The Effect Of Left Thoracotomy LVAD Implantation To LVAD Surgery And Followed Heart Transplantation Surgery

B. Deng[1], M. Kawabori[2], P. Shindgikar[1], C. Hironaka[1], Y. Zhan[2], F. Y. Chen[2], M. Lawrence[2], A. R. Vest[2], M. S. Kiernan[2], D. DeNofrio[2], G. S. Couper[2]

[1]Tufts University School of Medicine, Boston, MA
[2]Tufts Medical Center, Boston, MA

Introduction

- Left ventricular assist device (LVAD) implantations have traditionally been approached via full median sternotomy (FS).
- Recently, minimally invasive left thoracotomy (LT) approach has been popularized with improved surgical technique and emergence of smaller centrifugal LVAD devices.
- However, the surgical outcomes from a minimal surgical approach remains to be unclear.
- The primary objective was to compare outcomes among patients who received an LVAD via FS compared to LT as well as their subsequent outcomes following heart transplantation.

Methods

- This is a single center retrospective study.
- 126 patients who underwent primary LVAD implantation from Jan 2014 to June 2018 are included (FS group n=83, LT group n=43).
- 49 patients who underwent heart transplantation (HT) surgery are included (FS group n=27, LT group n=22).
- Preoperative patient demographics, intraoperative variables, hospital stay, postoperative survival and complications from LVAD implantation and heart transplantation were analyzed.

