Novel animal model of primary graft dysfunction in lung transplantation induced by histone administration.

Keita Nakao, MD[1], Masaki Anraku, MD, MSc, PhD[1], Tomonori Murayama, MD[1], Mitsuaki Kawashima, MD, PhD[1], Akihiro Hosoi, PhD[2], Jun Nakajima, MD, PhD[1]

[1] Department of Thoracic Surgery, Graduate School of Medicine, The University of Tokyo, Tokyo, Japan

[2] Department of Immunotherapeutics, The University of Tokyo Hospital, Tokyo, Japan

ISHLT 2018, Nice, France, 11st April, 2018

Backgrounds and Hypotheses

- It has been known that intranuclear histones are released into systemic circulation in various medical conditions caused by massive tissue damage, such as stroke or trauma[1].
- Recent studies have revealed circulating histone induced systemic inflammation and thrombosis and cause multiple organ injury[2,3,4,5].
- We hypothesized that a histone-associated lung injury caused in the process of brain deaths of donors is one of risk factors of primary graft dysfunction (PGD) after lung transplantation.
- In this study, we attempted to make novel PGD animal model in rat orthotopic lung transplantation by histone administration.

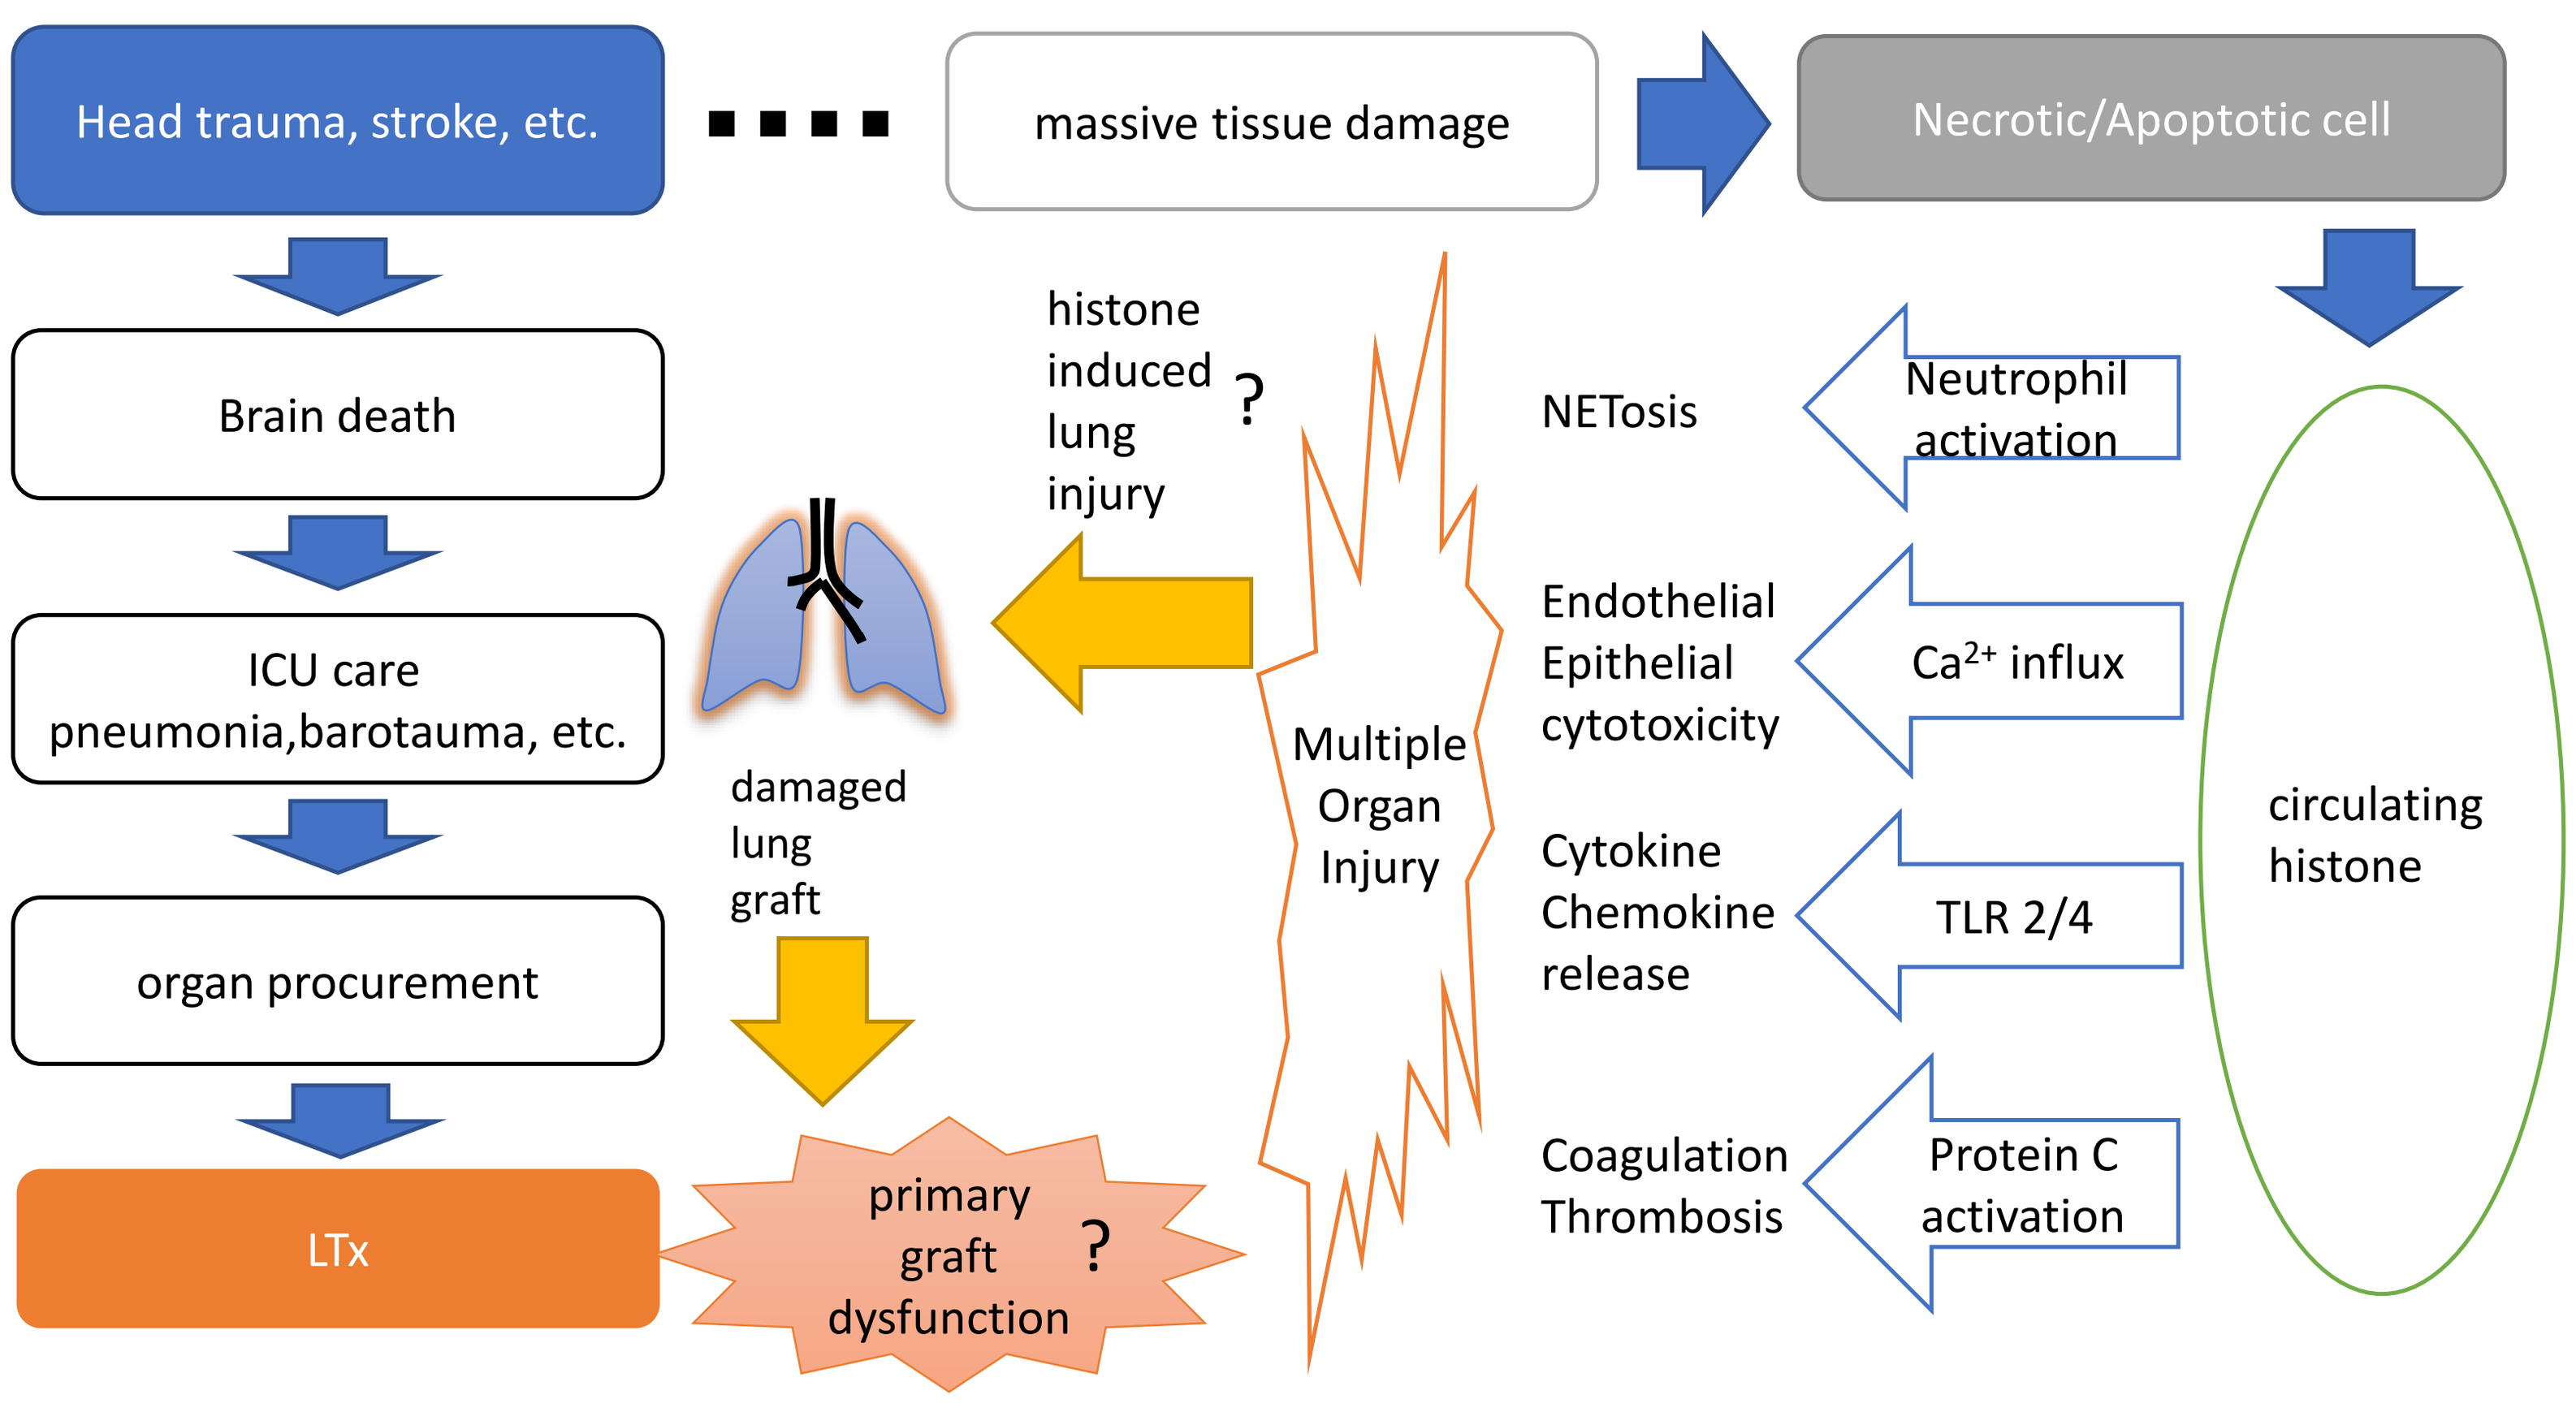
Head trauma, stroke, etc.
massive tissue damage
Necrotic/Apoptotic cell
Brain death
ICU care
pneumonia,barotauma, etc.
organ procurement
LTx
histone induced lung injury ?
damaged lung graft
primary graft dysfunction ?
Multiple Organ Injury
NETosis
Neutrophil activation
Endothelial Epithelial cytotoxicity
Ca2+ influx
Cytokine Chemokine release
TLR 2/4
Coagulation Thrombosis
Protein C activation
circulating histone

