Rapid Transition From Parenteral to Oral Treprostinil When Treating Pulmonary Arterial Hypertension is Feasible and Safe

H. Jarrett[1] A. K. Jonnalagadda[1] S. D. Liu[1] A. J. Bagnola[2] D. Lewis[2] O. A. Shlobin[2] C. F. Barnett[1]
[1]Medstar Heart and Vascular Institute, Washington Hospital Center, Washington, DC, USA
[2]Pulmonary Hypertension Program, Inova Fairfax Hospital, Falls Church, VA, USA

BACKGROUND

- Despite therapeutic advances in the past two decades, pulmonary arterial hypertension (PAH) still confers significant morbidity and mortality.
- Common complications related to delivery of parental prostacyclin analogues (PPA) used to treat PAH are serious systemic infection, catheter malfunction and vascular thrombosis.
- Until recently[1], inhaled prostacyclin therapy had been the only alternate mode of delivery. The approval of an oral prostacyclin analogue (Orenitram; United Therapeutics, Silver Spring, MD) has allowed for transition away from parental formulations.
- There is a paucity of data regarding the safety and feasibility of intravenous to oral prostacyclin transition[2]. We report our experience in performing such a transition.

METHODS

- Retrospective review of medical records from 2013-2017 revealed 100 patients with World Health Organization (WHO) Group 1 PAH patients on PPA therapy of whom 14 were transitioned to oral Orenitram (mean age 51, 93% female, 70% African-American; Table 1).
- The primary outcomes of interest were adverse side effects peri- and post-transition, length of time taken for transition. Pre- and post-transition New York Heart Association (NYHA) functional class, right heart catheterization hemodynamics and 6-minute walk distances (6MWD) were also evaluated.
- Mean values were compared using the student's t-test.

RESULTS

- Mean PPA dose was 47ng/kg/min (range 15-75).
- Mean final post transition total daily Orenitram dose was 27mg (range 9.75 – 60).
- Reasons for transition were:
 Hemodynamic stability (n=7)
 Infusion site pain /local irritation (n=4)
 Inability to tolerate home IV infusion (n=3)
- Median time taken for complete transition was 3 days (range 1-5).
- Pre- and post-transition adverse side effects were minor (Table 2).
- Pre and post right heart hemodynamics, NYHA functional class and 6MWD were similar.

CONCLUSION

- Rapid transition from parenteral treprostinil to oral Orenitram is feasible and safe in a low-risk cohort of patients with WHO Group 1 PAH.
- To date, no patients have required re-initiation of IV therapy.
- Dose-timing related side effects of abdominal pain and diarrhea were noted during the transition phase which resolved with subsequent adjustment.

Table 1: Baseline Characteristics	
Age in years (mean)	51.2
Female [N (%)]	13 (93)
Race [N (%)]	
- African American	9 (70)
- Caucasian	3 (21)
- Hispanic	1 (7)
Weight in kg (mean)	73.6
PAH etiology (%)	
- Idiopathic	8 (57)
- CTD	3 (22)
- Congenital	1 (7)
- HIV	2 (14)
Baseline WHO functional classification (%)	
- NYHA Class 1	3 (22)
- NYHA Class 2	10 (71)
- NYHA Class 3	1 (7)
- NYHA Class 4	0 (0)
Baseline 6MWD in meters (median; range)	406.5 (146 - 566)
Vital signs [median; range]	
- Systolic Blood Pressure (mmHg)	113 (93 – 165)
- Diastolic Blood Pressure (mmHg)	60 (52 – 89)
- Heart Rate (beats/minute)	85 (62 – 109)
- Oxygen Saturation (%)	97 (93 – 100)
Hypertension (%)	5 (36)
Lung Disease (%)	5 (36)
Coronary Disease (%)	0
Diabetes (%)	3 (22)
History of Arrhythmia (%)	2 (14)
Obstructive Sleep Apnea (%)	2 (14)
Hypothyroidism (%)	1 (7)
Liver Disease (%)	1 (7)
HIV (%)	2 (14)
Tobacco Use (%)	2 (14)
Background PAH therapy [N (%)]	
- Phosphodiesterase inhibitor	10 (71)
- Endothelin receptor antagonist	5 (36)
- Combination therapy	4 (29)
- Soluble guanylyl cyclase stimulator	1 (7)
Baseline Treprostinil dose ng/kg/min (mean ± sd)	47 ± 23.6
Time on Treprostinil in years (median; range)	3 (0 – 10)

