Temporal Trends in Primary Payer Status in Heart Transplantation in the United States

Matthew J. O’Connor, Kathryn Restaino, Danielle S. Burstein, Joseph W. Rossano, Christopher E. Mascio, Kimberly Y. Lin
Department of Pediatrics, Division of Pediatric Cardiology, Children’s Hospital of Philadelphia, Philadelphia, PA

BACKGROUND

- Heart failure and heart transplant impose a large burden on health care resources
- In general, pediatric patients have higher costs, LOS, and higher mortality compared to adults
- There has been a shift to public sources of healthcare reimbursement for heart failure and transplantation for adult patients (1)
- Outcomes are worse in adult patients funded through public insurance as well as patients that transition from private to public funding. (7)
- Data regarding financing for pediatric heart transplantation and if there have been changes in funding sources over time are scant.

METHODS

- The UNOS (United Network for Organ Sharing) database was queried for all patients < 18 at the time of heart transplant from 1994-2018
- Primary payer was identified at enrollment (TCR), at the time of transplant (TRR), and at follow up.
- Patients were classified as private, public, or ‘other’. Other included: self pay, donation, free care, or pending insurance status

Enrollment (TCR)
N= 12938 (124 missing information) = 12814 TCR patients

↓

Transplant (TRR)
N= 8385 (96 missing information) = 8289 TRR patients

↓ Death: 918 (11%)

1 year survival
N= 7467

→ 1 year follow up cohort
N=4290

Follow up documented between 12-14 months N = 6070
Insurance information missing: 1780

↓ All Death: 2516
Re-Transplant N=563 (median time to re-transplant 2723 days)
No follow up documented 1-5 years following transplant: 2132

Long term survival (1-5 years)
N = 5335 patients with 23,317 visits. 28% without insurance information
Median follow up time: 1826 days (0-0162)

RESULTS

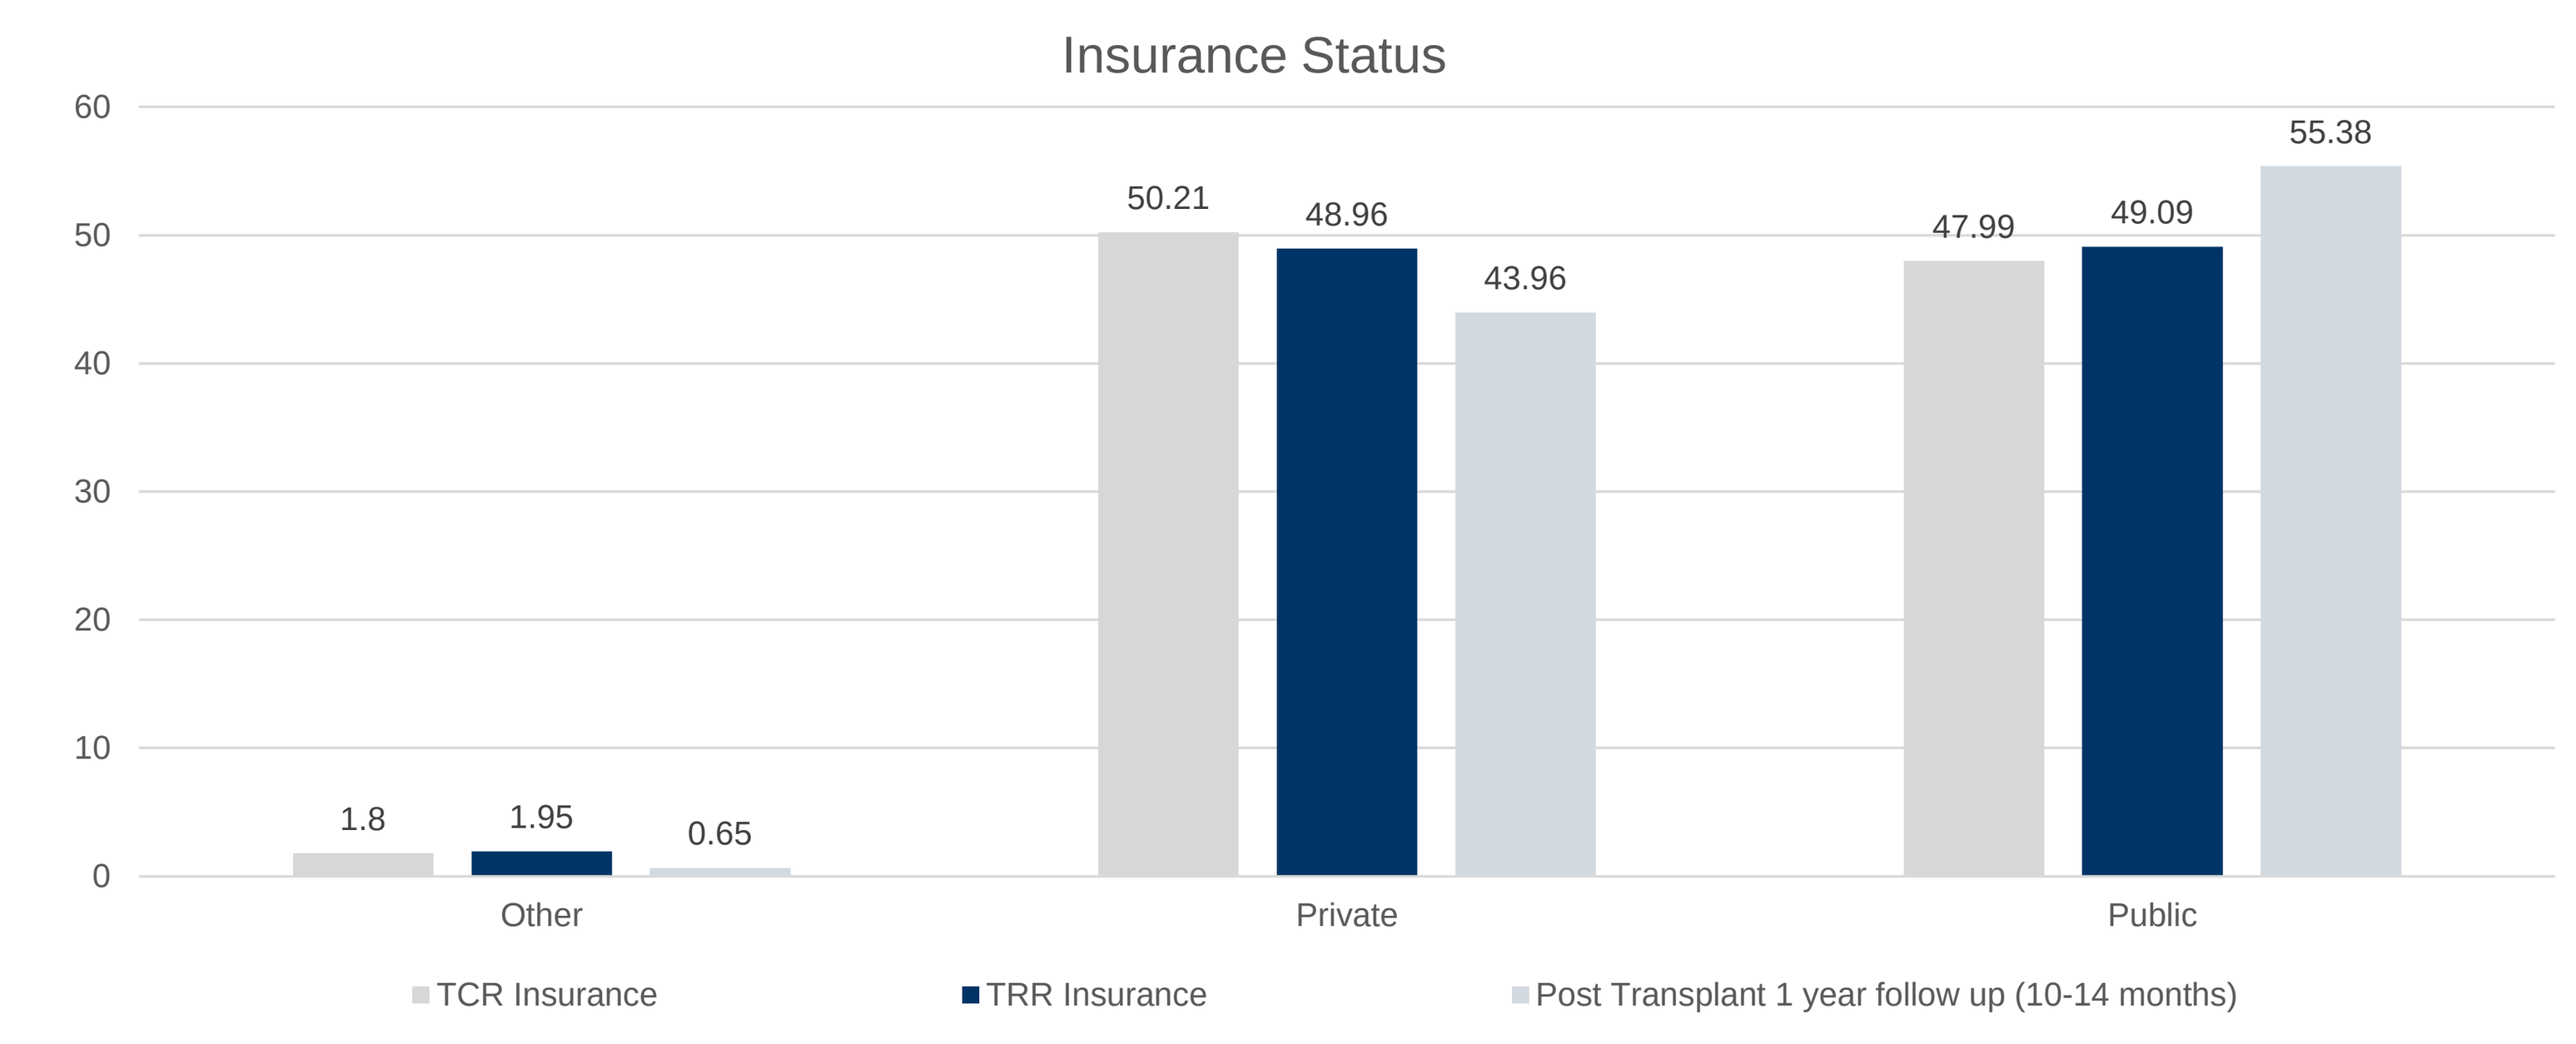
Insurance Status
60
50
40
30
20
10
0
1.8
1.95
0.65
Other
50.21
48.96
43.96
Private
47.99
49.09
55.38
Public
TCR Insurance
TRR Insurance
Post Transplant 1 year follow up (10-14 months)

