Measuring EBV in Lung Transplant Patients - Lessons of 16 Years of Follow Up

E. J. Gore[1], W. van der Bij[2], C. C. Leer[1], M. E. Erasmus[3], C. Gan[4], E. A. Verschuuren[5].

[1]Medical Microbiology and Virology, University Medical Centre Groningen, Groningen, Netherlands, [2]Pulmonary Diseases and Tuberculosis, University Medical Centre Groningen, Groningen, Netherlands, [3]Cardiothoracic Surgery, University Medical Centre Groningen, Groningen, Netherlands, [4]Pulmonary Diseases and Tuberculosis, University Medical Centre Groningen, Groningen, Netherlands, [5]University Medical Centre Groningen, Groningen, Netherlands,

Introduction

Currently there is debate regarding the use and usefulness of EBV viral loads in lung transplant patients. We routinely measured EBV DNA load from 2001 and studied the relation between EBV DNA load, chronic lung allograft dysfunction (CLAD), survival and post-transplant lymphoproliferative disease (PTLD). It has been suggested EBV can be used as a surrogate marker of immunocompetence (1) however this is not universally accepted (find ref). Other groups have previously described an increased risk of developing CLAD with exposure to EBV (2), we explored this theory with our cohort of 352 patients over 16 years.

PTLD is known to be associated with EBV (3) , Rituximab (RTx) is used to treat EBV derived CD20 positive tumours (3) as part of multi-drug chemotherapy, we report on the use of RTx for prophylactic treatment of elevated EBV viral loads before the development of PTLD.