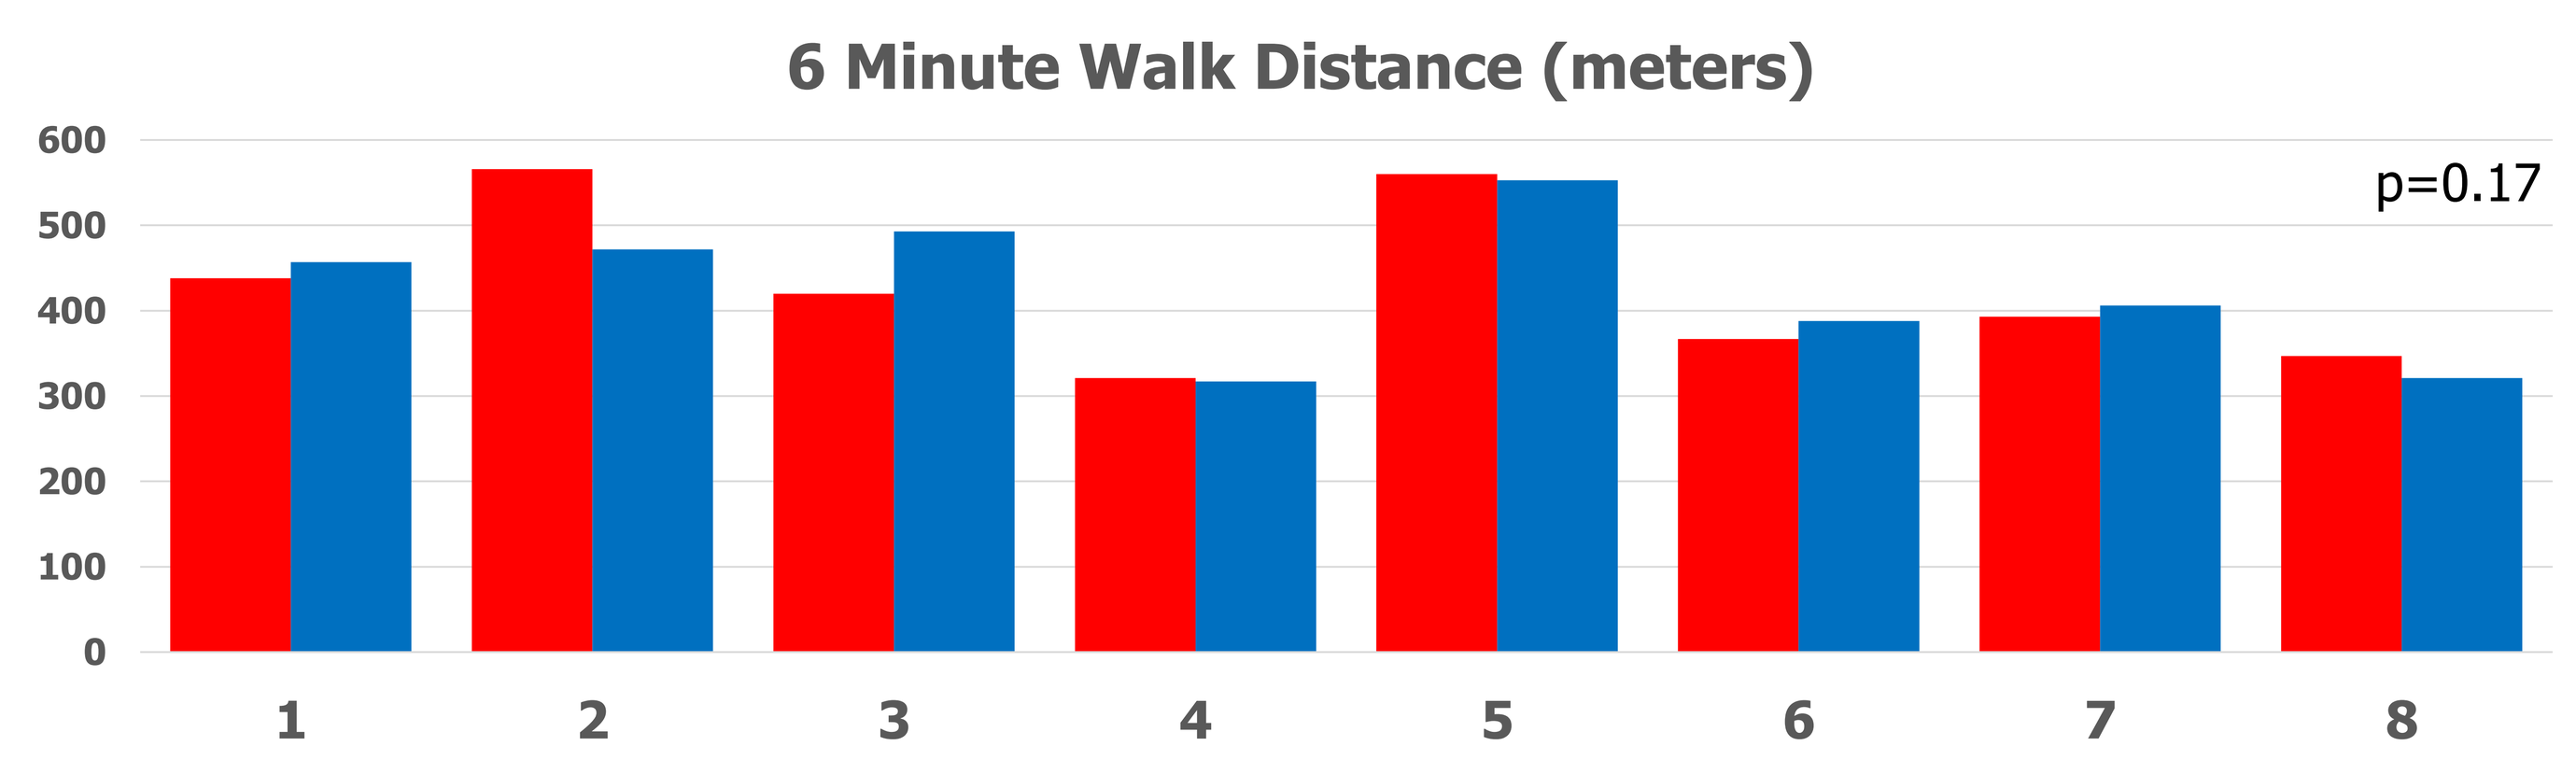
6 Minute Walk Distance (meters)
p=0.17
0
100
200
300
400
500
600
1
2
3
4
5
6
7
8

