LEFT ATRIAL VOLUME AFTER HEART TRANSPLANTATION – A PREDICTOR OF OUTCOMES?

Yael Peled, Jacob Lavee, Eilon Ram, Yigal Kassif, Leonid Sternik, Ehud Schwammenthal, Robert Klempfner and Boaz Tzur

Leviev Cardiothoracic and Vascular Center, Sheba Medical Center

OBJECTIVE

The left atrium (LA) in heart transplant (HT) recipients is a surgically created chamber from variable portions of donor and recipient. In the non-HT patients, there is strong evidence that left atrial volume (LAV) is a robust predictor of cardiovascular outcomes such as atrial fibrillation, stroke, heart failure and cardiovascular death. Thus, we thought to 1) characterize LA size in HT recipients and 2) investigate the implications of “new” LA volume on HT outcomes.

METHODS

First echocardiograms of 248 HT recipients between 2000-2018 were analyzed for LA volume index (LAVi). LAV was assessed employing the biplane Simpson method in the apical 4- and 2-chamber view and indexed to body surface area (LAVi). Patients were categorized by quartiles. The primary end points were: stroke, cardiovascular and all-cause mortality, cardiac allograft vasculopathy and heart failure.

RESULTS

Mean and median LAV were 69.3 mL (SD, 29.6) and 65.3 mL [IQR 48.2, 84.3], respectively, and, when indexed (LAVi), 37.2 mL/m2 (SD, 15.7) and 34.6 mL/m2 [IQR 25.9, 45.6], respectively. Baseline patient and donor clinical, demographic and hemodynamic parameters were similar. Outcomes, assessed by each LAVi quartile were shown to have similar cumulative probabilities. Kaplan-Meier analyses for each outcome, were similar too (Figure). Notably, with an emphasis on the enlarged LA volume of this cohort, the hazard of stroke, was low. Consistent with these findings, atrial fibrillation episodes were very rare (excluding perioperative/rejection) with a total of 7 patients experiencing episodes of paroxysmal atrial fibrillation.

