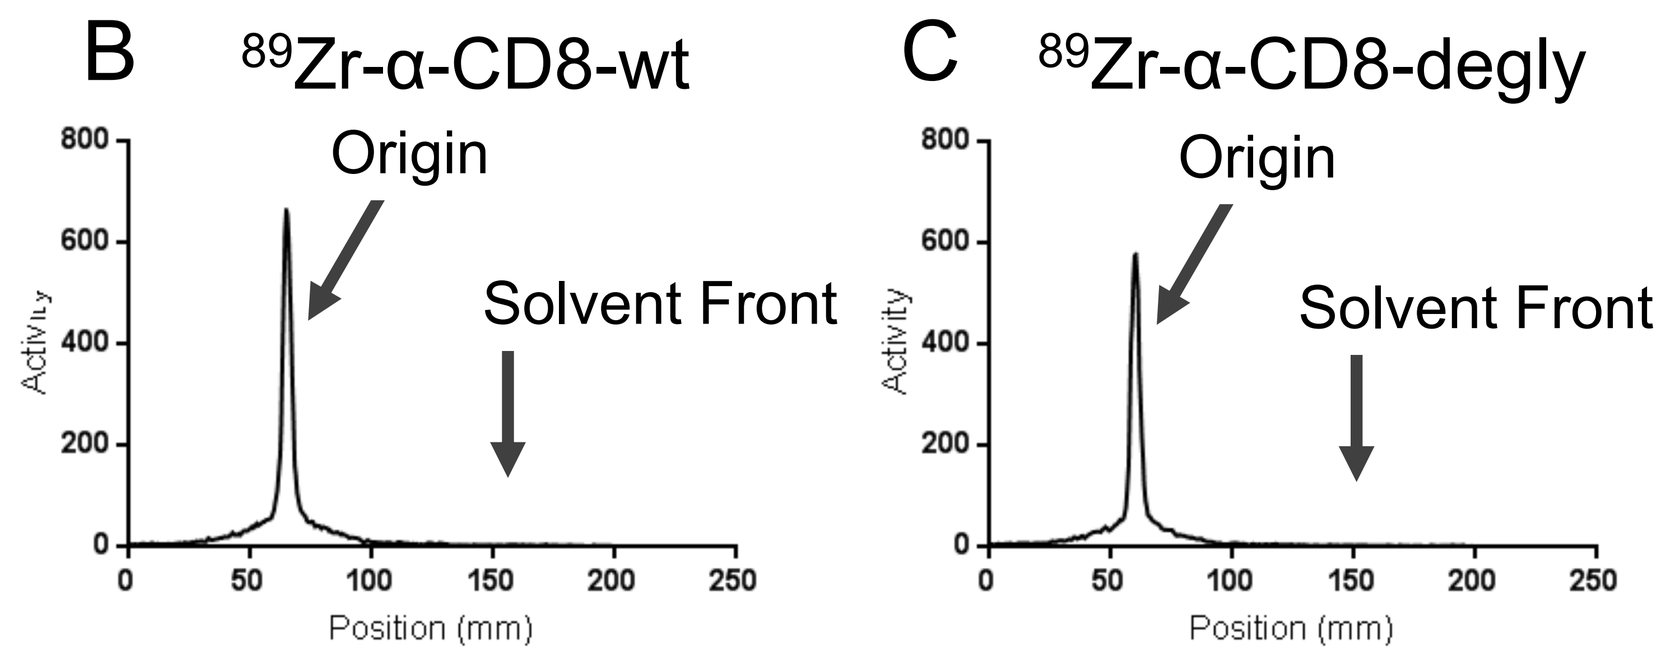
B 89Zr-α-CD8-wt
C 89Zr-α-CD8-degly
Origin
Solvent Front
Activity
Position (mm)