Material and Methods

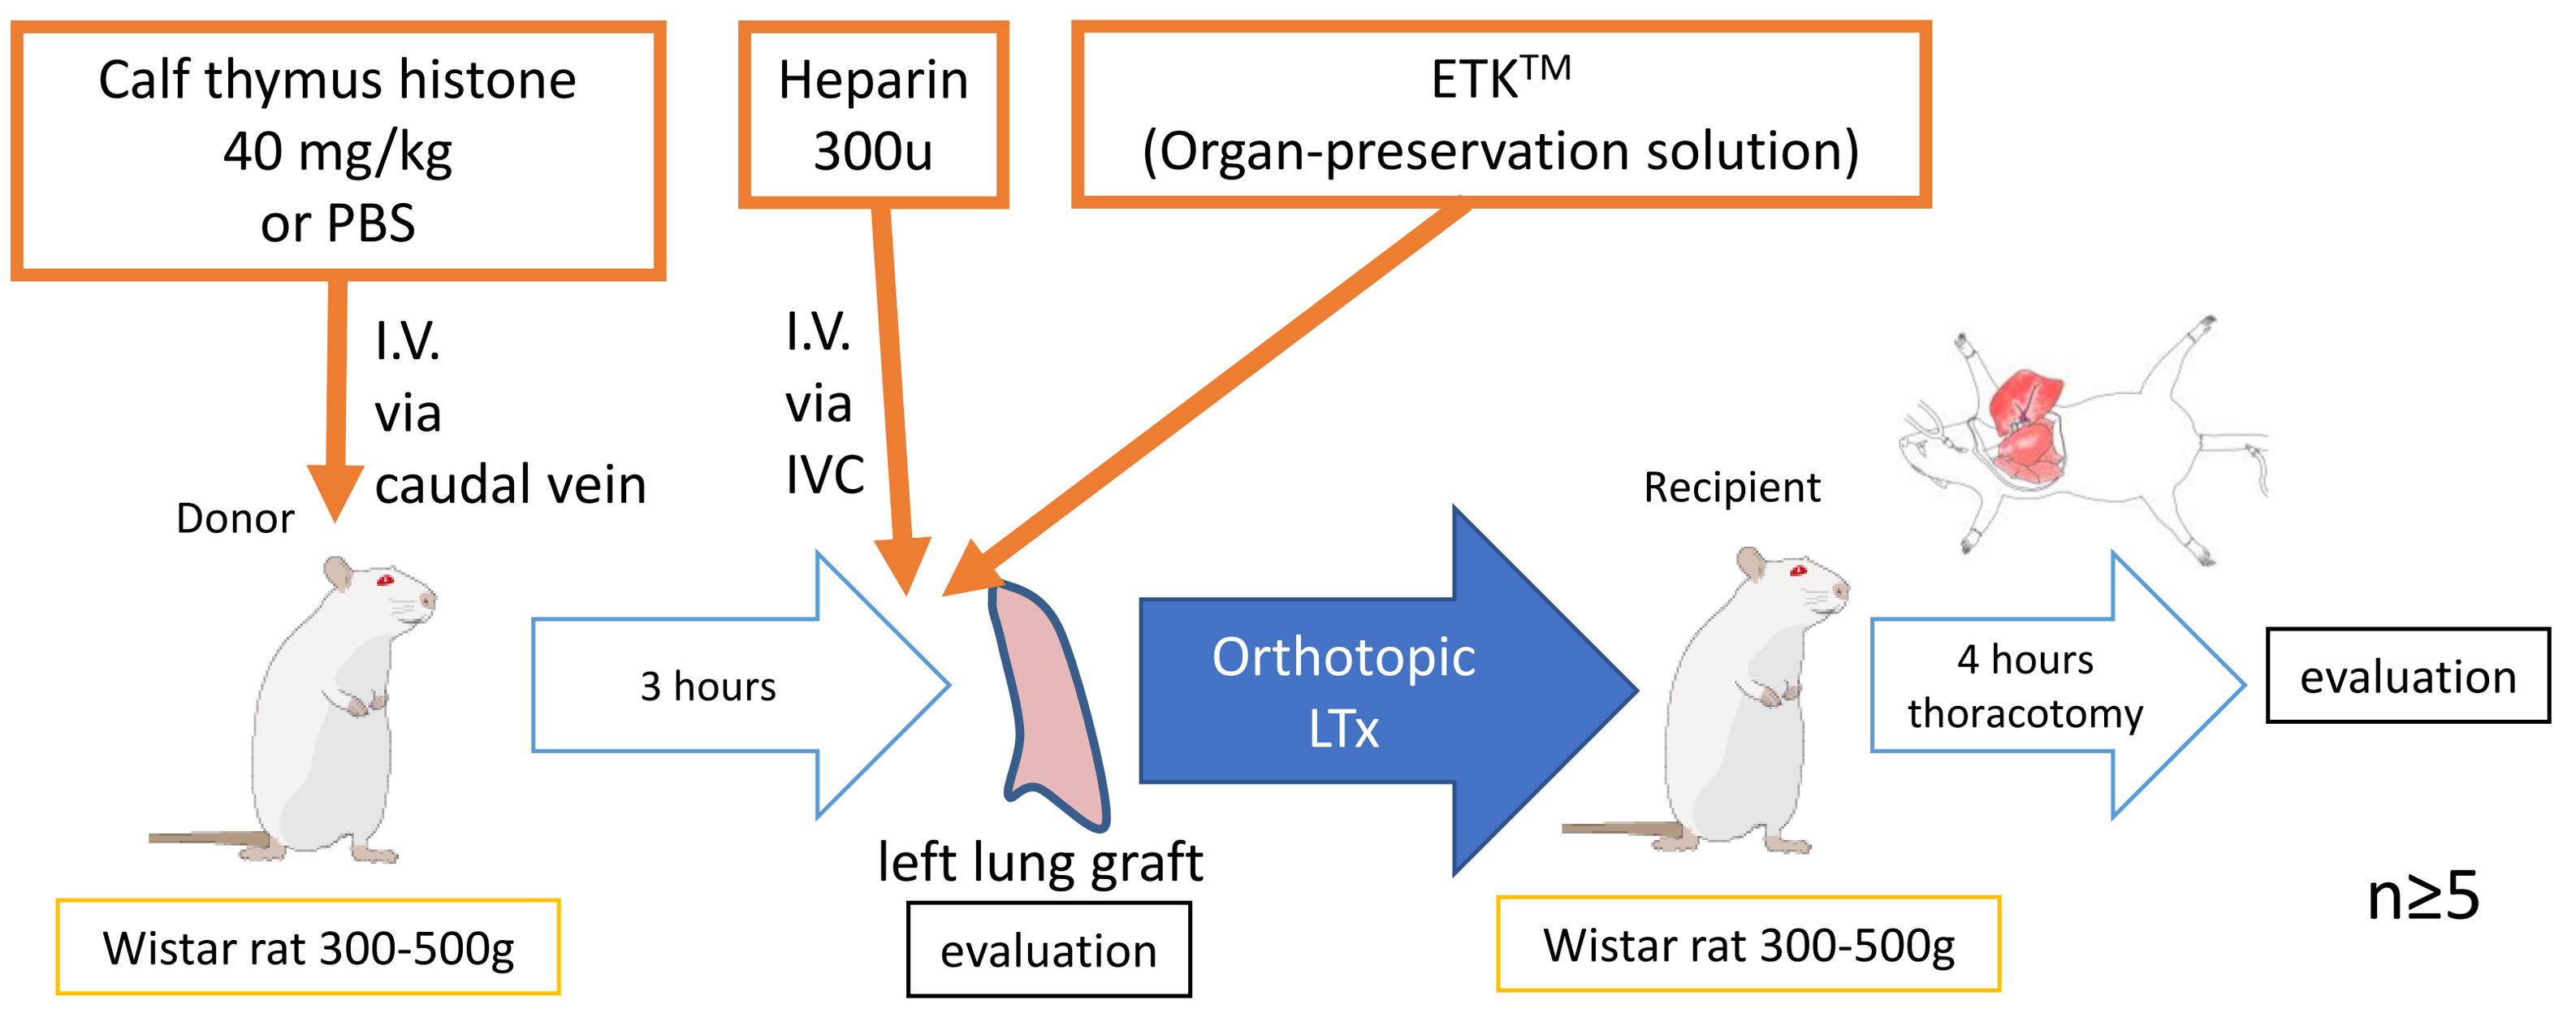
Calf thymus histone 40 mg/kg or PBS
Heparin 300u
ETKTM (Organ-preservation solution)
I.V. via caudal vein
I.V. via IVC
Donor
3 hours
left lung graft
evaluation
Orthotopic LTx
Recipient
4 hours thoracotomy
evaluation
Wistar rat 300-500g
Wistar rat 300-500g
n≥5

Evaluation **before** LTx 3 hours after histone injection
- Chest X-ray
- Arterial blood gas analysis from left appendage
- Real-time RT-PCR of homogenized lung tissue (CXCL1, TNF and HIF1-a/GAPDH)
- Wet to dry ratio
- Histology

Evaluation **4 hours after** LTx
- Arterial blood gas analysis every 1 hour from graft pulmonary vein
- Real-time RT-PCR of homogenized lung tissue (CXCL1, TNF and HIF1-a/GAPDH)
- X-ray
- Wet to dry ratio
- Histology
- Graft compliance

Results

BEFORE LTx

X-ray : No obvious change in lung transparency was seen.