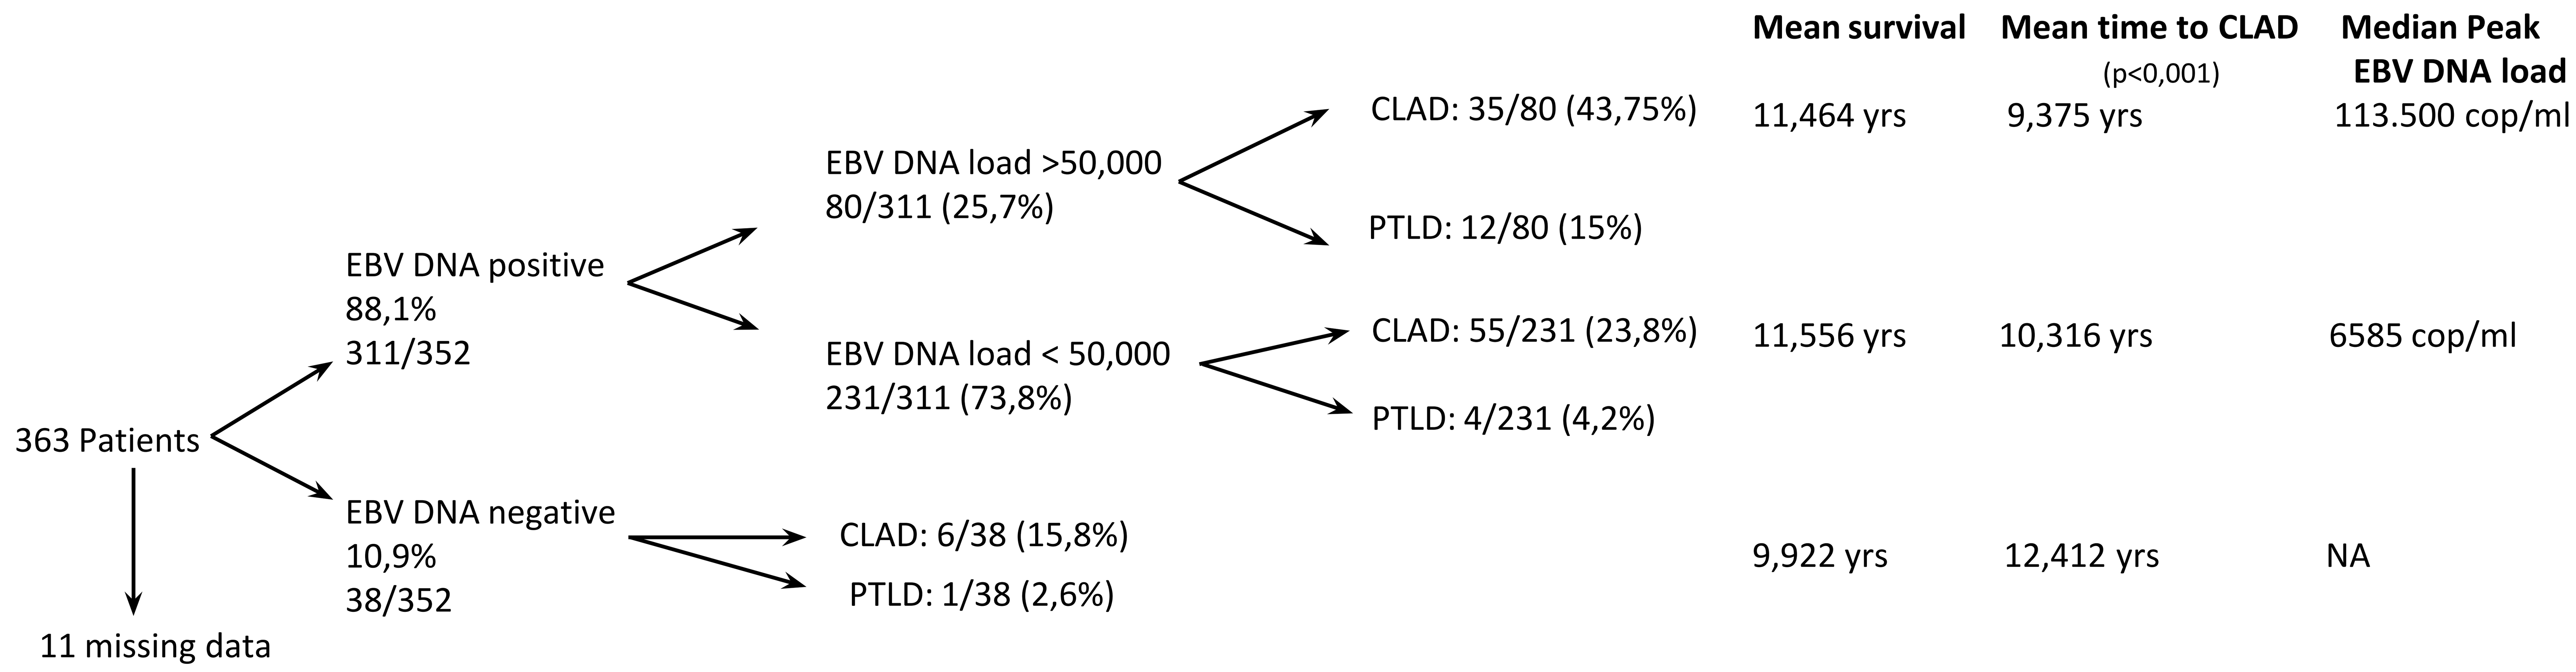
Mean survival
Mean time to CLAD
(p<0,001)
Median Peak
EBV DNA load
363 Patients
11 missing data
EBV DNA positive
88,1%
311/352
EBV DNA negative
10,9%
38/352
EBV DNA load >50,000
80/311 (25,7%)
EBV DNA load < 50,000
231/311 (73,8%)
CLAD: 35/80 (43,75%)
PTLD: 12/80 (15%)
11,464 yrs
9,375 yrs
113.500 cop/ml
CLAD: 55/231 (23,8%)
PTLD: 4/231 (4,2%)
11,556 yrs
10,316 yrs
6585 cop/ml
CLAD: 6/38 (15,8%)
PTLD: 1/38 (2,6%)
9,922 yrs
12,412 yrs
NA

Figure 1: Scheme showing study format and an overview of results.

