Is frailty associated with sarcopenia and dynapenia in lung transplant candidates?

Sunita Mathur[1], Manoela Ferreira[1], Nathalia Maia[1], Christian Martin[1], Noori Chowdhury[2], Anam Islam[2], Lianne Singer[2,3]

[1]Department of Physical Therapy, University of Toronto, [2]Toronto Lung Transplant Program, University Health Network, [3]Department of Medicine, University of Toronto, **Toronto, CANADA**

Contact: sunita.mathur@utoronto.ca

BACKGROUND:

- Frailty is a "dynamic state affecting an individual who experiences losses in one or more domains of human functioning, which increases the risk of adverse outcomes".[1] Chronic disease may accelerate the development of frailty.
- Frailty is a predictor of pre- and early post-transplant outcomes in lung transplant patients, and may be driven by the presence of sarcopenia (loss of muscle mass) or dynapenia (decline in muscle contractile function). [2,3]

OBJECTIVES:

1. To examine the association between physical frailty (Fried Frailty Index) and measures of sarcopenia and dynapenia in lung transplant candidates.
2. To examine the relationships between sarcopenia and dynapenia.

METHODS:

Prospective, longitudinal, single-centre, cohort study of people waiting for lung transplant (n =49).

Fried Frailty Index - Shrinking, weakness, fatigue, slowness, low physical activity.

Sarcopenia Measures: ultrasound of quadriceps for cross-sectional area and echogenicity (muscle quality)

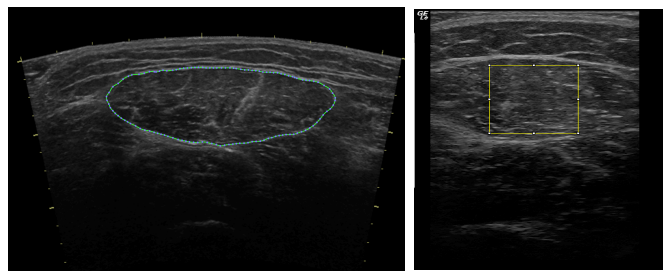

Figure 1 A. Cross-sectional area and **B.** Echogenicity of rectus femoris muscle. Freq = 10 MHz, Gain = 72, Depth = 6.0 cm.

Dynapenia Measures: Quadriceps **muscle strength** (isometric peak torque, Newton-metres), quadriceps **muscle power** from isotonic contractions (Watts)

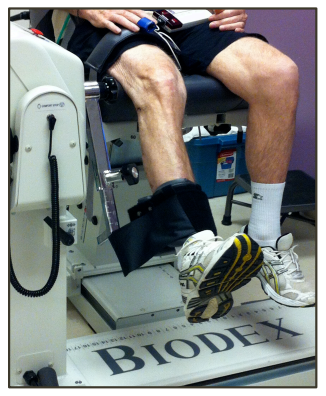
BIODEX

Figure 2. Biodex dynamometer was used to test quadriceps muscle strength (isometriv at 90deg knee flexion) and isotonic power at 20% of maximal load.

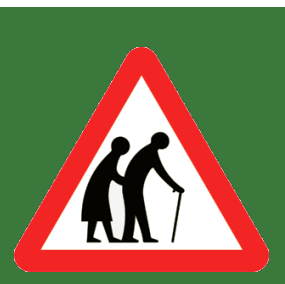

In frail people waiting for lung transplant, the main limitations are found in physical function like rising from a chair. Weak leg muscles are associated with reduced muscle size and quality.

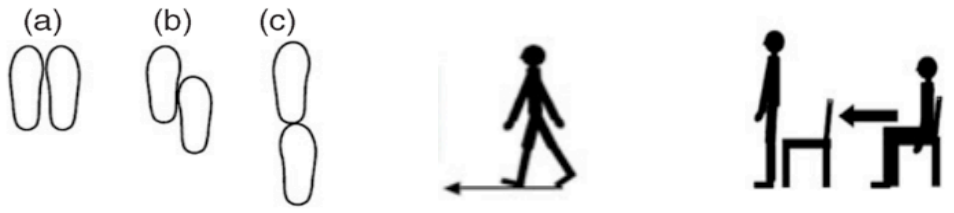
(a)
(b)
(c)
Balance – Tandem Stance
Walking Speed over 4 metres
5 Times Sit to Stand Test

Figure 3. Components of the Short Physical Performance Battery. Each is scored out of 4 points, for a maximum score of 12 points.

RESULTS

Table 1. Subject characteristics (n=49)

	Mean (Standard Deviation)
Age	59.6 (13) years
Sex	26 females, 22 males
FEV_1	1.14 (0.66) L
BMI	25.3 (4.0) kg/m^2
Fried Frailty Index (/5 points)	**n (%)**
i. Robust (0 points)	3 (3%)
ii. Pre-Frail (1 or 2 points)	33 (71%)
iii. Frail (3 or more points)	11 (22%)
Diagnosis	
i. Interstitial lung disease	25 (51%)
ii. Chronic obstructive pulmonary disease	17 (35%)
iii. Cystic fibrosis	3 (6%)
iv. Other*	4 (8%)

*Other diagnoses: pulmonary arterial hypertenison, graft vs host disease, bronchiectasis

Table 2. Spearman's correlations among frailty, sarcopenia and dynapenia variables

	SPPB Total	Chair Stands Test	Quadriceps strength	Quadriceps Power
Fried Frailty Index	-0.41*	0.30*	-0.28	-0.16
SPPB	--	--	0.05	0.10
Rectus Femoris CSA	0.04	--	0.44*	0.47*
Rectus Femoris echogenicity	-0.07	--	-0.38*	-0.58*

*significant *p*-value < 0.01

Table 3. Summary of frailty, sarcopenia and dynapenia measures (n=49).

	Median (interquartile range 25th -75th %tile)			
	Robust (n=3)	**Pre-frail (n=33)**	**Frail (n=11)**	**p-value**
SPPB Score (/12)	12 [12-12]	12 [11.5-12]	10 [10-12]	0.030*
i. Gait speed (m/sec)	1.1 [0.9-1.5]	1.0 [0.9-1.2]	0.9 [0.7-1.2]	0.130
ii. Chair stands time (sec)	7.4 [6.3-7.8]	8.9 [7.4-11.2]	10.1 [8.9-11.9	0.176
Quadriceps peak torque (Newton-metres)	84 [72-102]	111 [73-145]	75 [60-91]	0.014*
Quadriceps power (Watts)	171 [168-177]	230 [187-335]	195 [139-232]	0.030*
Rectus femoris cross-sectional area (cm^2)	6.2 [5.7-6.6]	6.8 [5.8-8.7]	6.1 [5.2-6.9]	0.085
Rectus femoris echogenicity (arbitrary units)	63 [59-65]	56 [49-66]	65 [46-67]	0.461

Mann-Whitney test used to compare pre-frail and frail groups. *significant *p*-value between Pre-frail and Frail subgroups

REFERENCES:

1. Gobbens RJ, et al. J Am Med Dir Assoc. 2010;11(5):338-43.
2. Cruz-Jentoft AJ et al. Age Ageing 2010;39:412-423.
3. Rozenberg D, et al. Transplantation. 2017; 101(9):2183-2191.

UHN Toronto General Toronto Western Princess Margaret Toronto Rehab Michener Institute COURAGE LIVES HERE
UNIVERSITY OF TORONTO
CIHR IRSC Canadian Institutes of Health Research Instituts de recherche en santé du Canada
The Toronto Lung Transplant Program