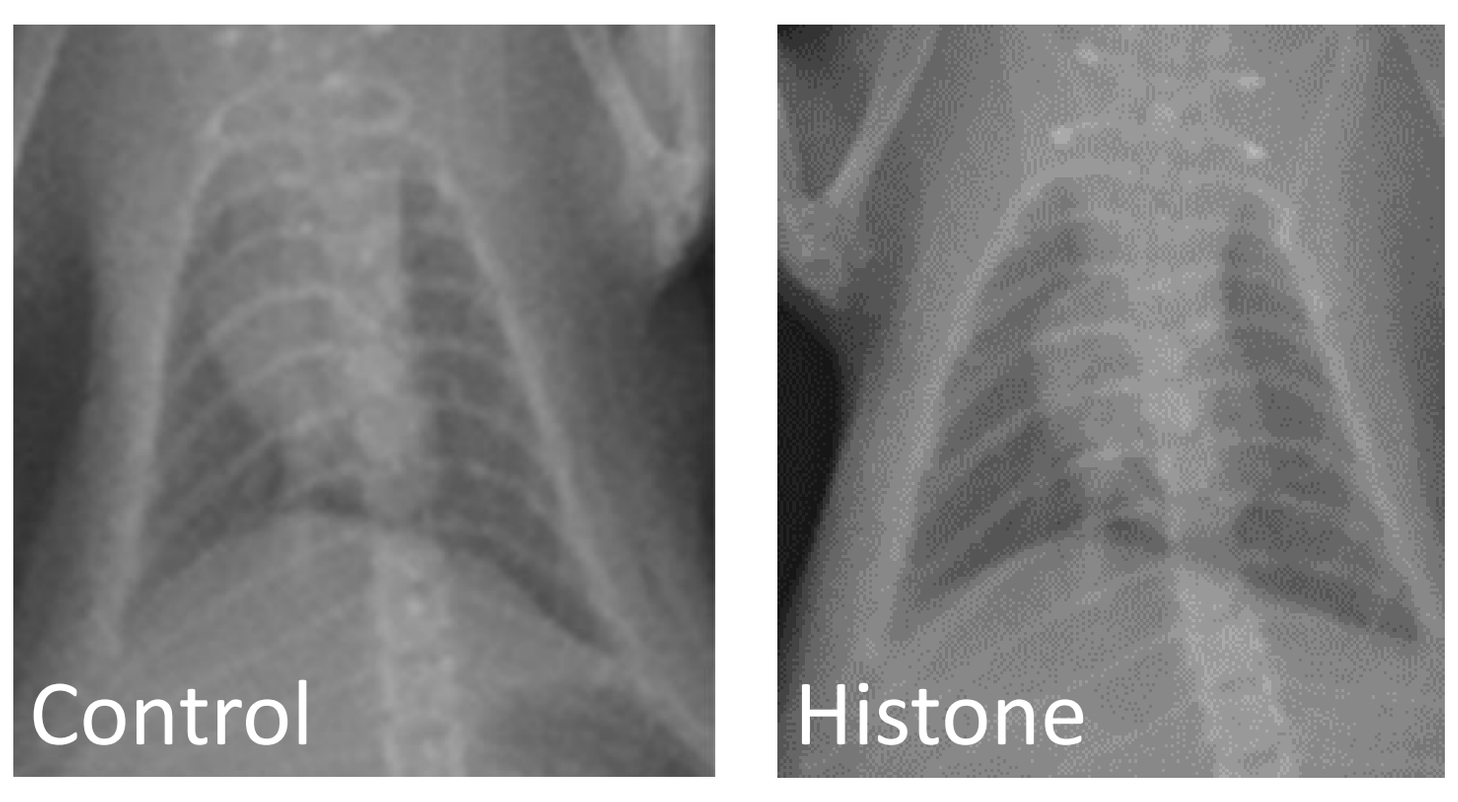
Control
Histone

ABG : There was no significant change in oxygenation.

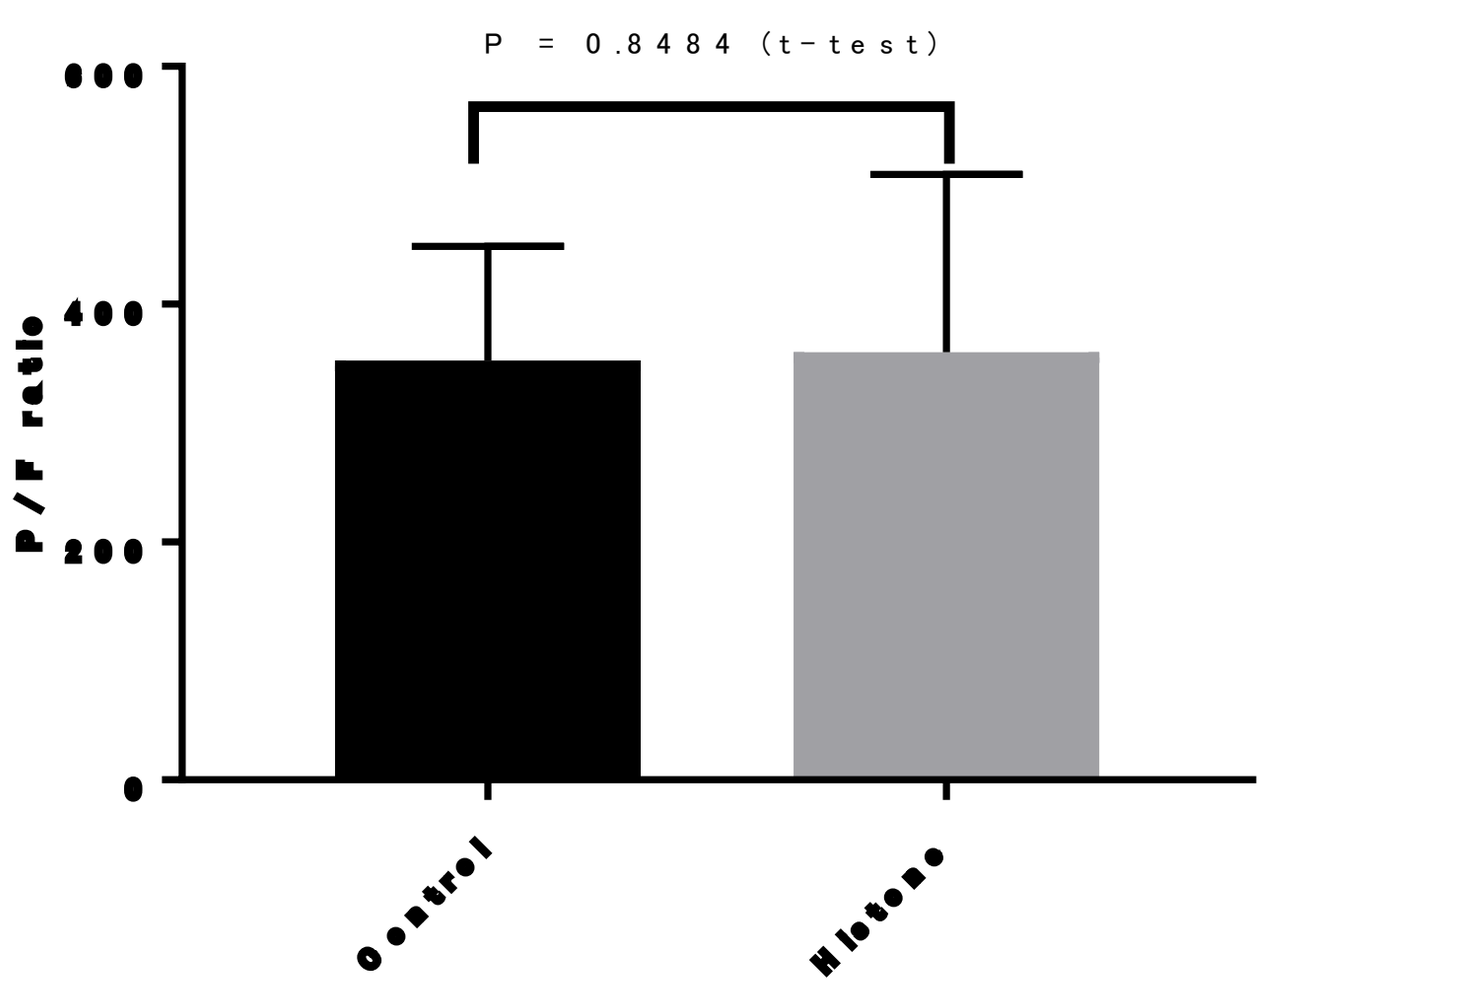
P = 0.8484 (t-test)
P/F ratio
Control
Histone

Gene expression :
Inflammatory cytokines were significantly highly expressed. Hypoxic cytokine was tend to be more expressed.

