The Effect of Dual Renin-angiotensin System Blockade Compared to Conventional Antihypertensive Combination on Blood Pressure and Renal Function After Heart Transplantation: A Randomized Controlled Trial

Lenka Hošková[1], Ivan Málek[1], Vojtěch Melenovský[1], Josef Kautzner[1], Ivan Netuka[1], Janka Franeková[2], Antonín Jabor[2]

[1]Heart Center, [2]Department of Laboratory Methods, **IKEM, Prague, Czech Republic**

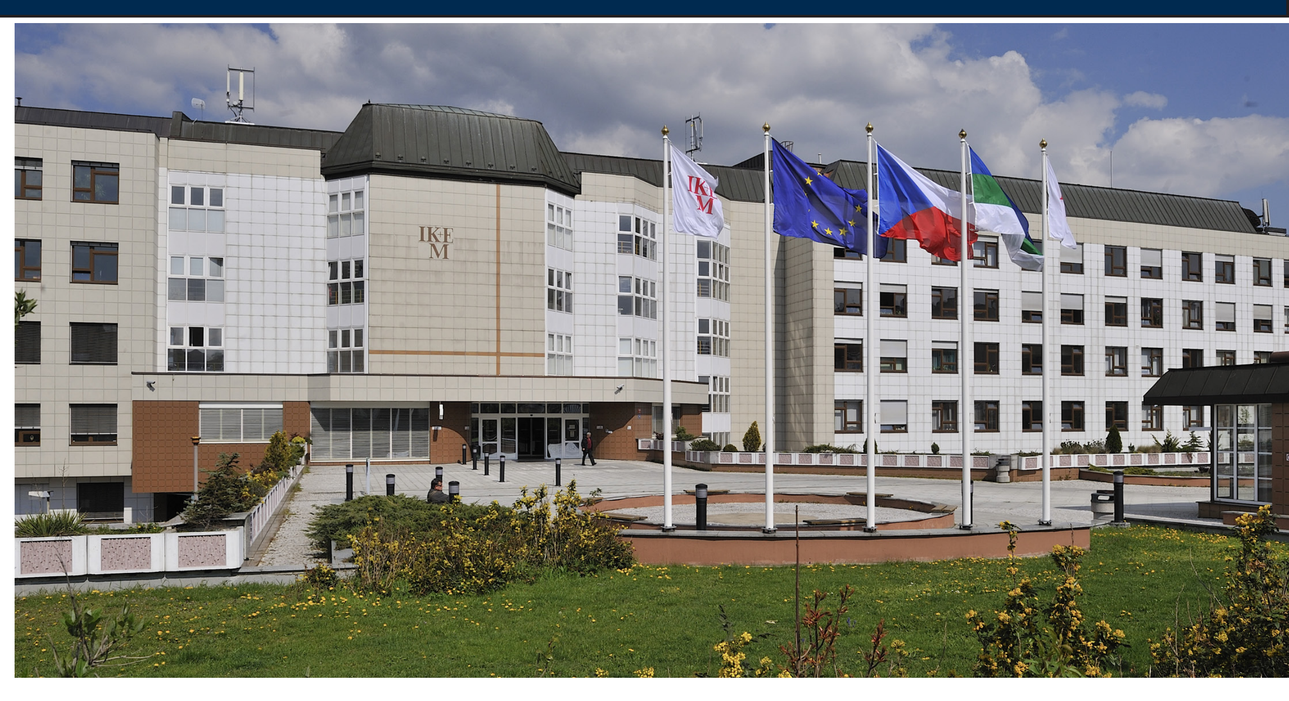

Purpose

Altered cardiovascular regulation and use of immunosuppressive drugs after heart transplantation (HTx) are associated with development of arterial hypertension (AHT) and nephrotoxicity. HTx-related AHT can be difficult to control and combination of antihypertensives is often necessary. We compared the effect of the standard antihypertensive medication (aHTM) with dual blockade of the renin-angiotensin system (dual RAS combination) on blood pressure control and renal functions.

Methods

Between 2010 and 2013, we enrolled 84 patients who developed AHT in the first six months after HTx. We randomized them in two groups: group 1 (44 patients) assigned to standard aHTM therapy consisting of 2-4 aHTM: calcium channel blocker (CCB), beta-blocker (BB), diuretic and ACE inhibitor; and group 2 (31 patients) assigned to combination of ACE inhibitor and angiotensin receptor blocker with CCB or BB or diuretic (2 to 4 aHTM), (Fig.1). The follow-up was 24 months. Systolic blood pressure (SBP) and diastolic blood pressure (DBP), glomerular filtration (GFR as CKD-EPI) and albuminuria (ACR albumin/creatinine) were serially measured (3,6,9,12,18, and 24th month).

Results

Both groups were comparable with respect to basic parameters such as age, sex, BMI and comorbidities. The mean levels of tacrolimus were not significantly different between groups. The blood pressure control was attained to similar level in both groups: SBP 127.9 ± 2.1 mmHg vs. 129.3 ± 2.3 mmHg, (Fig. 2) and DBP 82.3 ± 1.5 mmHg vs 82.7 ± 0.9 mmHg, (Fig. 3). After 24 months, GFR was maintained in group 2 (1.2 ± 0.05 ml/sec/1.73m^2), in contrast to group 1 where GFR dropped (1.0 ± 0.06 ml/sec/1.73m^2), p<0.05 (Fig 4). In comparison to group 1 (ACR 12.0 ± 3.2 g/mol), albuminuria was markedly lower in group 2 (ACR 2.8 ± 1.2 g/mol), p< 0.05 during the follow up period, (Fig.5). Three patients discontinued study because of hyperkalemia > 5,4 mmol/L (2 patients) and GFR<0,5 ml/s (1 patient).

