Usefulness of Near-Infrared Reflectance Spectroscopy to Assess Lower-Limb Perfusion on Femoral Veno-arterial Extracorporeal Membrane Oxygenation

717

Christine Chan, MS, CCP, Killian Patton-Rivera, MS, CCP, James Beck, BS, CCP, Kenmond Fung, MS, CCP, Garan Reshad, MD, Jiho Han, BS, Masahiko Ando, MD, Marisa Cevasco, MD, Paolo C. Colombo MD, Yoshifumi Naka, MD, Hiroo Takayama, MD, PhD, Koji Takeda, MD, PhD
Columbia University Medical Center, New York, NY, USA 10032

Objectives

The use of veno-arterial extracorporeal membrane oxygenation (V-A ECMO) has been on the rise to support patients with severe cardiopulmonary failure. Patients requiring V-A ECMO who receive femoral cannulation have an associated risk of lower-limb ischemia. Lower-limb ischemia can be detected with physical examinations showing signs of mottled skin, cold limb, and non-dopplerable pulse, however, it is not sensitive enough. We investigated the usefulness of near-infrared reflectance spectroscopy (NIRS) to detect lower-limb ischemia.

Methods

Between June 2016 and October 2017, 42 patients receiving femoral V-A ECMO were continuously monitored using the NIRS tissue oximeter. NIRS oximetry pads were placed on the patient’s left and right calf to monitor lower-limb tissue saturations. We evaluated the correlations between NIRS tissue saturations (StO2) and clinical indications of limb ischemia.