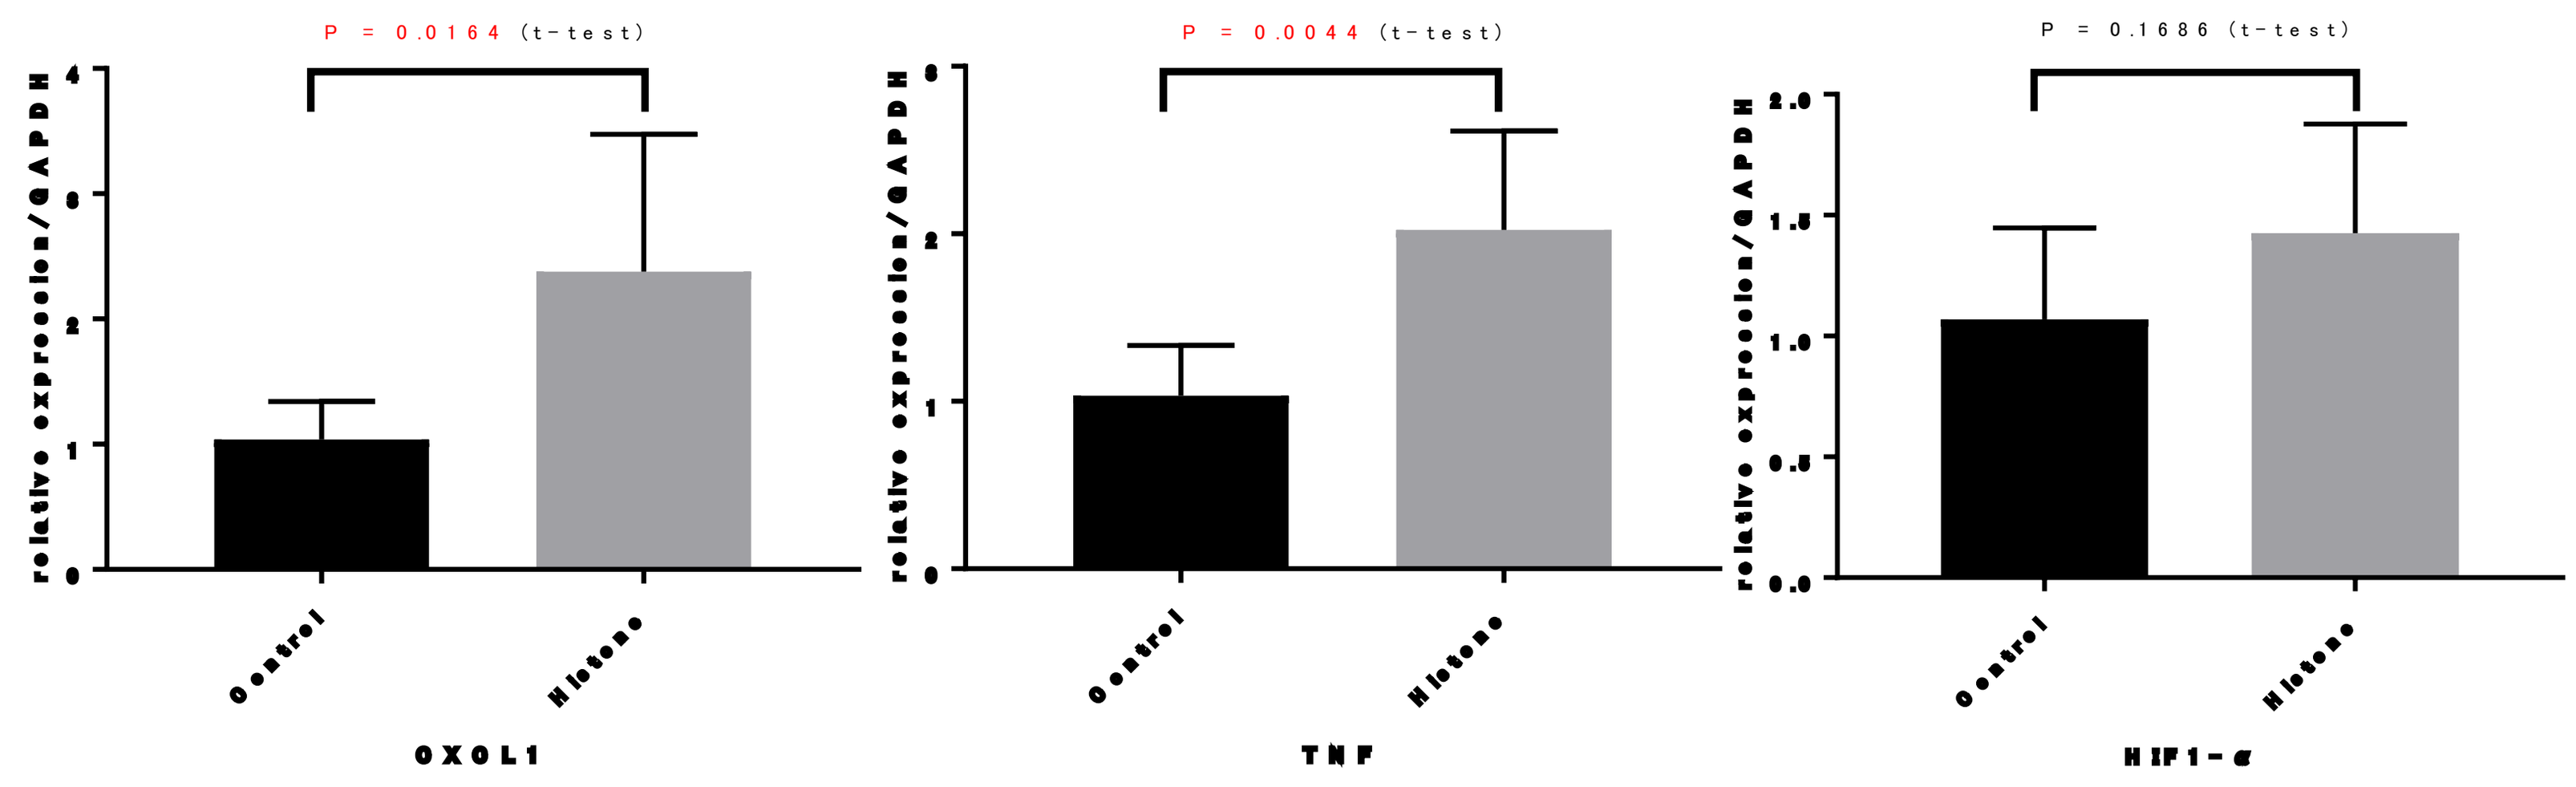
P = 0.0164 (t-test)
P = 0.0044 (t-test)
P = 0.1686 (t-test)
relative expression/GAPDH
Control
Histone
CXCL1
TNF
HIF1-α

Histology (H&E) : No obvious change was seen.

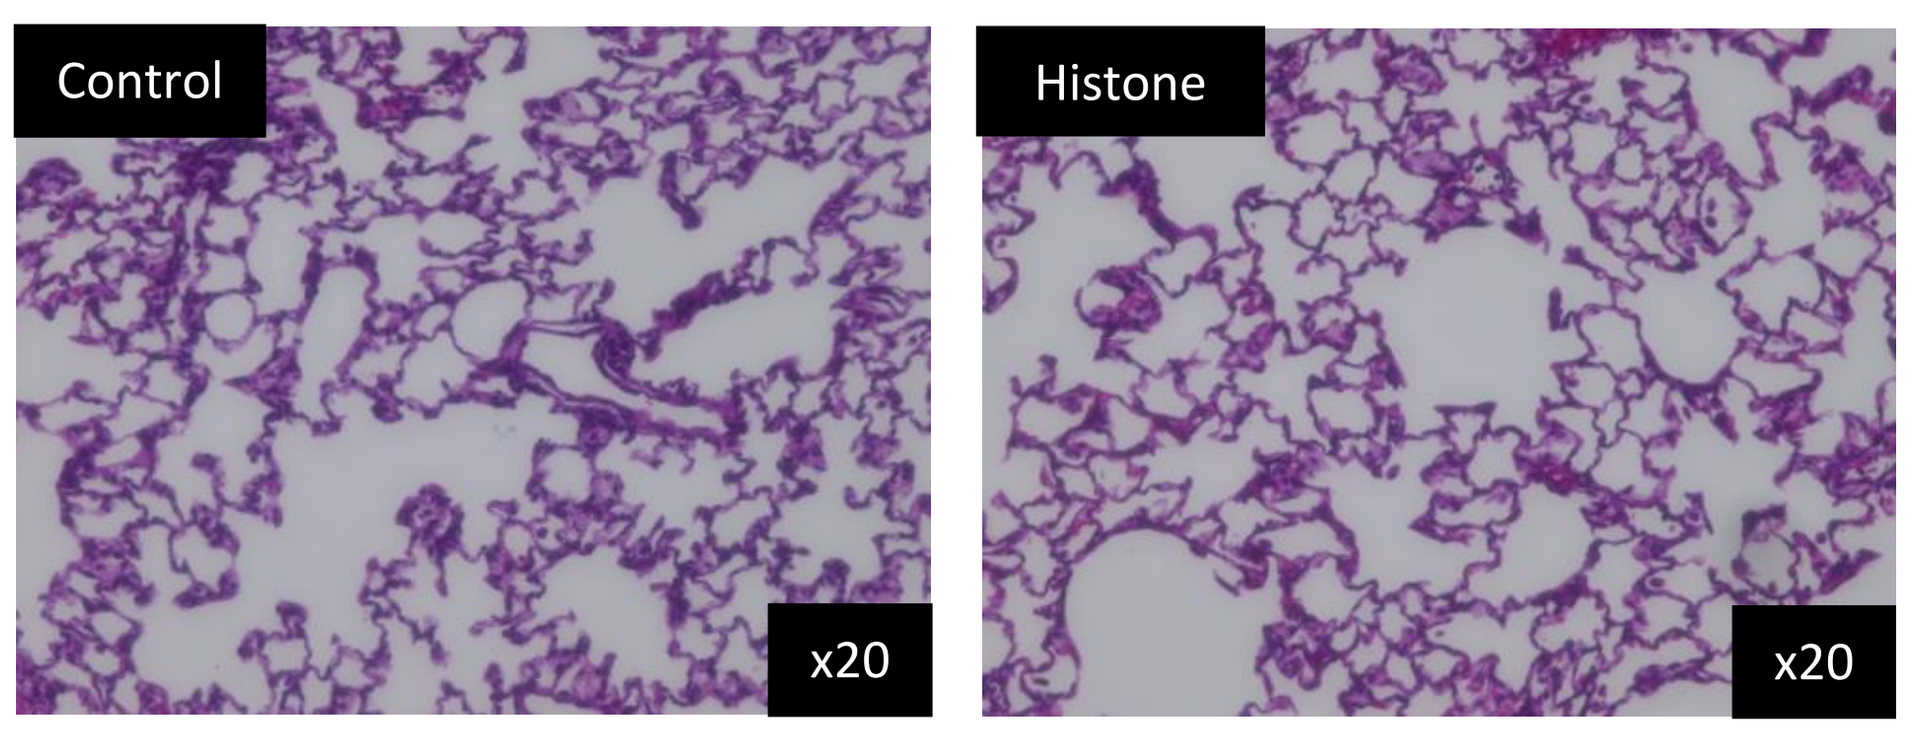
Control
Histone
x20
x20

W/D ratio :
No significant change was seen.