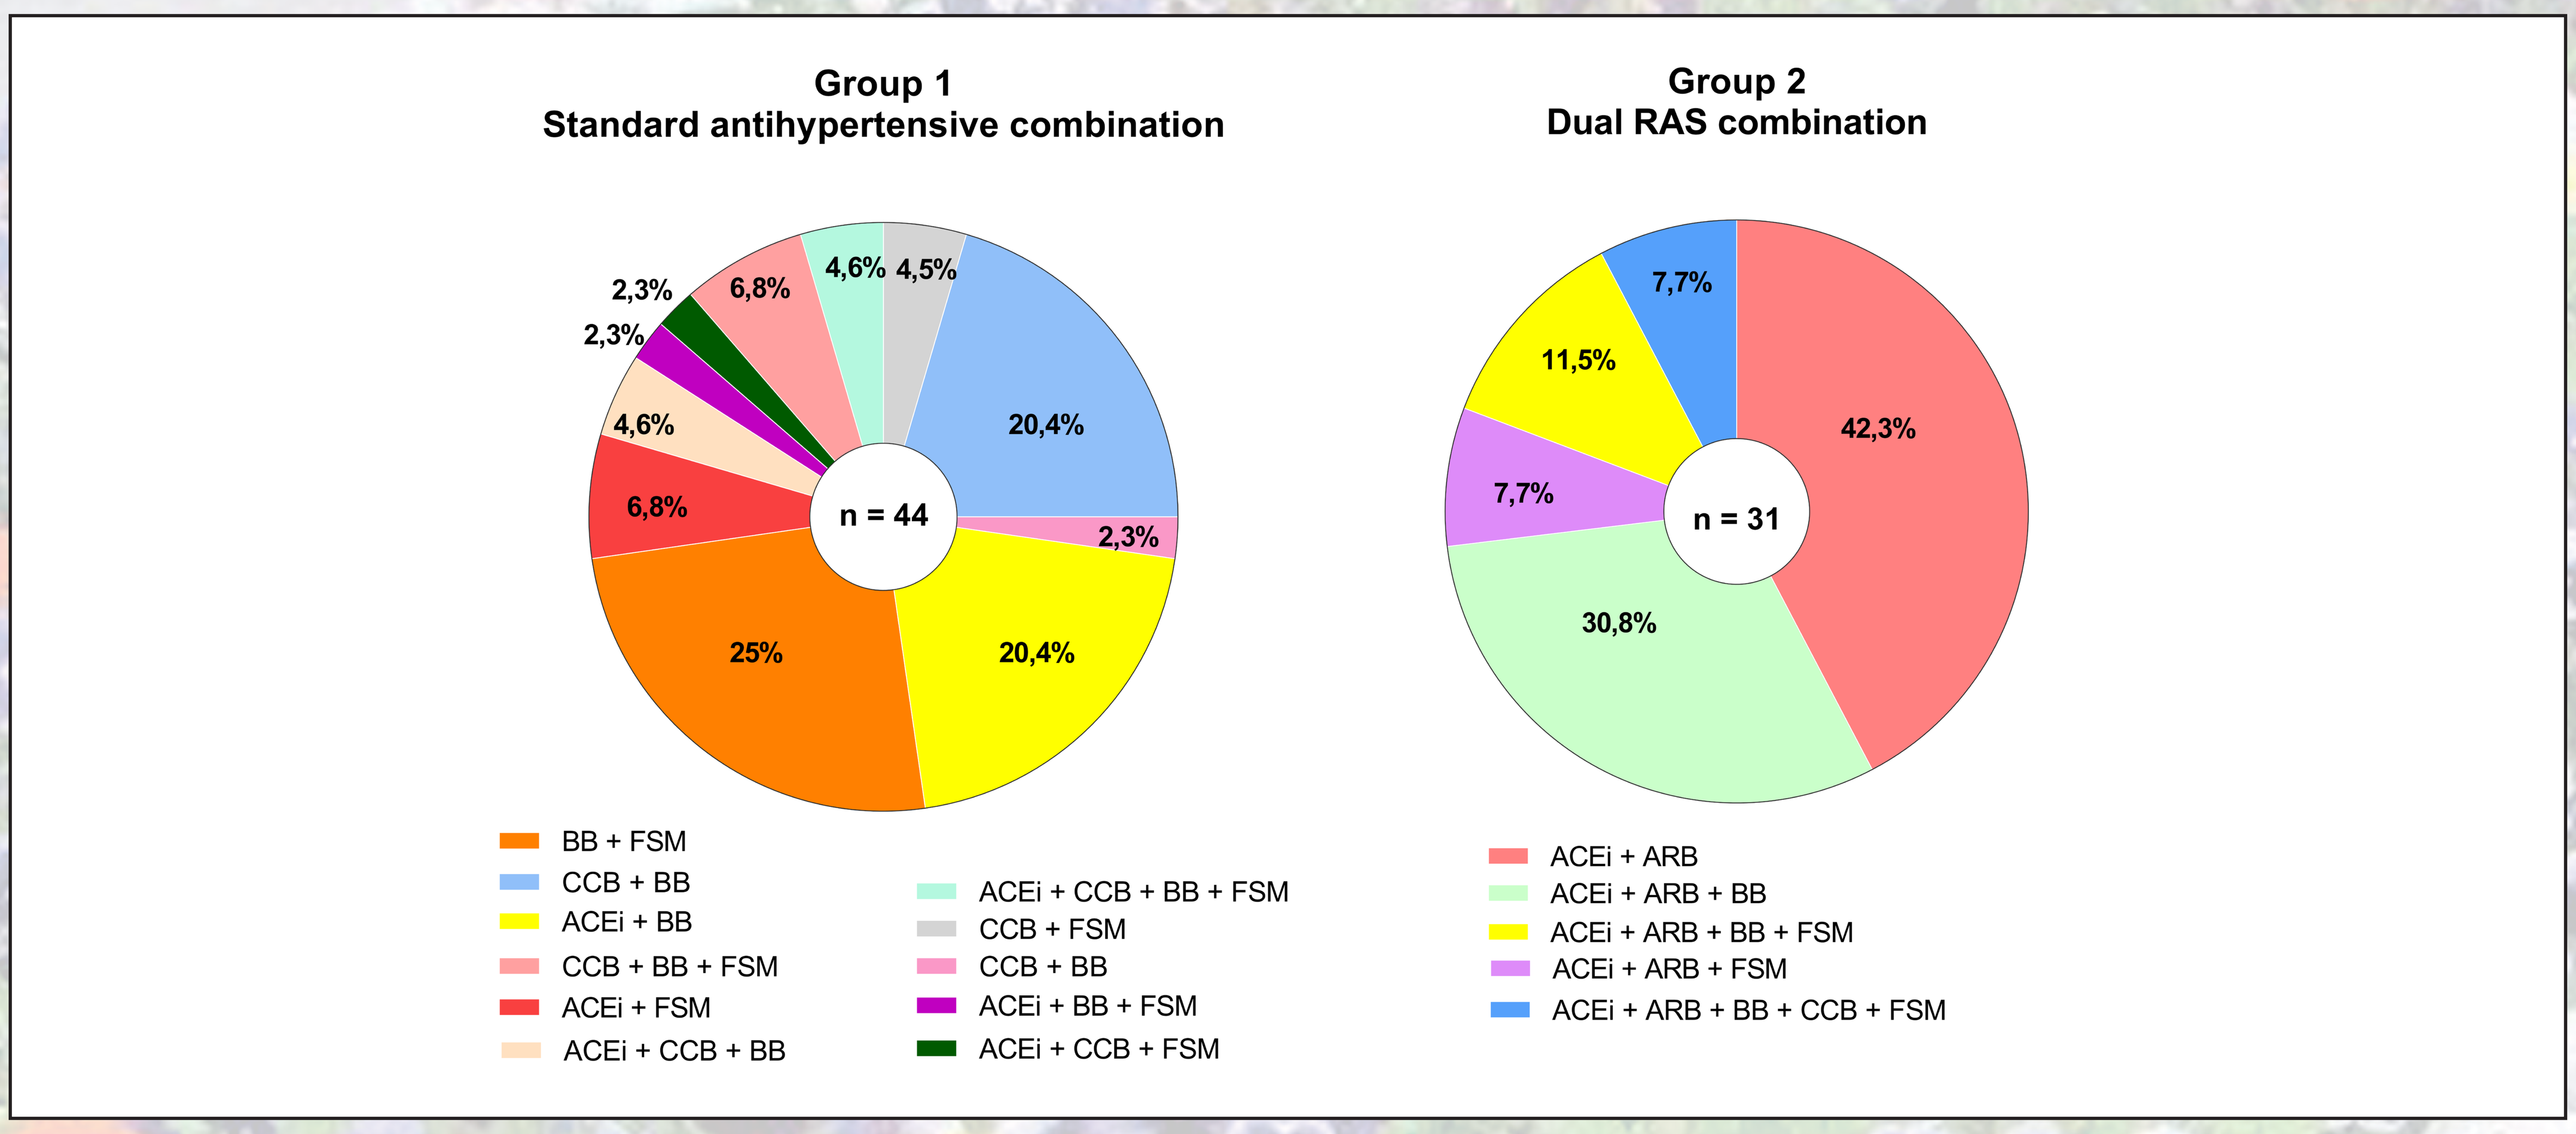
Group 1
Standard antihypertensive combination
25%
20,4%
20,4%
2,3%
6,8%
4,6%
2,3%
2,3%
6,8%
4,6%
4,5%
n = 44
BB + FSM
CCB + BB
ACEi + BB
CCB + BB + FSM
ACEi + FSM
ACEi + CCB + BB
ACEi + CCB + BB + FSM
CCB + FSM
CCB + BB
ACEi + BB + FSM
ACEi + CCB + FSM
Group 2
Dual RAS combination
42,3%
30,8%
7,7%
11,5%
7,7%
n = 31
ACEi + ARB
ACEi + ARB + BB
ACEi + ARB + BB + FSM
ACEi + ARB + FSM
ACEi + ARB + BB + CCB + FSM

Fig.1: Combination of antihypertensives
Group 1 (standard antihypertensive combination), Group 2 (dual RAS combination)
ACEi = angiotensin-converting enzyme inhibitor, ARB = angiotensin receptor blocker, BB = beta-blocker,CCB = calcium channel blocker, FSM = furosemide.