Kaplan-Meier Outcome-free Survival Curves Stratified by LAVi Quartiles

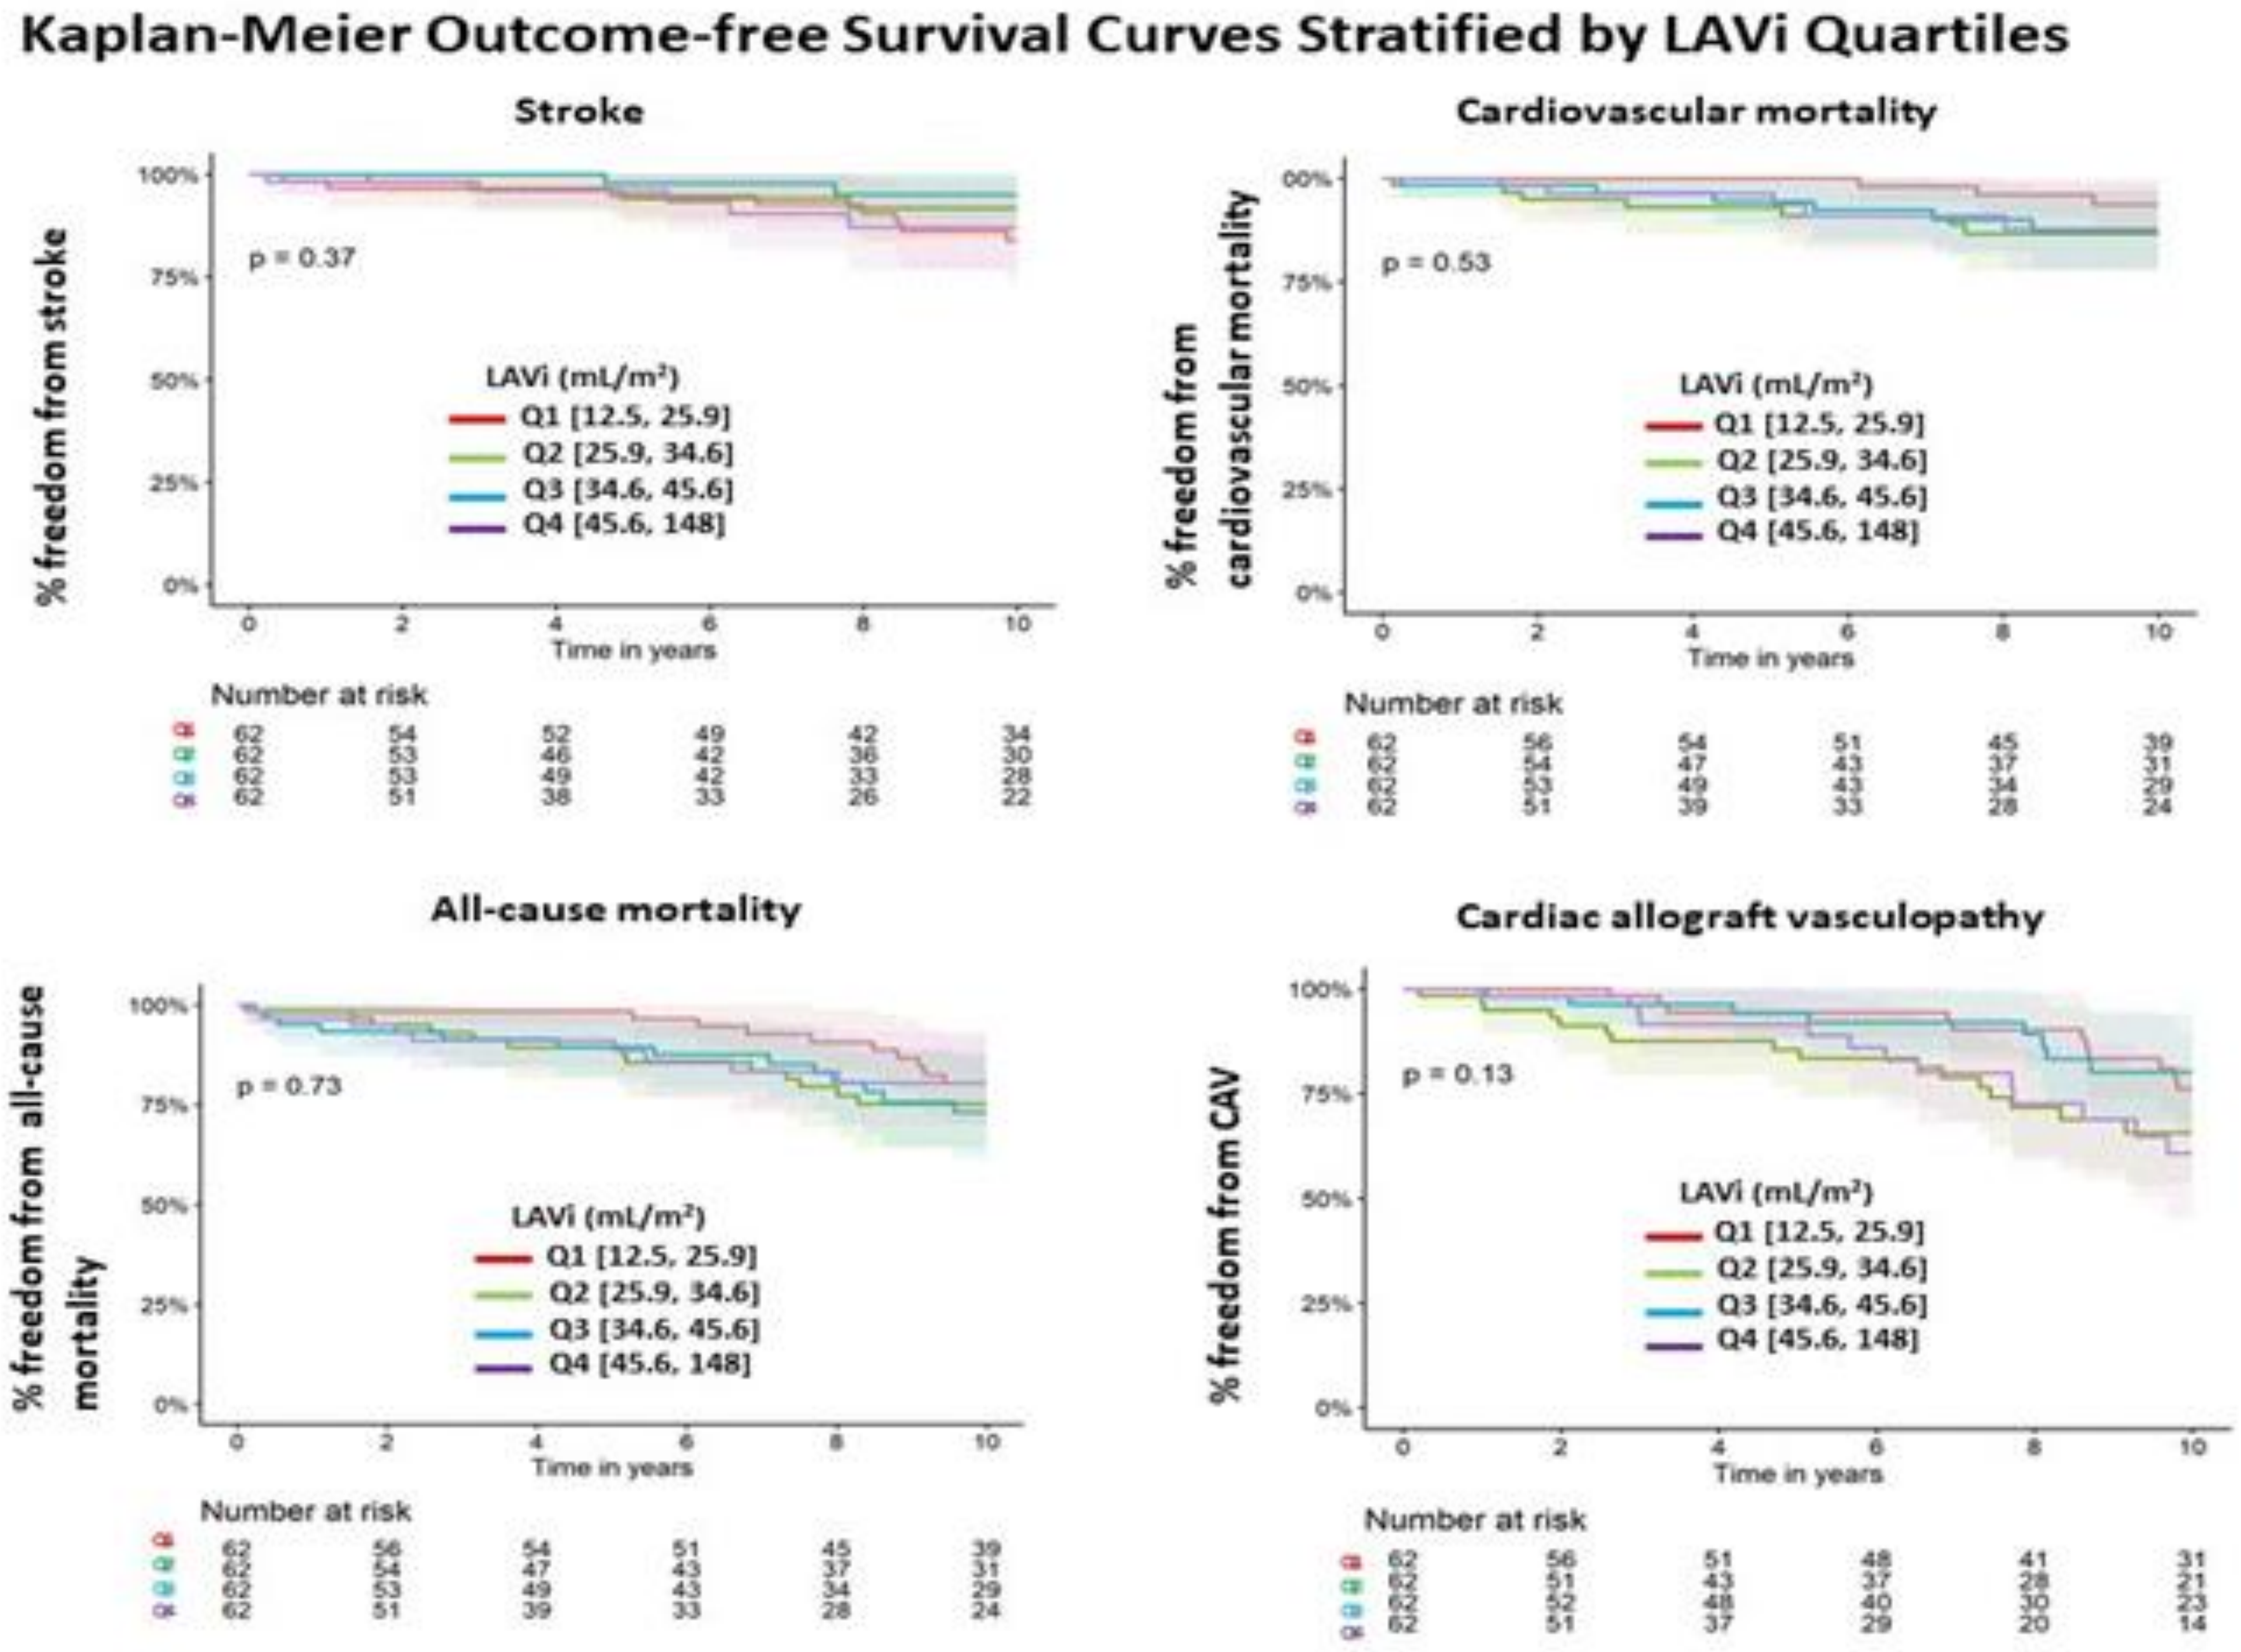
Stroke
% freedom from stroke
p = 0.37
LAVi (mL/m²)
Q1 [12.5, 25.9]
Q2 [25.9, 34.6]
Q3 [34.6, 45.6]
Q4 [45.6, 148]
Time in years
Number at risk
Cardiovascular mortality
% freedom from cardiovascular mortality
p = 0.53
All-cause mortality
% freedom from all-cause mortality
p = 0.73
Cardiac allograft vasculopathy
% freedom from CAV
p = 0.13

CONCLUSIONS

In contrast to what is known in non-HT patients, no association was demonstrated for LAV and cardiovascular adverse events. As all patients undergo a “MAZE procedure” during HT, our findings suggest that pulmonary vein isolation and cardiac denervation provide significant prophylaxis against cardiovascular events in HT patients, neutralizing the risks that are typically associated with significant enlargement of LA volumes.

No Disclosures