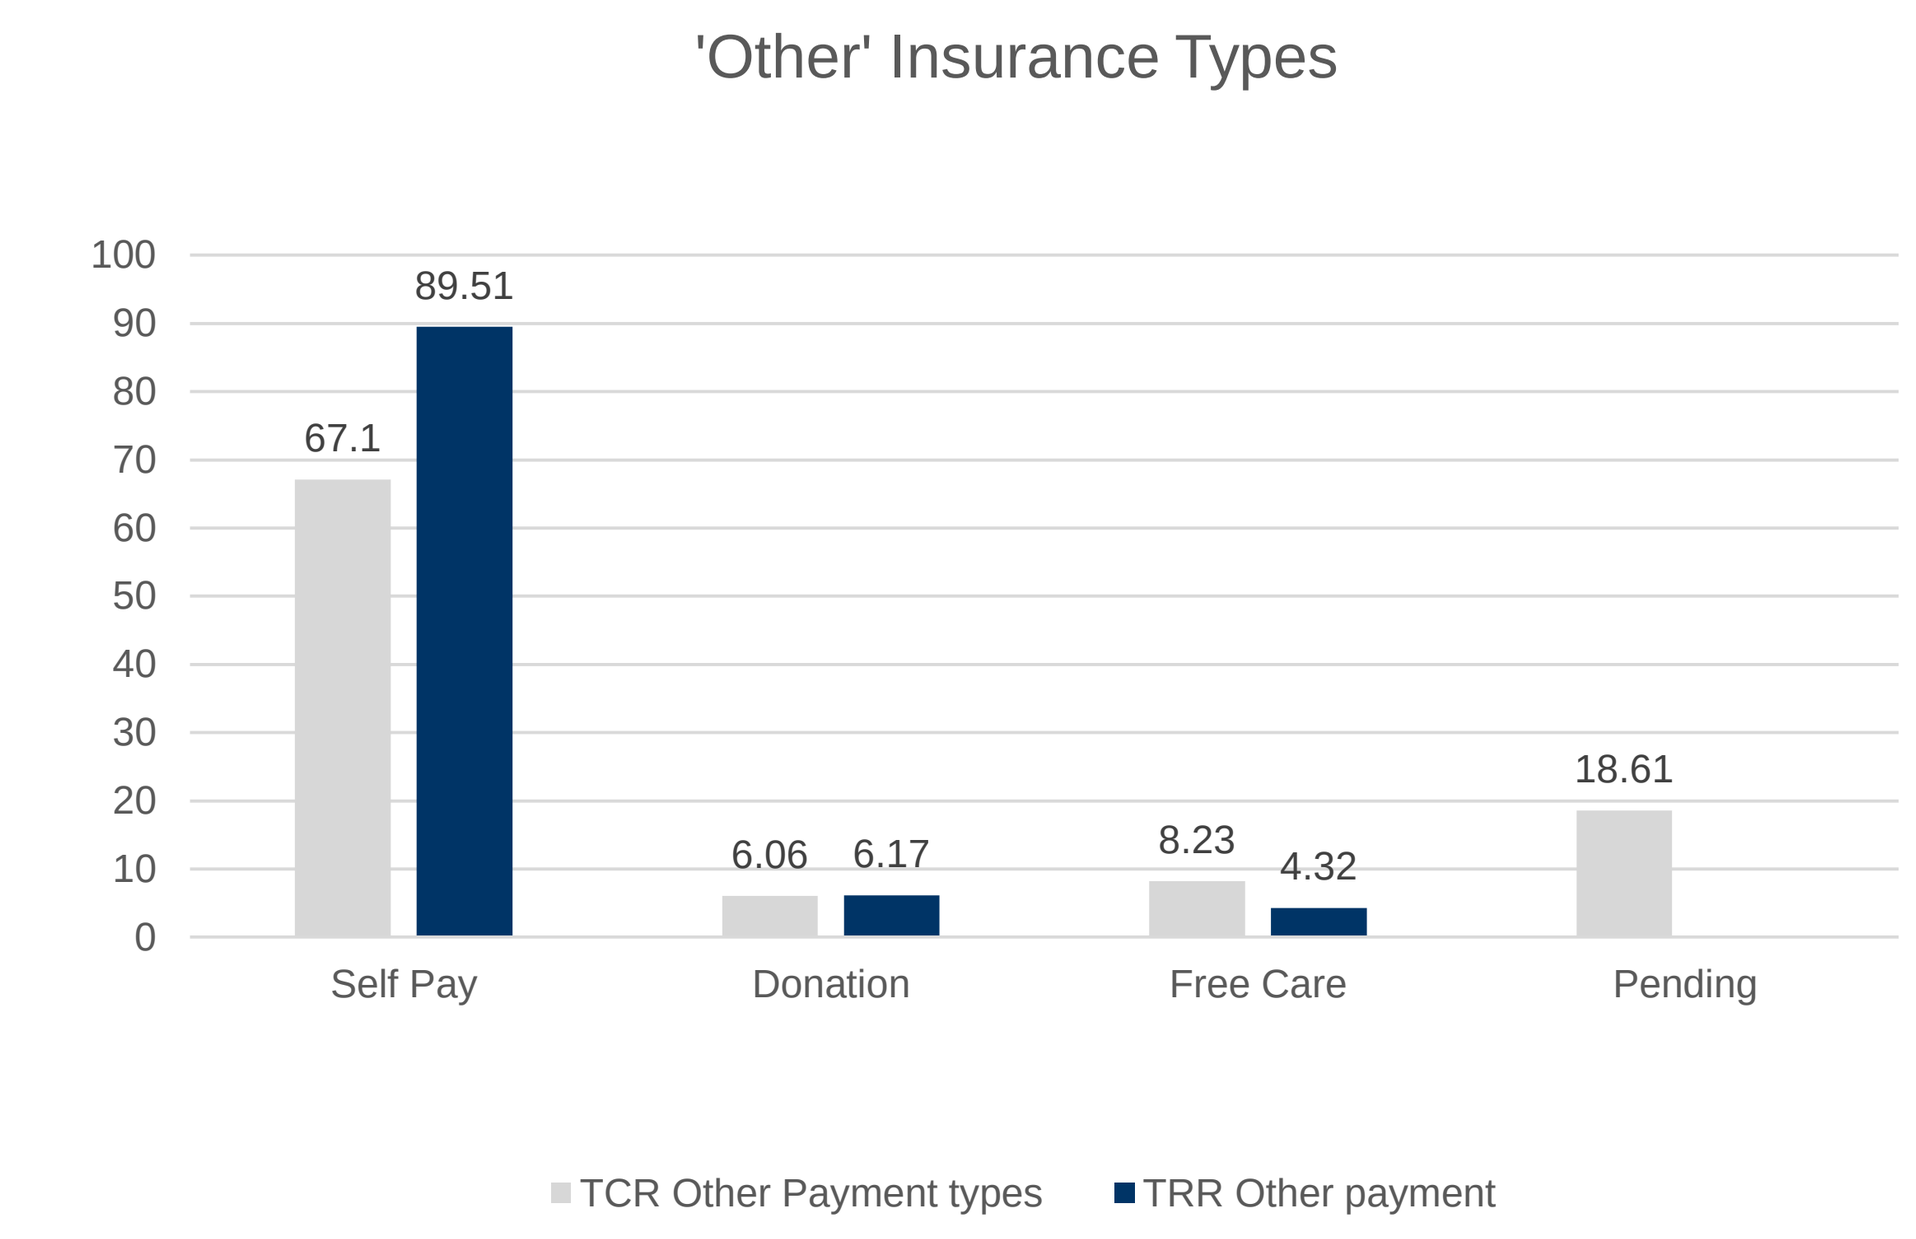
'Other' Insurance Types
67.1
89.51
Self Pay
6.06
6.17
Donation
8.23
4.32
Free Care
18.61
Pending
TCR Other Payment types
TRR Other payment