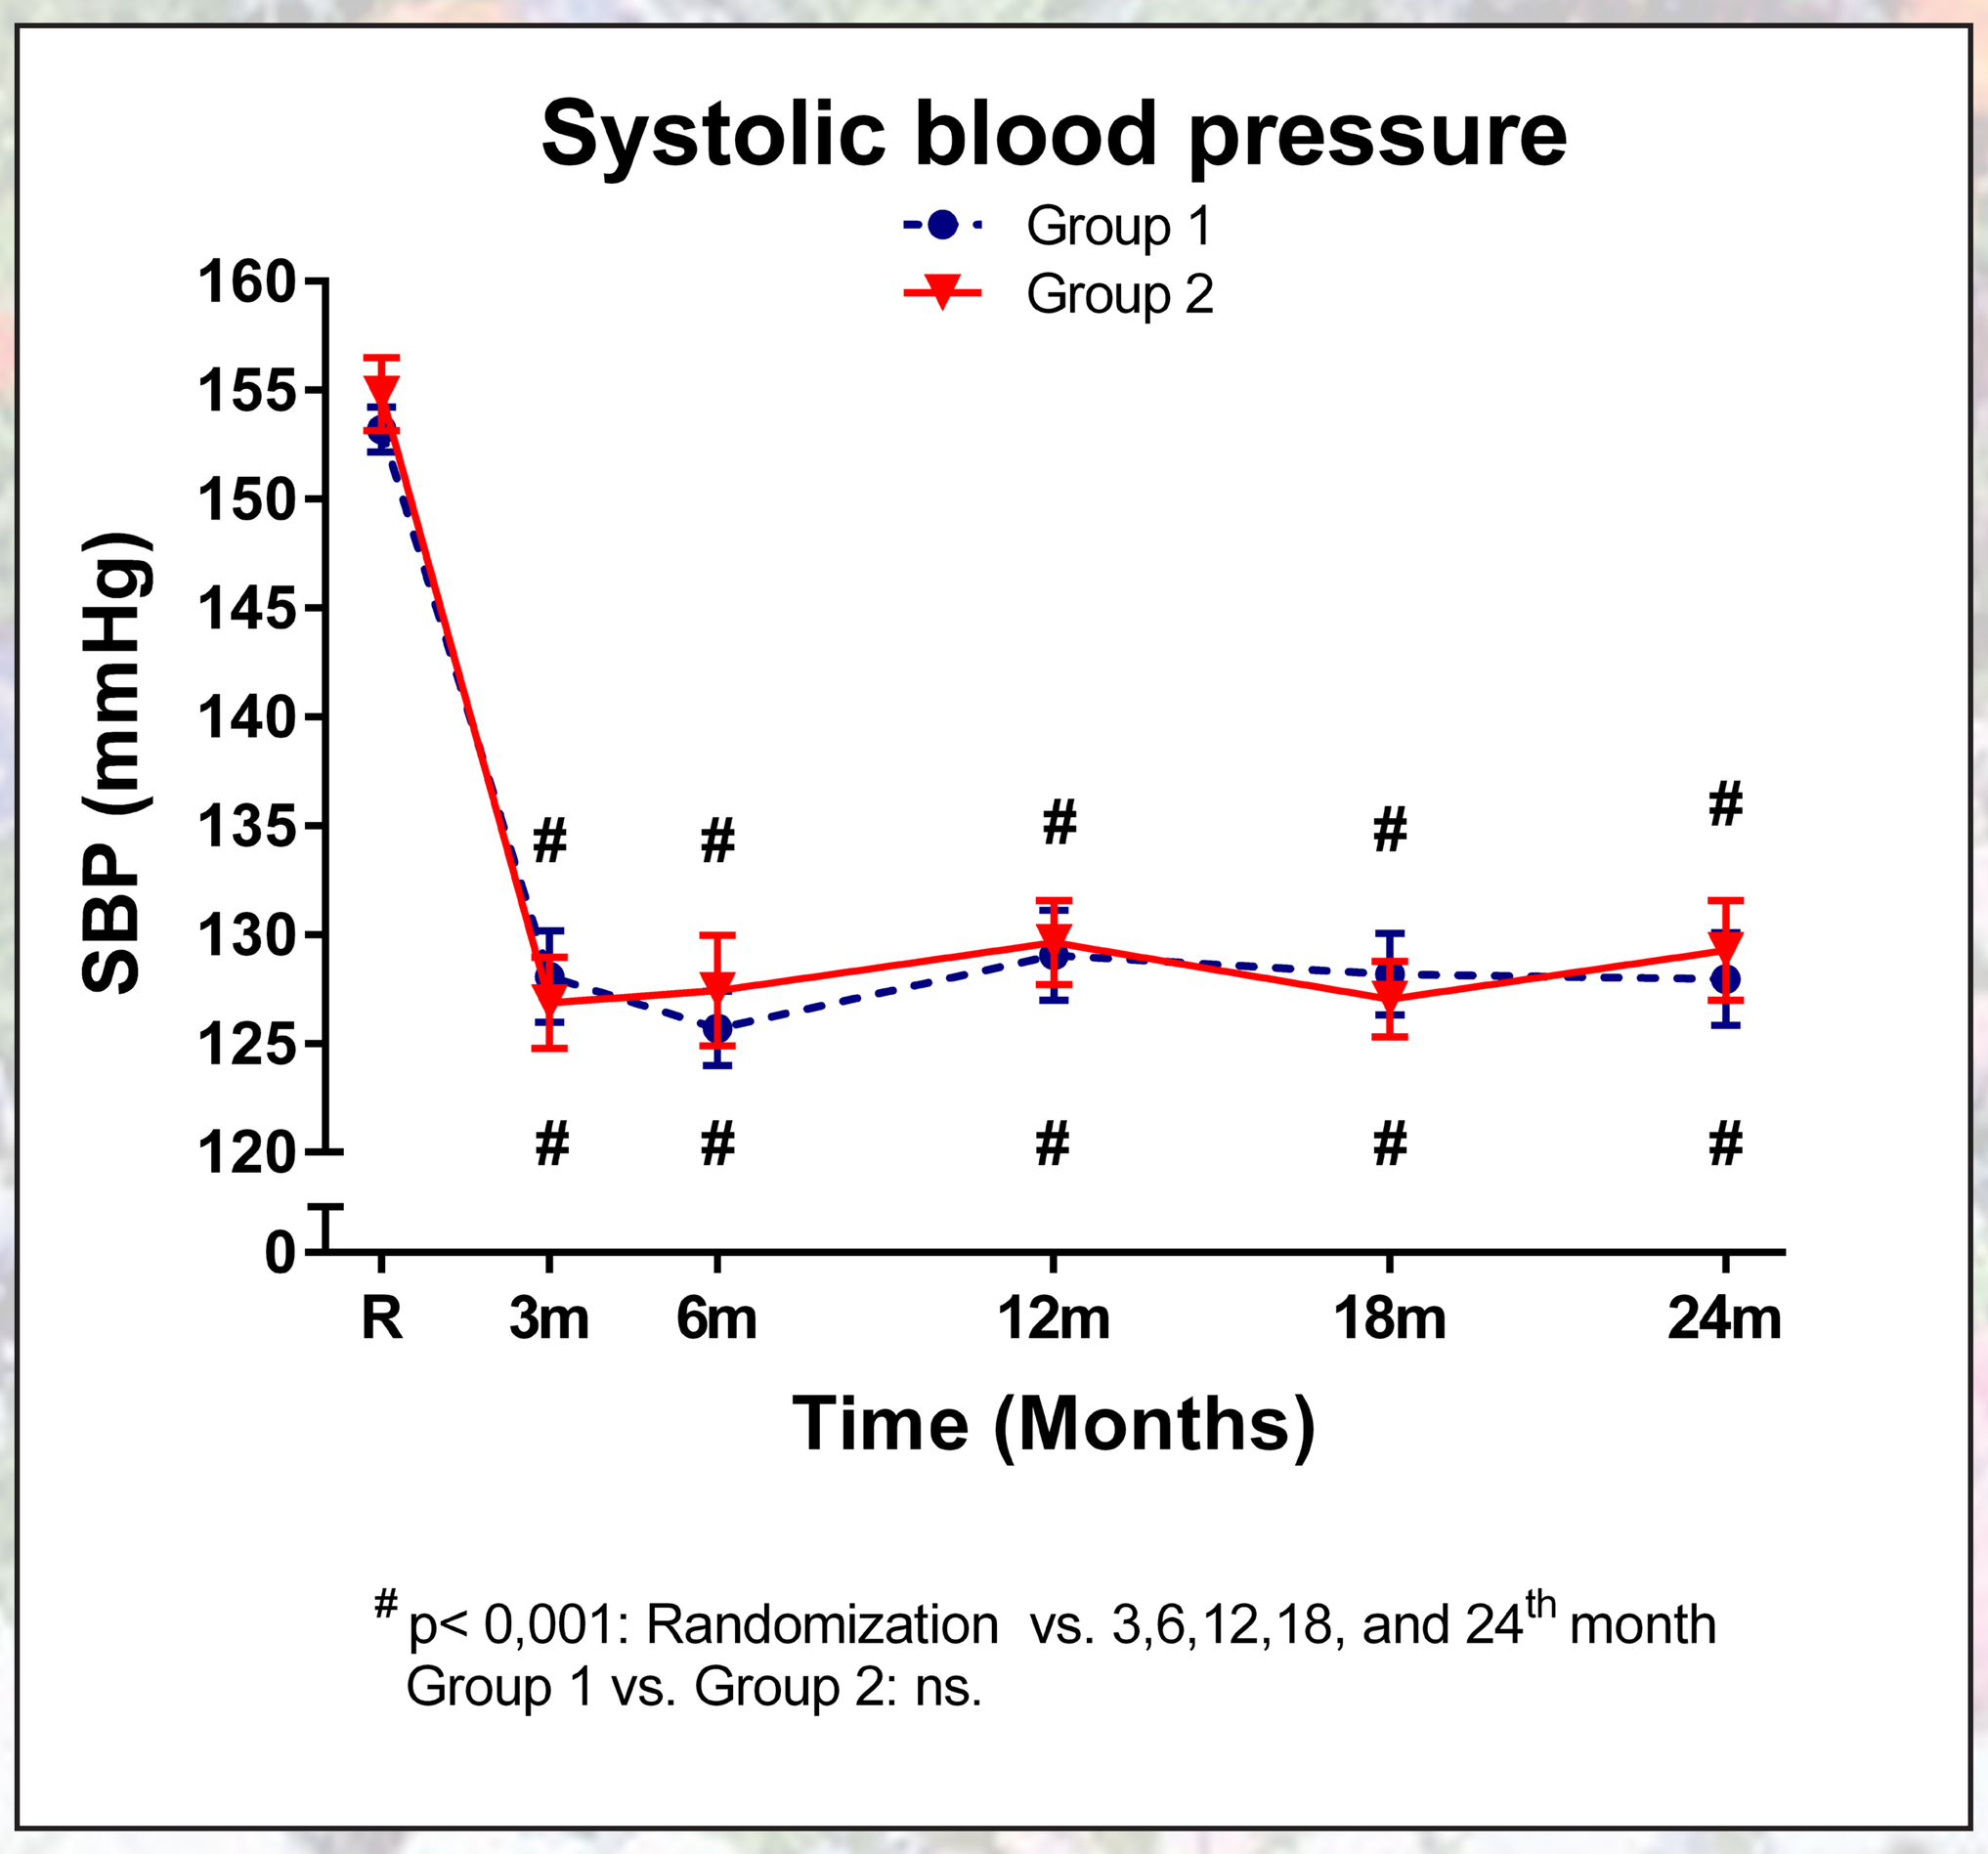
Systolic blood pressure
Group 1
Group 2
SBP (mmHg)
Time (Months)
R 3m 6m 12m 18m 24m
p< 0,001: Randomization vs. 3,6,12,18, and 24th month
Group 1 vs. Group 2: ns.