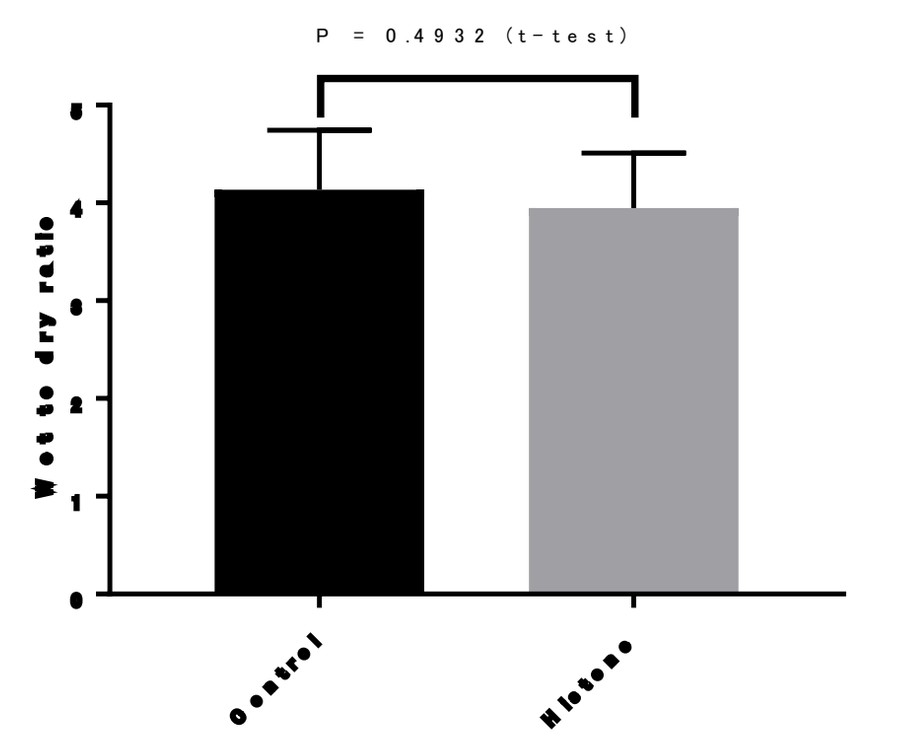
P = 0.4932 (t-test)
Wet to dry ratio
Control
Histone

AFTER LTx

Appearance : The graft was partially congested in Histone group.

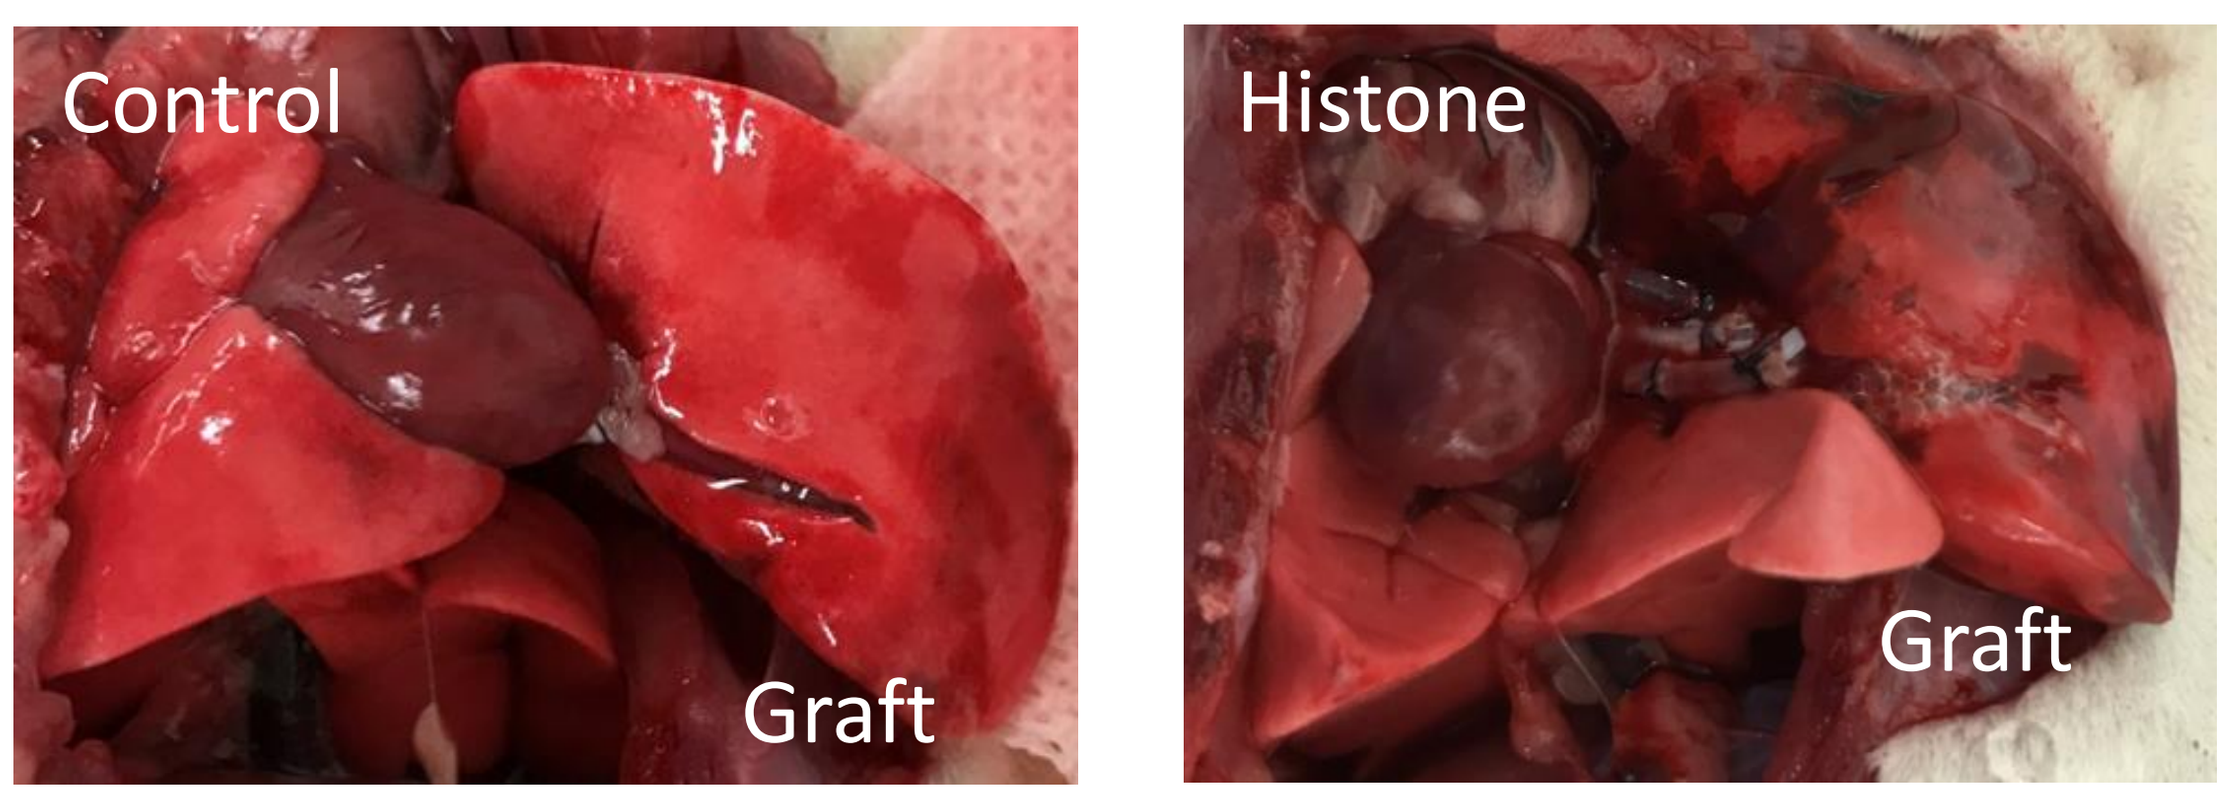
Control
Graft
Histone
Graft

X-ray : Lung transparency was decreased in Histone group.

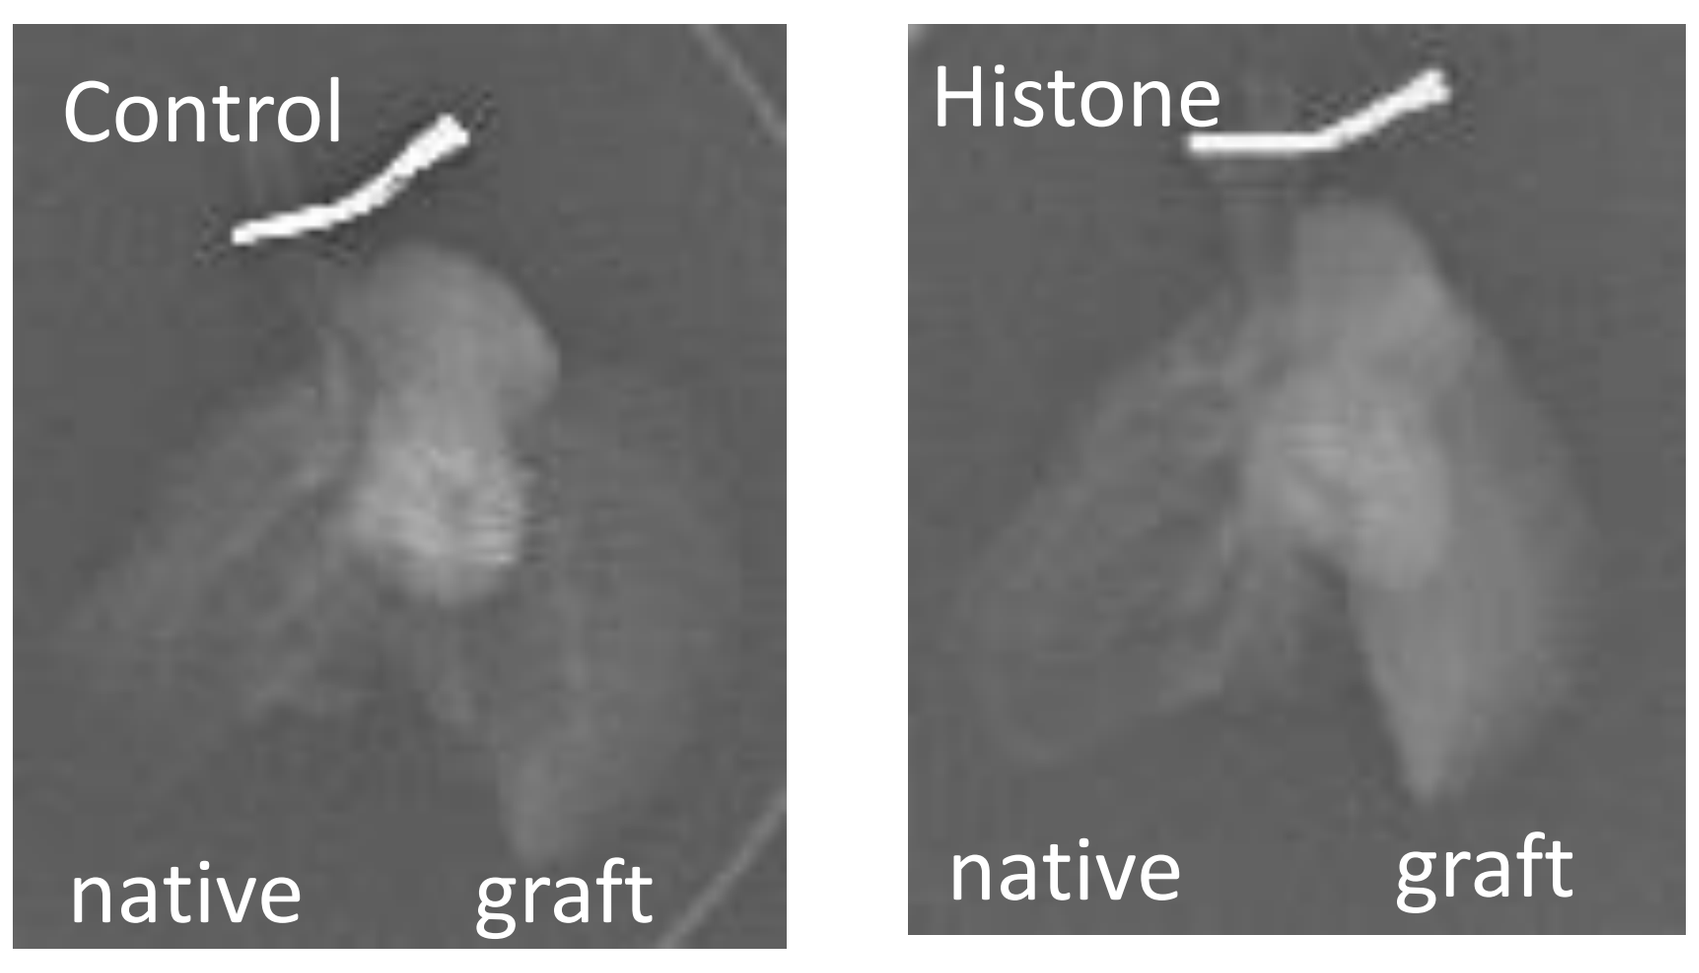
Control
native
graft
Histone
native
graft