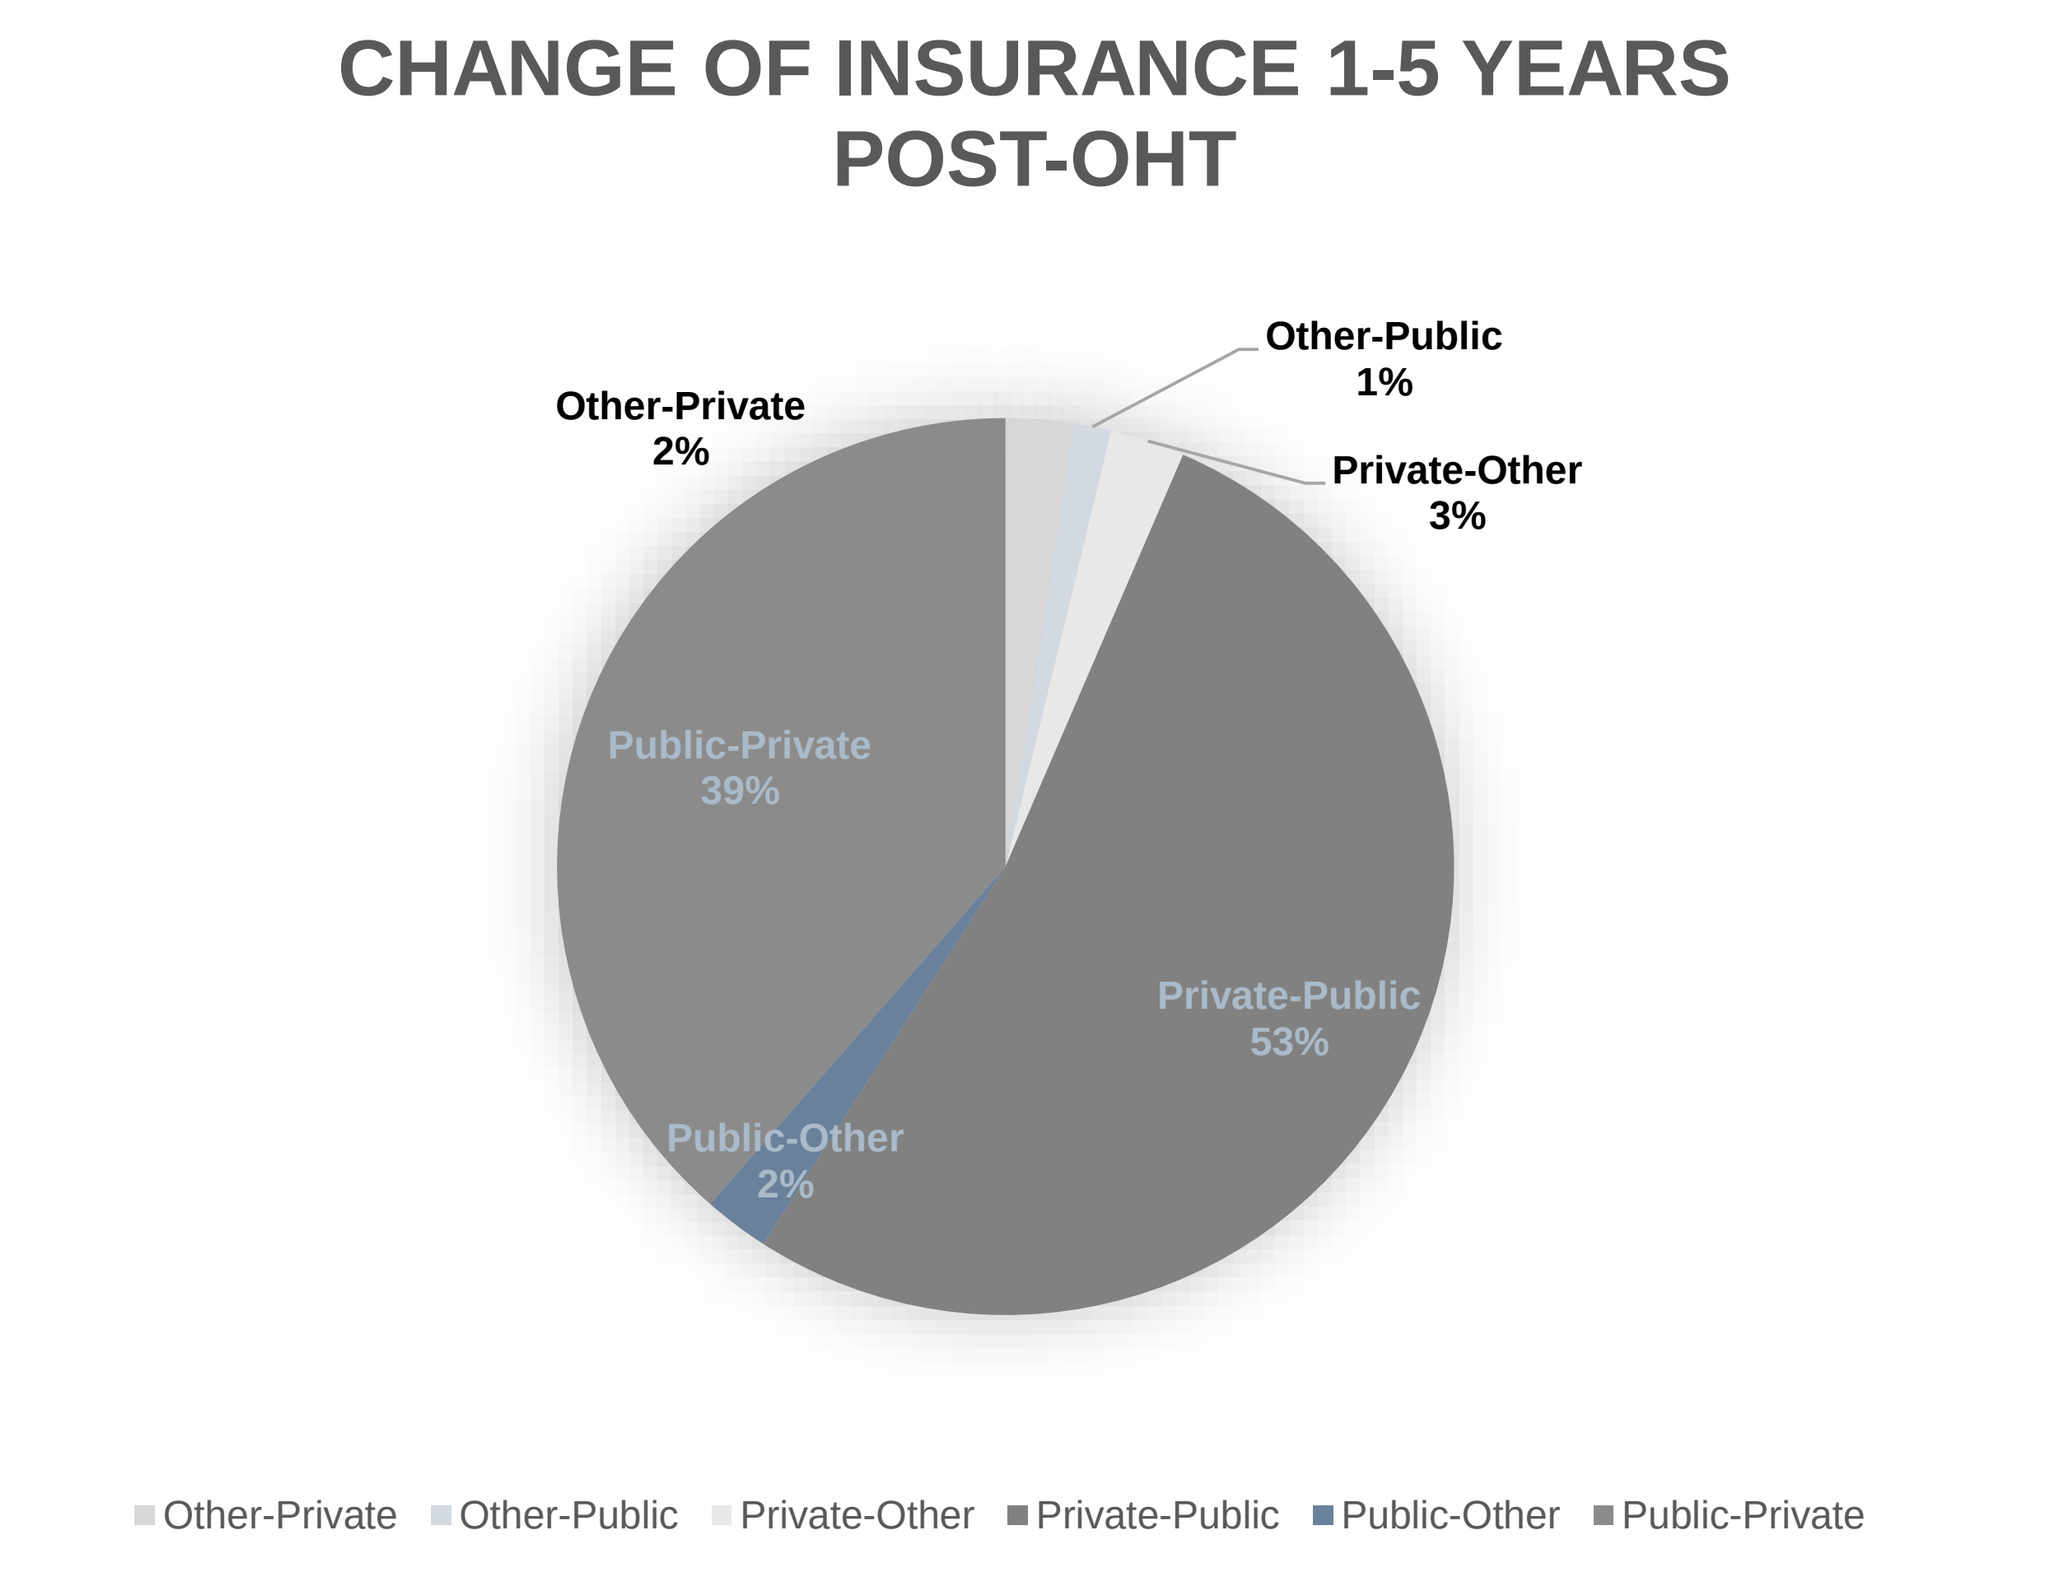
CHANGE OF INSURANCE 1-5 YEARS POST-OHT
Other-Private 2%
Other-Public 1%
Private-Other 3%
Public-Private 39%
Private-Public 53%
Public-Other 2%
Other-Private
Other-Public
Private-Other
Private-Public
Public-Other
Public-Private