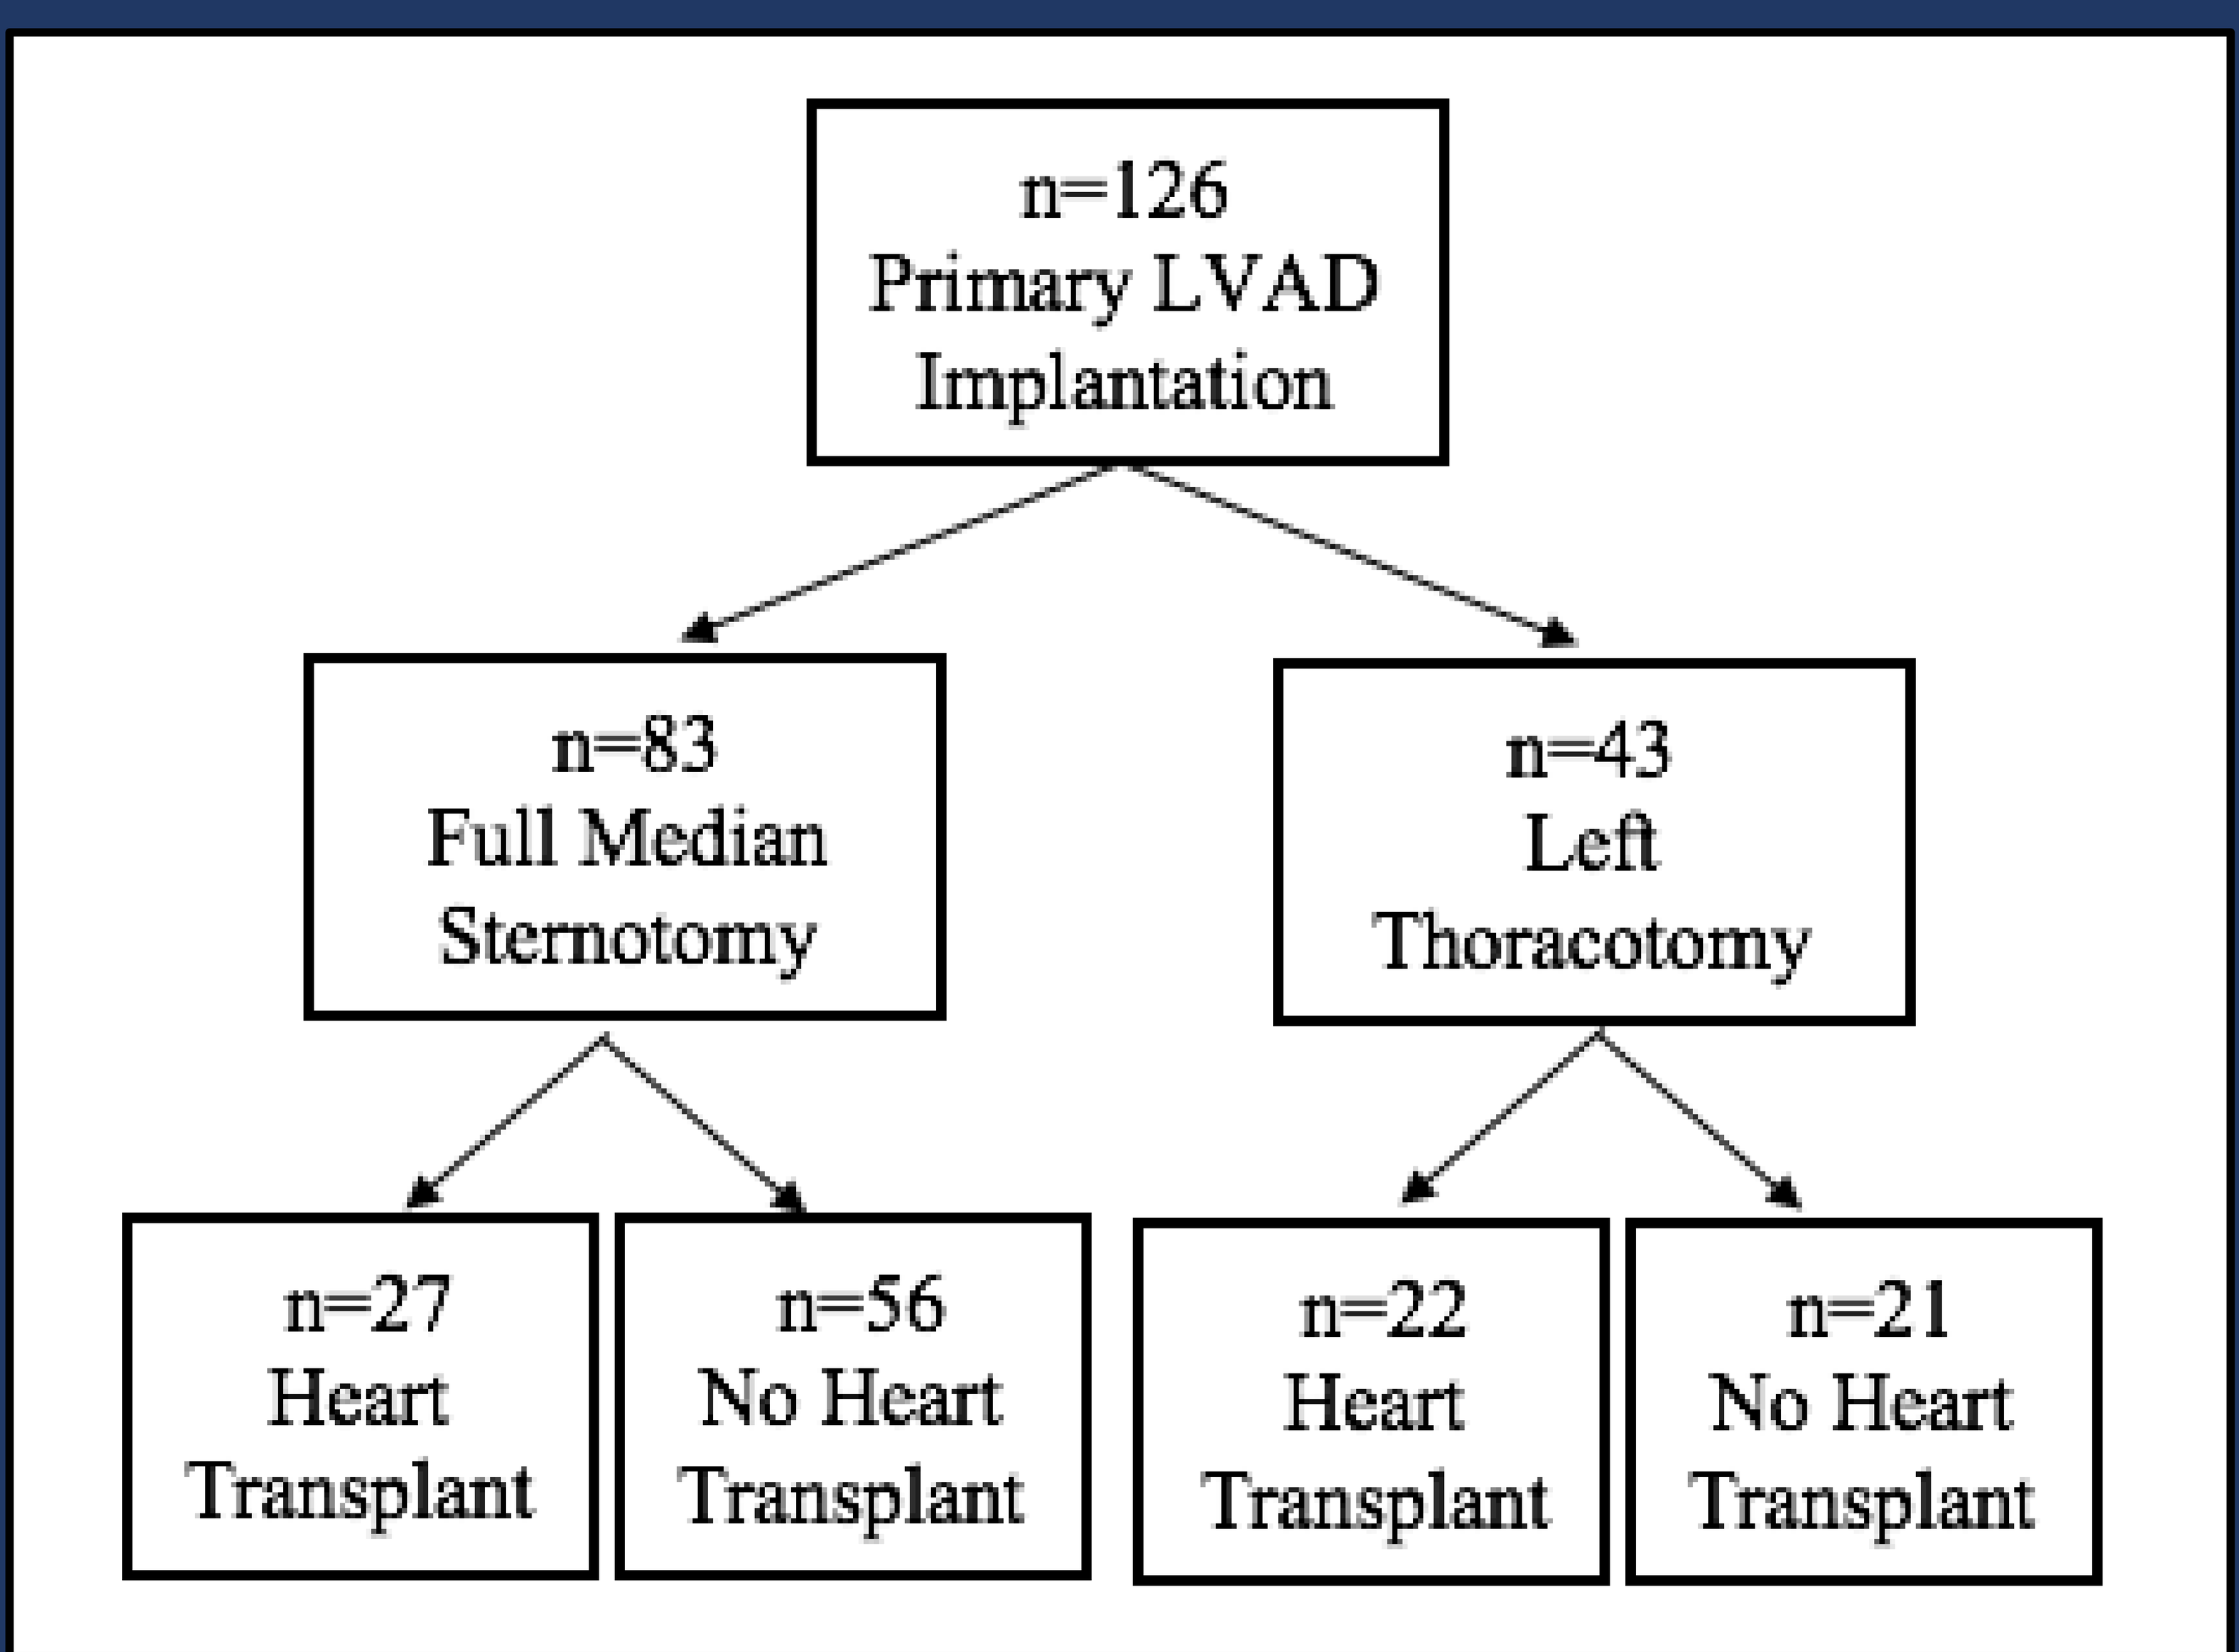
n=126
Primary LVAD
Implantation
n=83
Full Median
Sternotomy
n=43
Left
Thoracotomy
n=27
Heart
Transplant
n=56
No Heart
Transplant
n=22
Heart
Transplant
n=21
No Heart
Transplant

