Post-Discharge Quality of Life with a Long-Term Ventricular Assist Device: Focus on Anxiety, Depression and Post-Traumatic Stress Disorder

Sladen RN [a], Shulman MA [b], Javaid A [c], Amlani AM [c], Hupf JC [c], Naka Y [d], McGiffin D [e], Bergin P [f], Takayama H [d], Buckland M [b], Takeda K [d], Hodgson CL [g], Yozefspolskaya M [c], Myles PS [b], Colombo PC [c]

Departments of Anesthesiology (Critical Care Medicine) [a], Surgery (Cardiothoracic Surgery) [d] and Medicine (Cardiology) [c]
at Columbia University Medical Center New York NY, USA
Departments of Anaesthesiology and Perioperative Medicine [b], Cardiothoracic Surgery [e] and Cardiology [f]
at The Alfred Hospital and Department of Epidemiology [g] at Monash University Melbourne, Australia

PURPOSE

Reported outcomes after left ventricular assist device (LVAD) implantation as destination therapy (DT) or bridge to transplant (BTT) commonly include 6-month mortality, hospital readmission and severe adverse events (SAEs). We have less information about patients' perceptions of their quality of life (QOL) after hospital discharge.

We conducted an IRB-approved prospective two-center international pilot study to assess anxiety, depression and post-traumatic stress disorder (PTSD) and relate them to the European Quality of Life Visual Analog Scale (EQ-VAS).

METHODS

124 patients ≥ 30 days after hospital discharge post-implantation were assessed during on-site clinic visits or by telephonic follow up at home.

Four validated questionnaires were administered:

1. Generalized Anxiety Disorder Scale (**GAD-7**)[1], a seven-item 0-3 scale assessing patient-perceived symptoms of anxiety (maximum score 21).

2. Patient Health Questionnaire (**PHQ-9**)[1], a nine-item 0-3 scale assessing patient-perceived symptoms of depression (maximum score 27).

GADS-7 scores ≥ 10 and PHQ-9 scores ≥ 14 warrant further evaluation and possible therapeutic intervention.

3. Impact of Event Scale – Revised (**IES-R**)[2] a 22-item 0-4 scale (maximum 88) reflecting self-described symptoms of PTSD. Scores ≥ 24 imply clinical concern for PTSD; ≥ 33 probable PTSD and ≥ 37 severe PTSD.

3. EQ Visual Analog Scale (**EQ-VAS**)[3] requires the patient to say or point to a score on a thermometer-like scale describing their health that day between 0 (worst imaginable) and 100 (best imaginable).

RESULTS

Demographic Data:

Mean patient age was 57.5±15.2 yrs, with 20% between 20-50 yrs and 27% between 70-90 yrs; only 15% of patients were female. 47% of patients were >1 year post-implantation, 56% of patients were DT, and 67% of patients had a HeartMate III LVAD.

GAD-7 and PHQ-9:

Scores warranted further evaluation for anxiety in 18% of patients and for depression in 8% of patients. A small number exhibited suicidal ideation. Symptoms of severe anxiety were more likely in patients < 60 yrs (p <0.01).

GAD-7 (Anxiety)