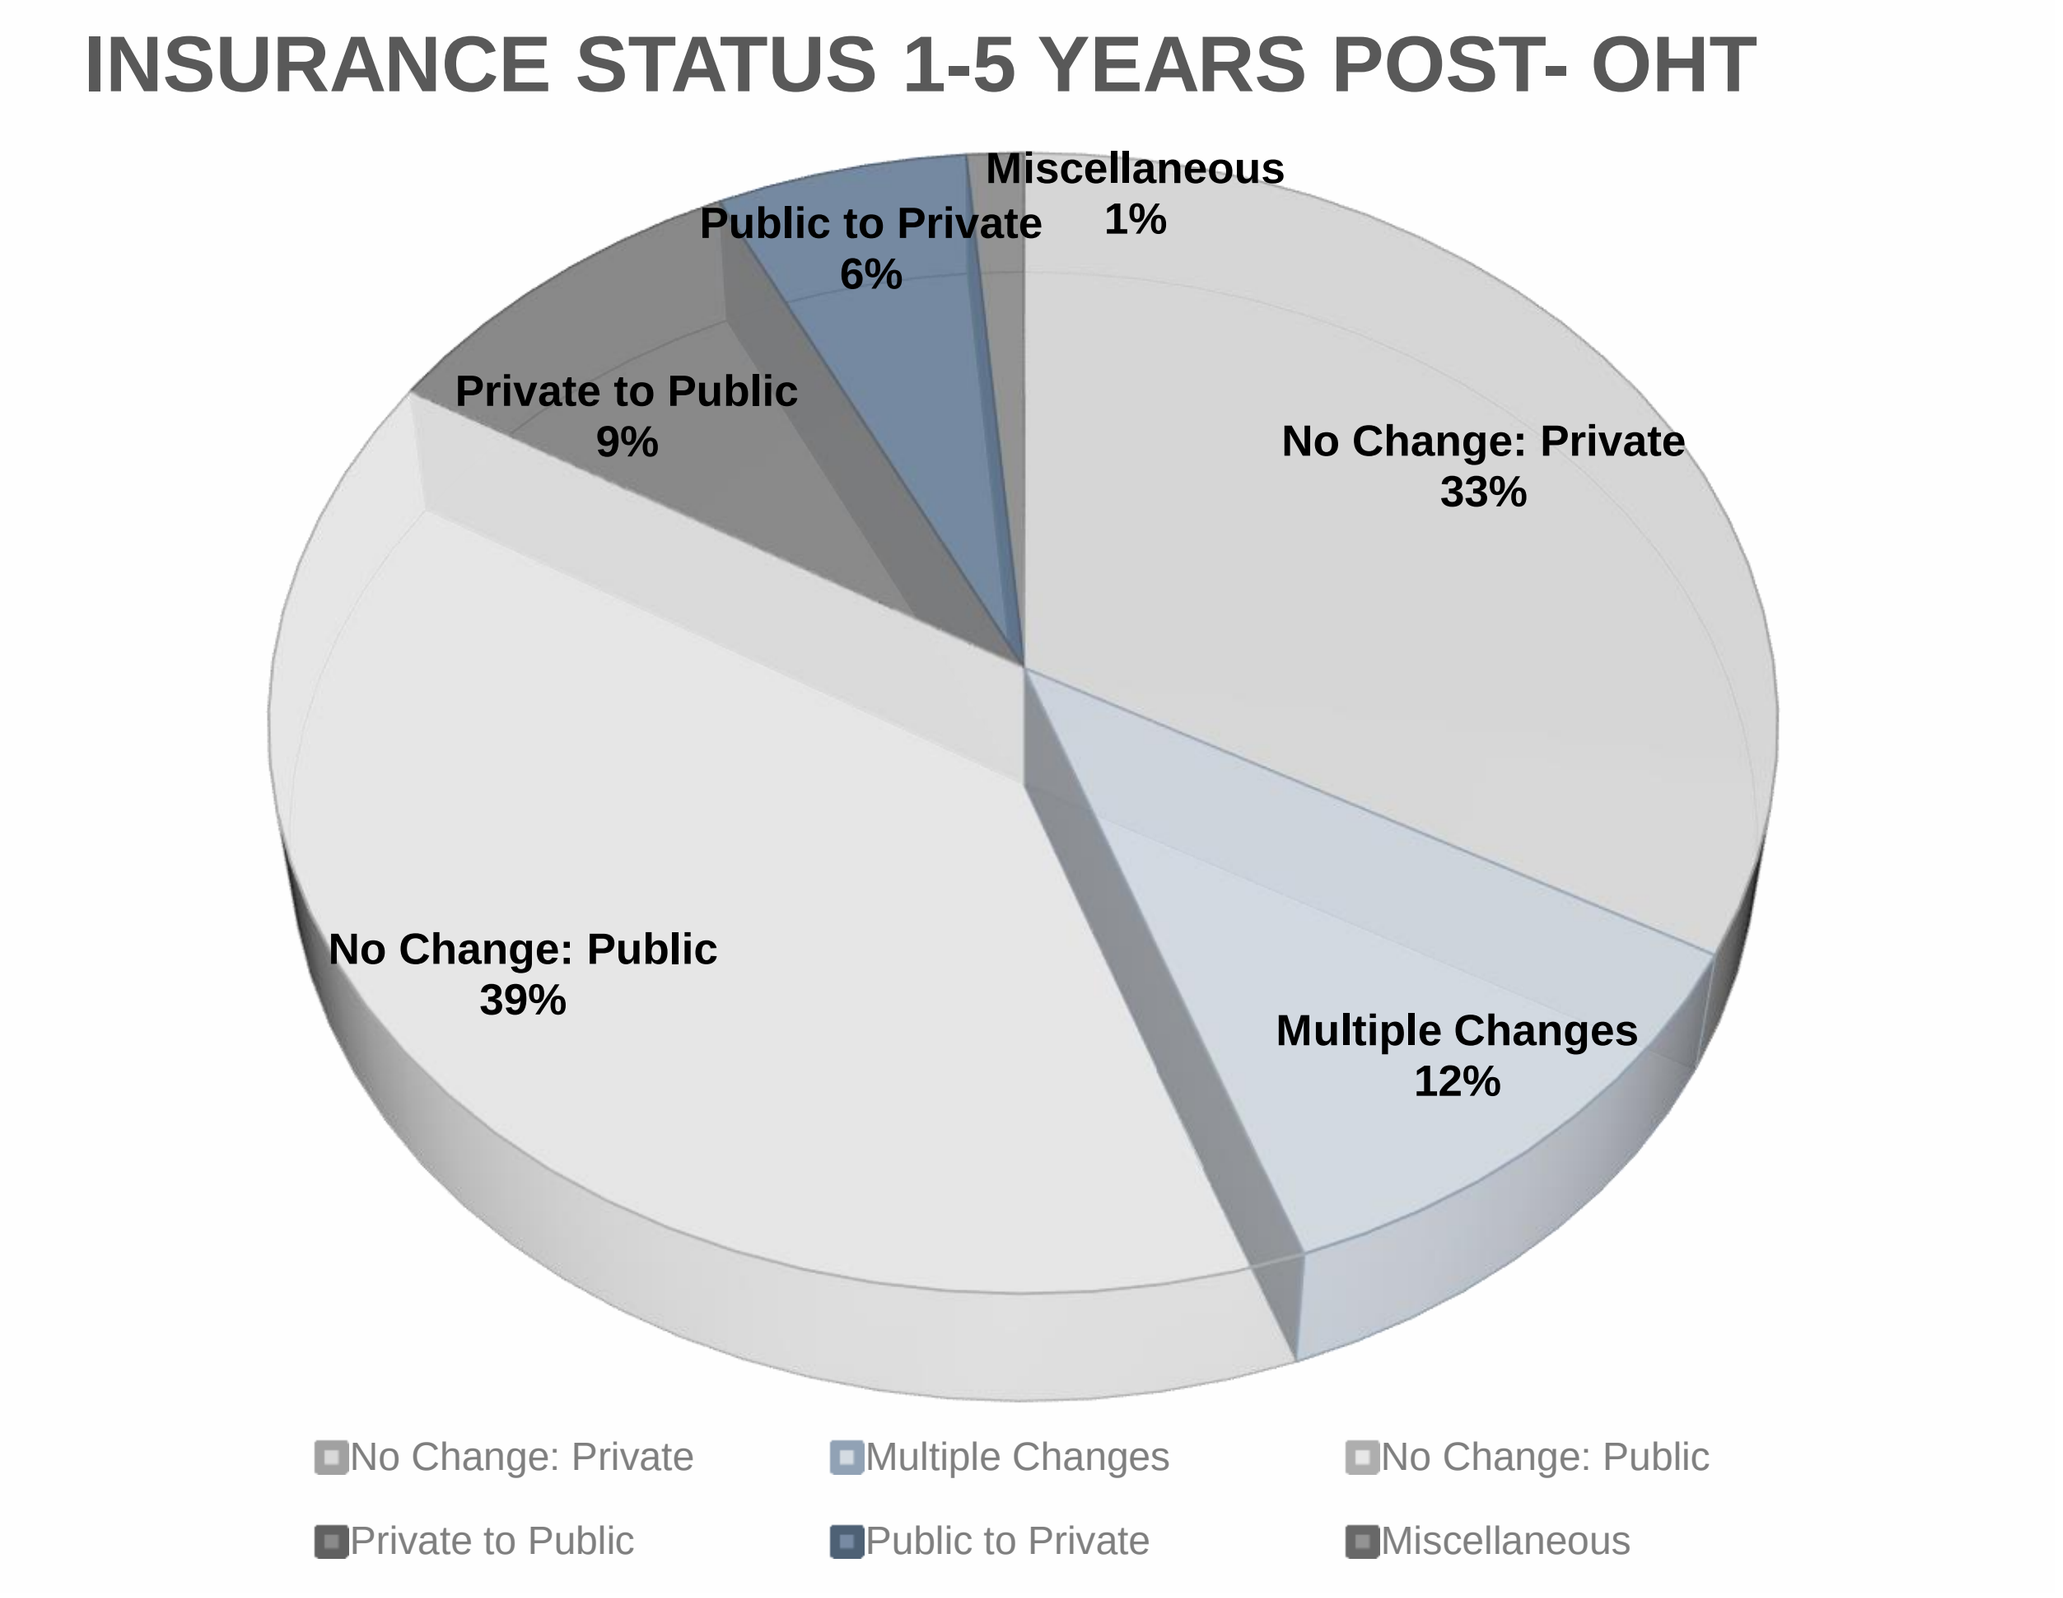
INSURANCE STATUS 1-5 YEARS POST- OHT
Miscellaneous 1%
Public to Private 6%
Private to Public 9%
No Change: Private 33%
No Change: Public 39%
Multiple Changes 12%
No Change: Private
Multiple Changes
No Change: Public
Private to Public
Public to Private
Miscellaneous