W/D ratio :
No significant change was seen.

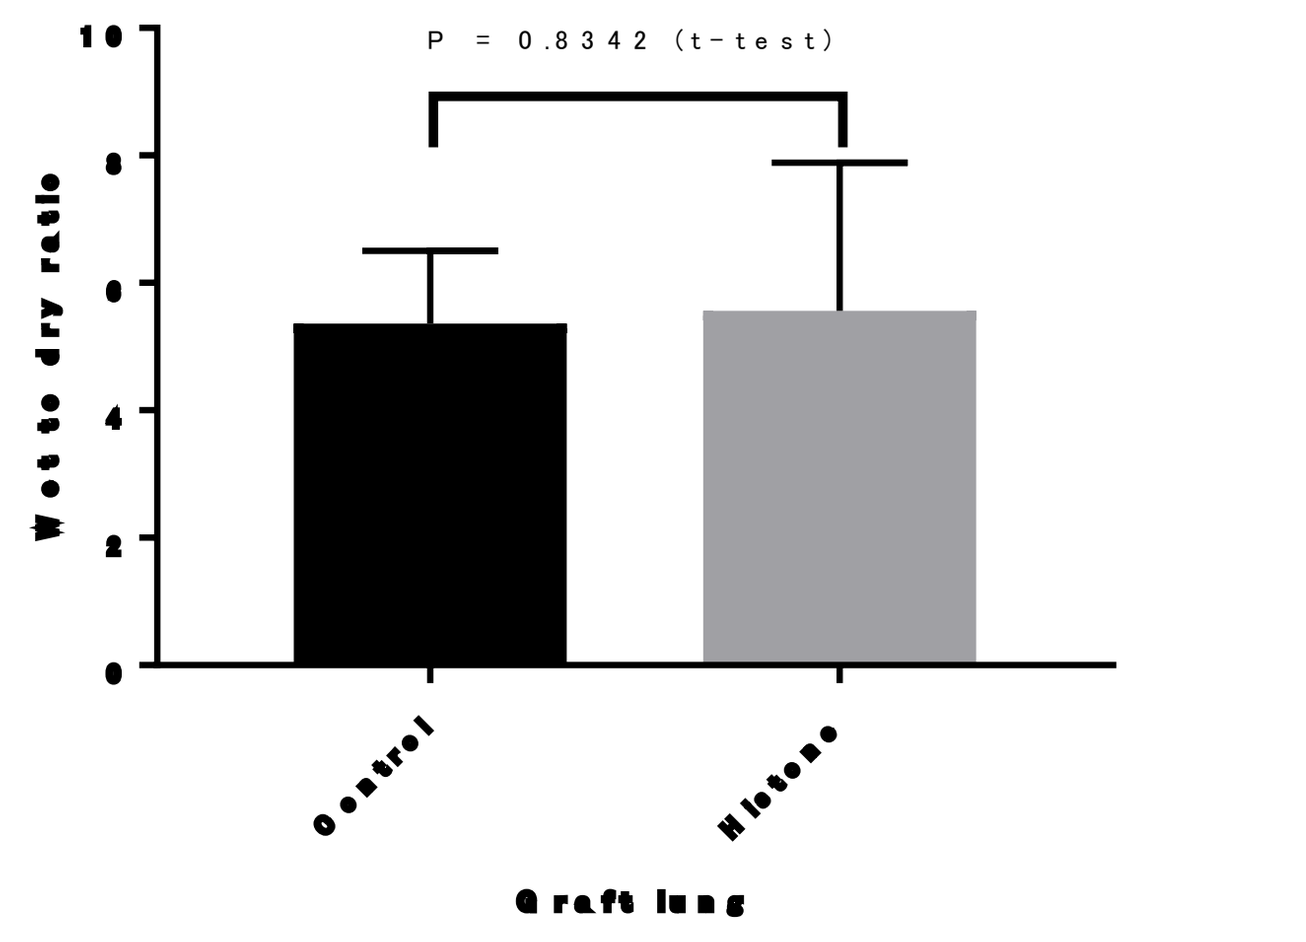
P = 0.8342 (t-test)
Wet to dry ratio
Control
Histone
Graft lung

Histology : Inflammatory infiltration was seen in Histone group.

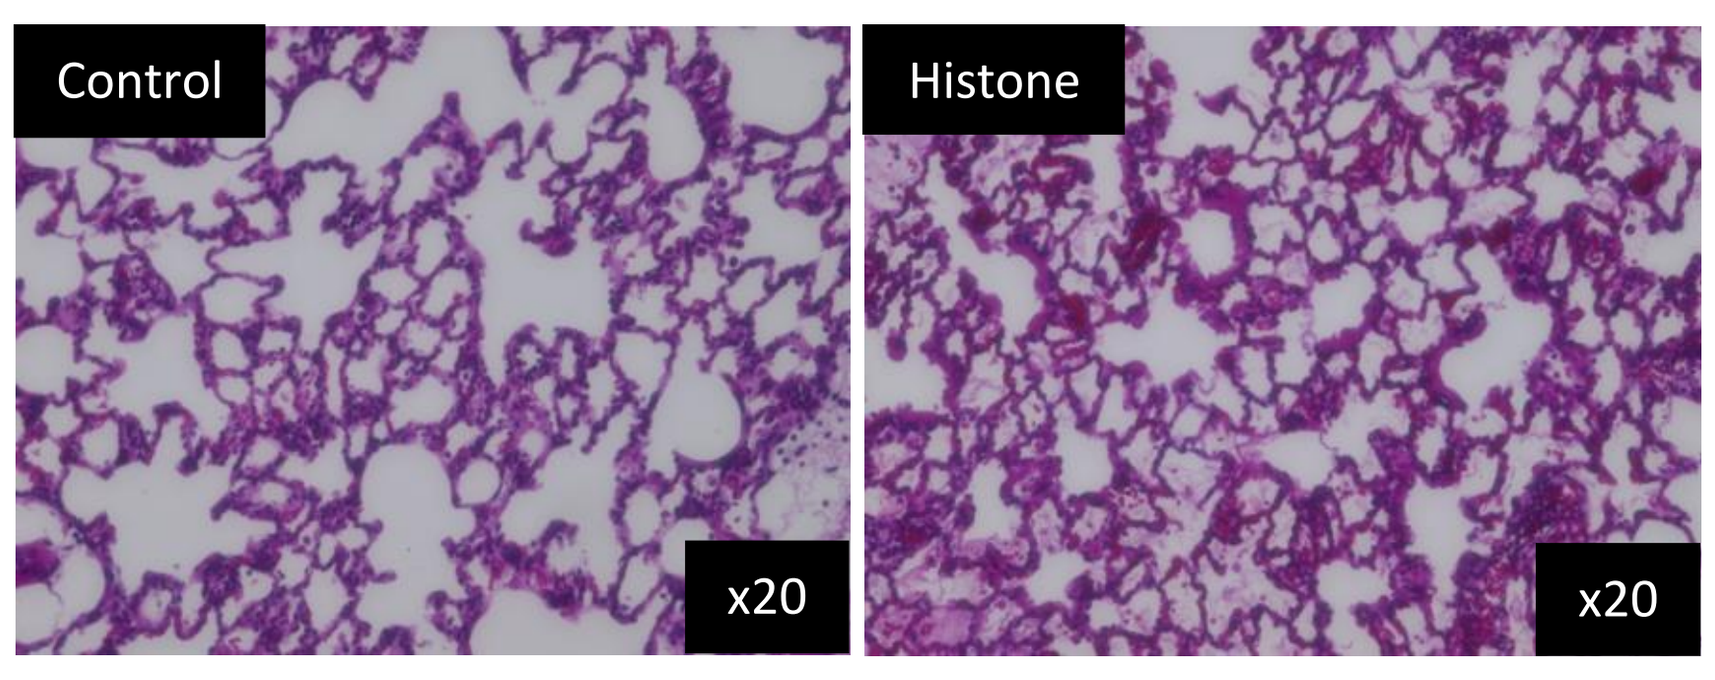
Control
Histone
x20
x20

Graft Compliance :
No significant change was seen.