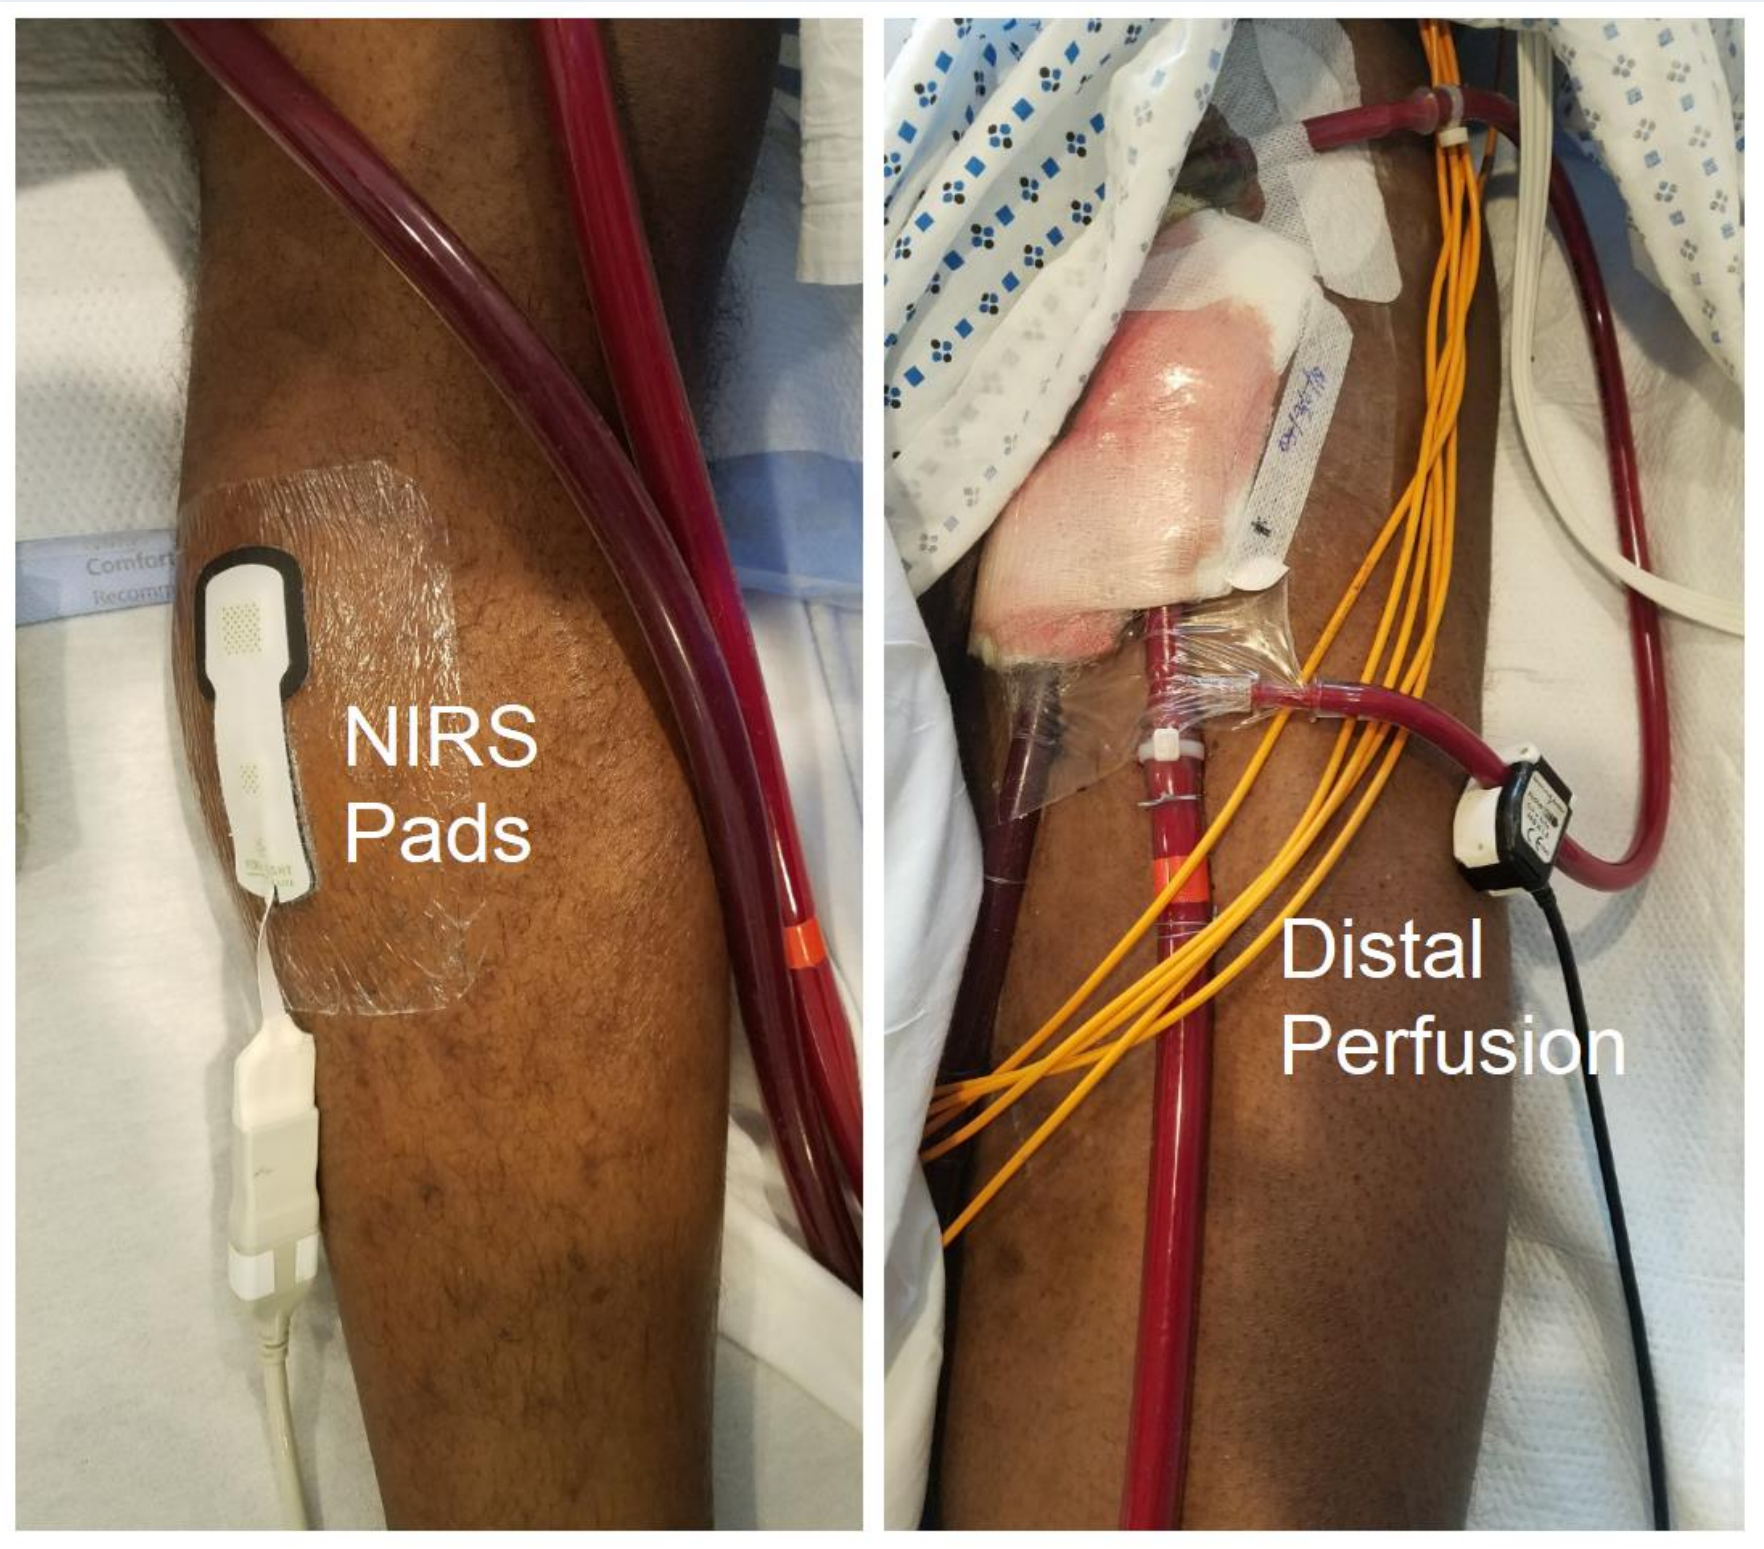
NIRS
Pads
Distal
Perfusion

Results

8 patients (19%) had StO2 below 50% that persisted for longer than four minutes, and all 8 patients had clinical indications of lower-limb ischemia: cold limb, mottled skin, and pulseless Doppler ultrasound. After interventions, all patients’ StO2 increased above 50% and clinical signs of lower-limb ischemia resolved. The StO2 difference between the cannulated leg and contralateral leg was 26% for patients with lower-limb ischemia, and 5% for patients without lower-limb ischemia (p<0.0001).

All authors have no conflicts of interest to disclose

Patient Demographics

Characteristics	n=42
Gender	
Male	24 (57%)
Female	18 (43%)
Age	55 ± 12 (22-77)
BSA	2.02 ± 0.29 (1.42-2.69)
Days on ECMO	9 ± 10 (1-65)
ECMO Flow (LPM)	3.3 ± 0.7 (2.0-4.7)

Primary Indication	n=42
Cardiogenic Shock	15 (36%)
Acute Cardiopulmonary Decompensation	3 (7%)
Pulmonary Hypertension/Respiratory Insufficiency	7 (17%)
Postcardiotomy Shock	7 (17%)
Primary Graft Failure - Cardiac	7 (17%)
Primary Graft Failure - Pulmonary	3 (7%)

Cannula Sizes

Arterial	
14 fr	1 (2.5%)
15 fr	27 (64%)
17 fr	11 (26%)
18 fr	1 (2.5%)
19 fr	1 (2.5%)
23 fr	1 (2.5%)
Venous	
19 fr	4 (10%)
21 fr	5 (12%)
23 fr	30 (71%)
24 fr	1 (2.5%)
25 fr	1 (2.5%)
27 fr	1 (2.5%)
Distal Perfusion	
none	15 (36%)
6 fr	2 (5%)
8 fr	25 (59%)

Diagnostic Test Evaluation

	Value	95% CI
Sensitivity	100%	63.1% to 100%
Specificity	97.1%	84.7 to 99.93%
PPV	88.9%	53.7% to 98.2%
NPV	100%	
Accuracy	97.60%	87.4% to 99.9%

Conclusions

Advancements in NIRS technology have improved its accuracy and can provide clinician with an additional criterion to detect lower-limb ischemia of the cannulated leg for patients on femoral V-A ECMO with continuous, non-invasive monitoring.