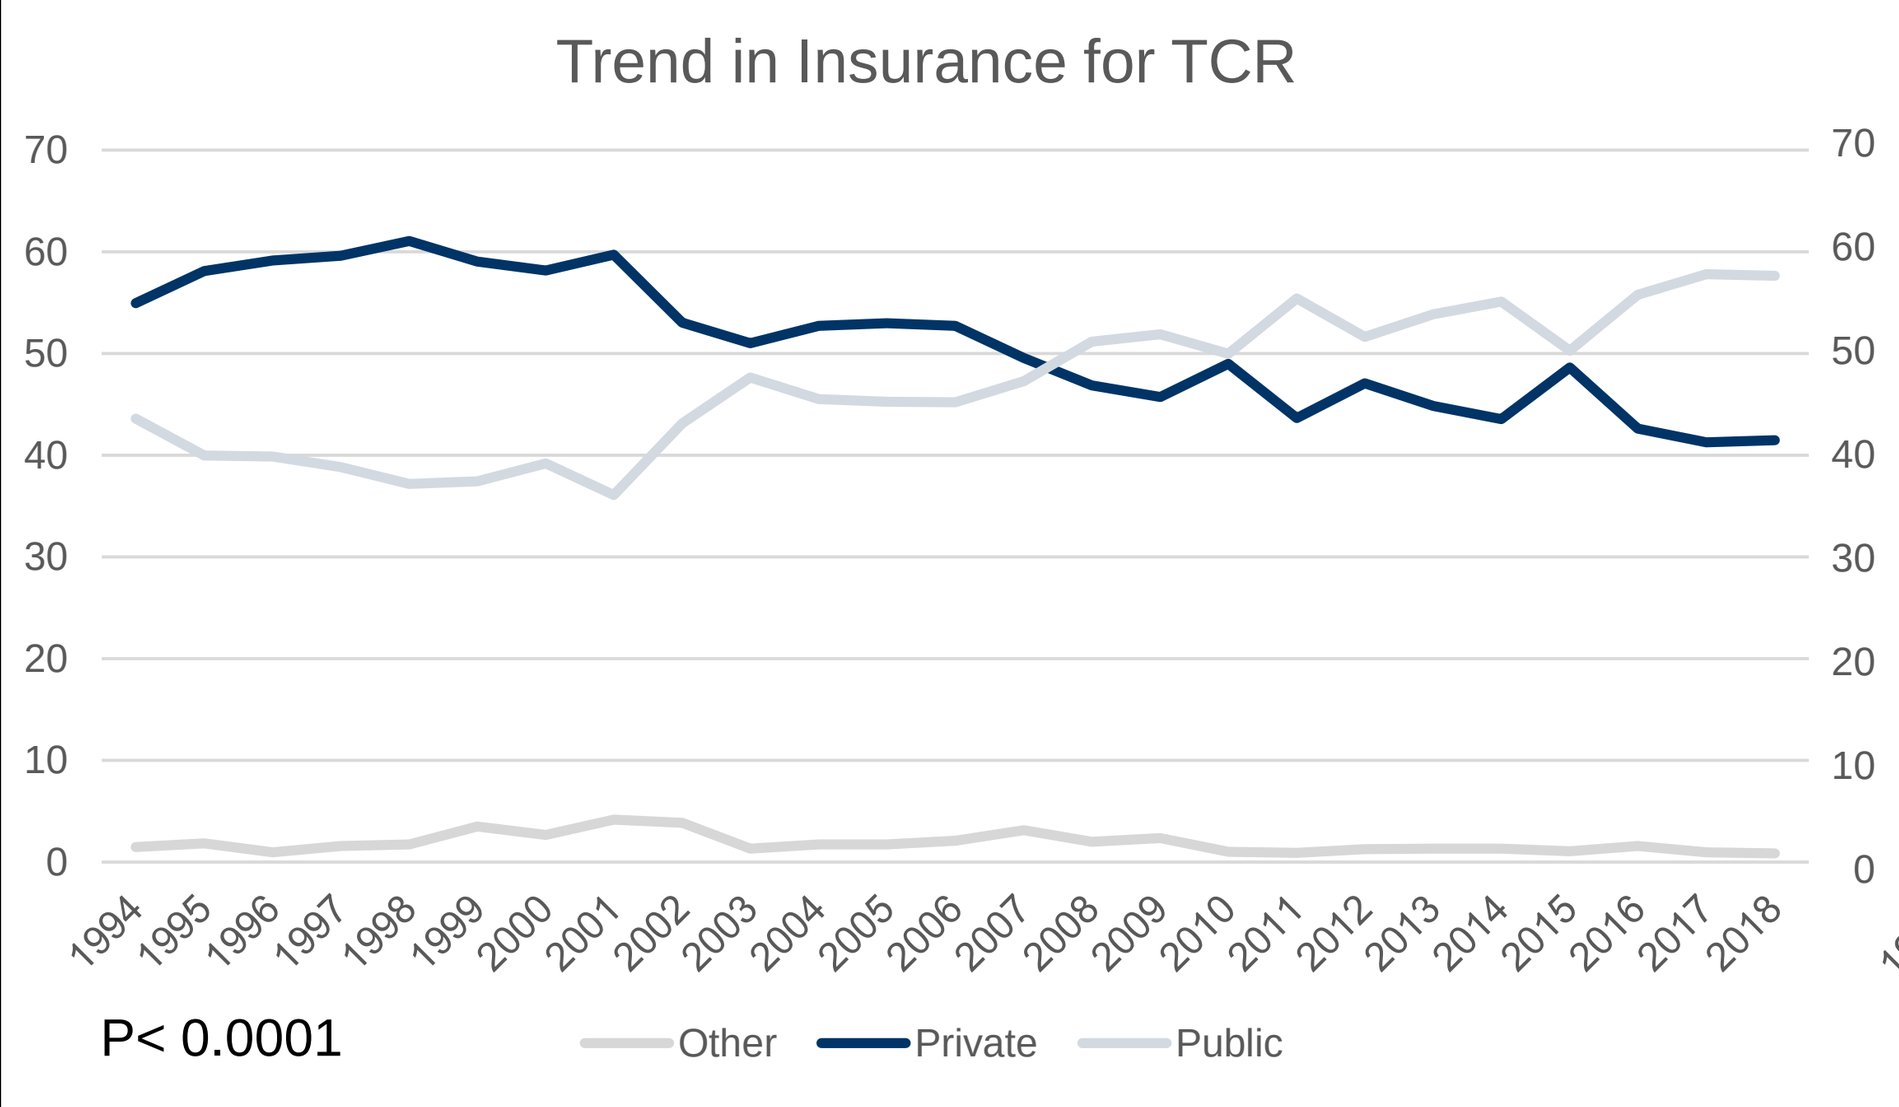
Trend in Insurance for TCR
P< 0.0001
Other
Private
Public