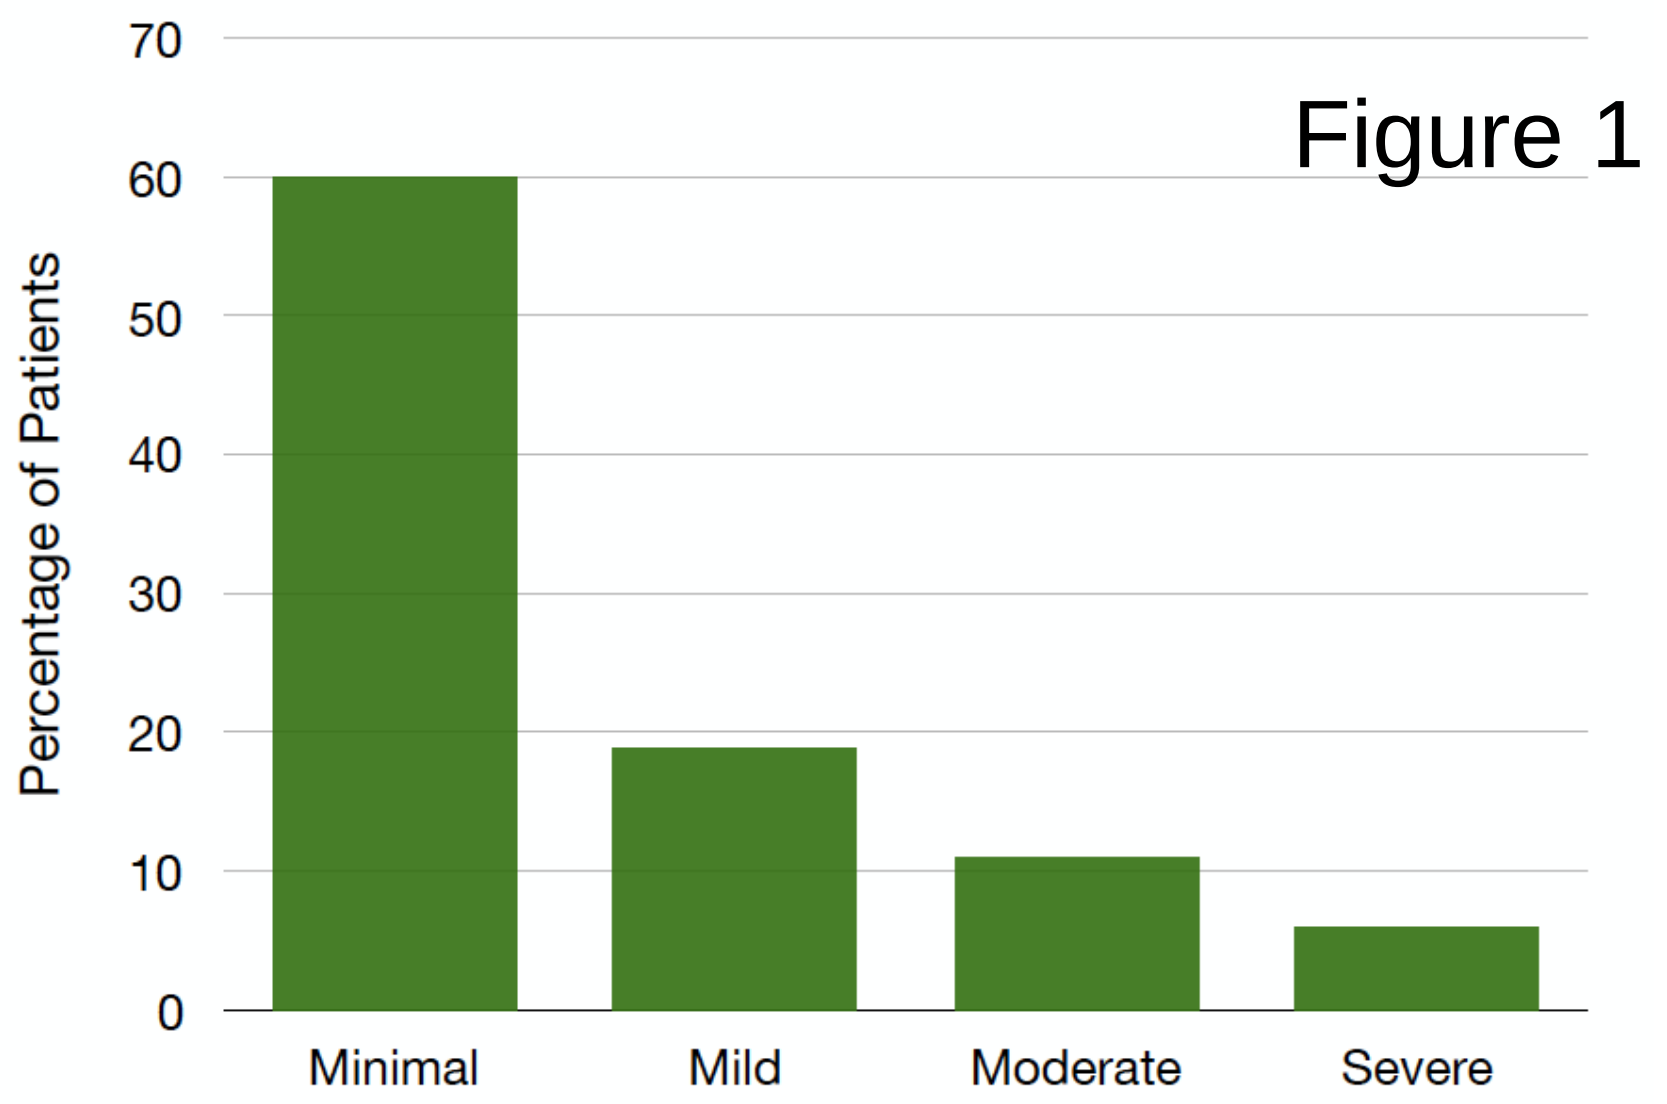
Percentage of Patients
0
10
20
30
40
50
60
70
Minimal
Mild
Moderate
Severe