Fig.2: Systolic blood pressure (SBP)
Group 1 (standard antihypertensive combination), Group 2 (dual RAS combination).

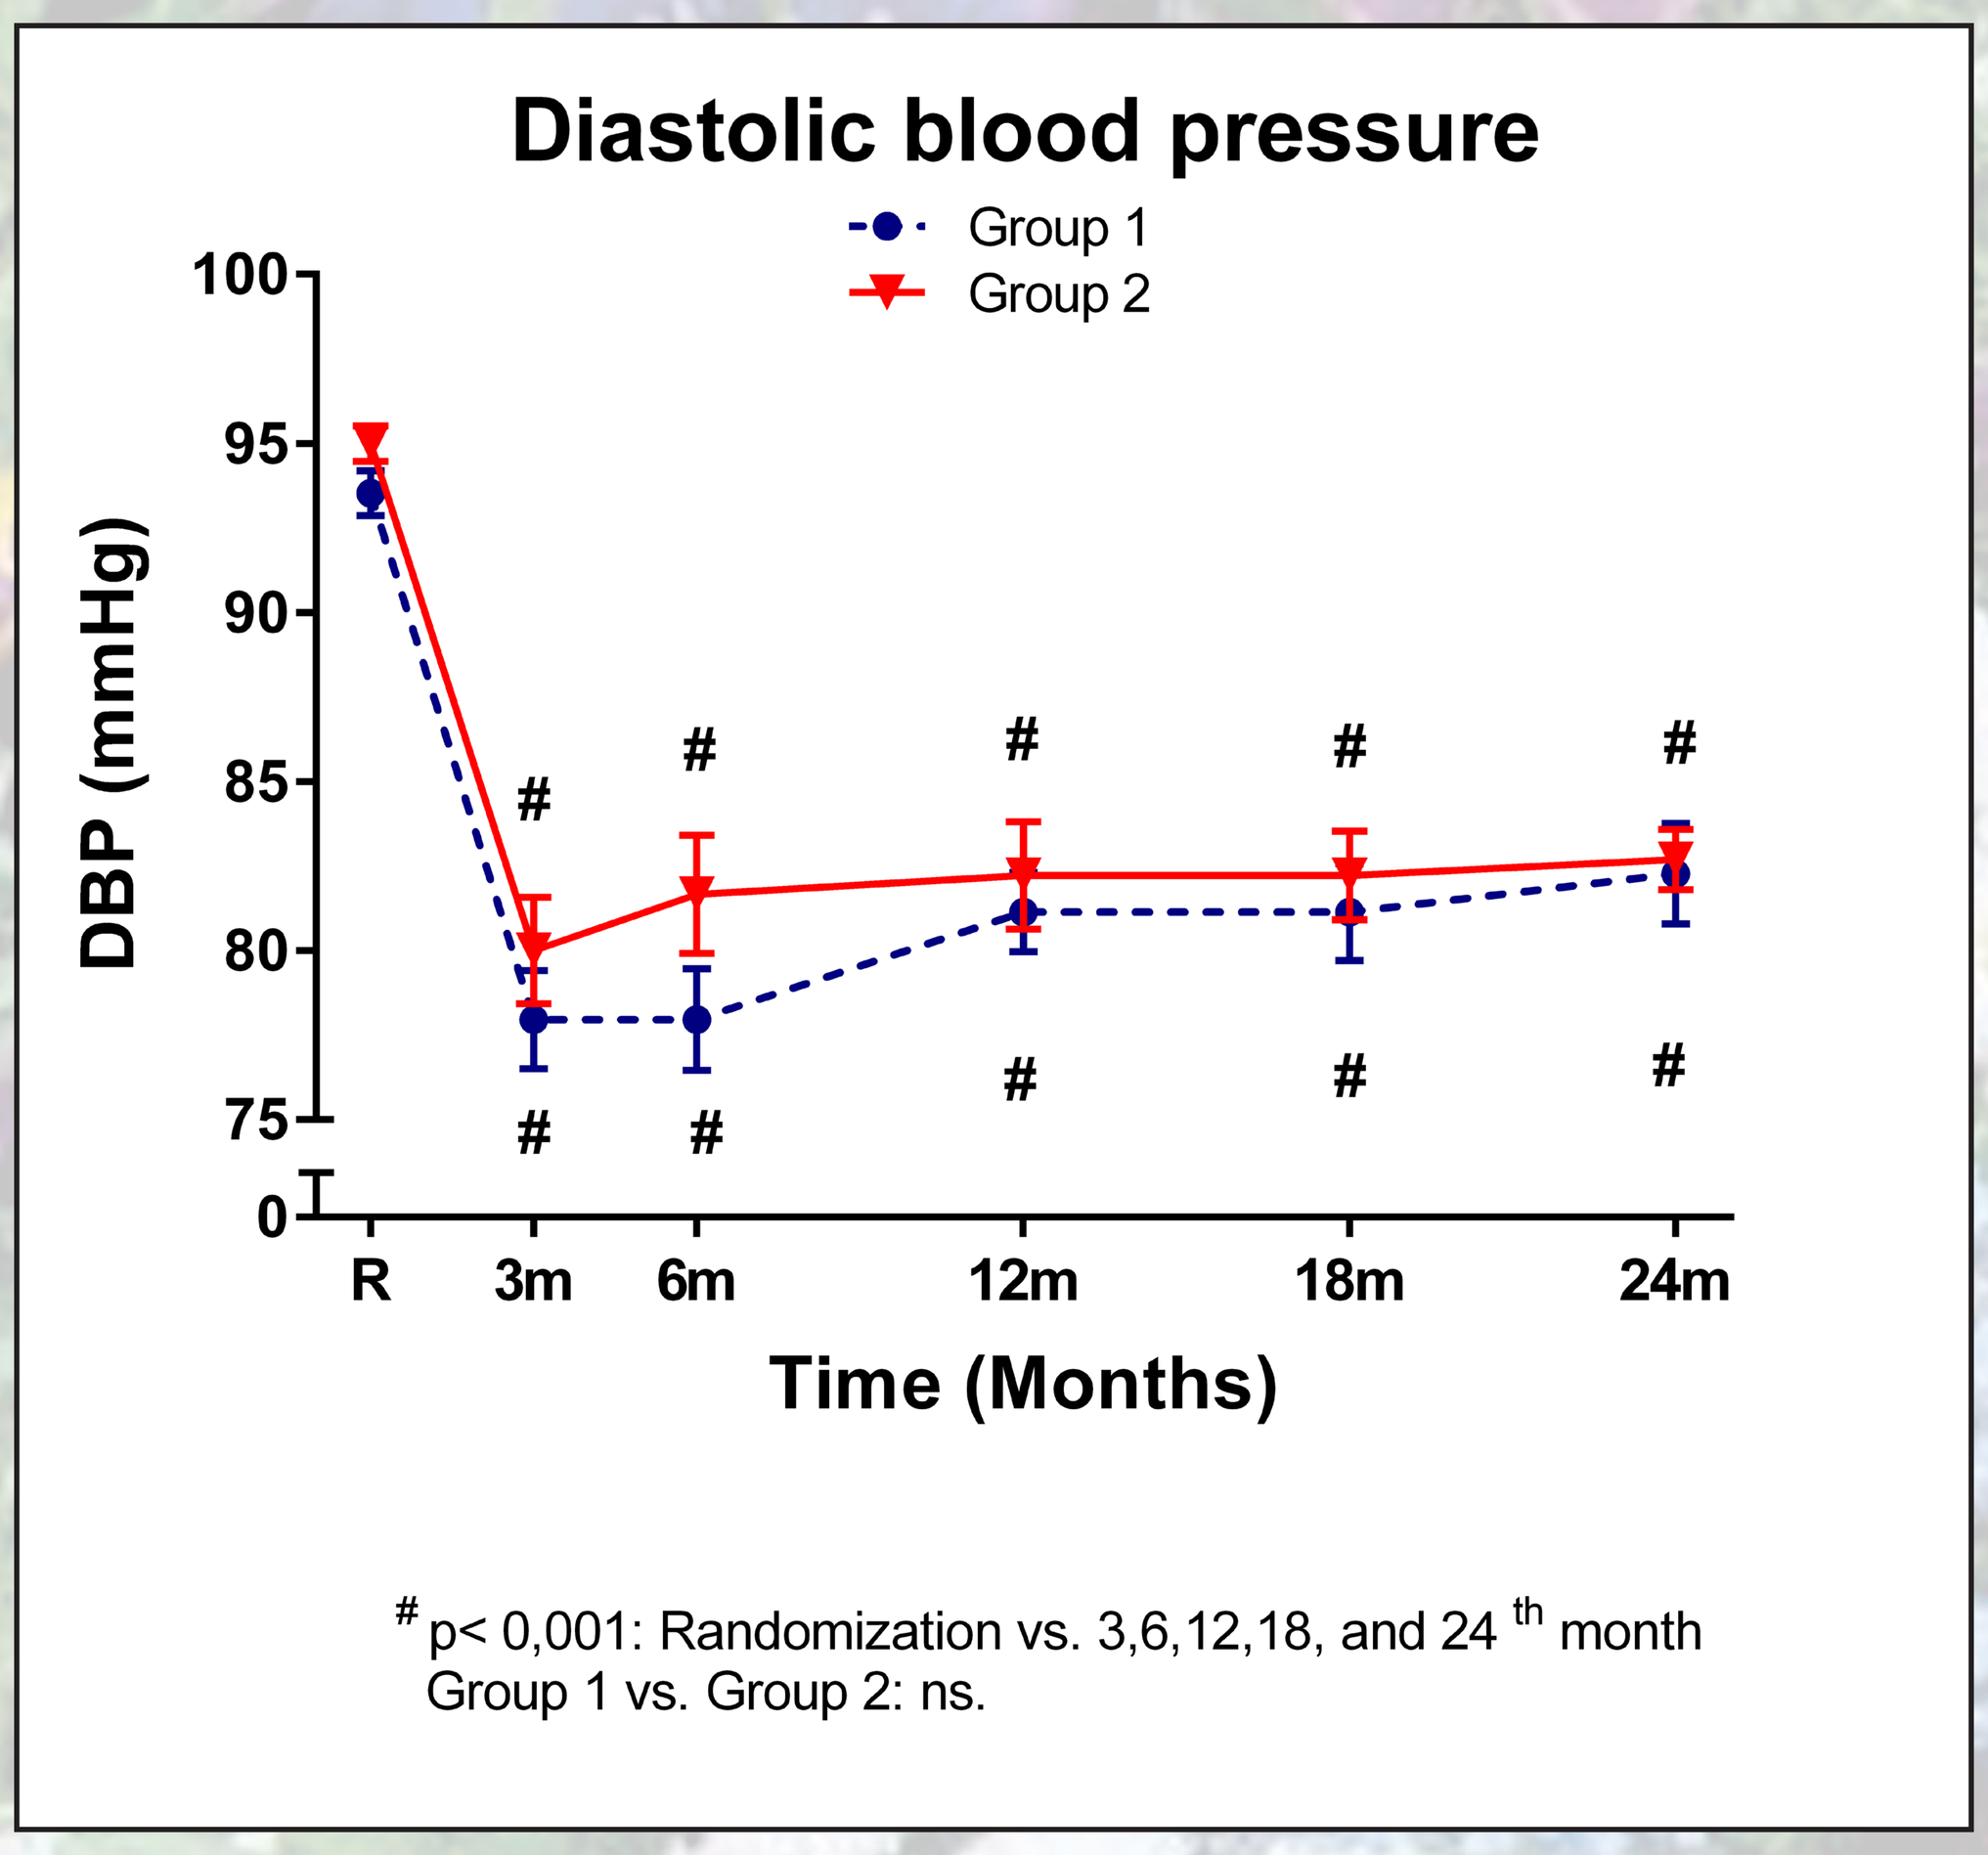
Diastolic blood pressure
Group 1
Group 2
DBP (mmHg)
Time (Months)
R 3m 6m 12m 18m 24m
p< 0,001: Randomization vs. 3,6,12,18, and 24 th month
Group 1 vs. Group 2: ns.

Fig.3: Diastolic blood pressure (DBP)
Group 1 (standard antihypertensive combination), Group 2 (dual RAS combination).