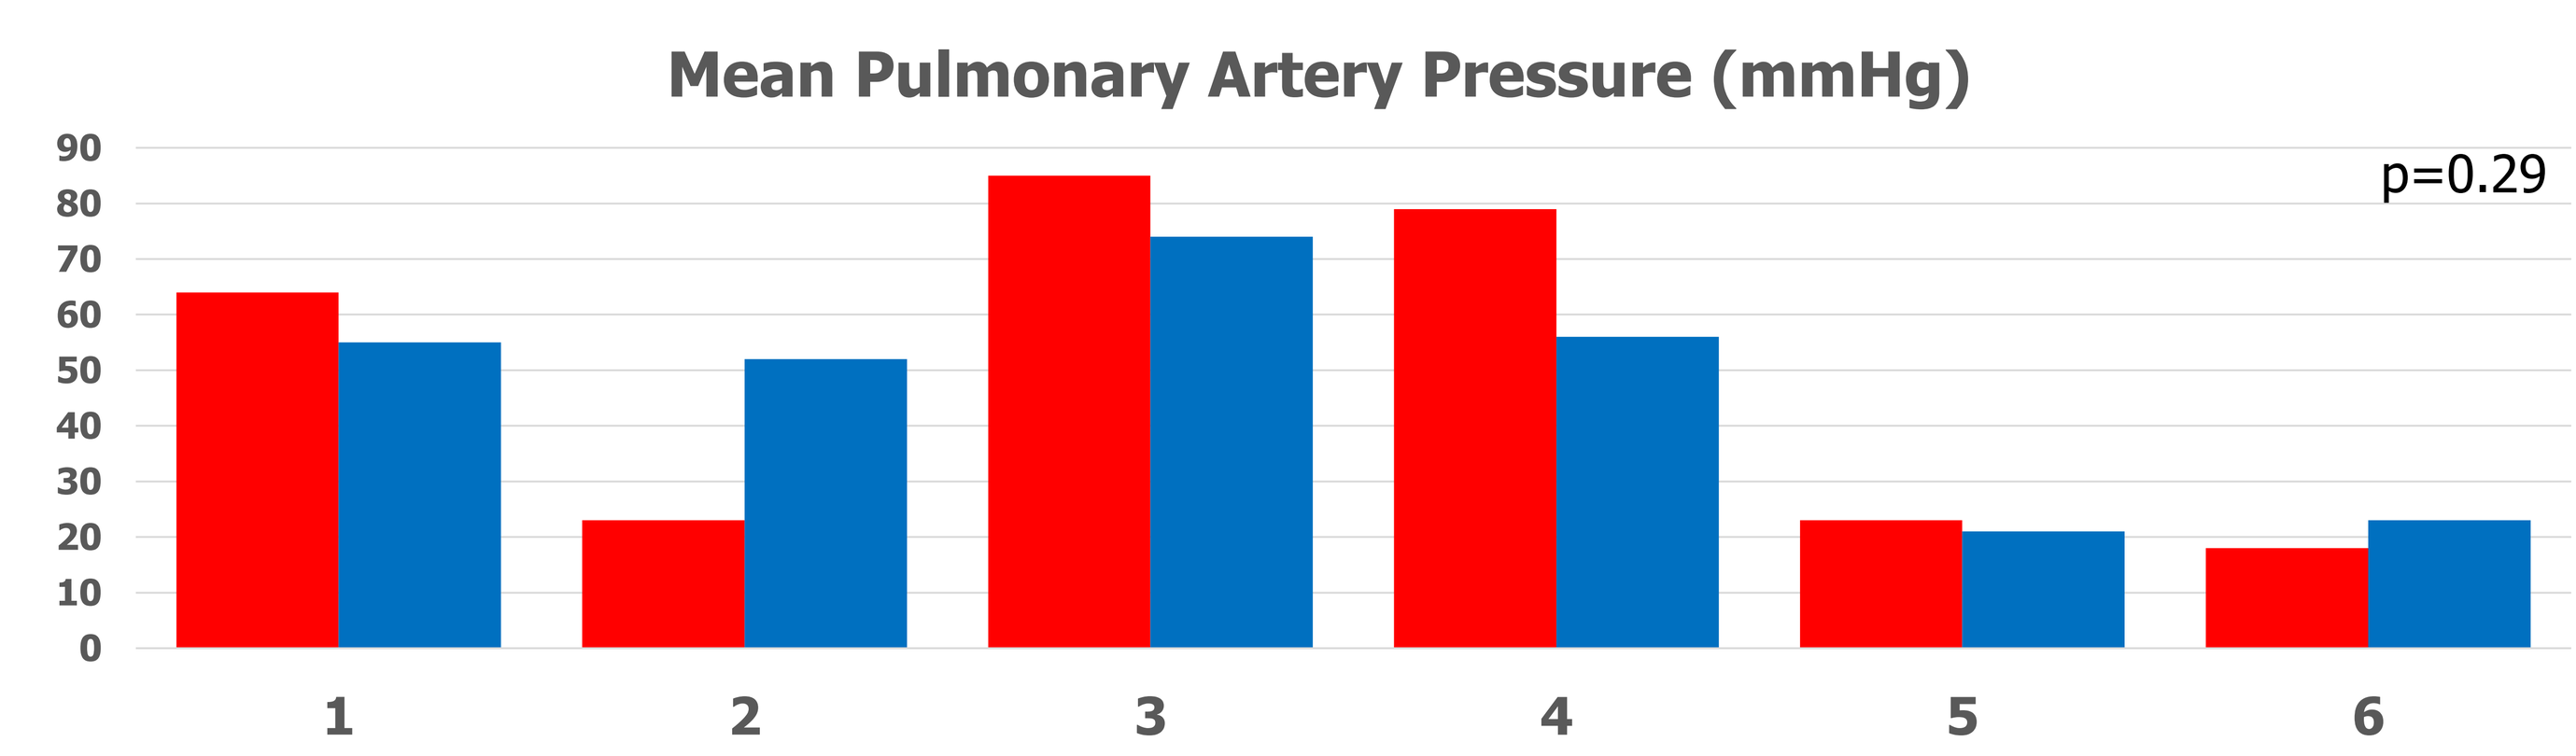
Mean Pulmonary Artery Pressure (mmHg)
p=0.29
0
10
20
30
40
50
60
70
80
90
1
2
3
4
5
6