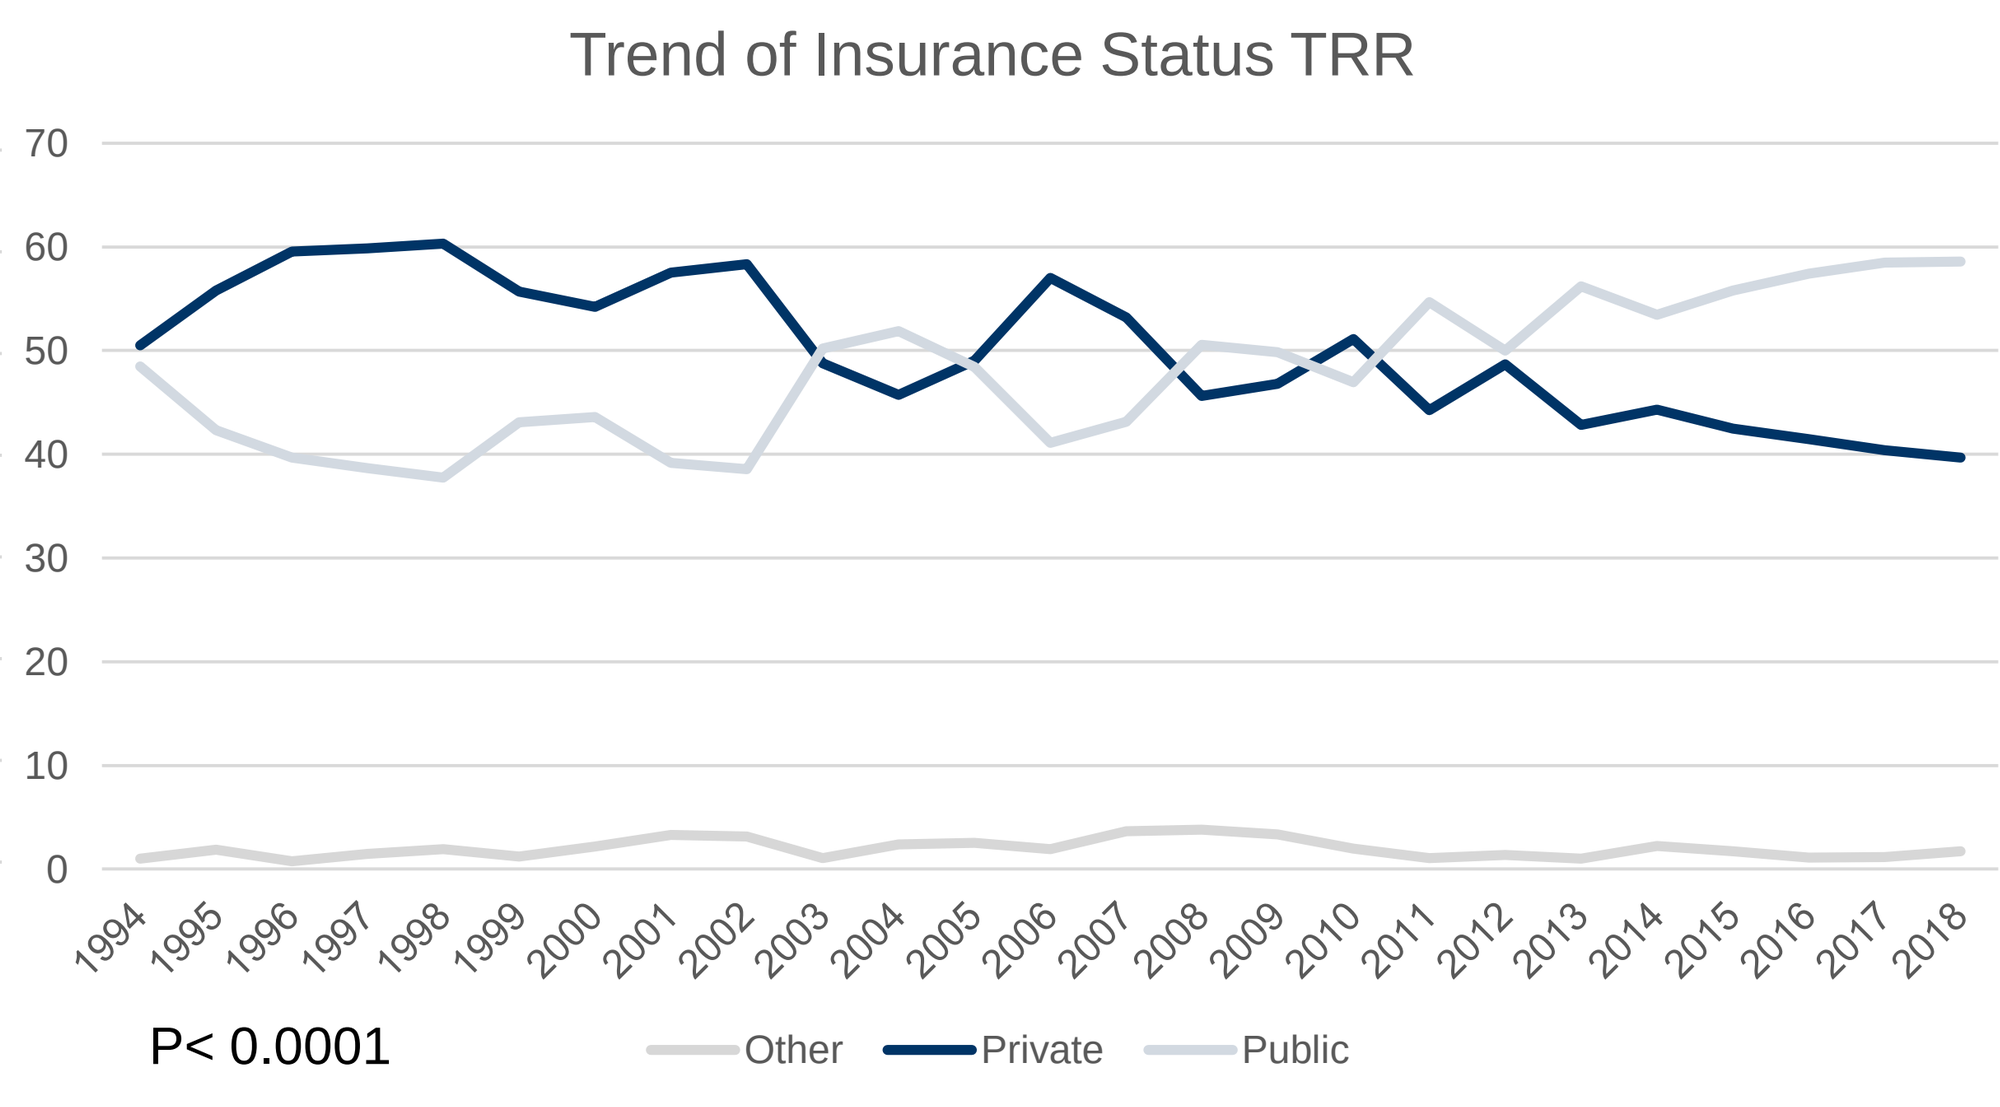
Trend of Insurance Status TRR
P< 0.0001
Other
Private
Public