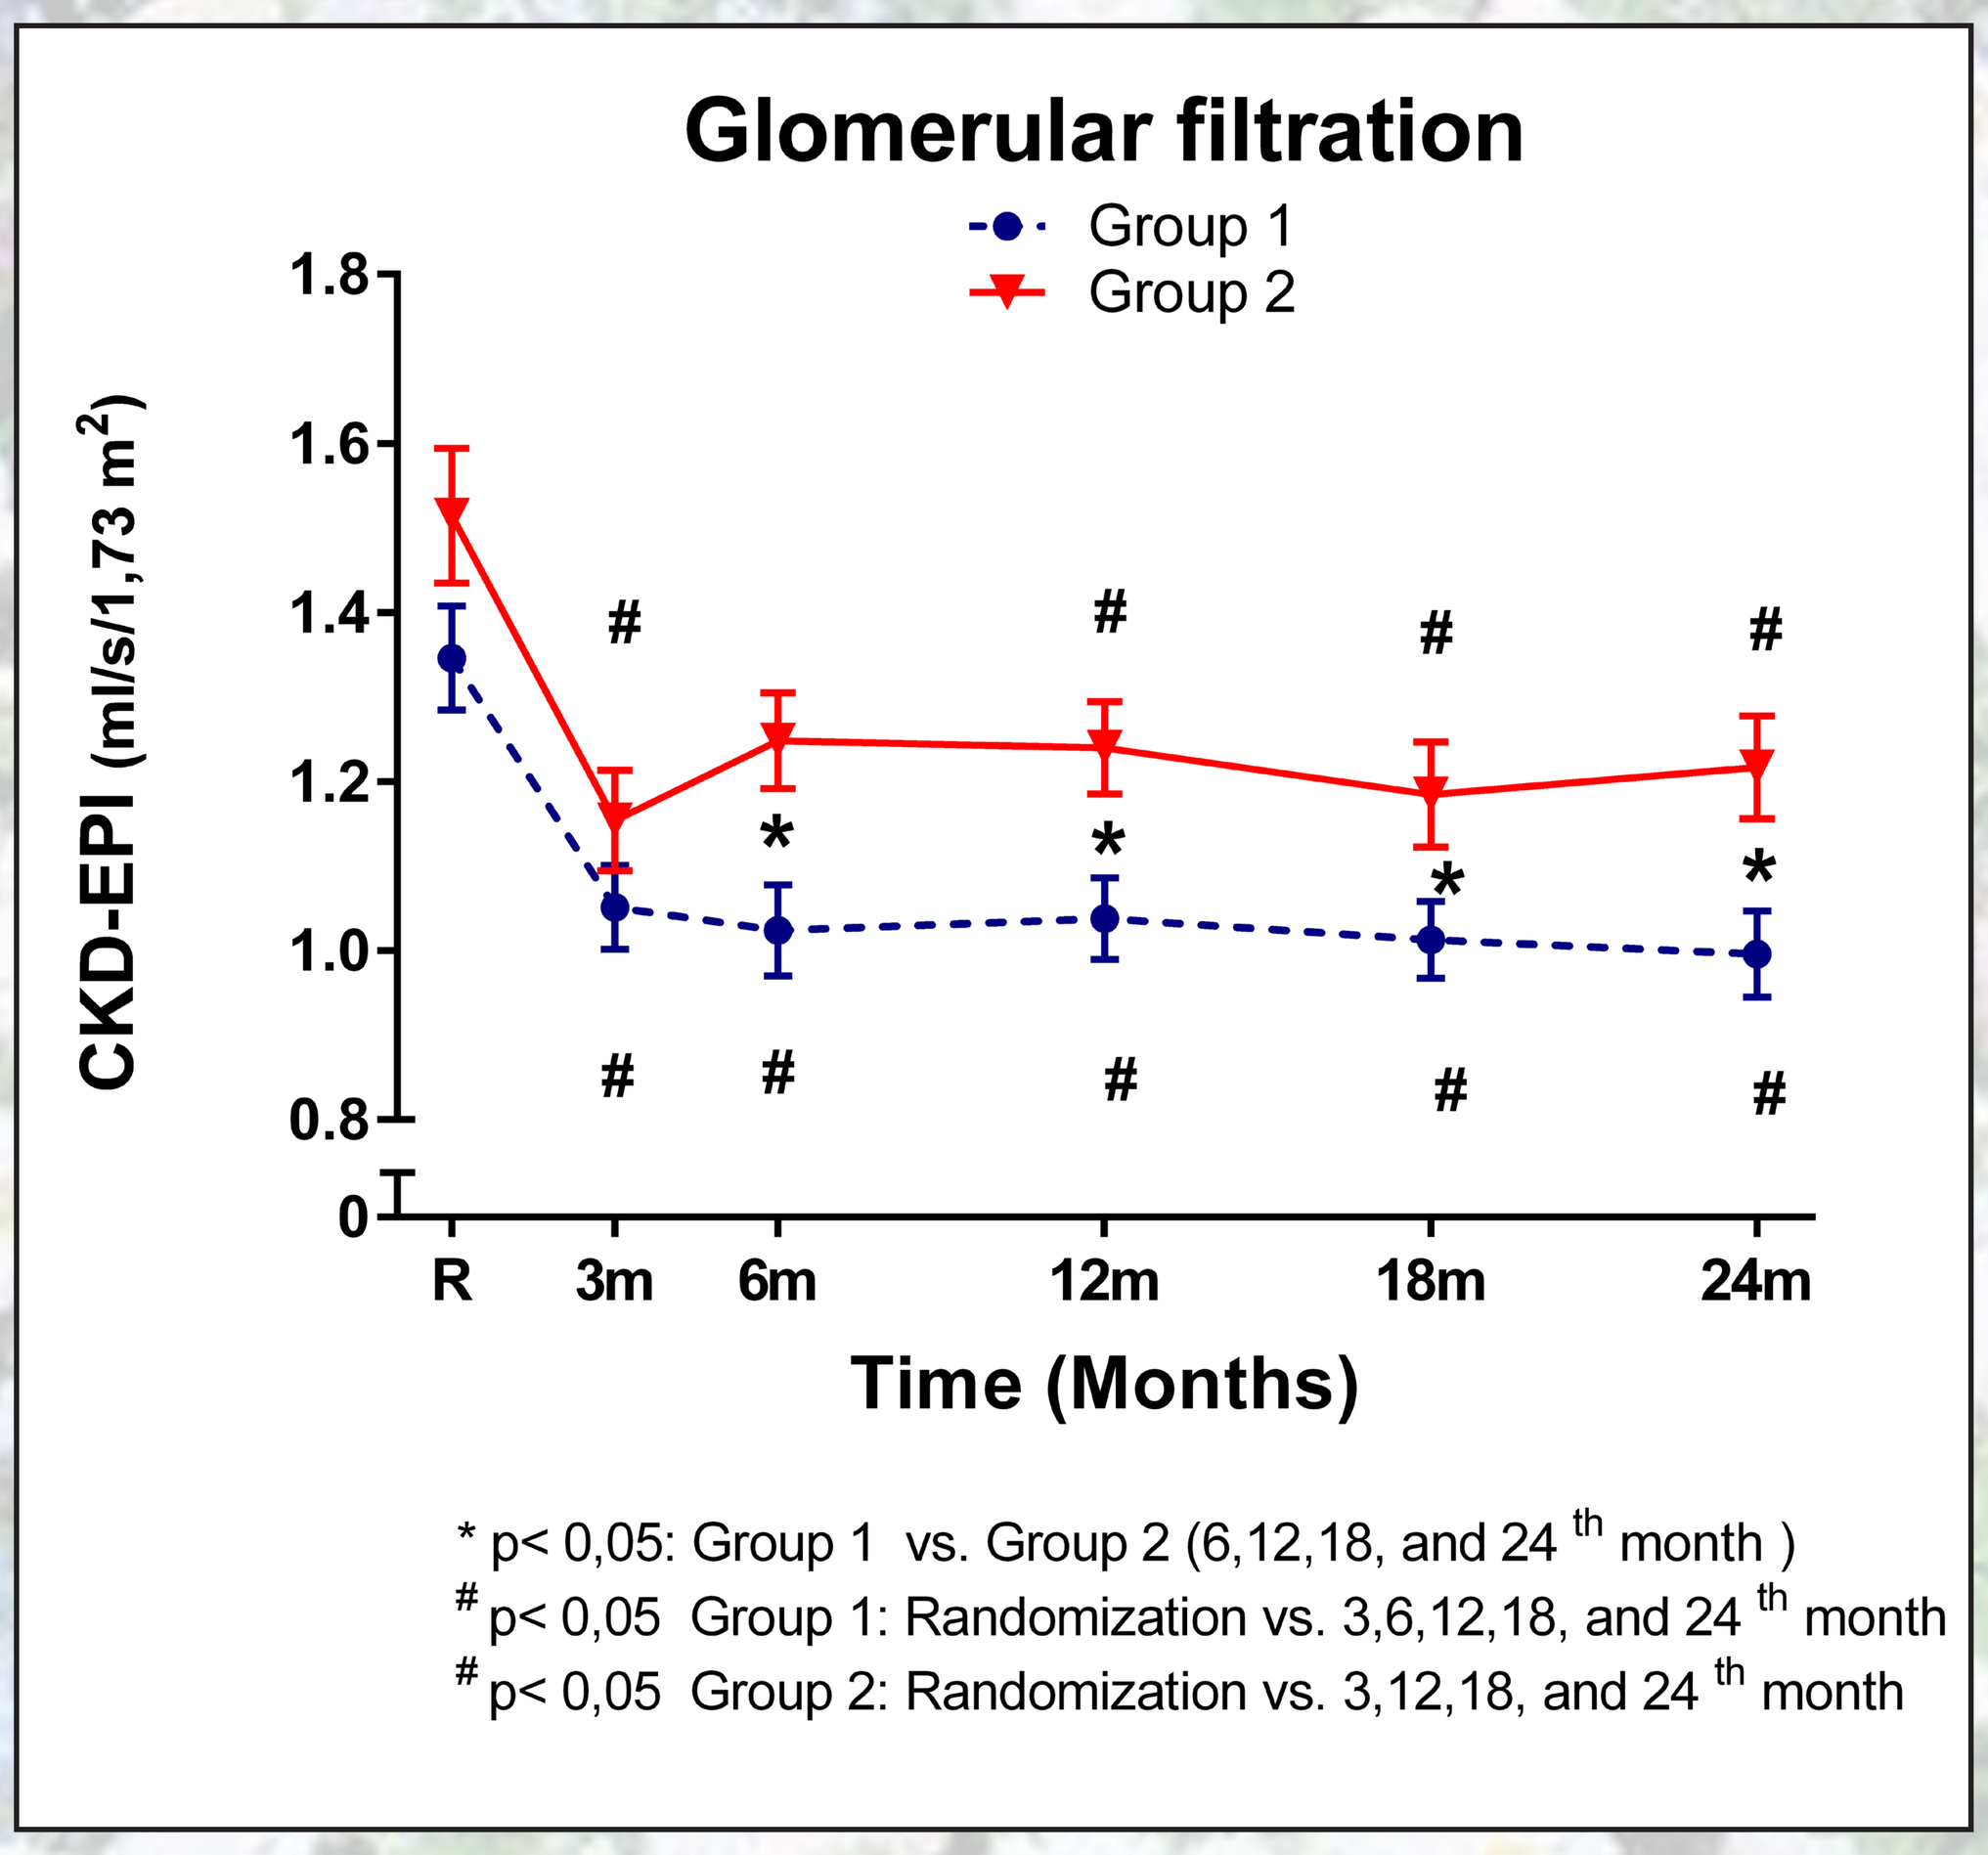
Glomerular filtration
Group 1
Group 2
CKD-EPI (ml/s/1,73 m2)
Time (Months)
R 3m 6m 12m 18m 24m
* p< 0,05: Group 1 vs. Group 2 (6,12,18, and 24 th month)
p< 0,05 Group 1: Randomization vs. 3,6,12,18, and 24 th month
p< 0,05 Group 2: Randomization vs. 3,12,18, and 24 th month