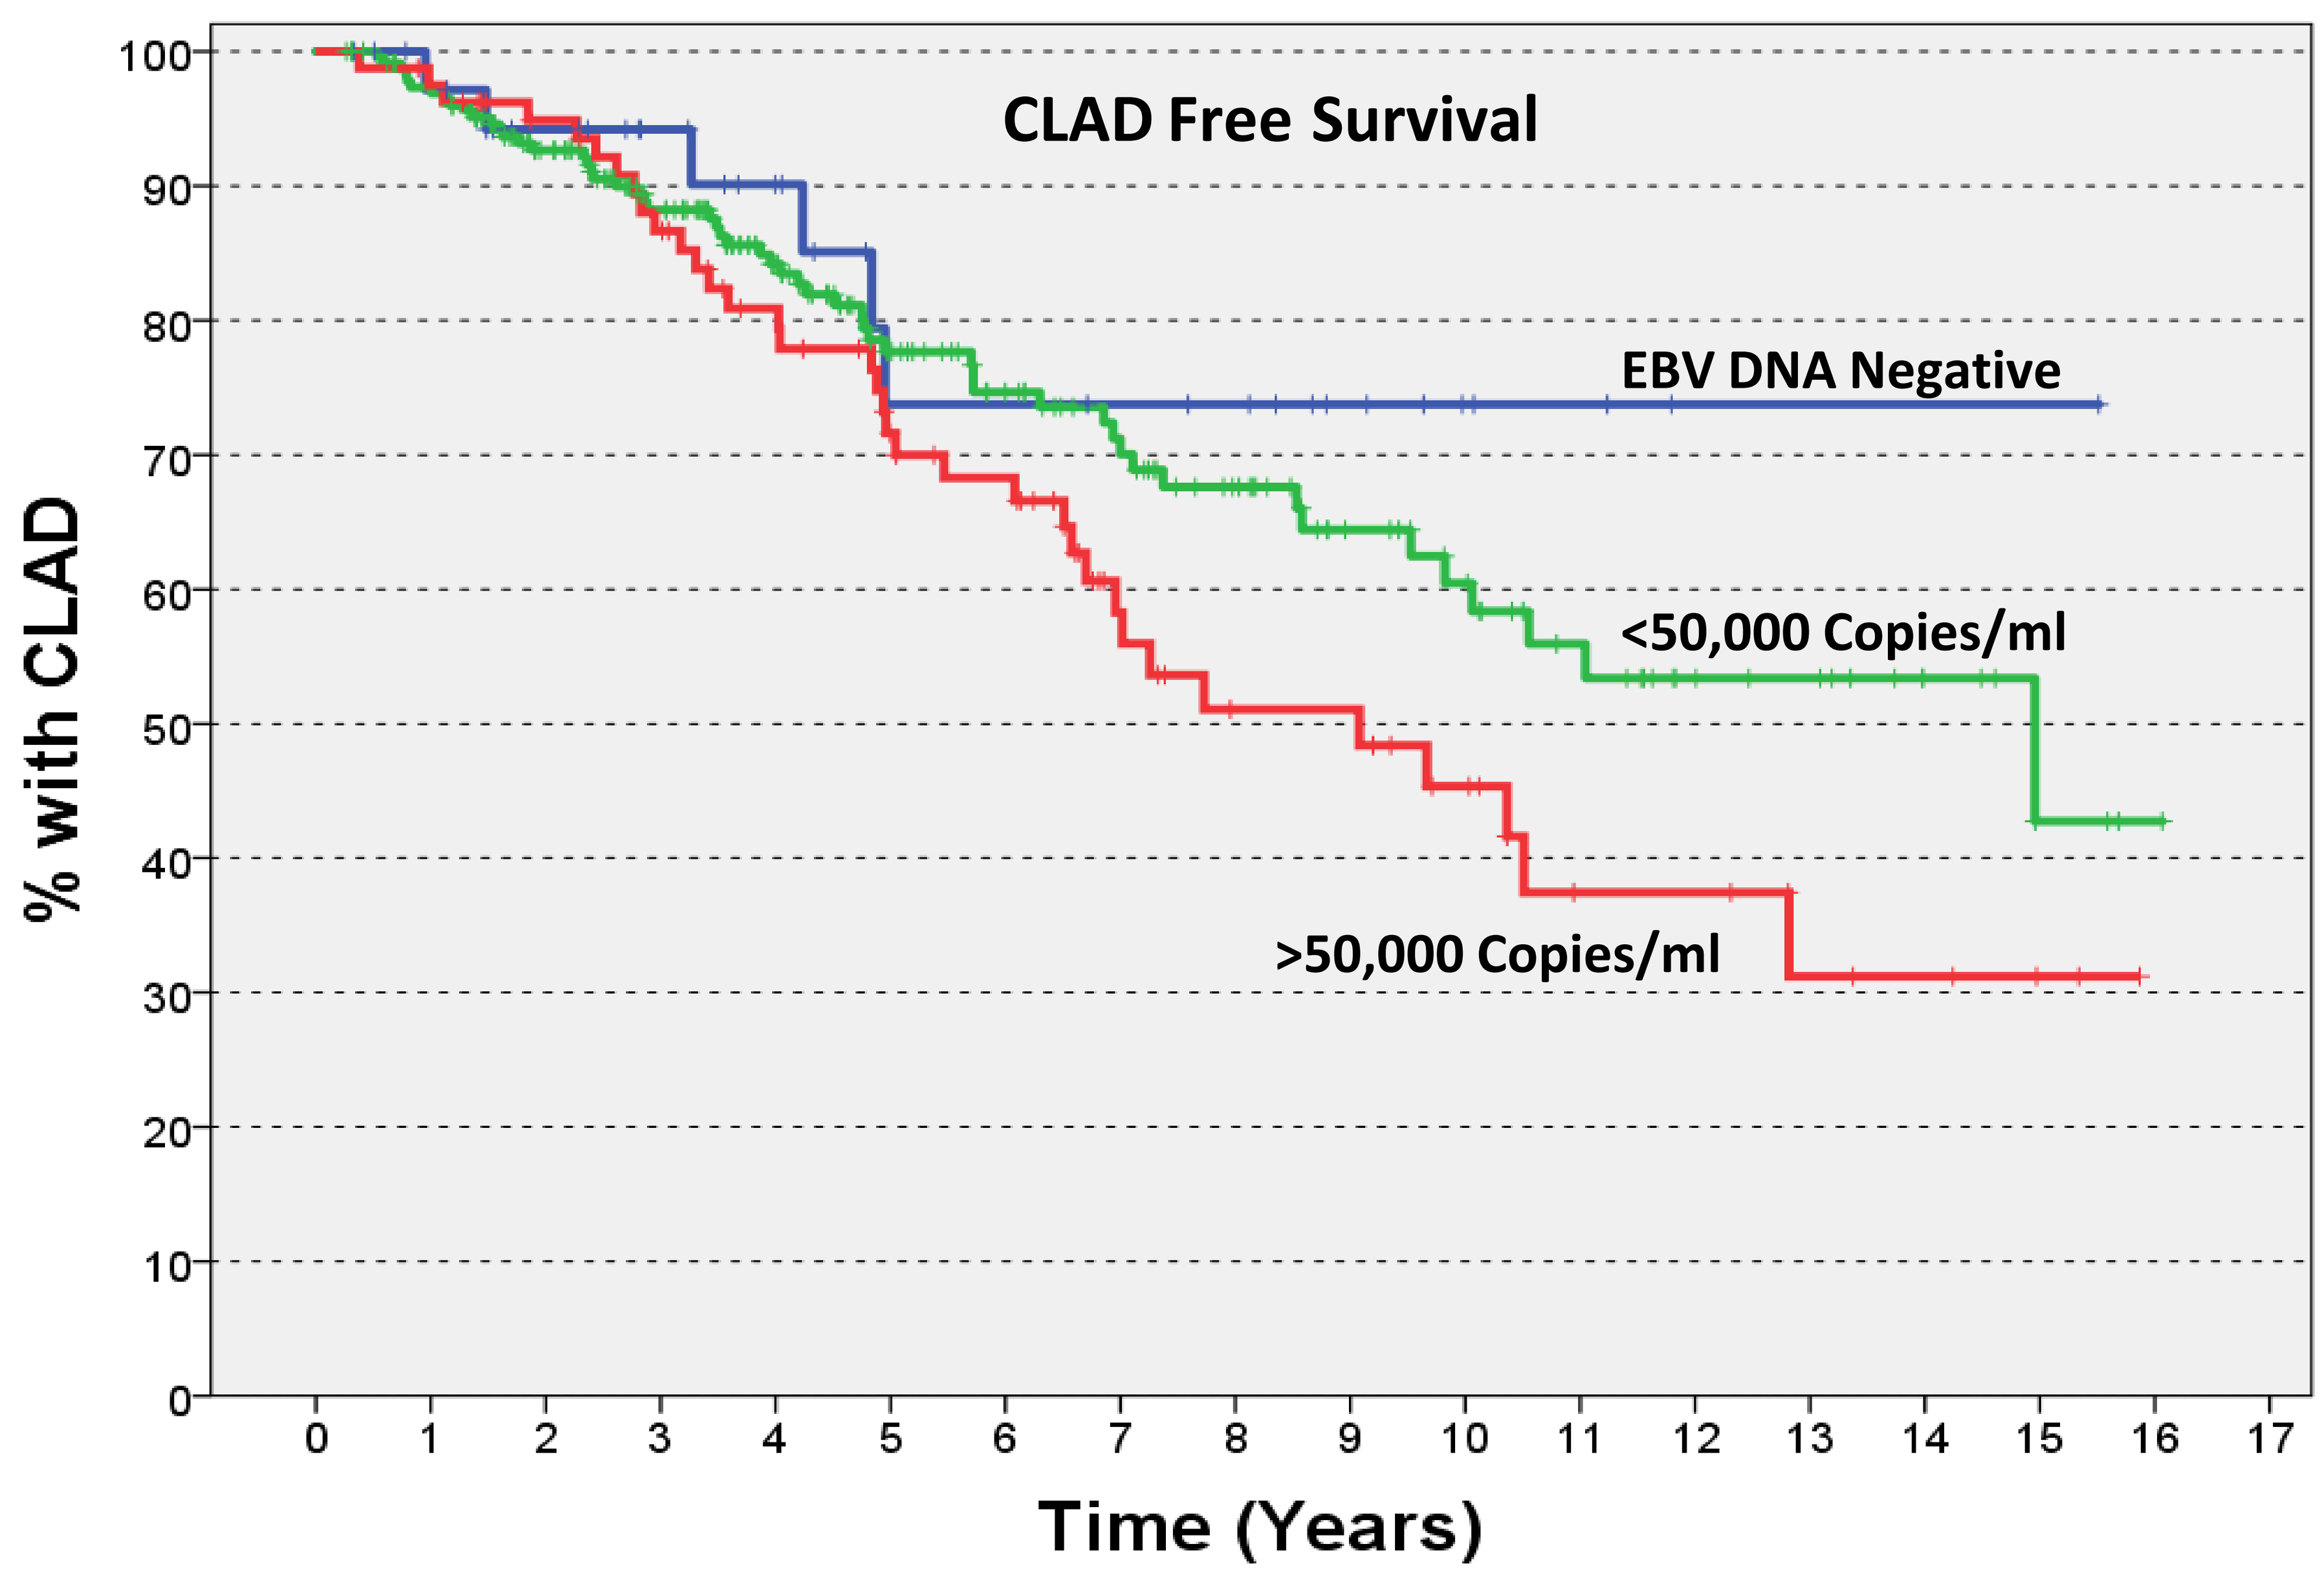
CLAD Free Survival
EBV DNA Negative
<50,000 Copies/ml
>50,000 Copies/ml
% with CLAD
Time (Years)