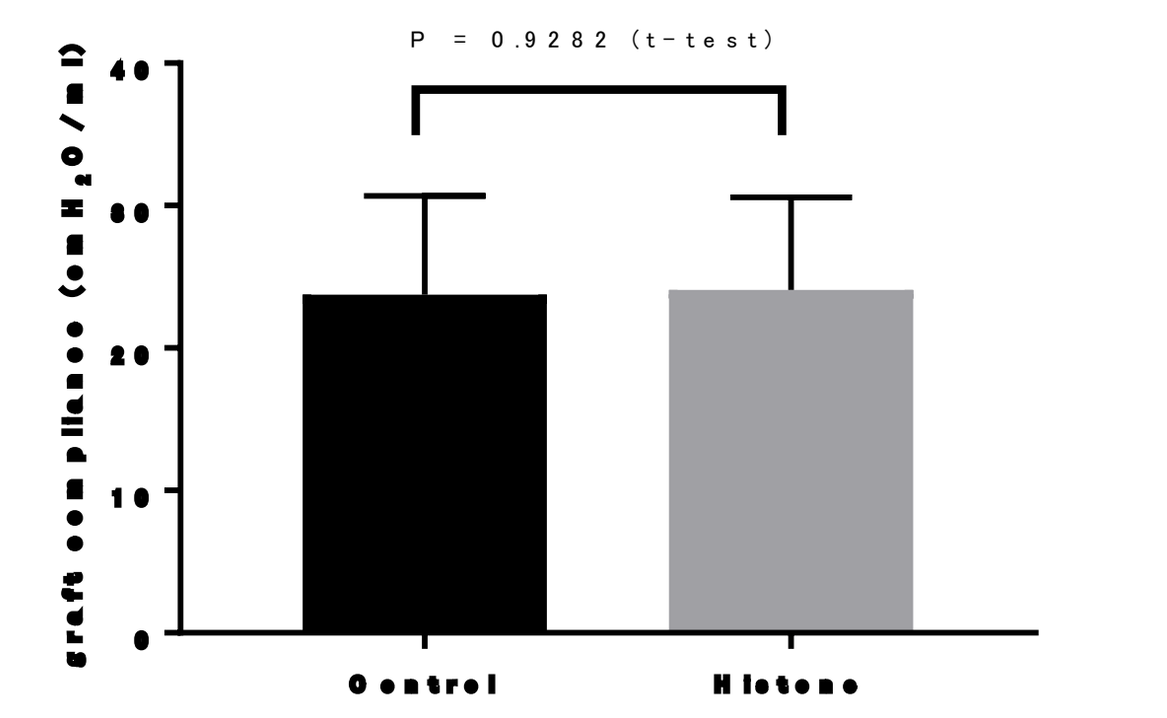
P = 0.9282 (t-test)
Control
Histone

Gene expression :
Inflammatory and hypoxia cytokine had tendency to be expressed in Histone group. But, individual differences were large and there was no significant difference.

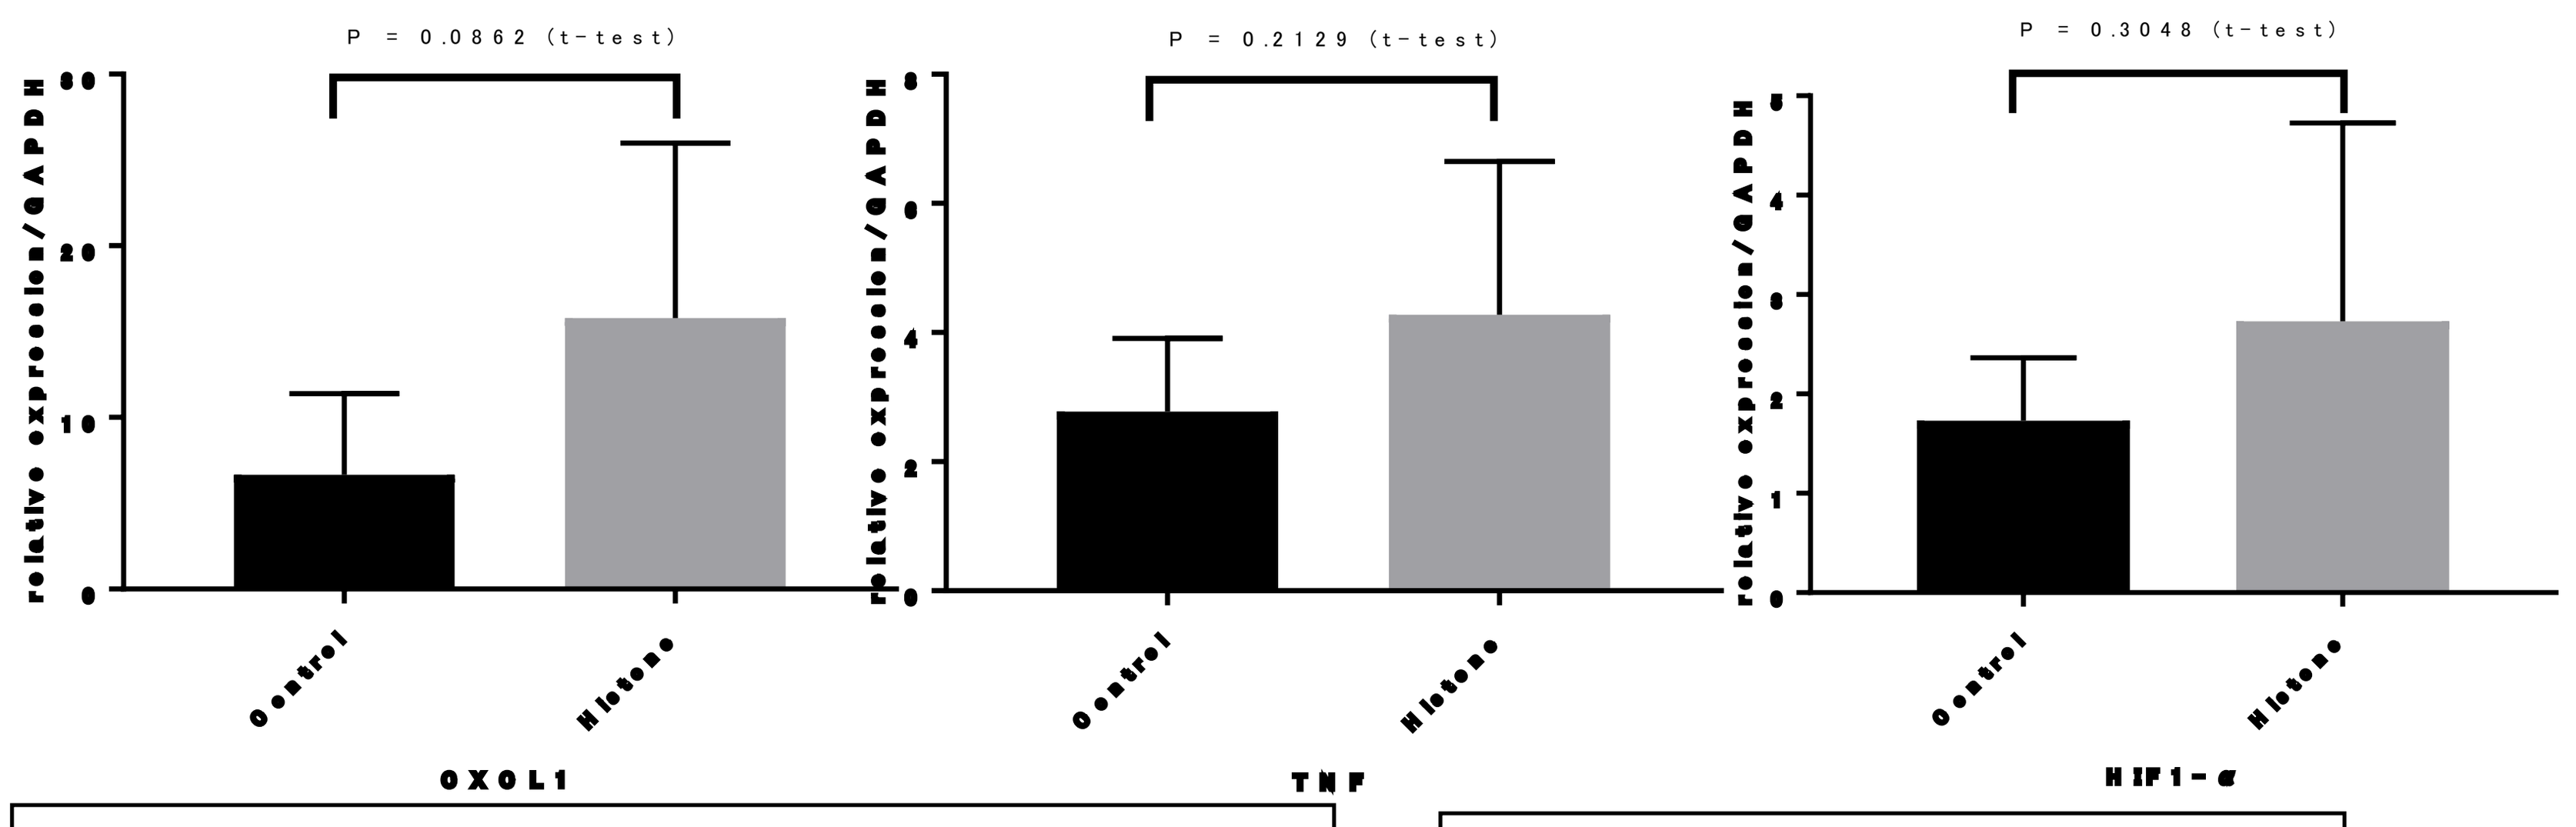
P = 0.0862 (t-test)
P = 0.2129 (t-test)
P = 0.3048 (t-test)
relative expression/GAPDH
Control
Histone
CXCL1
TNF
HIF1-α

ABG from graft PV :
Oxygenation in Histone group remained worse 4 hours after reperfusion.