V-A ECMO with/without Distal Perfusion & Other Interventions

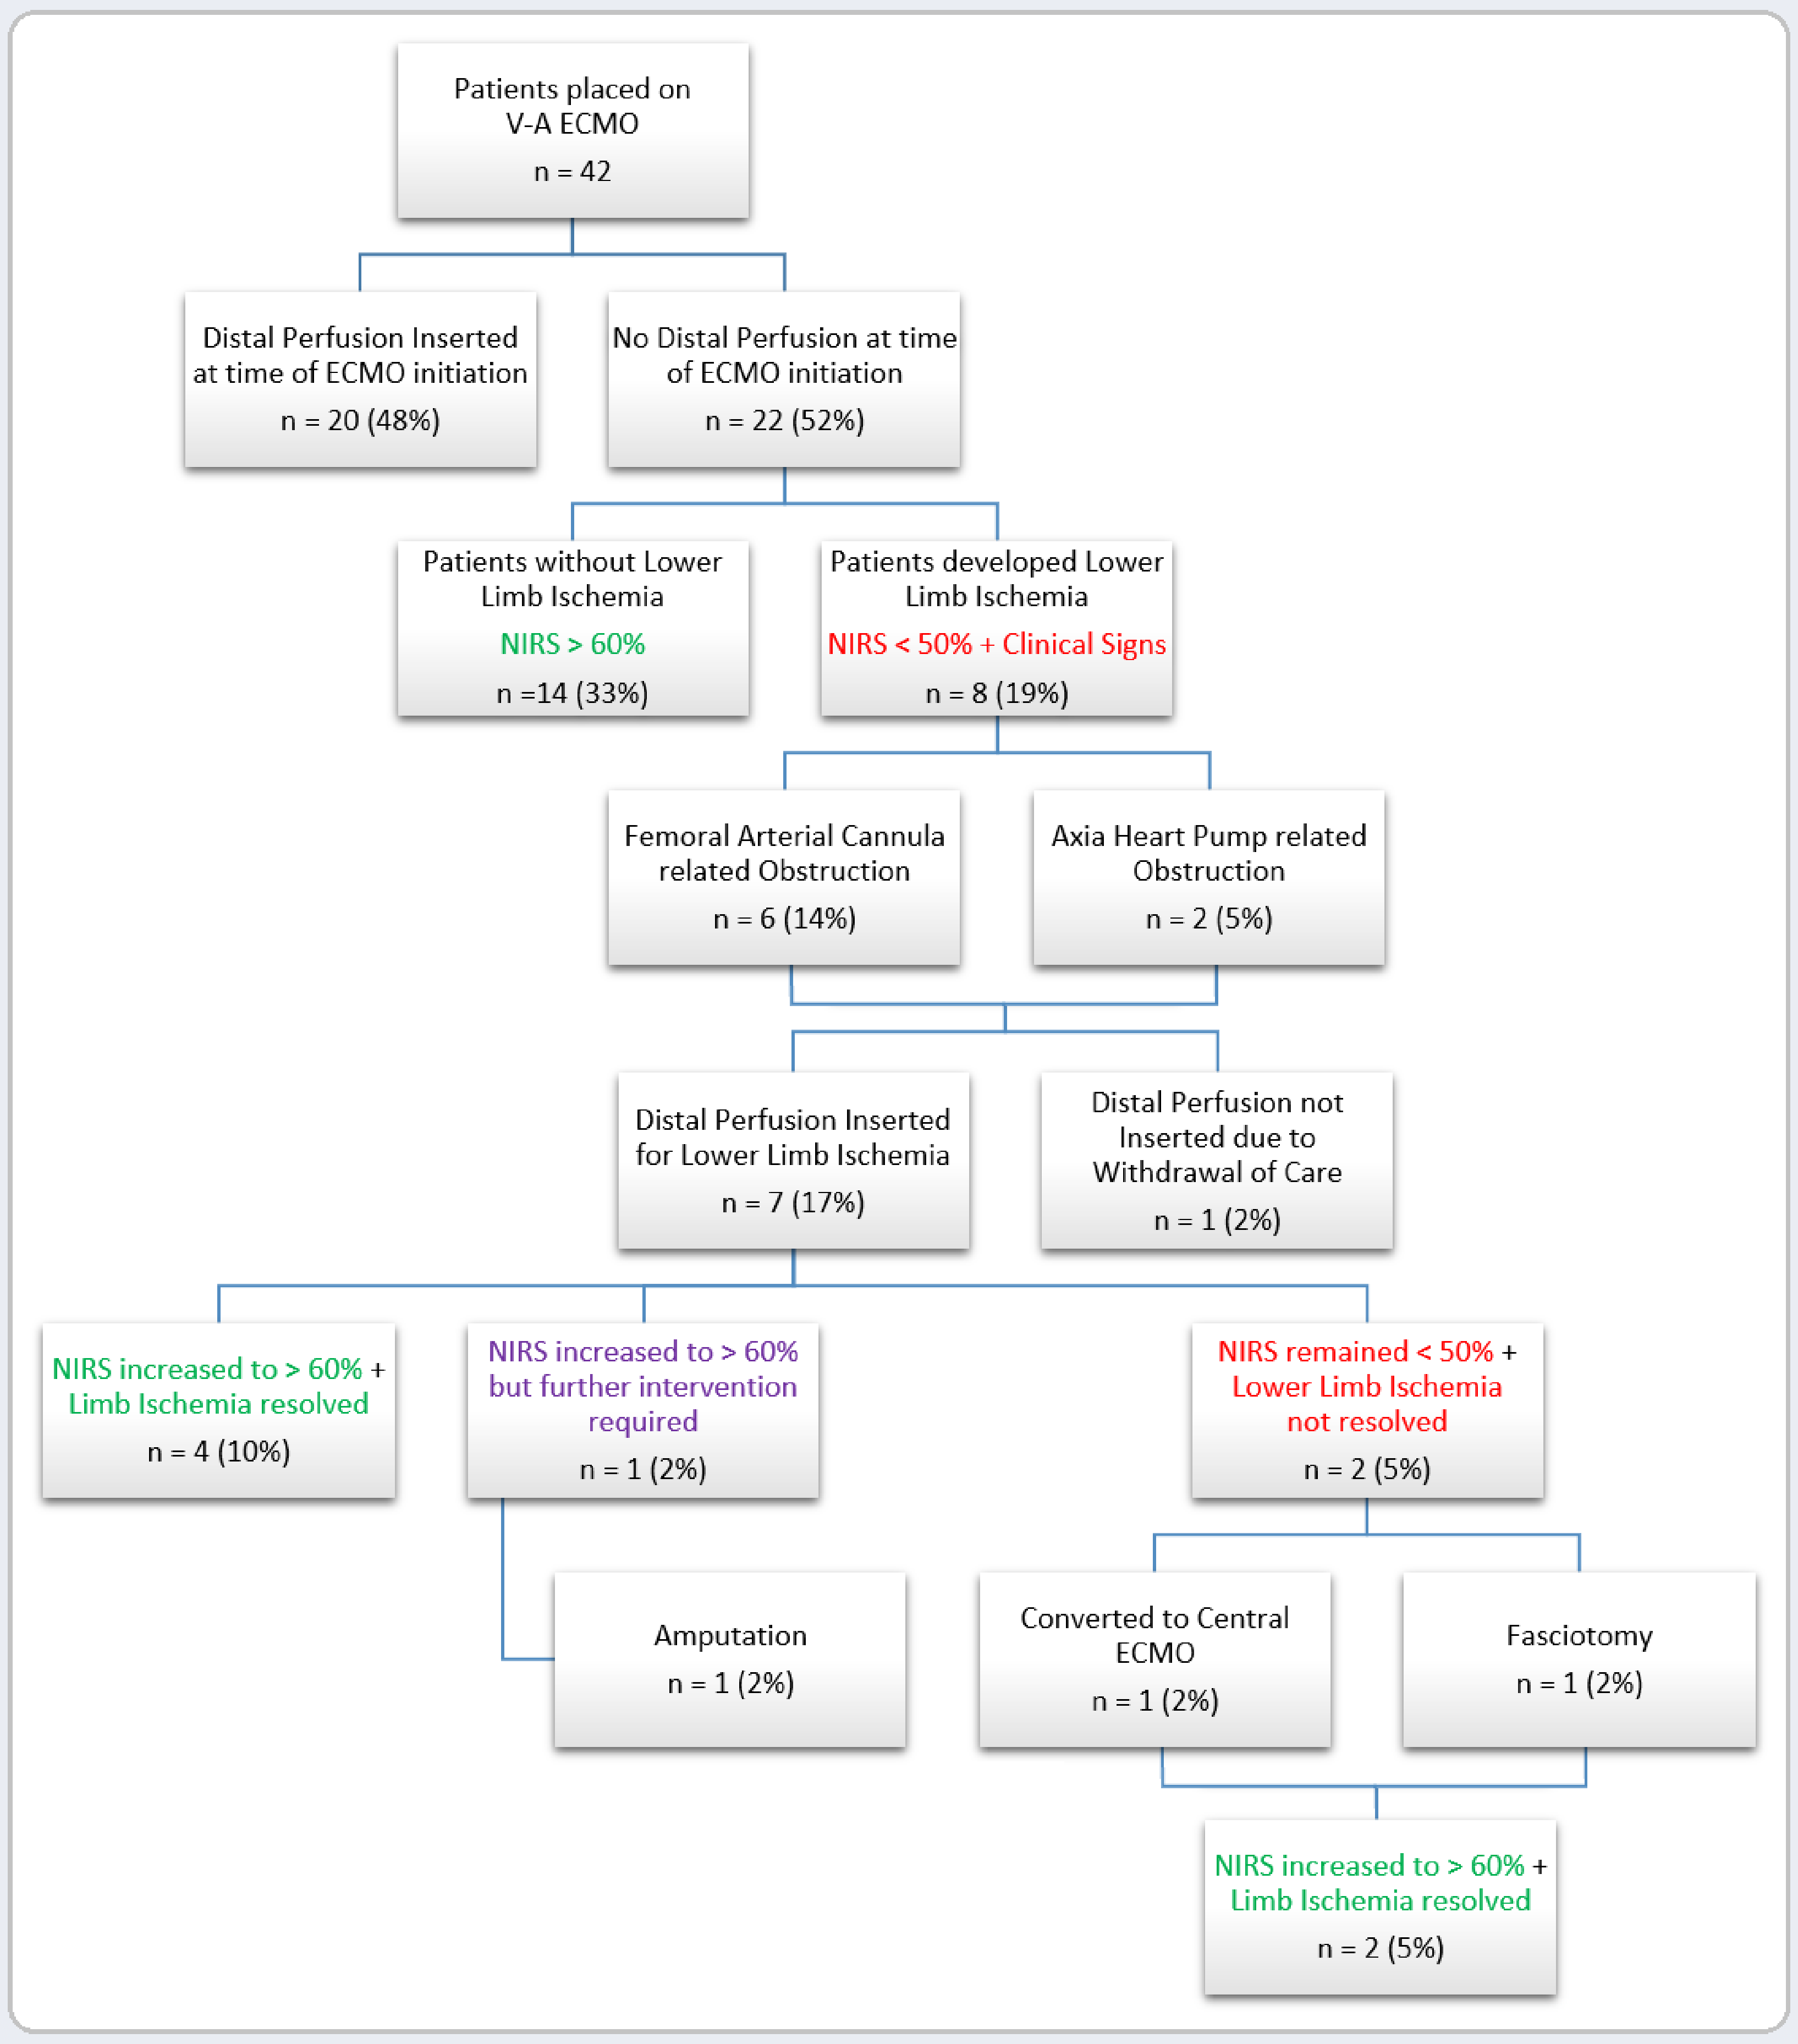
Patients placed on V-A ECMO
n = 42
Distal Perfusion Inserted at time of ECMO initiation
n = 20 (48%)
No Distal Perfusion at time of ECMO initiation
n = 22 (52%)
Patients without Lower Limb Ischemia
NIRS > 60%
n =14 (33%)
Patients developed Lower Limb Ischemia
NIRS < 50% + Clinical Signs
n = 8 (19%)
Femoral Arterial Cannula related Obstruction
n = 6 (14%)
Axia Heart Pump related Obstruction
n = 2 (5%)
Distal Perfusion Inserted for Lower Limb Ischemia
n = 7 (17%)
Distal Perfusion not Inserted due to Withdrawal of Care
n = 1 (2%)
NIRS increased to > 60% + Limb Ischemia resolved
n = 4 (10%)
NIRS increased to > 60% but further intervention required
n = 1 (2%)
NIRS remained < 50% + Lower Limb Ischemia not resolved
n = 2 (5%)
Amputation
n = 1 (2%)
Converted to Central ECMO
n = 1 (2%)
Fasciotomy
n = 1 (2%)
NIRS increased to > 60% + Limb Ischemia resolved
n = 2 (5%)