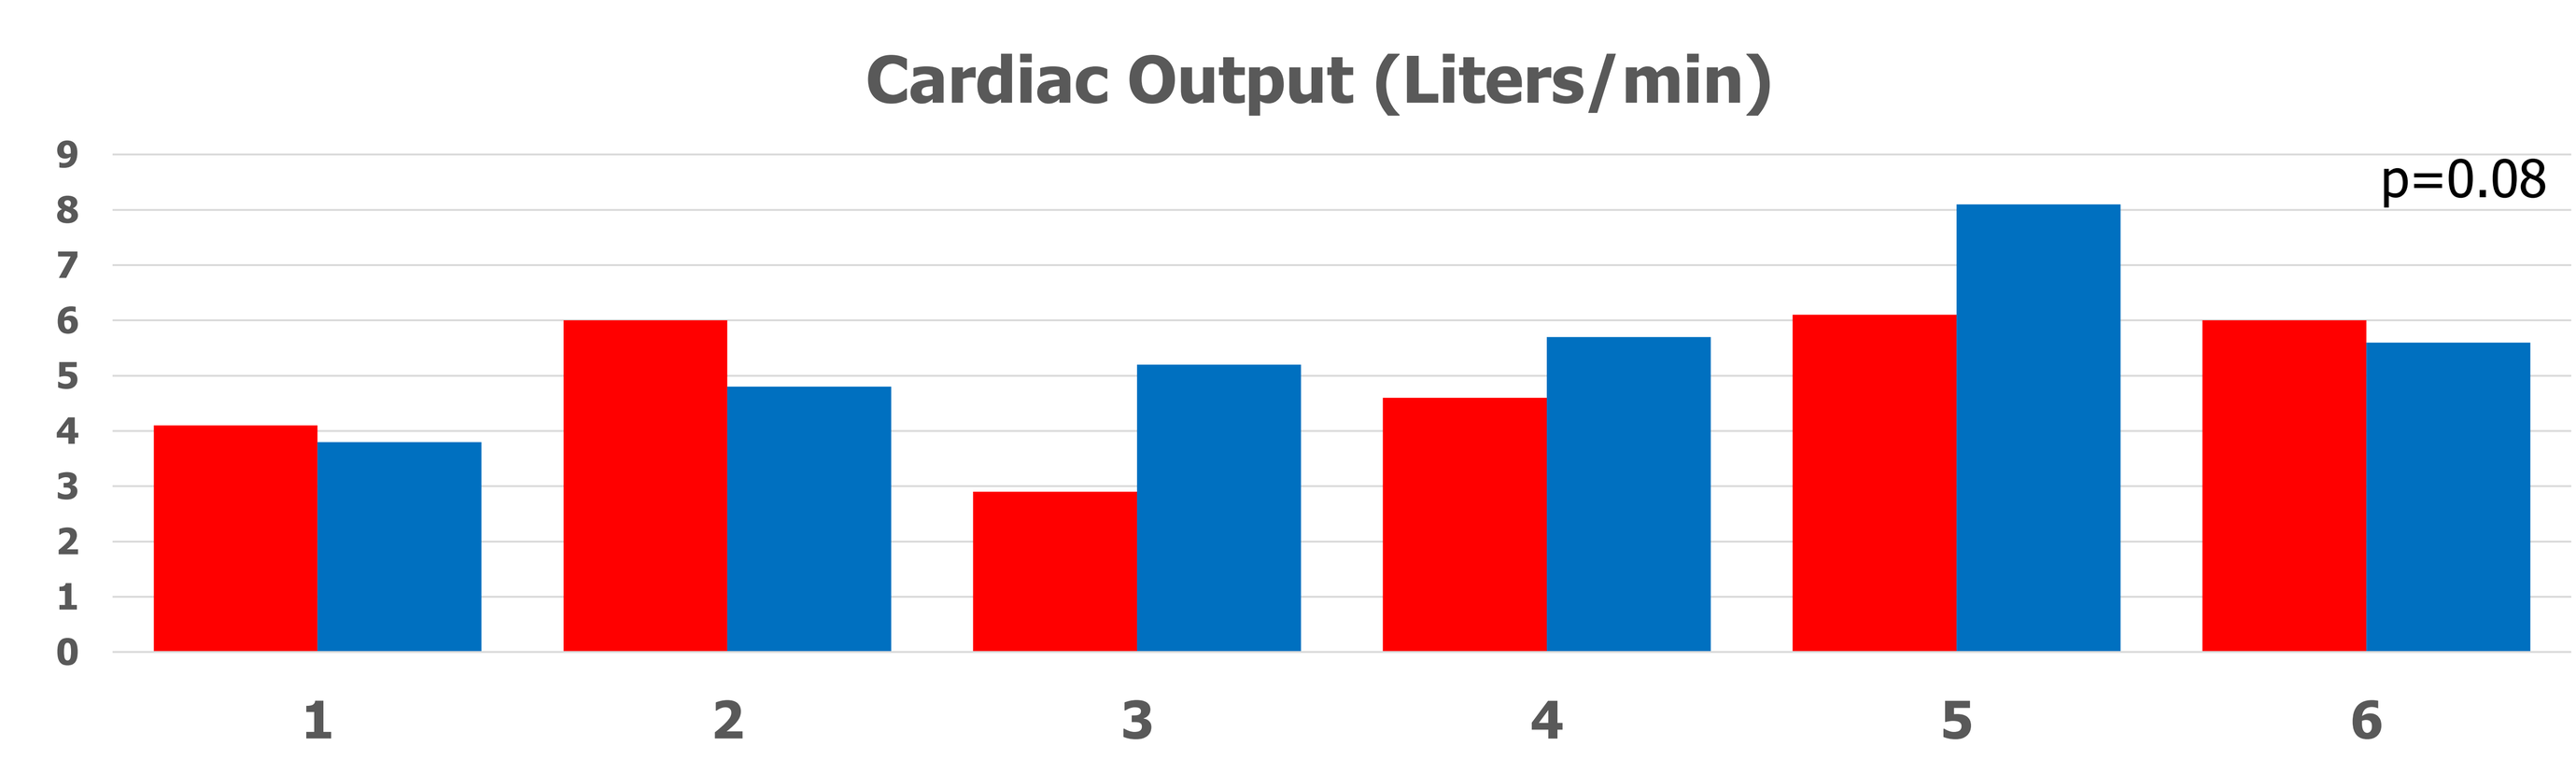
Cardiac Output (Liters/min)
p=0.08
0
1
2
3
4
5
6
7
8
9
1
2
3
4
5
6