Specific Activity: WT: 5.03 ± 0.06 mCi/mg Degly: 4.95 ± 0.24 mCi/mg

α-CD8-degly exhibits targeted spleen uptake

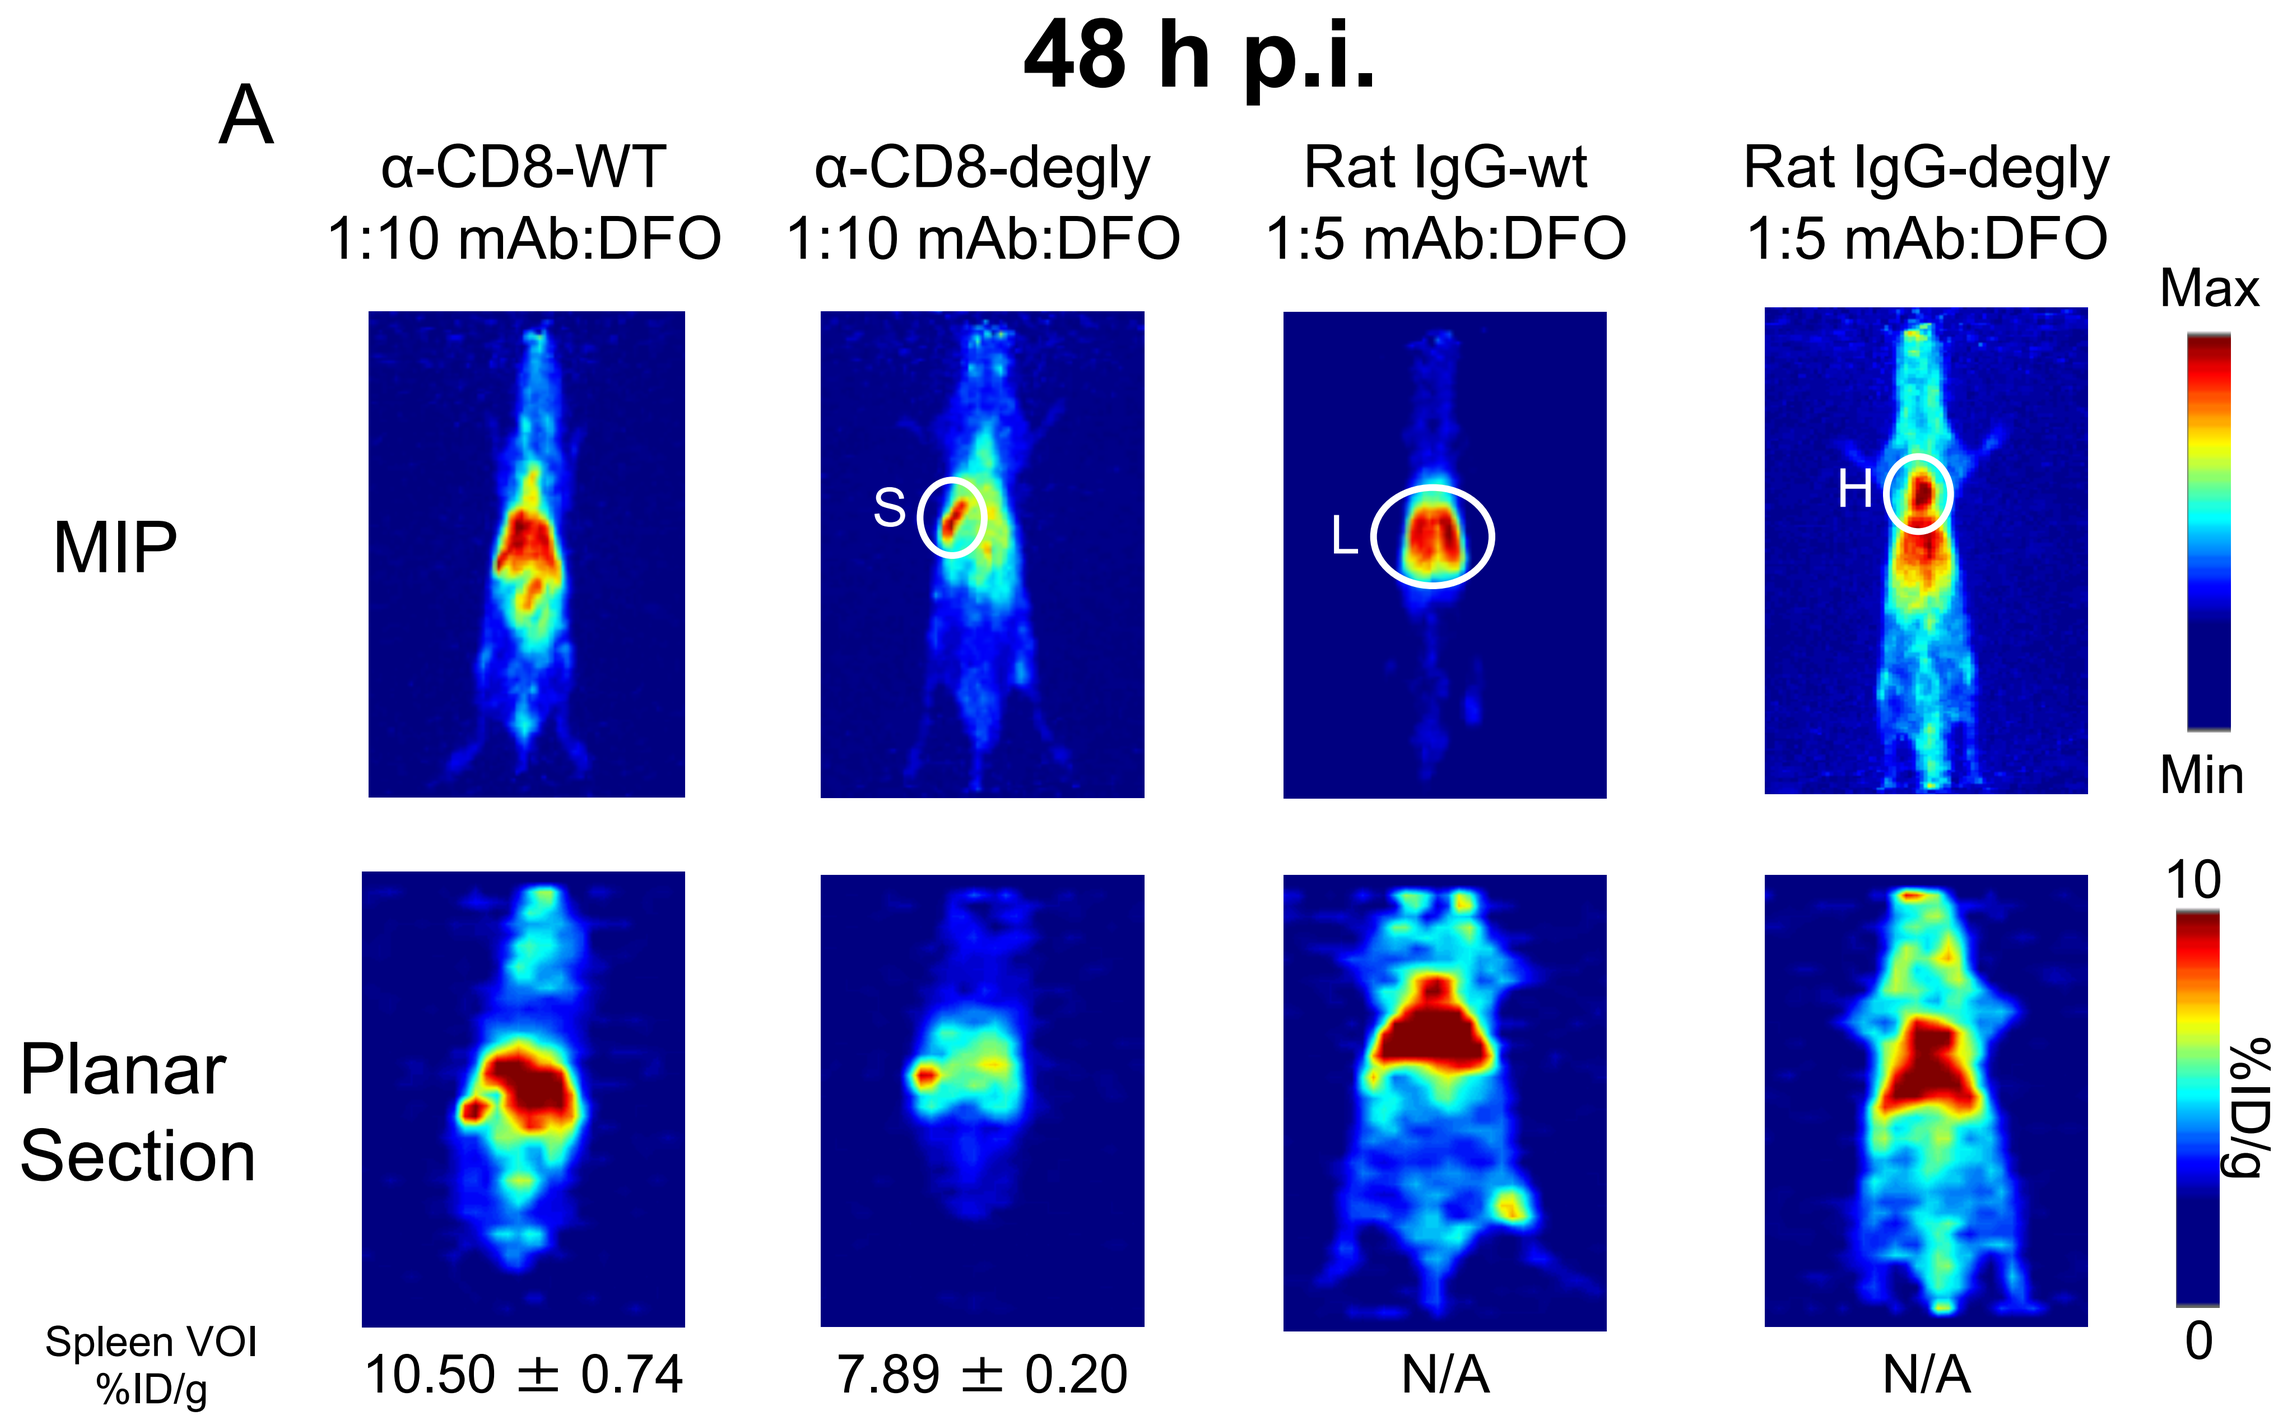
48 h p.i.
A
α-CD8-WT 1:10 mAb:DFO
α-CD8-degly 1:10 mAb:DFO
Rat IgG-wt 1:5 mAb:DFO
Rat IgG-degly 1:5 mAb:DFO
MIP
Planar Section
Max
Min
10
0
%ID/g
S
L
H
Spleen VOI %ID/g
10.50 ± 0.74
7.89 ± 0.20
N/A
N/A