NIRS Saturation Ipsilateral vs Contralateral Leg

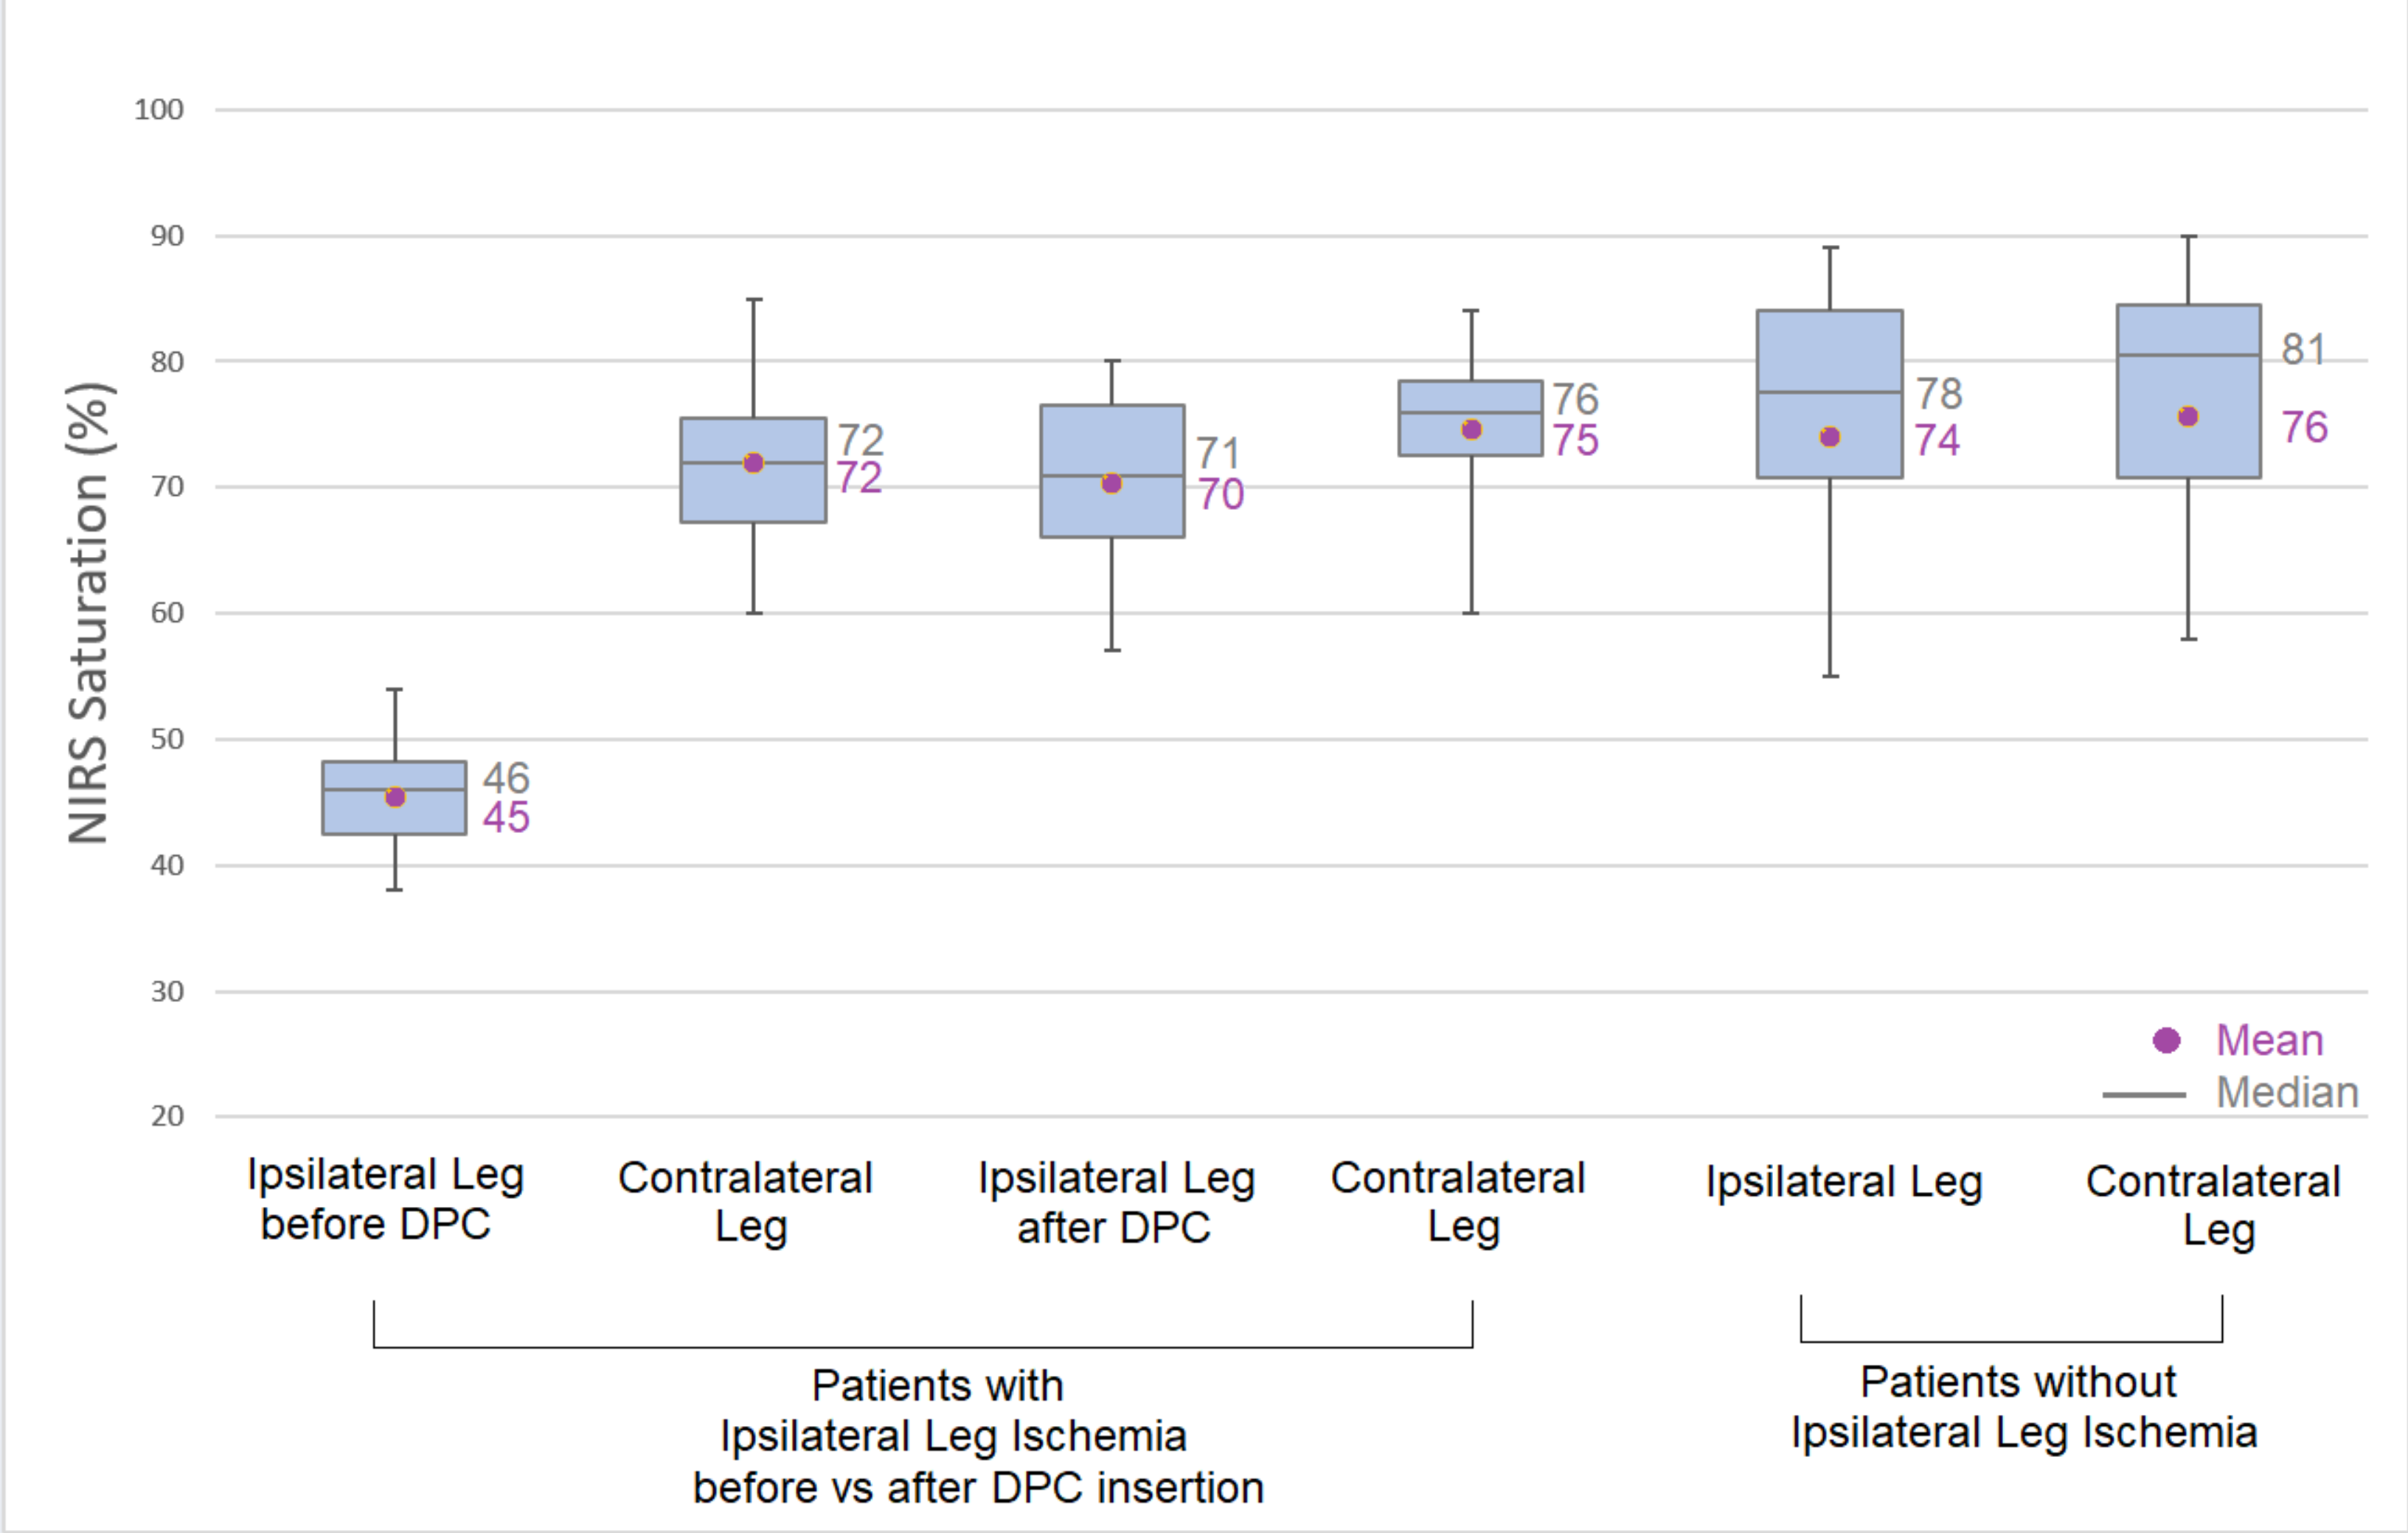
NIRS Saturation (%)
100
90
80
70
60
50
40
30
20
46
45
72
72
71
70
76
75
78
74
81
76
Mean
Median
Ipsilateral Leg before DPC
Contralateral Leg
Ipsilateral Leg after DPC
Contralateral Leg
Ipsilateral Leg
Contralateral Leg
Patients with Ipsilateral Leg Ischemia before vs after DPC insertion
Patients without Ipsilateral Leg Ischemia