Figure 2: Kaplan-Meier plot of peak whole blood EBV values against CLAD free survival. EBV has a statistically significant effect on the development of CLAD (Chi-square p<0,001)

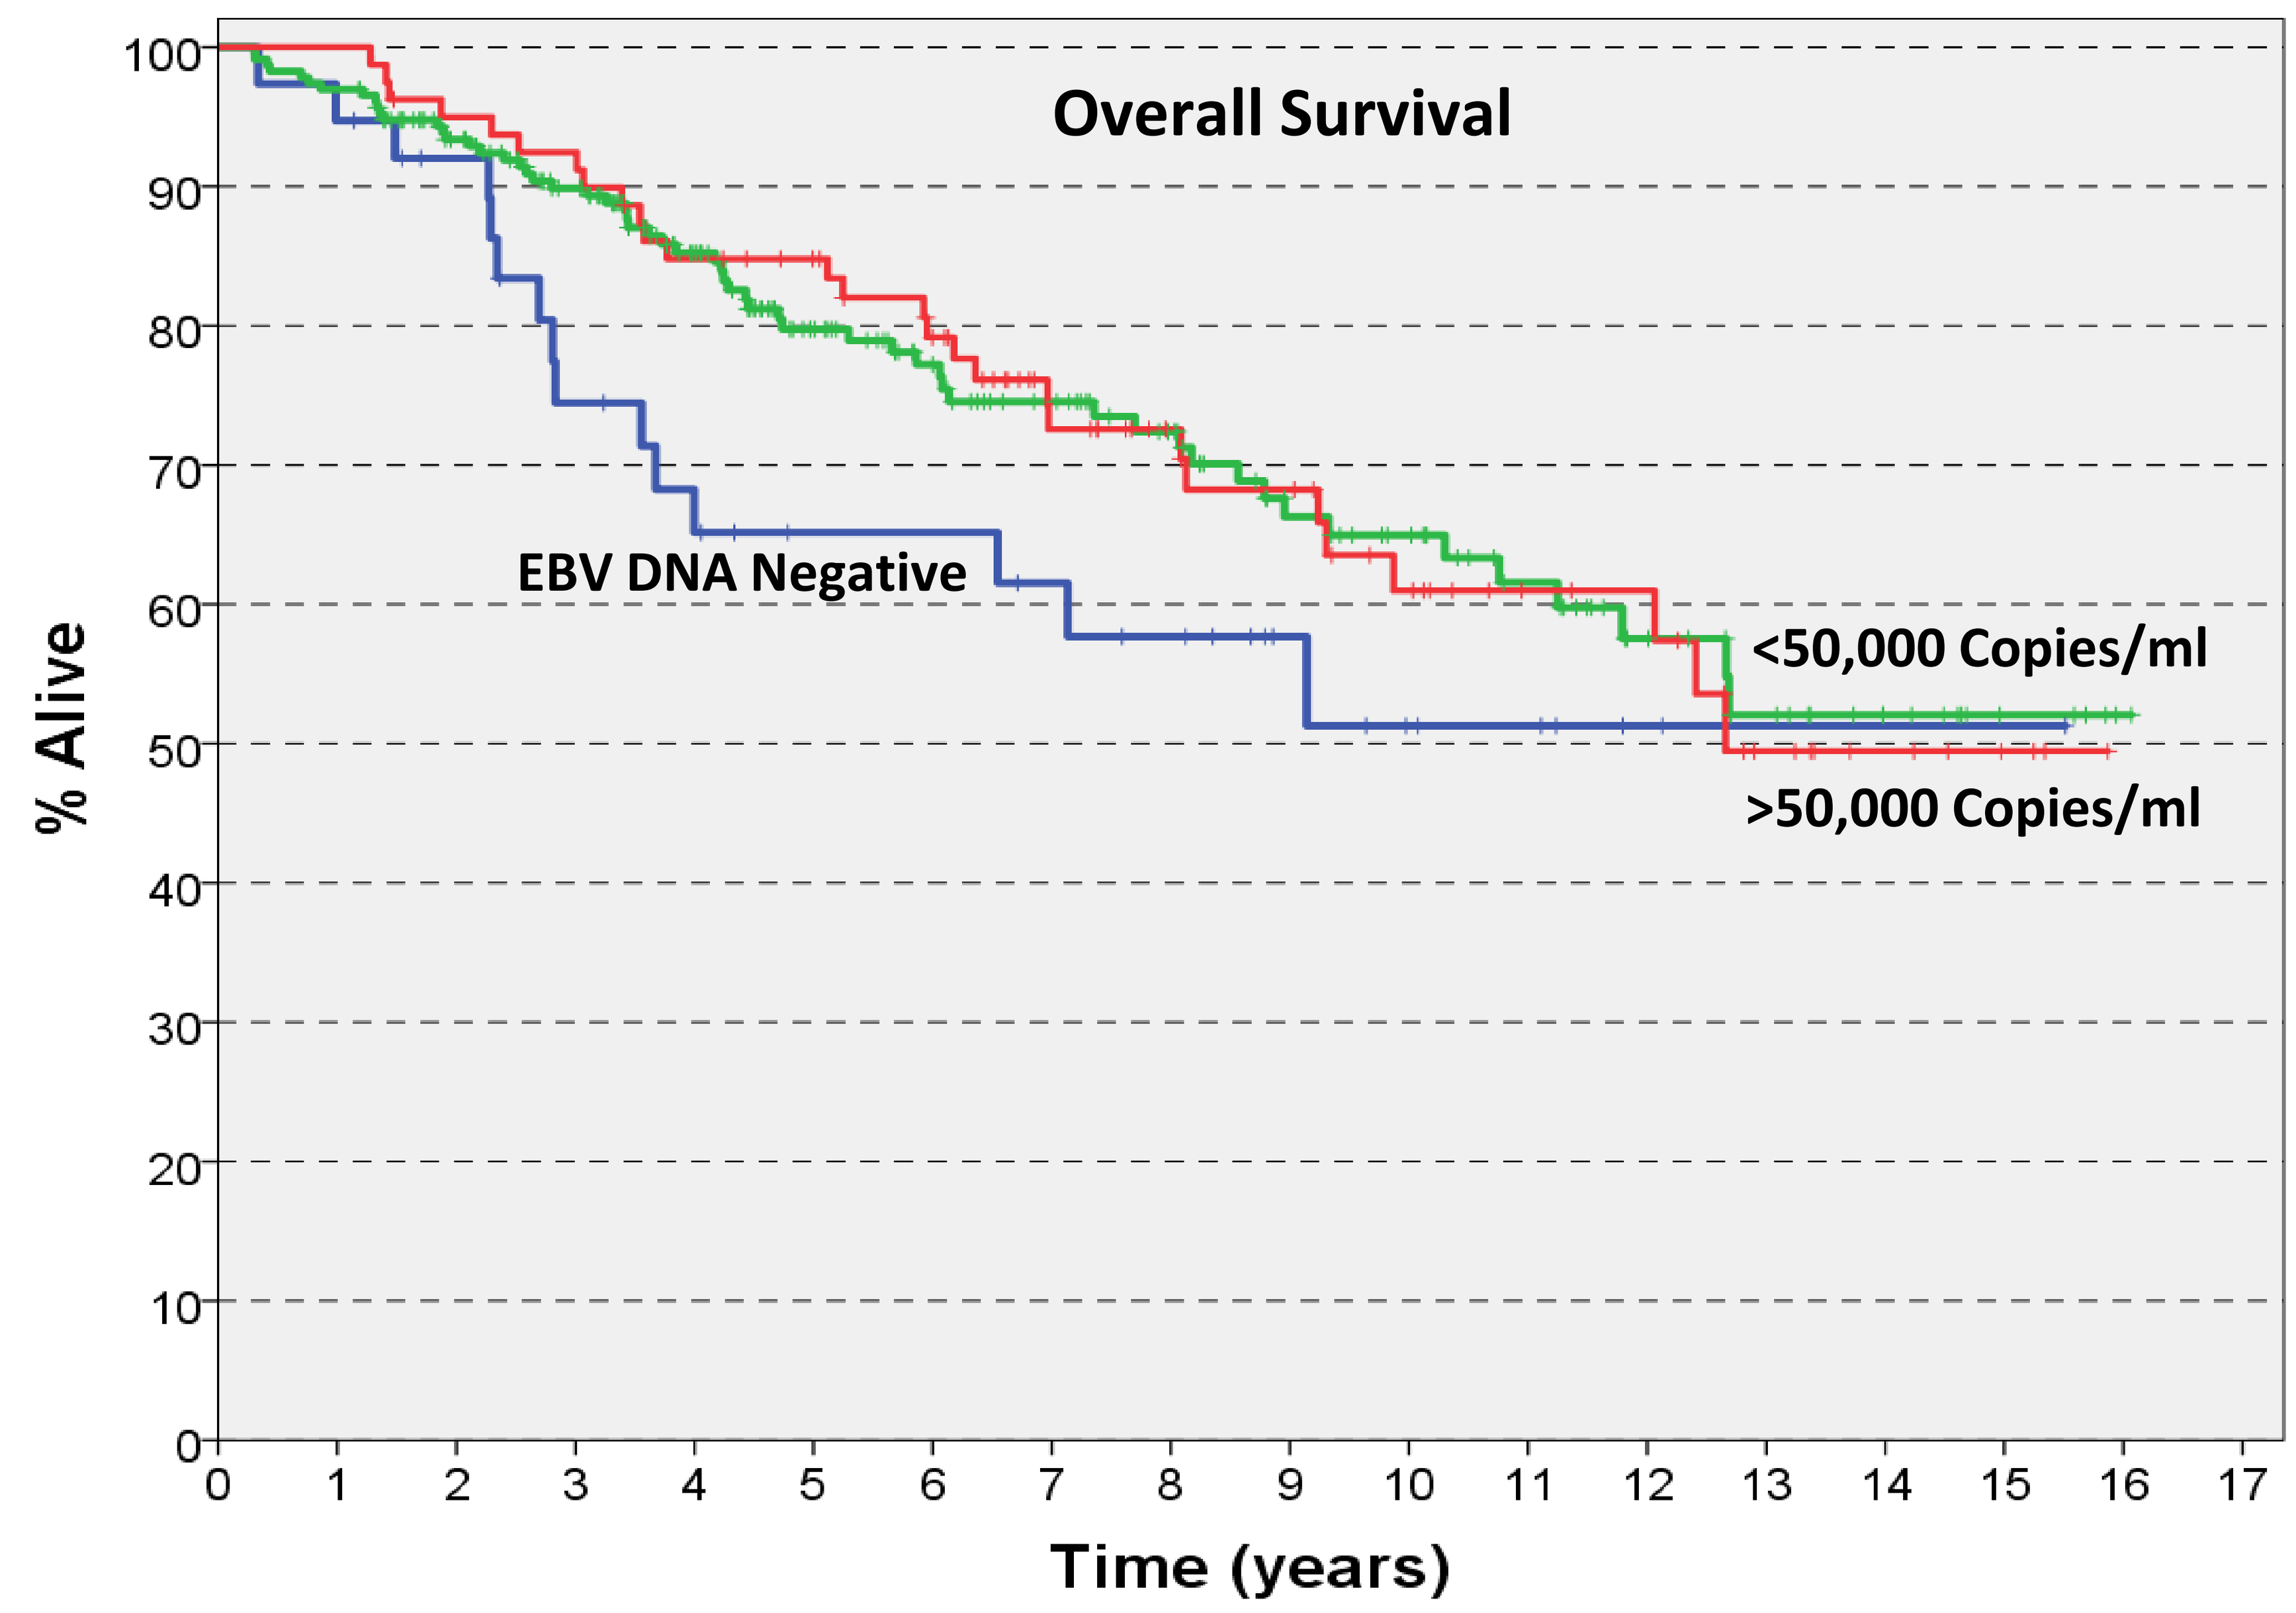
Overall Survival
EBV DNA Negative
<50,000 Copies/ml
>50,000 Copies/ml
% Alive
Time (years)