Figure 1

PHQ-9 (Depression)

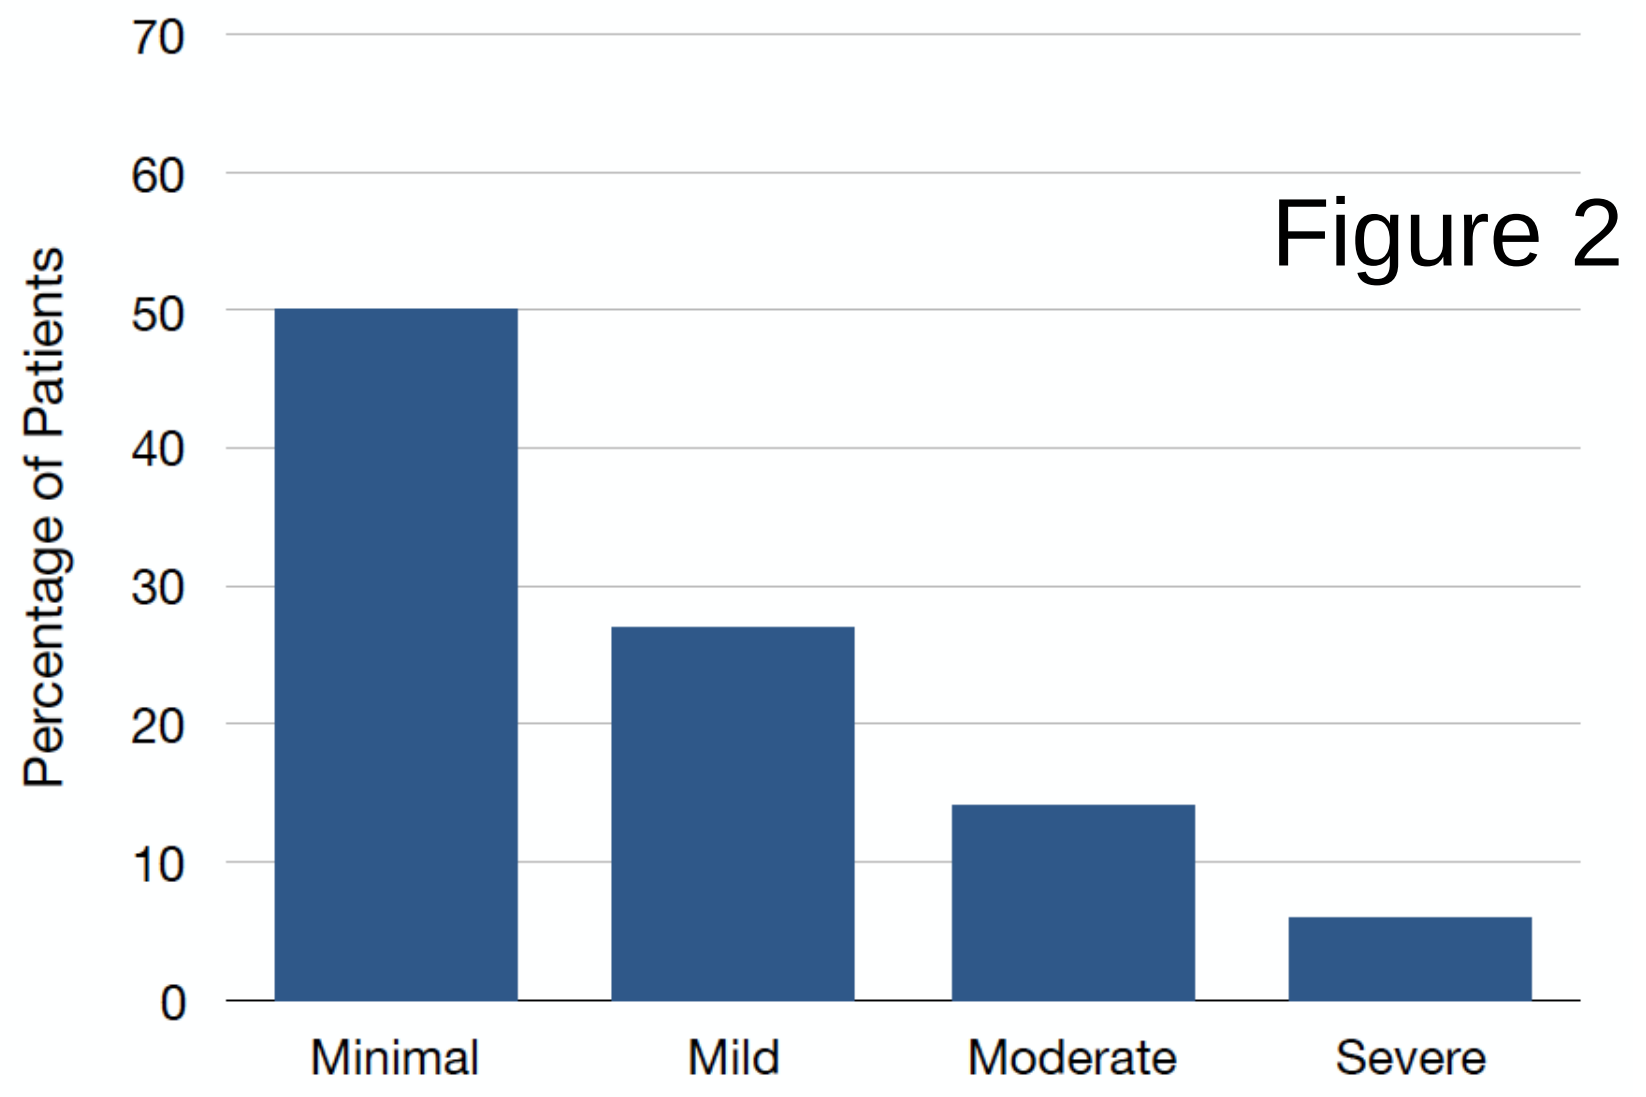
Percentage of Patients
0
10
20
30
40
50
60
70
Minimal
Mild
Moderate
Severe