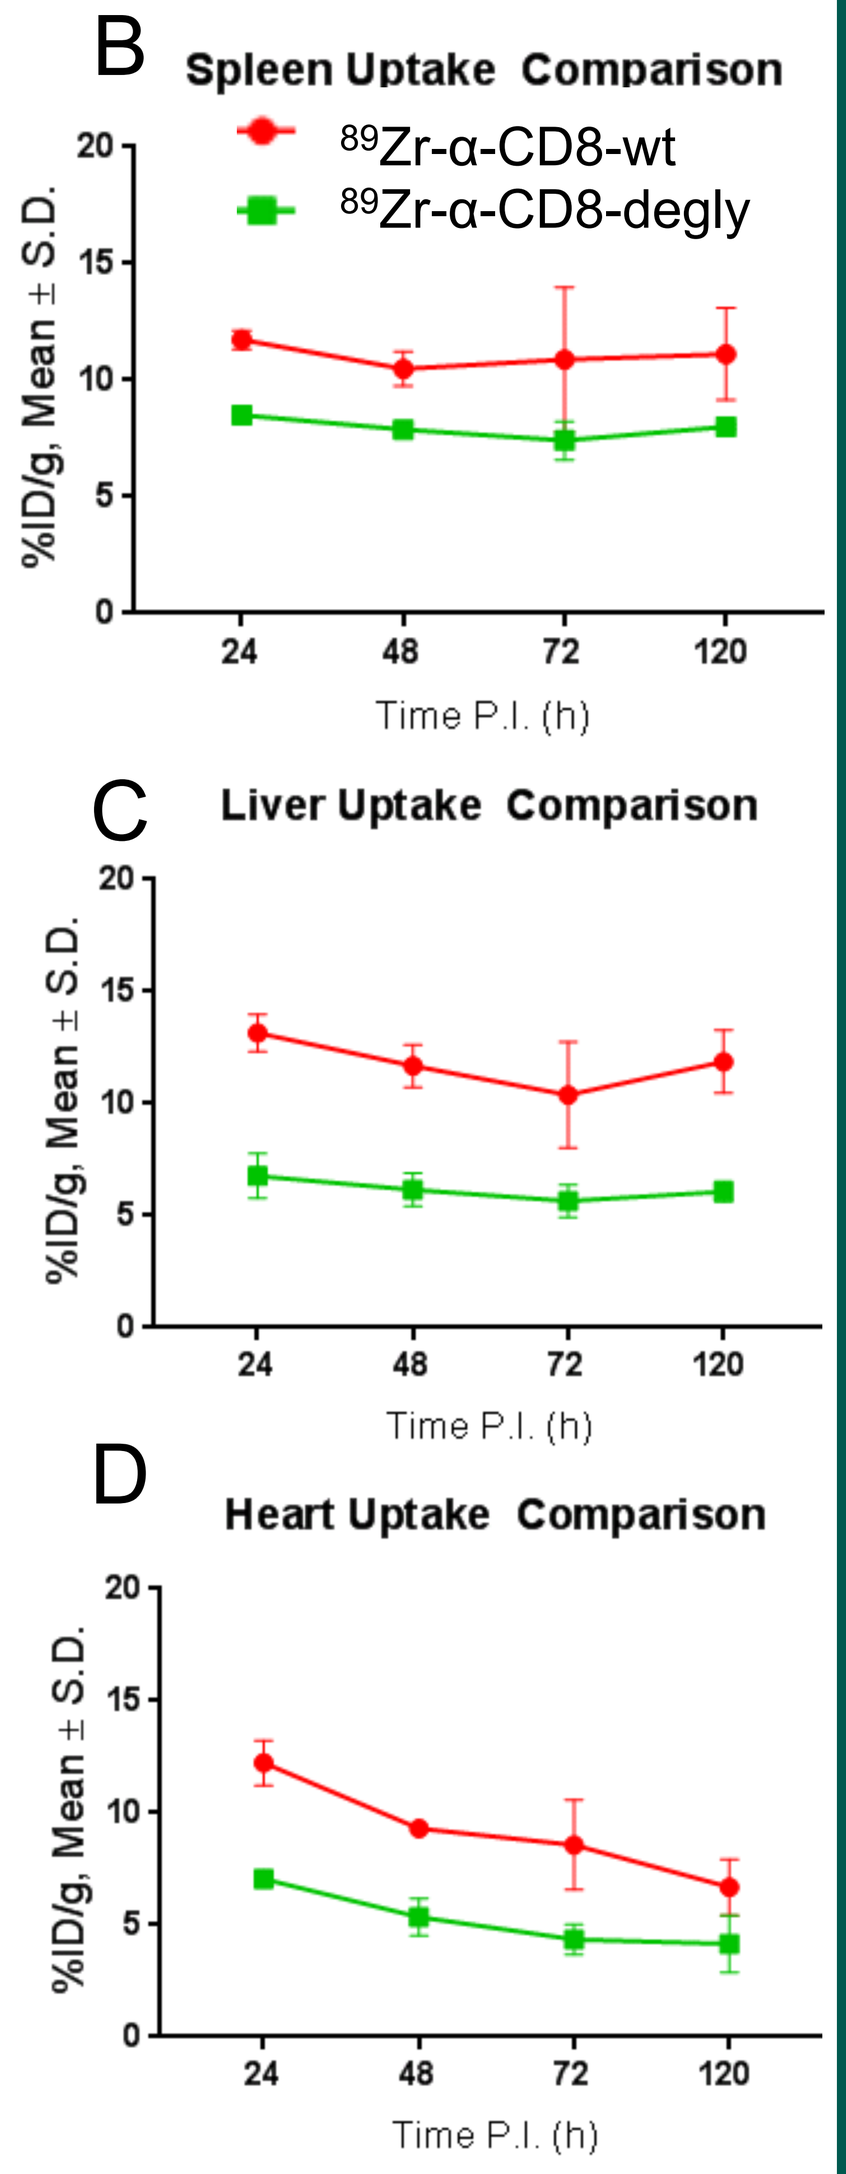
B Spleen Uptake Comparison
89Zr-α-CD8-wt
89Zr-α-CD8-degly
C Liver Uptake Comparison
D Heart Uptake Comparison
%ID/g, Mean ± S.D.
Time P.I. (h)