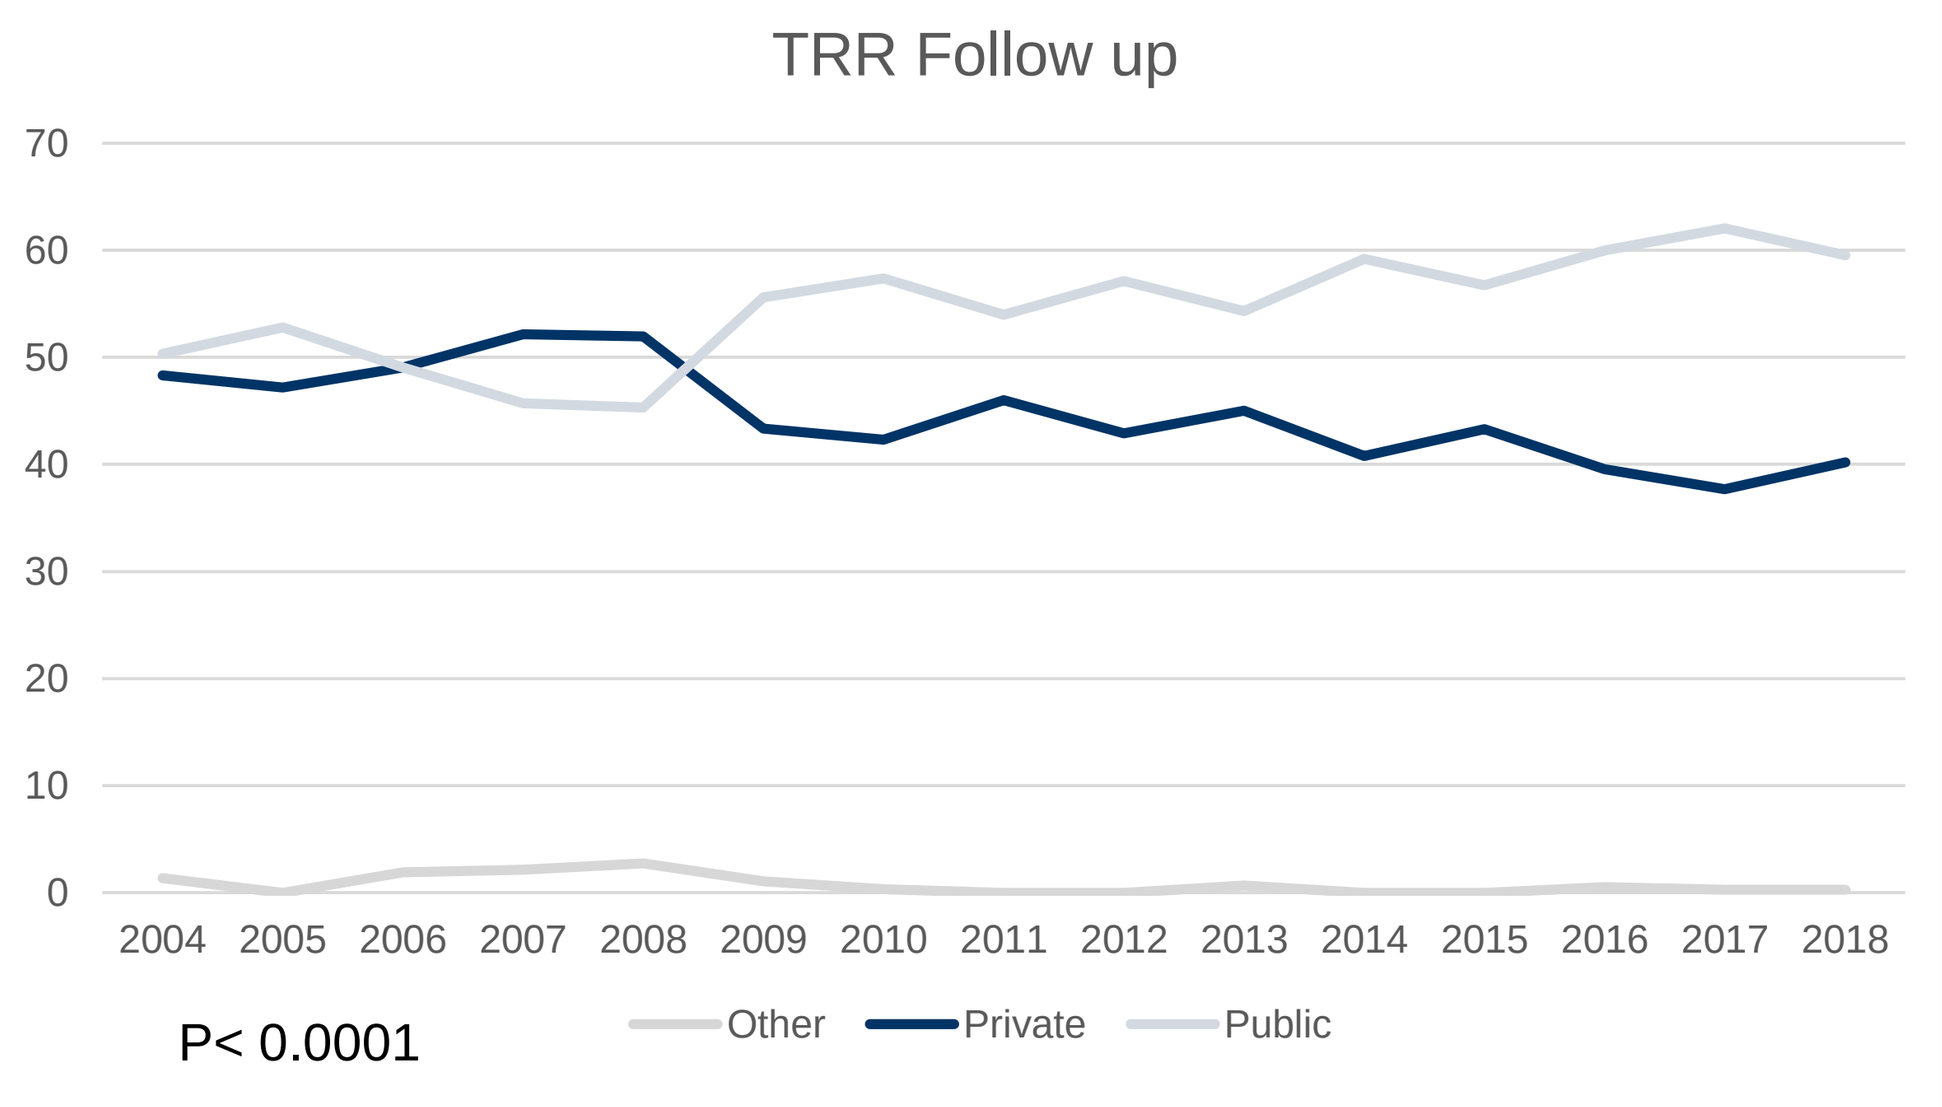
TRR Follow up
P< 0.0001
Other
Private
Public