Figure 2

IES-R:

23% of patients had scores indicating concern for PTSD, and in 80% of these their scores indicated it was moderate to severe (Figure 2).

The risk of anxiety, depression and PTSD was highest in patients with EQ-VAS < 40 (p<0.05) (Figure 2).

There were no differences in the risk of anxiety, depression or PTSD related to gender or LVAD strategy (DT or BTT)

RESULTS

IES-R Score (PTSD)

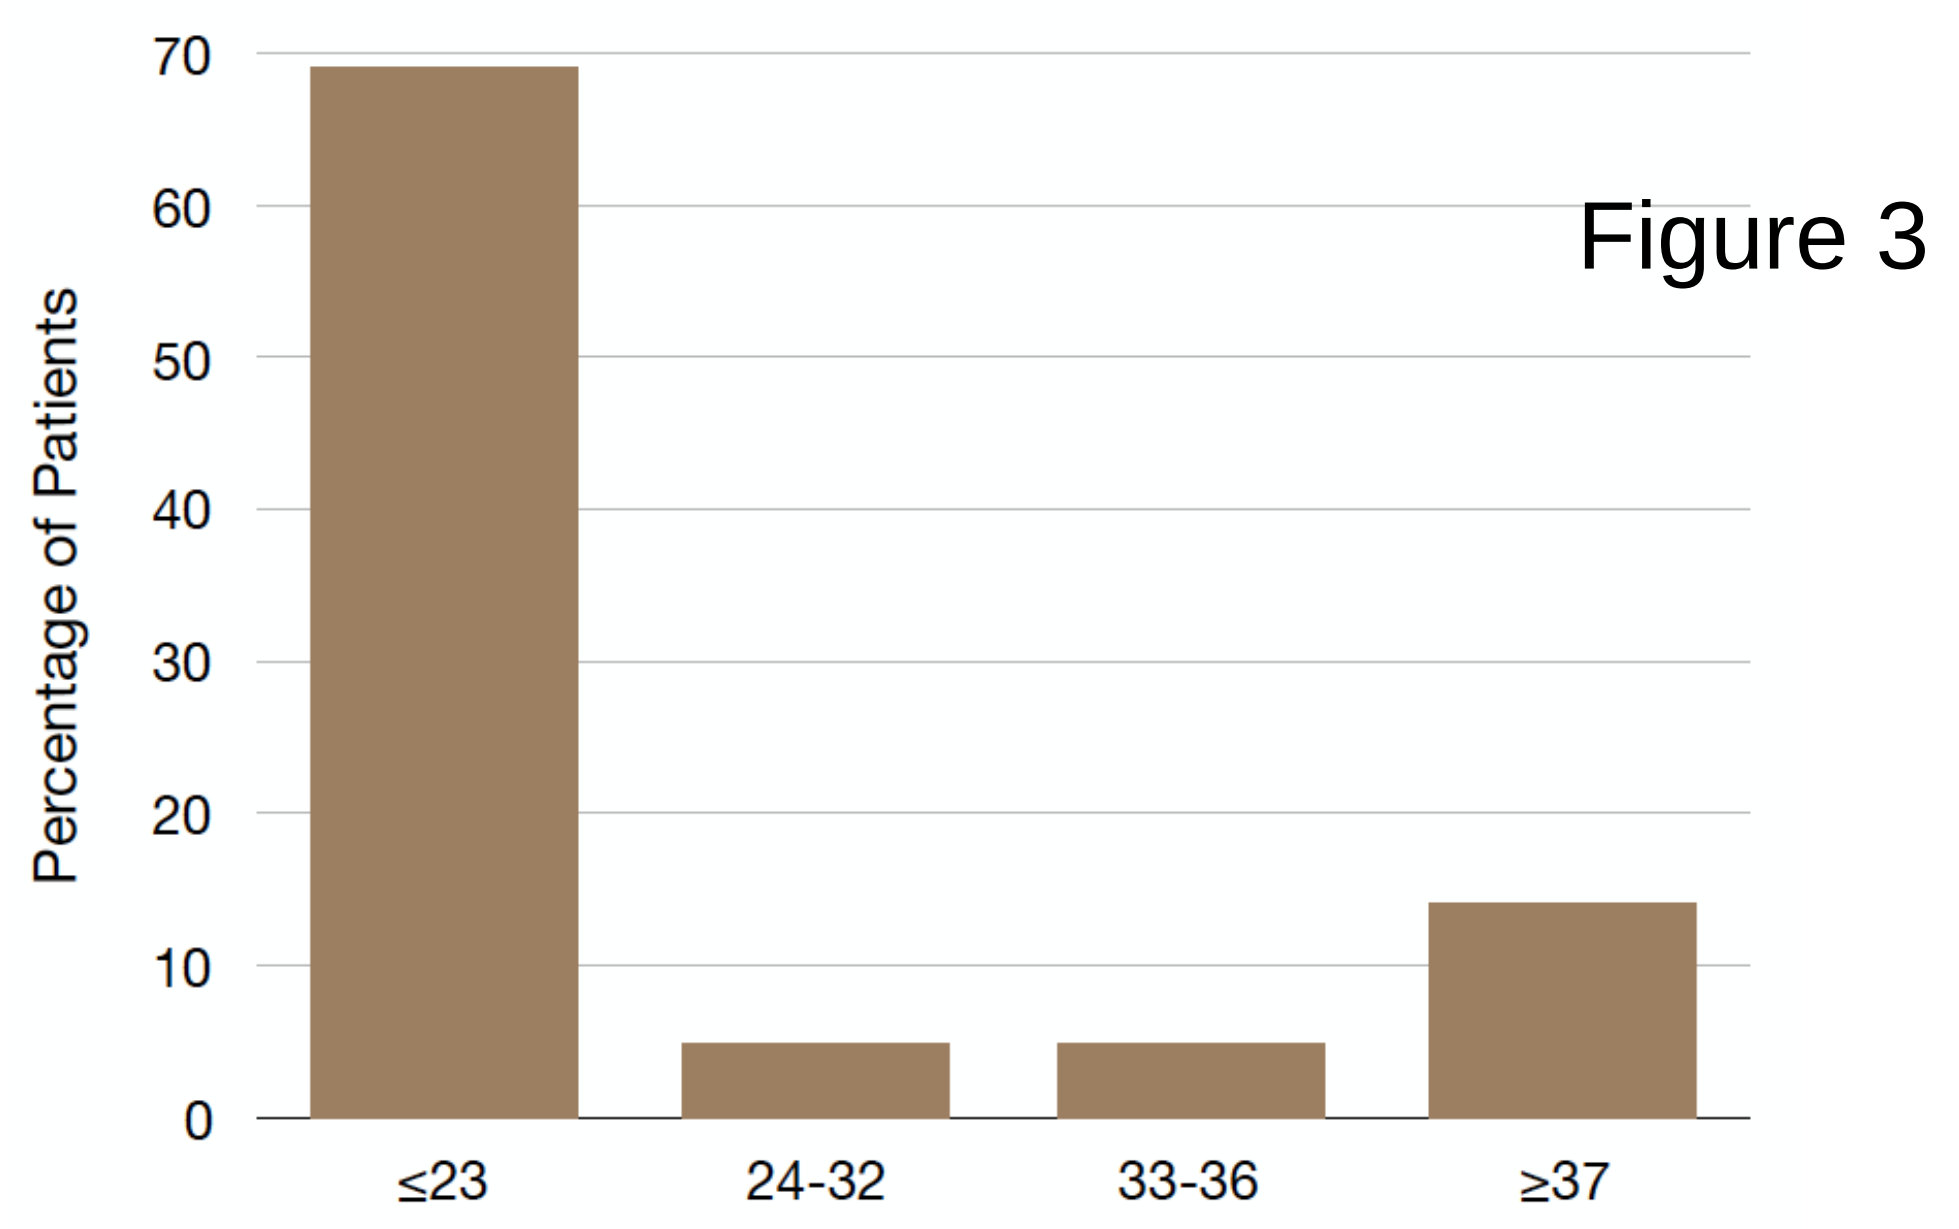
Percentage of Patients
0
10
20
30
40
50
60
70
≤23
24-32
33-36
≥37