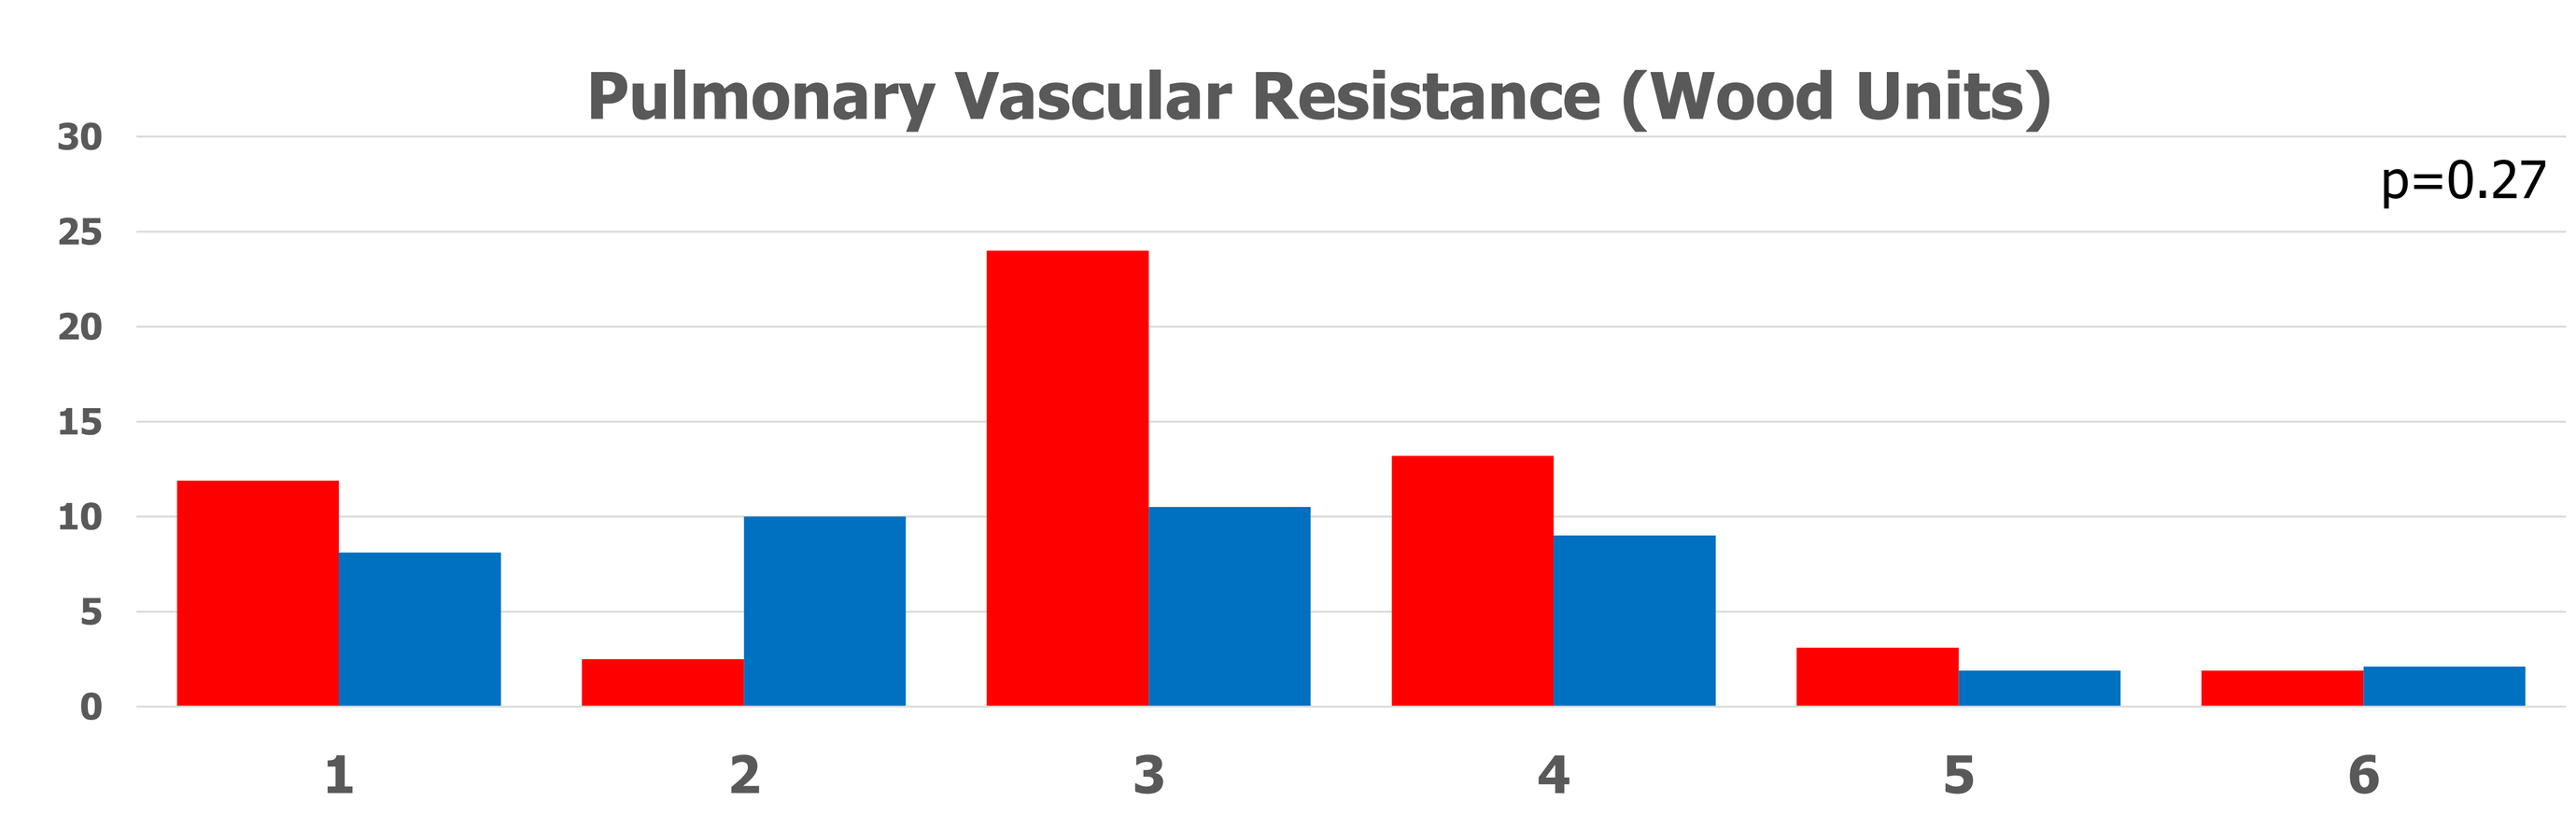
Pulmonary Vascular Resistance (Wood Units)
p=0.27
0
5
10
15
20
25
30
1
2
3
4
5
6

Table 2: Adverse Effects		
	Pre-transition (n=14)	Post-transition (n=14)
Headache (%)	21	29
Diarrhea (%)	43	43
Nausea (%)	14	7
Flushing (%)	29	14
Pain (%)	0	0
Jaw pain (%)	14	7
Limb pain (%)	14	0
Abdominal pain (%)	0	7
Hypokalemia (%)	7	0

References
1. Jing ZC *et al.* Efficacy and safety of oral treprostinil monotherapy for the treatment of pulmonary arterial hypertension: a randomized, controlled trial. Circulation 2013 Feb;127(5): 624-33
2. Chakinala MM. *et al.* Transition from parenteral to oral treprostinil in pulmonary arterial hypertension. J Heart Lung Transplant 2017 Feb;36(2):193-201

Disclosures: None