Figure 1: Schematic illustrating number of participants in the study who were followed based on procedures they underwent.

Results

- Preoperatively, the FS group had higher right atrial pressures ($p<0.05$).
- Intraoperative data showed LT group had 24% longer LVAD implant surgical time ($p<0.05$).
- Kaplan Meier curve showed better survival in LT group after LVAD implantation ($p=0.04$). The 1-year survival rate after heart transplantation was 100% for both groups.
- For post LVAD complications, FS group had more gastrointestinal bleeding events ($p<0.05$).
- There were no significant differences in post heart transplantation complications.
- Lastly, average length of hospital stay after LVAD implantation and heart transplantation showed no significance, respectively (25.0±24.9 vs 19.3±12.0, $p=0.18$),(28.2±41.9 vs 21.1± 13.7, $p=0.46$).

Conclusion

- Our study showed LT group was associated with improved survival after LVAD implantation.
- However, our study has limitation of containing bias from retrospective, non-randomized study design.
- Further, larger studies are warranted to prove the effects of minimally invasive LVAD implantation surgery.

Table 1A

Complications after LVAD Implantation (events per patient support year)			
	Full Sternotomy	Left Thoracotomy	p Value
GI Bleeding	0.43	0.08	***p < 0.01***
AKI	0.11	0.04	p = 0.15
Ischemic Stroke	0.1	0.15	p = 0.33
Hemorrhagic Stroke	0.05	0	p = 0.12
Other Neuro Dysfunction	0.05	0.02	p = 0.40
Right Heart Failure	0.36	0.42	p = 0.55

Table 1B

Complications after Heart Transplantation (events per patient support year)			
	Full Sternotomy	Left Thoracotomy	p Value
GI Bleeding	0.04	0.02	p = 0.10
AKI	0.19	0.1	p = 0.23
Ischemic Stroke	0.01	0.05	p = 0.21
Hemorrhagic Stroke	0	0	p = 1.00
Other Neuro Dysfunction	0.01	0	p = 0.48
Right Heart Failure	0.12	0.2	p = 0.28

Table 1A, 1B: This data illustrates significant post-operative complications after LVAD implantation (1A) and heart transplantation (1B) for both FS and LT groups. Complications determined based on common post-operative complications after LVAD implantation.