Figure 3: Kaplan-Meier plot of peak whole blood EBV values against survival. Interestingly EBV load has minimal effect on overall long term survival.

Materials and Methods

All lung transplant recipients (N=355) transplanted between June 2001 and May 2016 surviving over 1 months routine serial EBV DNA loads in whole blood were investigated. Paediatric patients, defined as <16 at transplant, patients who died within 30 days of transplant and re-transplants were excluded from analysis. Median number of samples was 29 (range 1-88). Patient outcome with regards to overall survival (OS), development of CLAD and PTLD was noted and analysed. CLAD was defined using recent guidelines (4,5). Statistics were performed using SPSS 23 (SPSS Inc, Chicago, IL) p-values <0.05 were taken as significant. Pearson's Chi-square test and the Wald test were used as appropriate. Viral impact on CLAD free survival and overall survival was assessed using Kaplan-Meier analysis and the log rank test.

Results

Of 356 patients 38 remained EBV DNA negative with 1 (EBER negative) PTLD in this group. 231 had peak EBV DNA levels between 1.000 and 50.000 copies/mL of whom 4 developed PTLD. 80 had peak levels above 50.000 copies/mL and 12 developed PTLD. Of these 80 patients 7 patients received pre-emptive RTx based on high and rising EBV-DNA load which might have prevented development of PTLD in 6 (figure 1). There is a reduction in the OR for developing PTLD following RTx use with a 60% reduction in risk 95% CI (0.054-3.063) (p>0.05). EBV DNA load was significantly associated with PTLD (p<0,0001) >50.000 having the most risk 95% CI (2.379-24.489) (p=<0.001) and with CLAD (p<0,001, figure 2). But surprisingly not with overall survival (figure 3).

Conclusion

Monitoring of EBV viral loads can identify patients with a higher risk of developing complications such as PTLD and CLAD. It also enables the use of pre-emptive Rituximab based on EBV DNA load which might prevent PTLD. Surprisingly no significant relationship between EBV DNA load and survival was seen.

References

(1) Bakker NA, Verschuuren EAM, Erasmus ME, Hepkema BG, Veeger, Nic J G M , Kallenberg, Cees G M, et al. Epstein-Barr Virus DNA load monitoring late after lung transplantation: a surrogate marker of the degree of immunosuppression and a safe guide to reduce immunosuppression. Transplantation 2007;83.

(2) Engelmann I, Welte T, Führer T, Simon AR, Mattner F, Hoy L, et al. Detection of Epstein–Barr virus DNA in peripheral blood is associated with the development of bronchiolitis obliterans syndrome after lung transplantation. Journal of Clinical Virology 2009;45(1):47-53.

(3) Dan L Longo, Daan Dierickx, Thomas M Habermann. Post-Transplantation Lymphoproliferative Disorders in Adults. The New England Journal of Medicine 2018 Feb 8,;378(6):549.

(4) Verleden GM, Raghu G, Meyer KC, Glanville AR, Corris P. A new classification system for chronic lung allograft dysfunction. The Journal of heart and lung transplantation : the official publication of the International Society for Heart Transplantation 2014 Feb;33(2):127.

(5) Vos R, Verleden SE, Verleden GM. Chronic lung allograft dysfunction: evolving practice. Current opinion in organ transplantation 2015 Oct;20(5):483.

Relevant Financial Relationship Disclosure Statement

No relationships to disclose: E.J. Gore: None. W. van der Bij: None. C.C. Leer: None. M.E. Erasmus: None. C. Gan: None. E.A. Verschuuren: None