Balb/c male mice were imaged from 24 to 120 h p.i. with either ^{89}Zr-α-CD8-wt, ^{89}Zr-α-CD8-degly, or their corresponding non-specific IgG isotype control. (A) Comparisons of ^{89}Zr-α-CD8-wt, ^{89}Zr-α-CD8-degly, and their corresponding controls exhibited targeted spleen uptake in both ^{89}Zr-α-CD8-wt and ^{89}Zr-α-CD8-degly. Additionally, time activity uptake curves are shown for the spleen (B), liver (C) and heart (D) for both tracers.

S: Spleen; L: Liver; H: Heart; VOI: Volume of Interest

^{89}Zr-α-CD8 exhibits specific uptake

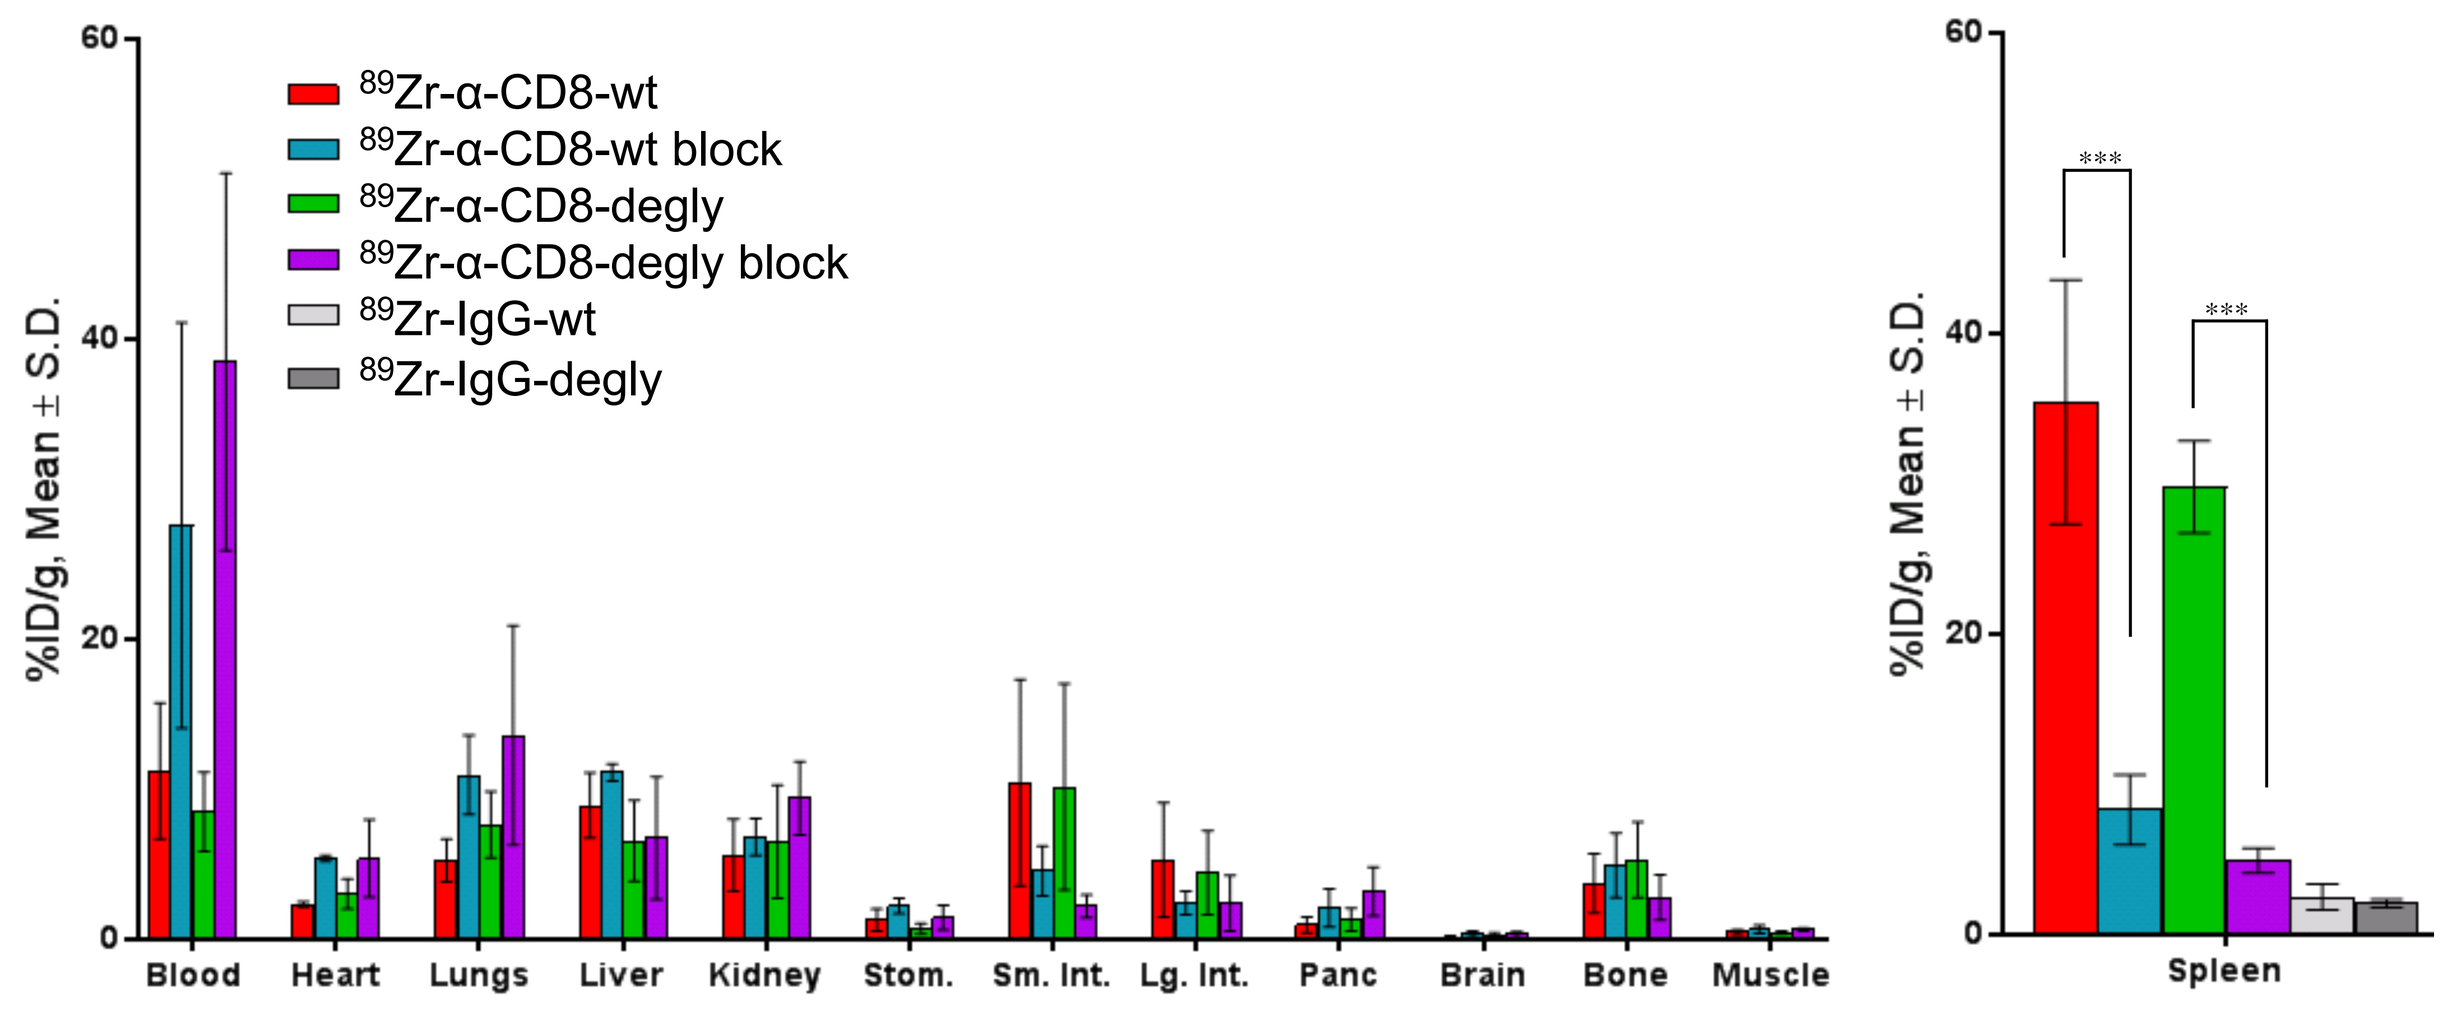
89Zr-α-CD8-wt
89Zr-α-CD8-wt block
89Zr-α-CD8-degly
89Zr-α-CD8-degly block
89Zr-IgG-wt
89Zr-IgG-degly
%ID/g, Mean ± S.D.
Blood
Heart
Lungs
Liver
Kidney
Stom.
Sm. Int.
Lg. Int.
Panc
Brain
Bone
Muscle
Spleen

To examine tracer specificity, mice were imaged with 40-50 µCi (1.48-1.85 MBq), 8-10 µg of tracer. A blocking cohort of mice received excess non-radiolabeled antibody (10×, 80-100 µg). The significant decrease in spleen uptake for both the α-CD8-wt and α-CD8-degly blocked cohorts suggests α-CD8 specificity is not affected by the glycosylation status of the mAb.