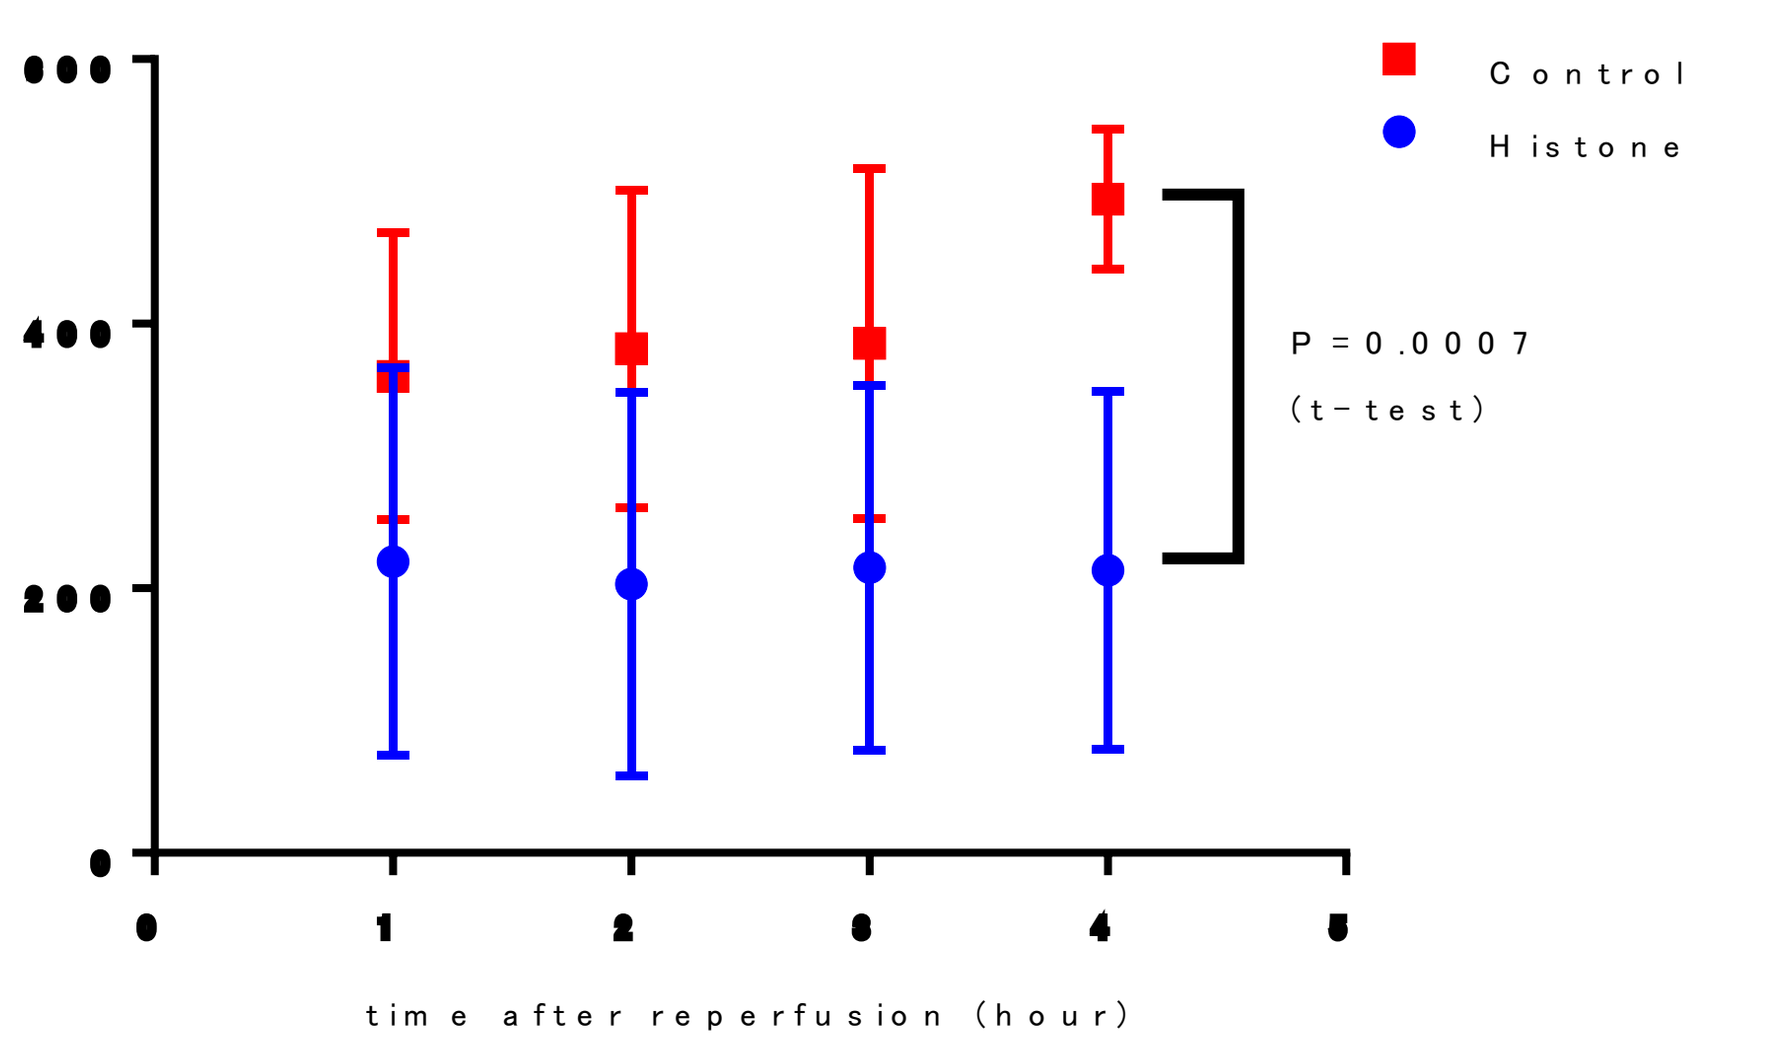
Control
Histone
P = 0.0007
(t-test)
time after reperfusion (hour)

ISHLT PGD grading :
PGD score were worse in Histone group.

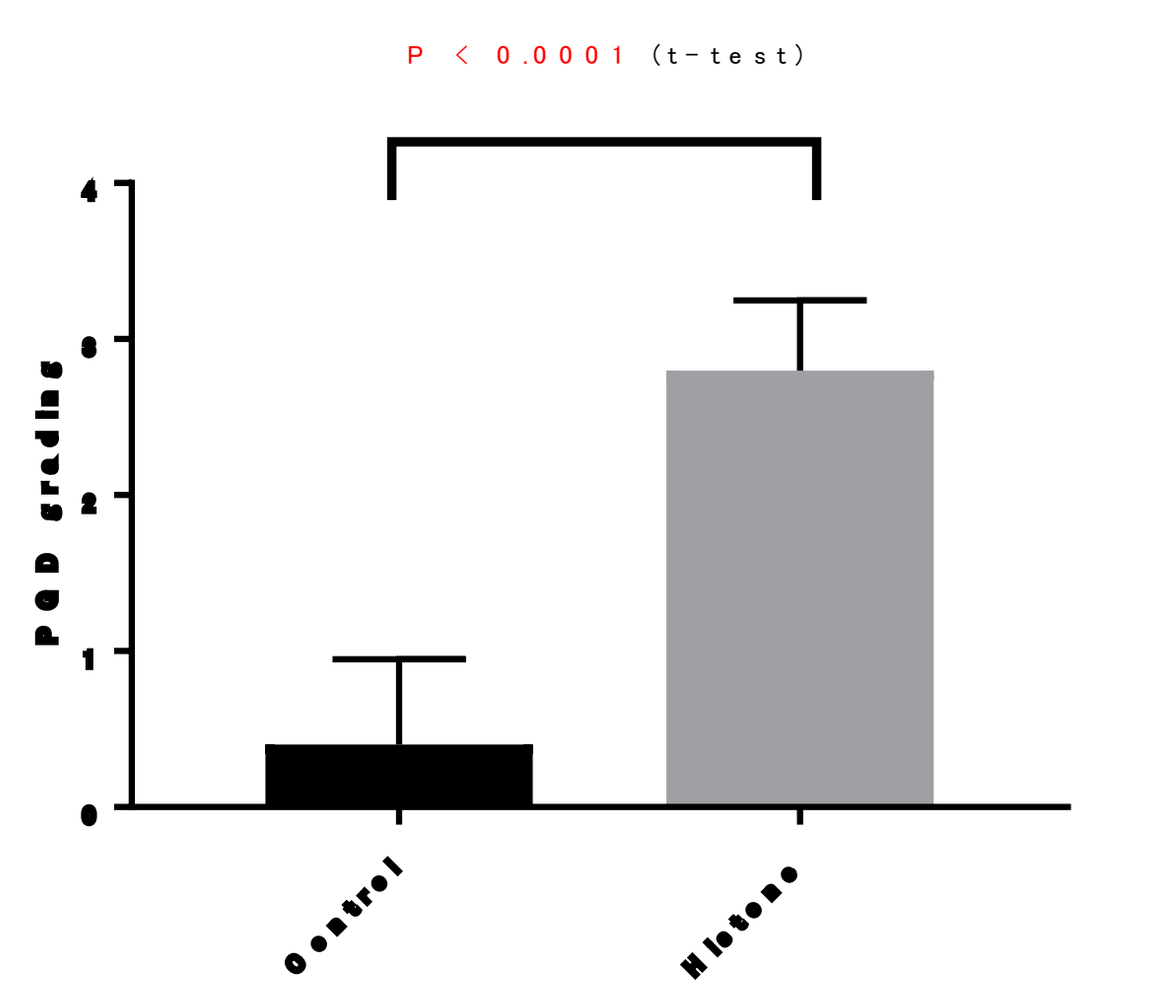
P < 0.0001 (t-test)
PGD grading
Control
Histone

Discussion

- Before lung transplantation, the lung function was not damaged in terms of arterial blood gas analysis or chest X-ray as we usually do in graft assessment. Gene expression may represent the state of inflammation and hypoxia in the forefront.
- After lung transplantation, the lung function decreased markedly in lung transparency and chronological change of arterial blood gas analysis from graft PV though oxygenation in the control group improved as the influence of reperfusion injury disappeared.
- Preclinical influence of histone administration on lung graft may be seen in gene expression and it may become apparent after lung transplantation.
- Further investigation is necessary as to what is the cause of this result in the various influences of histones. We would like to use it as a clue to development of treatment of pulmonary disorders induced by circulating histone and other damage-associated molecular patterns (DAMPs) in the donor's body.

Conclusion

- Circulating histone-associated lung injury in the donor could cause similar conditions of PGD after lung transplantation in our model.
- It may be underrecognized in the current clinical donor assessment as the lung injury may not be immediately apparent.

References

1. Geiger S, Holdenrieder S, Stieber P, et al. Nucleosomes in Serum of Patients with Early Cerebral Stroke. *Cerebrovasc Dis*. 2006;21:32-37.
2. Silk E, Zhao H, Weng H, Ma D. The role of extracellular histone in organ injury. *Nat Publ Gr*. 2017.
3. Alhamdi Y, Abrams ST, Cheng Z, et al. Circulating Histones Are Major Mediators of Cardiac Injury in Patients With Sepsis*. *Crit Care Med*. 2015;43(10):2094-2103.
4. Alhamdi Y, Zi M, Abrams ST, et al. Circulating Histone Concentrations Differentially Affect the Predominance of Left or Right Ventricular Dysfunction in Critical Illness. *Crit Care Med*.
5. Kawai C, Kotani H, Miyao M, et al. Circulating Extracellular Histones Are Clinically Relevant Mediators of Multiple Organ Injury. 2016. doi:10.1016/j.ajpath.2015.11.025