Figure 3

EQ-VAS Distribution

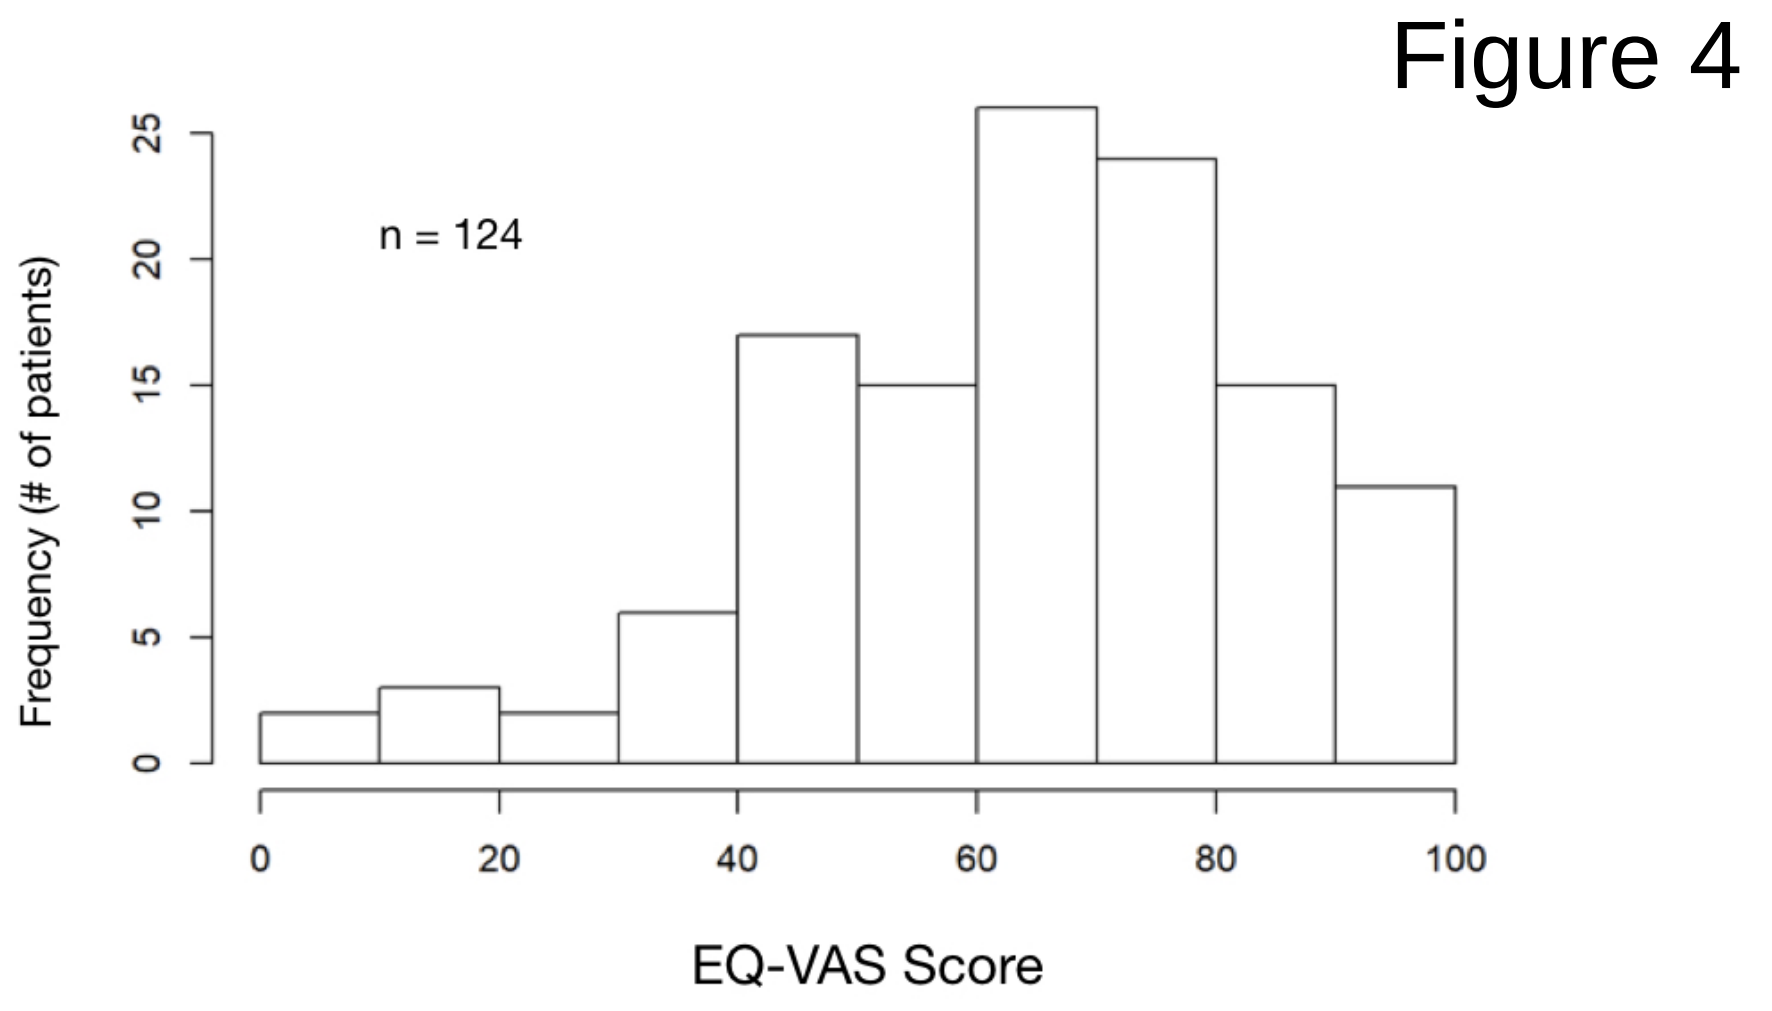
n = 124
Frequency (# of patients)
0
5
10
15
20
25
0
20
40
60
80
100
EQ-VAS Score

Figure 4

CONCLUSIONS

The majority of LVAD out-patients reported low levels of anxiety, depression and did not exhibit symptoms of PTSD.

However, a small but important minority exhibit symptoms of anxiety, depression and PTSD that warrant evaluation for therapeutic intervention.

A low EQ-VAS (< 40) may be helpful in identifying patients at risk, but a high EQ-VAS does not exclude it.

Routine assessment of functional ability and health status is feasible in post-discharge LVAD patients.

Standardized, regular assessments may help to identify patients in urgent need of evaluation for severe anxiety, depression (including suicidal ideation) and PTSD.

REFERENCES

1. Kroenke K et al. *Psychosom Med* 2016;78:716-27
2. Bienvenu OJ et al. *Chest* 2013;144:24-31
3. Feng Y et al. *Qual Life Res* 2014;23:977-89