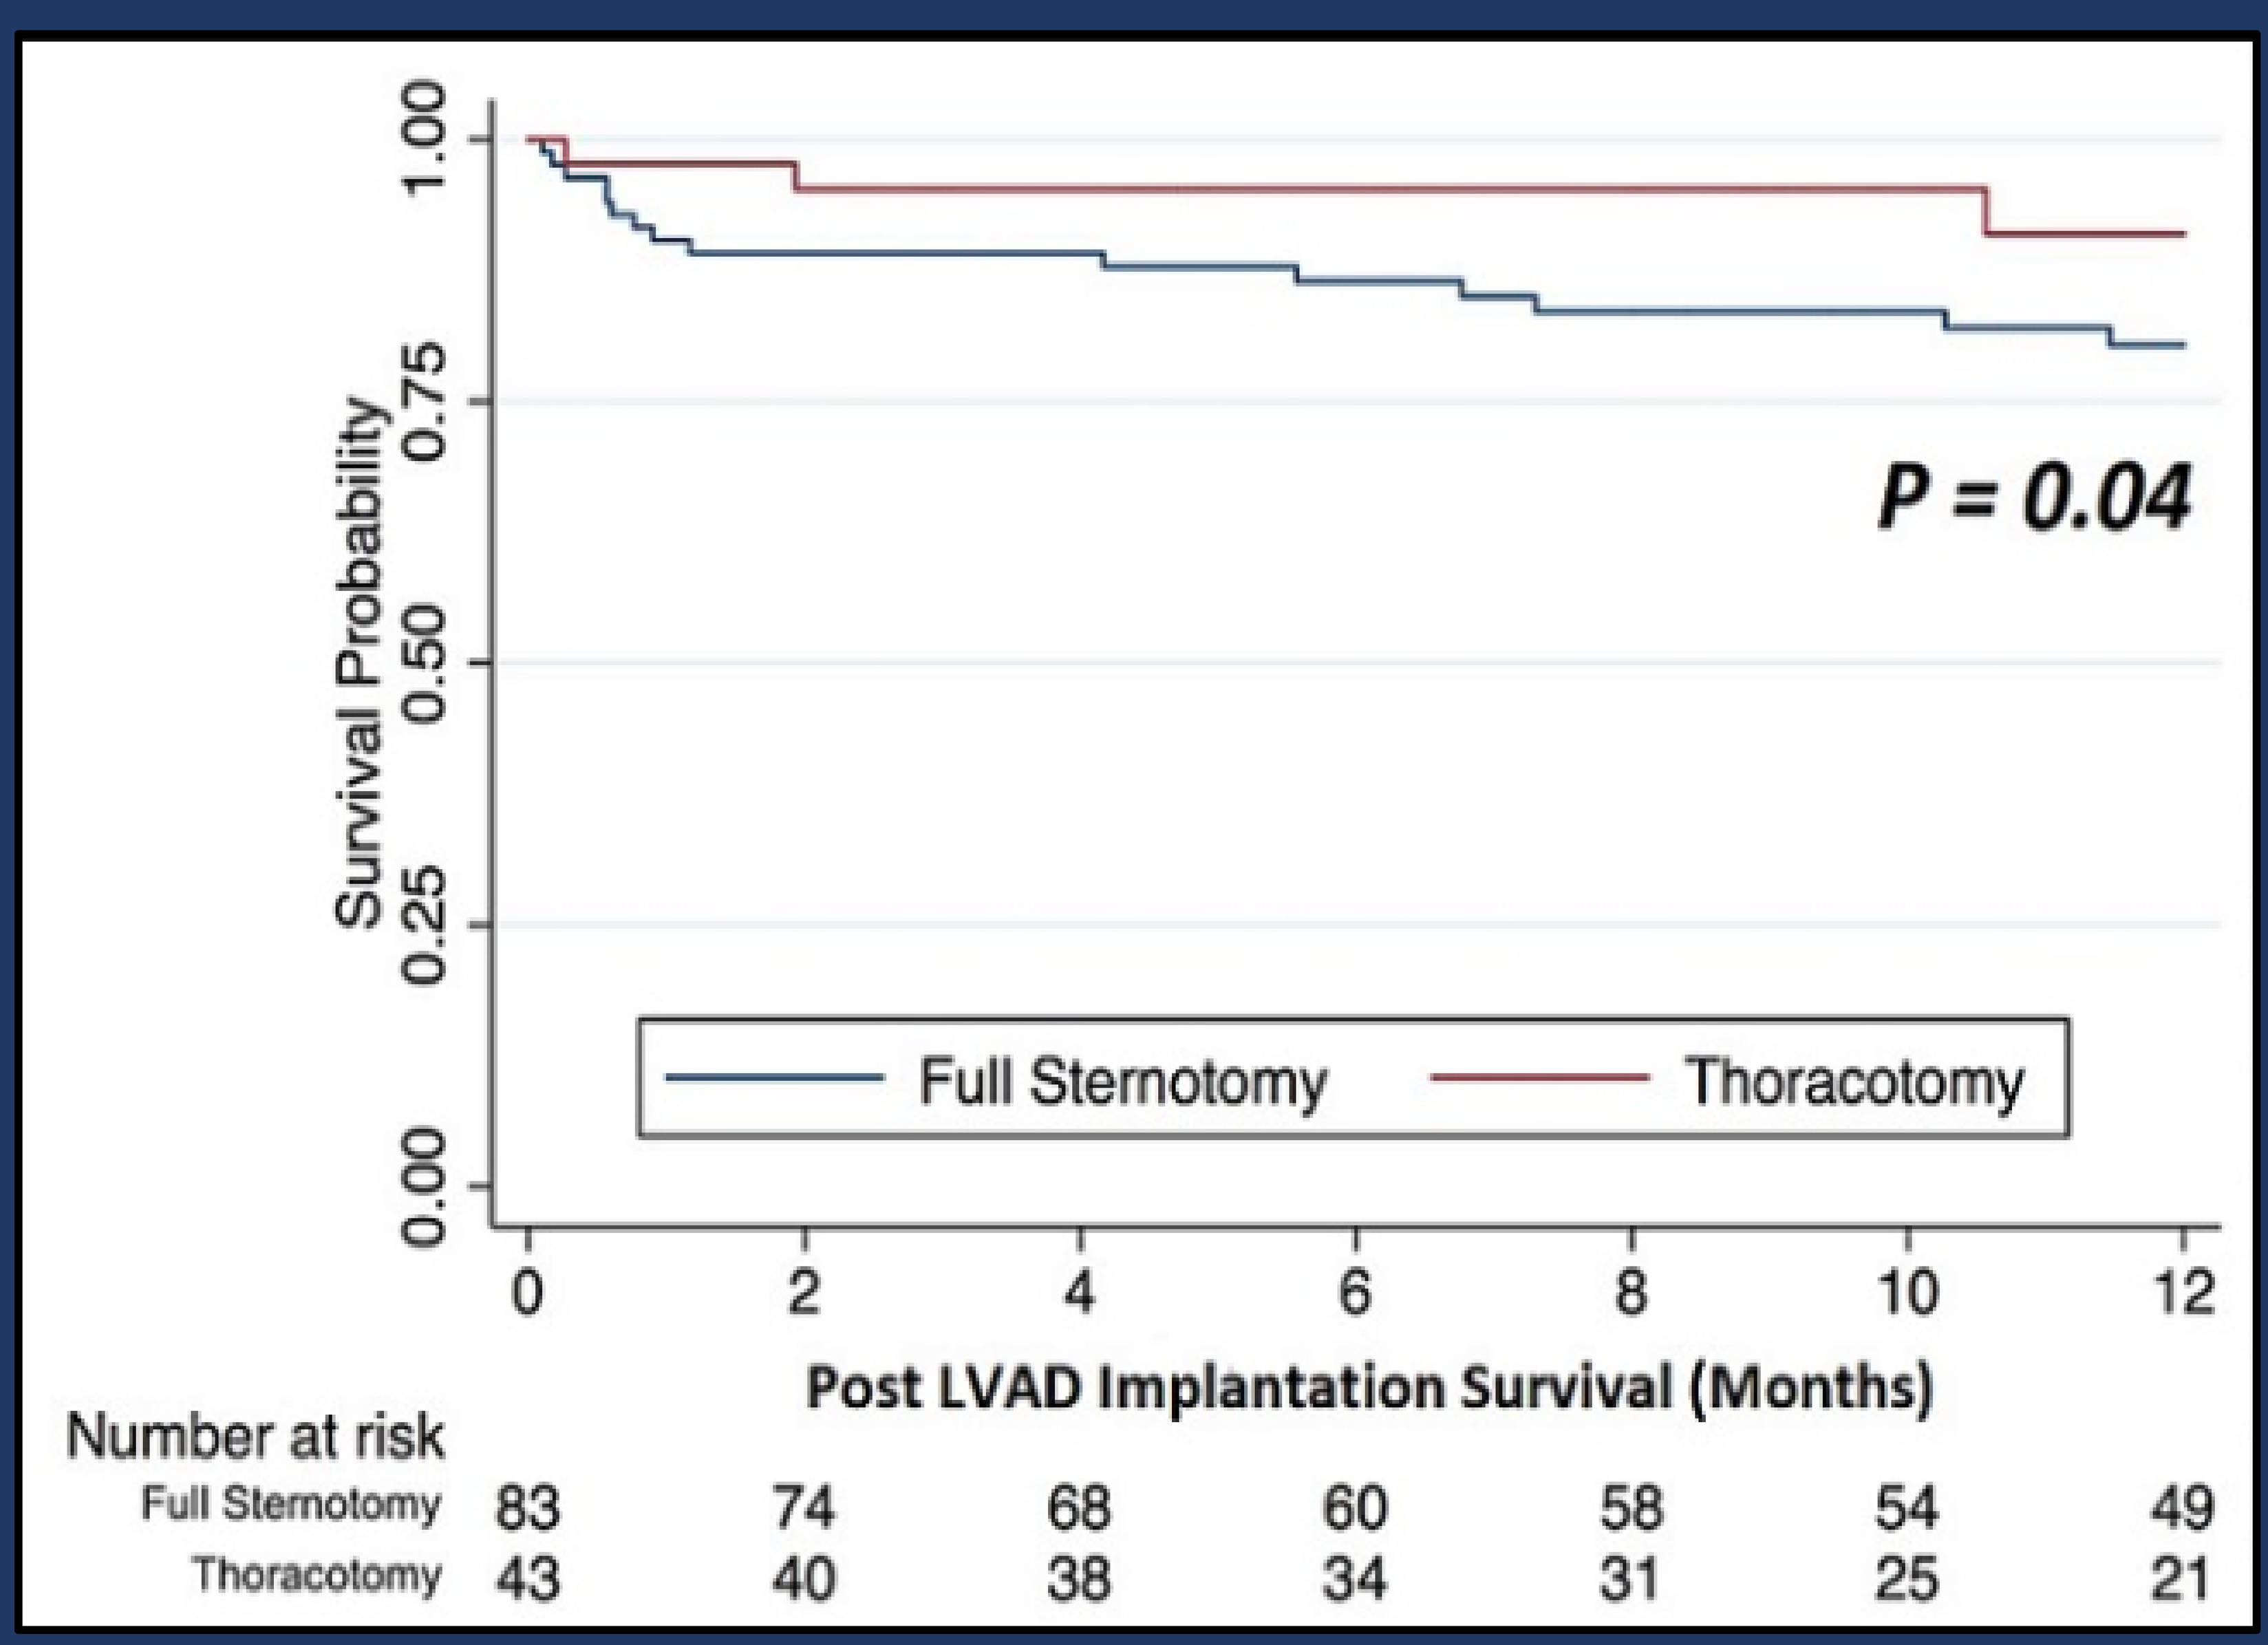
Survival Probability
0.00
0.25
0.50
0.75
1.00
P = 0.04
Full Sternotomy
Thoracotomy
0
2
4
6
8
10
12
Post LVAD Implantation Survival (Months)
Number at risk
Full Sternotomy 83 74 68 60 58 54 49
Thoracotomy 43 40 38 34 31 25 21

Figure 2: Survival after a left thoracotomy approach for LVAD implantation was greater than for a full sternotomy approach.

References

1. Strueber M, O'Driscoll G, Jansz P, Khaghani A, Levy WC, Wieselthaler GM. Multicenter evaluation of an intrapericardial left ventricular assist system. J Am Coll Cardiol 2011;57:1375–82.
2. Sabashnikov A, Mohite PN, Zych B, etal. Outcomes and predictors of early mortality after continuous-flow left ventricular assist device implantation as a bridge to transplantation. ASAIO J 2014;60:162–9.
3. Popov AF, Hosseini MT, Zych B, Simon AR, Bahrami T. HeartWare left ventricular assist device implantation through bilateral anterior thoracotomy. Ann Thorac Surg 2012;93:674–6.

Disclosures

No authors have any conflicts of interest or any financial relationships with commercial entities to this direction.