NIRS Difference between Ipsilateral & Contralateral Leg

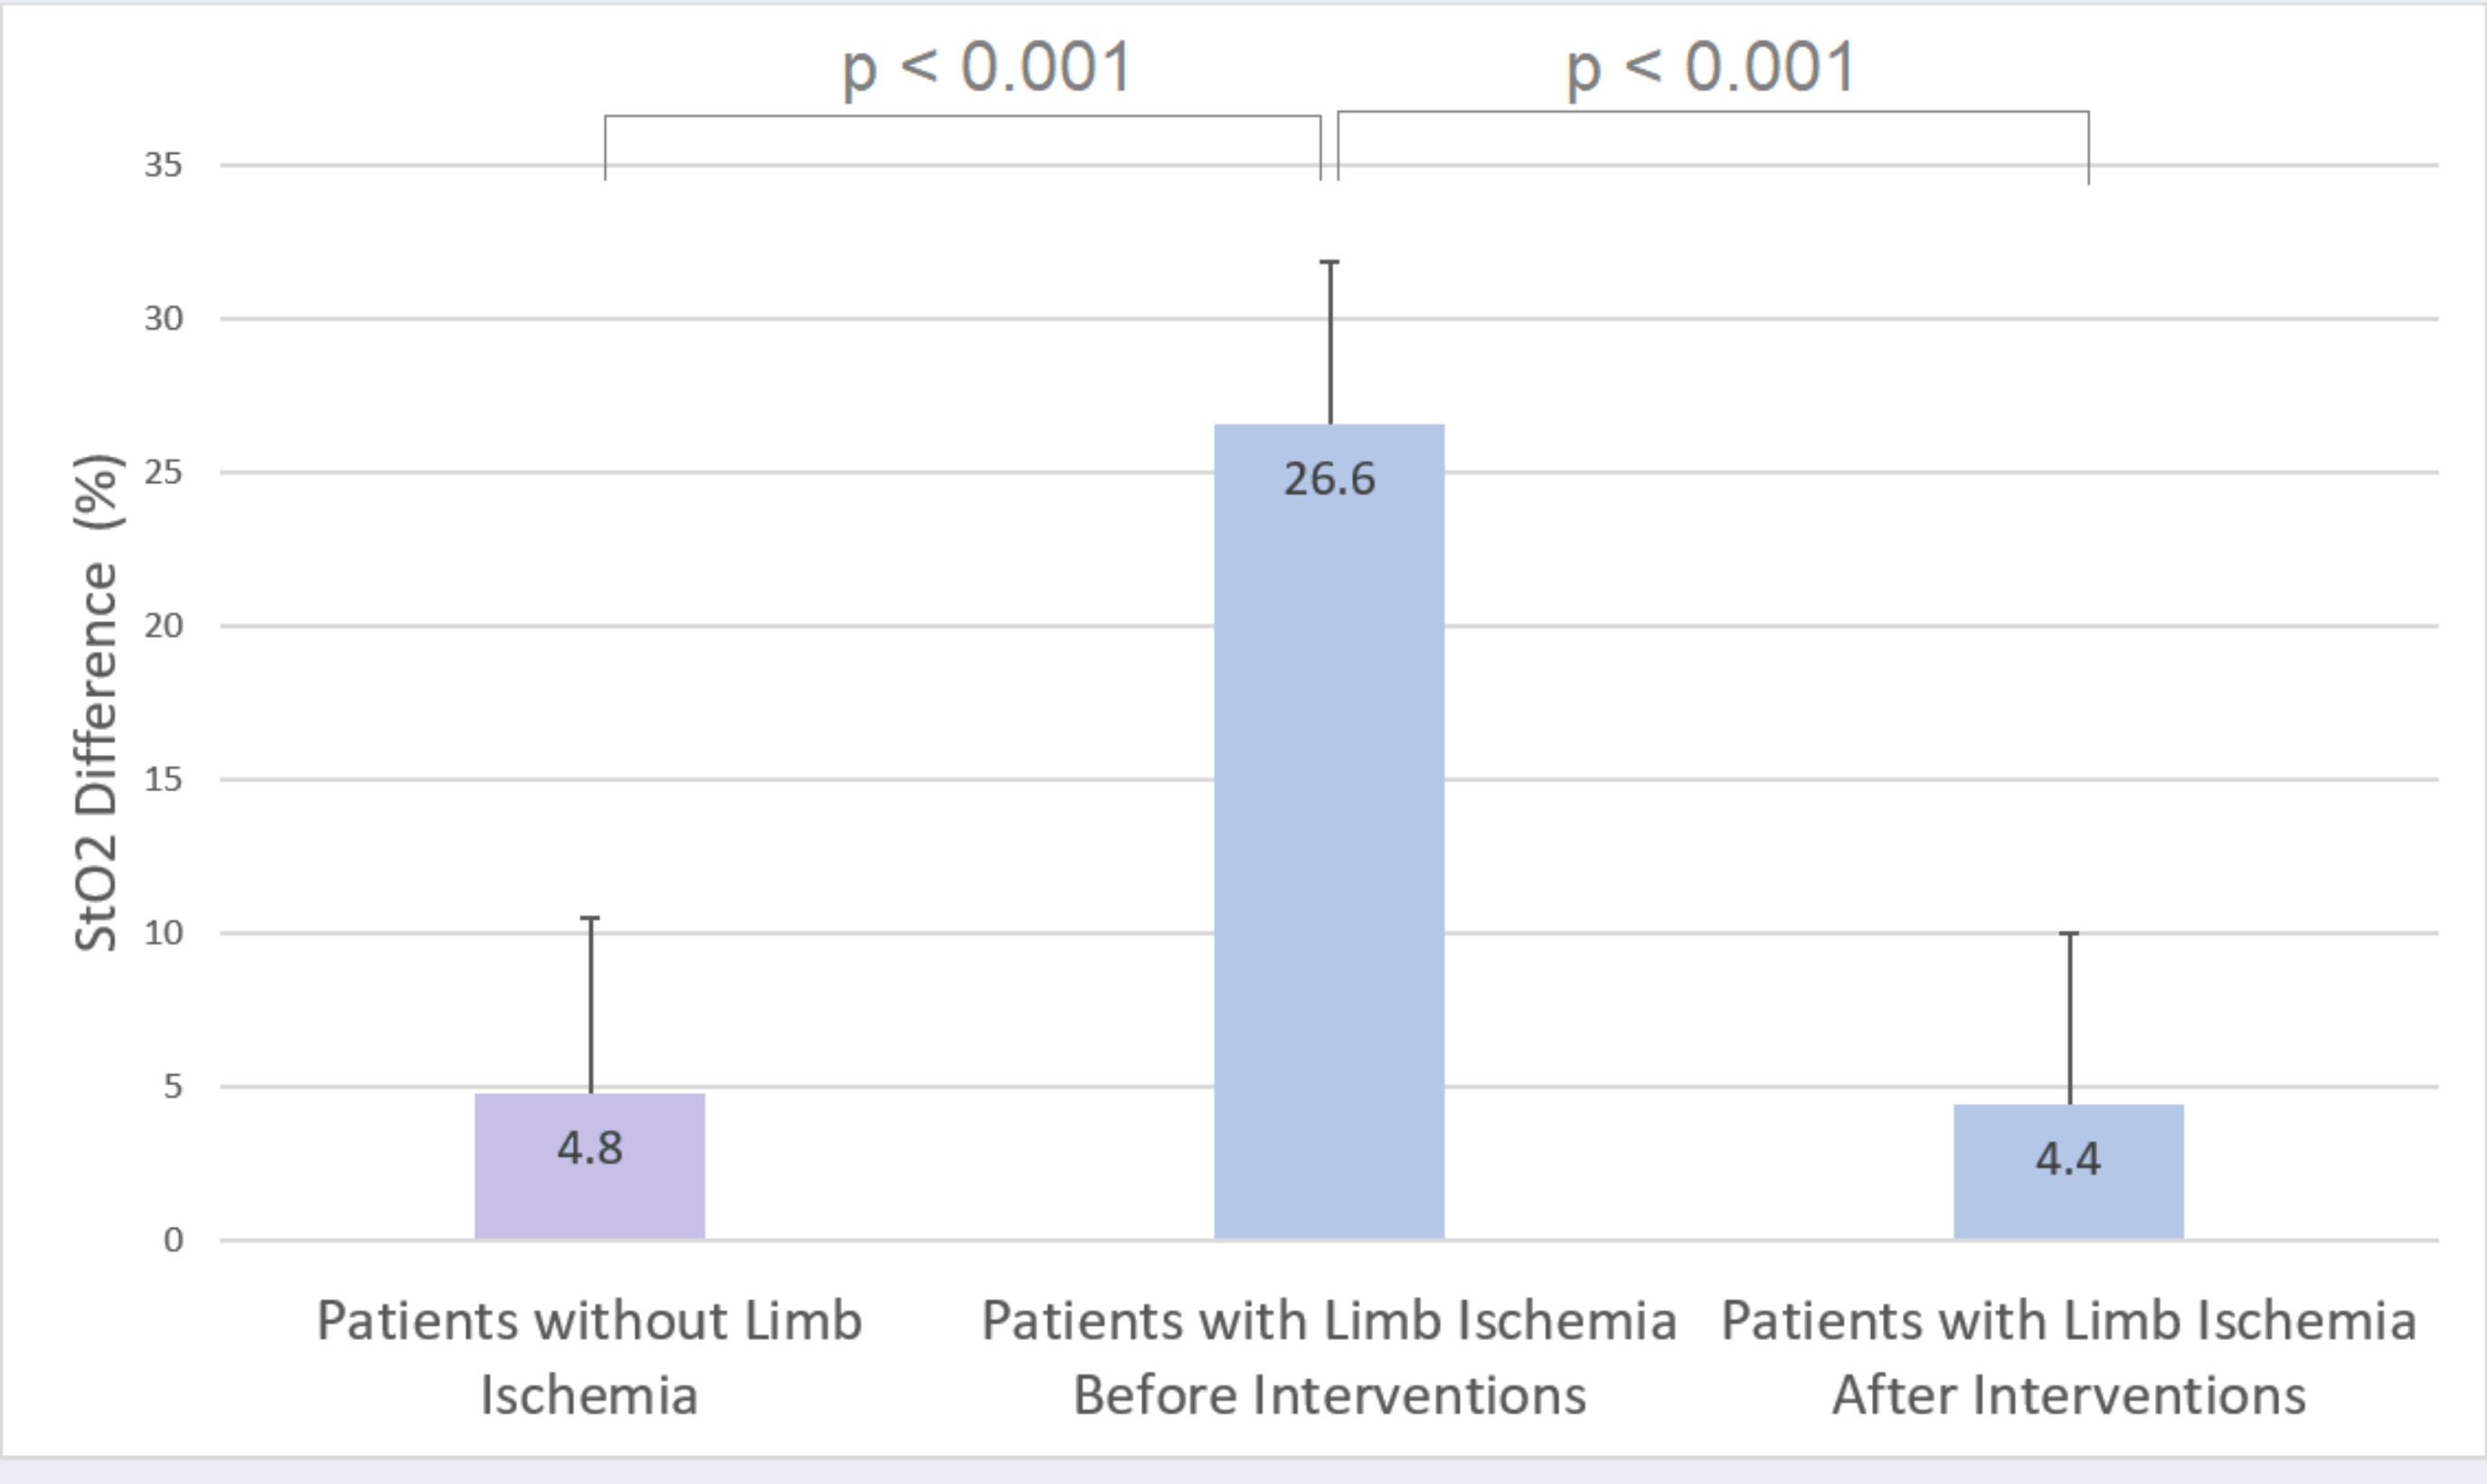
p < 0.001
p < 0.001
StO2 Difference (%)
35
30
25
20
15
10
5
0
4.8
26.6
4.4
Patients without Limb Ischemia
Patients with Limb Ischemia Before Interventions
Patients with Limb Ischemia After Interventions