Treatment with α-CTLA-4 & α-PD-1

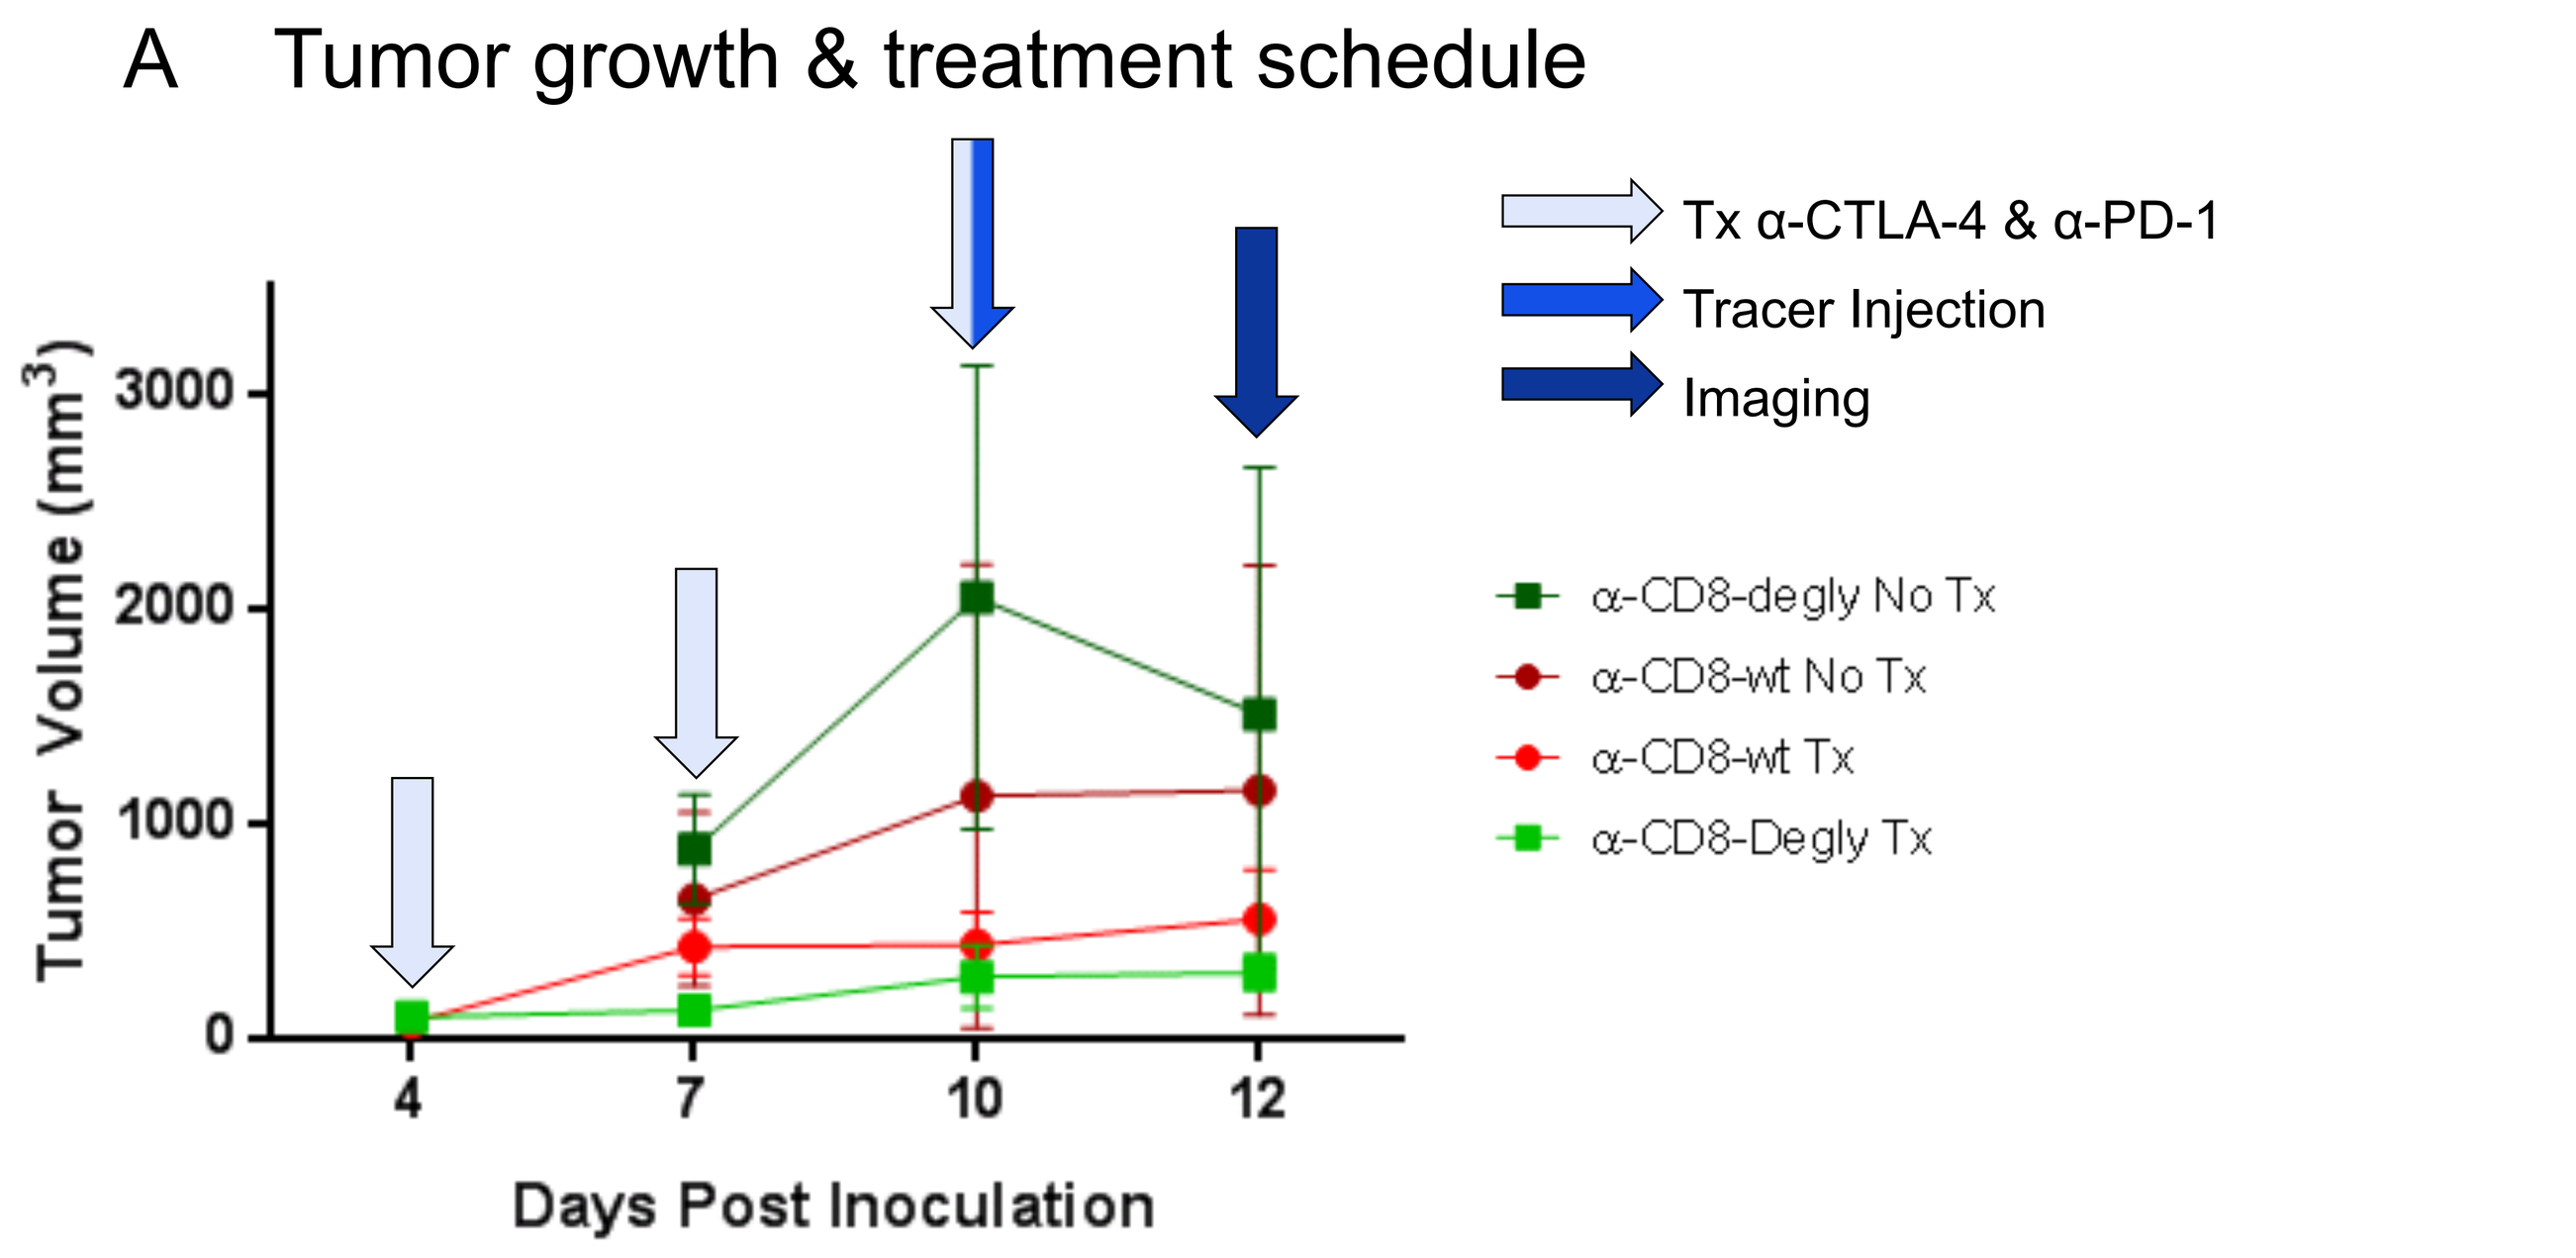
A Tumor growth & treatment schedule
Tx α-CTLA-4 & α-PD-1
Tracer Injection
Imaging
α-CD8-degly No Tx
α-CD8-wt No Tx
α-CD8-wt Tx
α-CD8-Degly Tx
Tumor Volume (mm3)
Days Post Inoculation