Results

Table 1

TCR →TRR

	Other	Private	Public	Total
Other	128 76.65 79.01	15 8.98 0.37	24 14.37 0.59	167
Private	29 0.7 17.9	3794 91.36 93.66	330 7.95 8.12	4153
Public	5 0.13 3.09	242 6.12 5.97	3708 93.75 91.29	3955
Total	162	4051	4062	8275

P< 0.0001

Table 2

TRR → Follow Up

	Other	Private	Public	Total
Other	9 21.43 32.14	21 50 1.11	12 28.57 0.51	42
Private	12 0.6 42.86	1682 83.93 89.18	310 15.47 13.05	2004
Public	7 0.31 25	183 8.16 9.7	2054 91.53 86.45	2244
Total	28	1886	2376	4290

P< 0.0001

Rows: 1. Frequency, 2, Row % 2. Column %

Ex: 8% of patients with private insurance at the time of enrollment transitioned to public insurance by the time of transplant.

15% of patients with private insurance at transplant had transitioned to public funding by 1 year follow up.

Conclusions

- There is a statistically significant trend towards increasing prevalence of public insurance coverage in pediatric heart transplantation
- When patients change primary payer, they are more likely to change to public insurance
- Further study is needed to determine if these trends are associated with outcomes, particularly for those patients with public insurance coverage

REFERENCES

1. Defilippis, Ersilia M., et al. “Emerging Trends in Financing of Adult Heart Transplantation in the United States.” JACC: Heart Failure, vol. 7, no. 1, 2019, pp. 56–62., doi:10.1016/j.jchf.2018.10.001.
2. Axelrod, David A. “Economic and Financial Outcomes in Transplantation.” Current Opinion in Organ Transplantation., vol. 18, no. 2, 2013, pp. 222–228., doi:10.1097/MOT.0b013e32835f0757.
3. Nandi D, Lin KY, O’Connor MJ, et al. Hospital charges for pediatric heart failure-related hospitalizations from 2000 to 2009. Pediatr Cardiol. 2016;37(3):512–518.
4. Boucek, Dana M., et al. “Resource Utilization for Initial Hospitalization in Pediatric Heart Transplantation in the United States.” The American Journal of Cardiology, vol. 121, no. 8, 2018, pp. 981–985., doi:10.1016/j.amjcard.2018.01.007.
5. Godown, Justin, et al. “Changes in Pediatric Heart Transplant Hospitalization Costs Over Time.” Transplantation, vol. 102, no. 10, 2018, pp. 1762–1767., doi:10.1097/tp.0000000000002202.
6. Wittlieb-Weber, Carol A., et al. “Pediatric Versus Adult Cardiomyopathy and Heart Failure–Related Hospitalizations: A Value-Based Analysis.” Journal of Cardiac Failure, vol. 21, no. 1, 2015, pp. 76–82., doi:10.1016/j.cardfail.2014.10.011.
7. Tumin, Dmitry, et al. “Health Insurance Trajectories and Long-Term Survival After Heart Transplantation.” Circulation: Cardiovascular Quality and Outcomes, vol. 9, no. 5, 2016, pp. 576–584., doi:10.1161/circoutcomes.116.003067.
8. Dipchand, Anne I. “Current State of Pediatric Cardiac Transplantation.” Annals of Cardiothoracic Surgery, vol. 7, no. 1, 2018, pp. 31–55., doi:10.21037/acs.2018.01.07.
9. Trivedi, J., Ising, M., Fox, M., Cannon, R., van Berkel, V., & Slaughter, M. (2018). Solid-Organ Transplantation and the Affordable Care Act: Accessibility and Outcomes. The American Surgeon., 84(12), 1894–1899.
10. Breathett, K., Allen, L. A., Helmkamp, L., Colborn, K., Daugherty, S. L., Khazanie, P., … Peterson, P. N. (2017). The Affordable Care Act Medicaid Expansion Correlated With Increased Heart Transplant Listings in African-Americans But Not Hispanics or Caucasians. JACC: Heart Failure, 5(2), 136–147. doi: 10.1016/j.jchf.2016.10.013
11. Gates, Jason, et al. “Uninsurance among Children, 1997–2015 : Long-Term Trends and Recent Patterns.” Urban Institute, 4 May 2020,
12. Wayda, Brian, et al. “Socioeconomic Disparities in Adherence and Outcomes After Heart Transplant.” Circulation: Heart Failure, vol. 11, no. 3, 2018, doi:10.1161/circheartfailure.117.004173.