Fig 4: Glomerular filtration
Glomerular filtration was calculated with CKD-EPI formula. CKD-EPI (The Chronic Kidney Disease Epidemiology Collaboration).

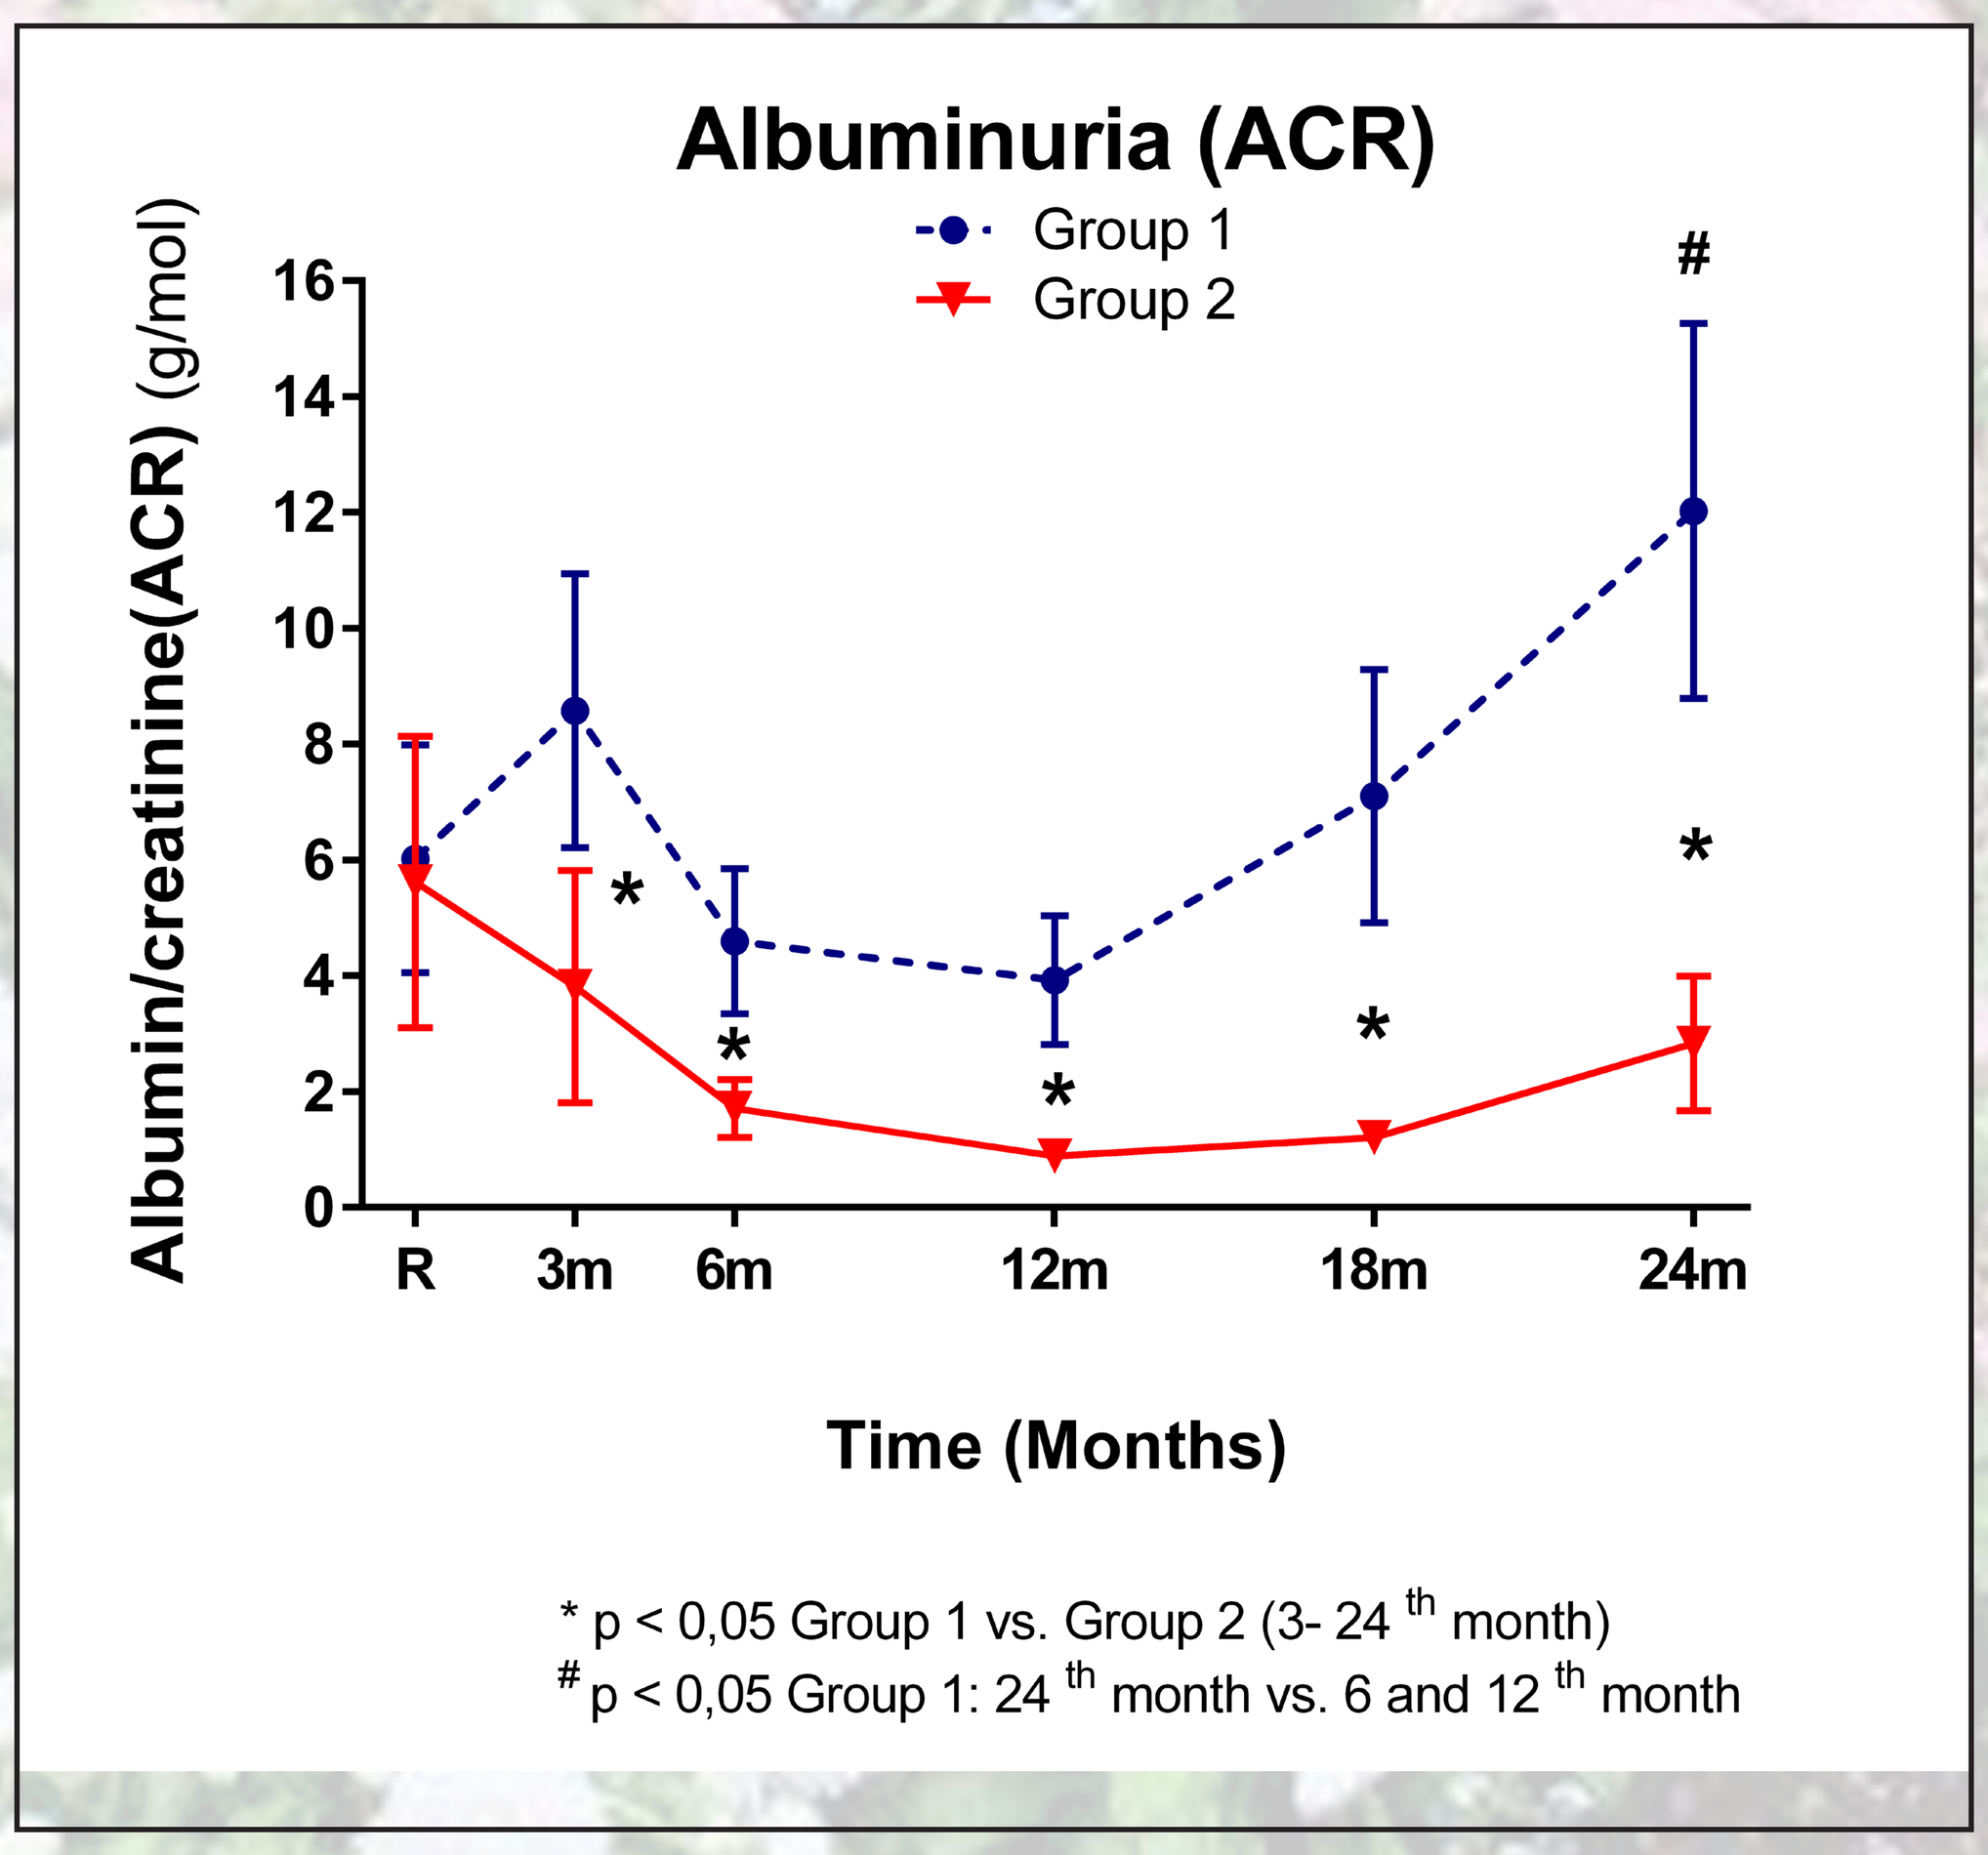
Albuminuria (ACR)
Group 1
Group 2
Albumin/creatinine(ACR) (g/mol)
Time (Months)
R 3m 6m 12m 18m 24m
* p < 0,05 Group 1 vs. Group 2 (3- 24 th month)
p < 0,05 Group 1: 24 th month vs. 6 and 12 th month

Fig. 5. Albuminuria (ACR)
ACR was calculated as albumin to creatinine ratio.

Conclusion:

Blood pressure was effectively controlled in both treatment groups. However combination of aHTM with dual RAS blockade significantly reduced albuminuria and maintained of glomerular filtration rate. This study indicated that dual RAS blockade might be more renoprotective in some HTx patients.

Disclosure Statement: Presenting author and co-authors have no relationships to disclosure.

Supported by Ministry of Health, Czech Republic - conceptual development of research organization (Institute for Clinical and Experimental Medicine – IKEM, IN 00023001).