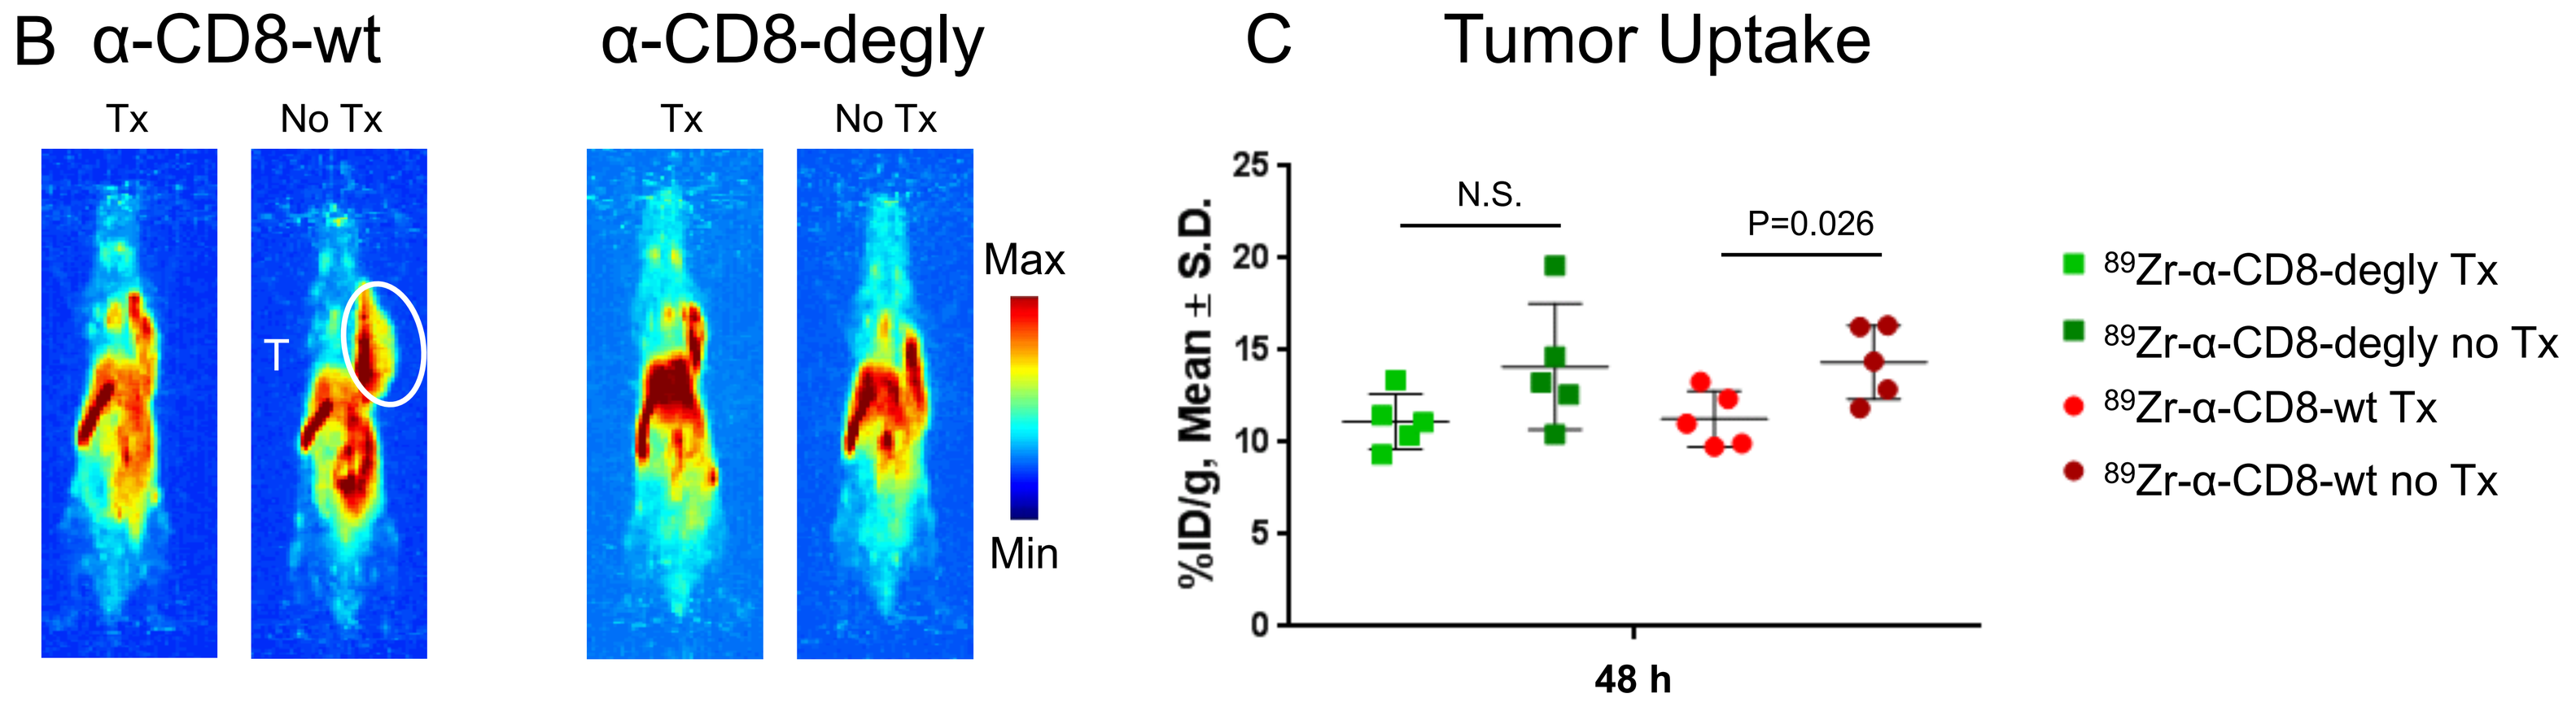
B α-CD8-wt
α-CD8-degly
Tx
No Tx
T
Max
Min
C Tumor Uptake
N.S.
P=0.026
%ID/g, Mean ± S.D.
48 h
89Zr-α-CD8-degly Tx
89Zr-α-CD8-degly no Tx
89Zr-α-CD8-wt Tx
89Zr-α-CD8-wt no Tx

D

Treatment	Tumor % ID/g (Mean ± SD)	Spleen % ID/g (Mean ± SD)
α-CD8-Degly Tx	11.14 ± 1.50 %	13.43 ± 1.55 %
α-CD8-Degly No Tx	14.12 ± 3.43 %	14.28 ± 2.00 %
α-CD8-Wt Tx	11.29 ± 1.53 %	14.11 ± 1.19 %
α-CD8-WT No Tx	14.36 ± 2.00 %	13.24 ± 1.55 %

To examine changes in CD8+ infiltrates in the tumor, mice were treated with CTLA-4 and PD-1. (A) Mice were inoculated on day 0 and treated intraperitoneally on days 4, 7 and 10 with 100 µg of α-CTLA-4 and 200 µg of α-PD-1. On day 10, mice were injected with either ^{89}Zr-α-CD8-wt or ^{89}Zr-α-CD8-degly and imaged on day 12. The tumor (T) growth exhibited by the two treated cohorts was similar. (B-D) The tumor uptake was significantly higher in the ^{89}Zr-α-CD8-wt untreated group, spleen uptakes were not significantly different between the two tracers.

Conclusions & Future Directions

- Full-length α-CD8-degly does not deplete the CD8+ T cell population.
- ^{89}Zr-labeled α-CD8-degly and α-CD8-wt exhibited similar pharmacokinetic profiles and specific uptake.
- Further examination of tumors with IHC, flow cytometry and qPCR for viable CD8+ T cell expression following treatment and imaging will be performed.

Acknowledgements

We would like to thank NIH/NCI R37 CA220482 (NTV/HMG), NIH T32 CAA09531 (JMW), The Microscopy, Imaging and Cytometry Resources Core (MICR) supported, in part, by NIH Center grant P30 CA022453 to the Karmanos Cancer Institute at Wayne State University, and the Perinatology Research Branch of the National Institutes of Child Health and Development at Wayne State University.